Efficacy and safety of larotrectinib as first-line treatment for paediatric patients with TRK fusion cancer

421

Daniel Orbach[1], C. Michel Zwaan[2], Catherine M. Albert[3], Noah Federman[4], Alberto Pappo[5], Vadim Bernard-Gauthier[6], Natascha Neu[7], Esther De La Cuesta[8], Theodore W. Laetsch[9], Yi-Zhuo Zhang[10]

[1]SIREDO Oncology Center (Care, Innovation and Research for Children and AYA with Cancer), Institut Curie, Paris, France; [2]Prinses Máxima Centrum, Utrecht, the Netherlands and Erasmus MC, Rotterdam, the Netherlands; [3]Seattle Children's Hospital and University of Washington School of Medicine, Seattle, WA, USA; [4]David Geffen School of Medicine, University of California, Los Angeles, CA, USA; [5]St. Jude Children's Research Hospital, Memphis, TN, USA; [6]Bayer Pharmaceuticals, Inc., Toronto, Canada; [7]Chrestos Concept GmbH & Co. KG, Essen, Germany; [8]Bayer Pharmaceuticals, Inc., Whippany, NJ, USA; [9]The Children's Hospital of Philadelphia, University of Pennsylvania, Philadelphia, PA, USA; [10]Sun Yat-sen University Cancer Center, Guangzhou, China.

BACKGROUND

- *NTRK* gene fusions are rare oncogenic drivers in a variety of adult and paediatric tumour types.[1]
- Larotrectinib is the first-in-class, highly selective, central nervous system (CNS)-active TRK inhibitor approved for tumour-agnostic use in adult and paediatric patients with TRK fusion cancer. Its approval was based on tumour response and durable efficacy in terms of survival.[2,3]
- Here, we report data on paediatric patients with TRK fusion cancer treated with larotrectinib in the first-line systemic setting.

METHODS

- Paediatric patients with systemic treatment-naïve non-primary CNS TRK fusion cancer treated in 2 larotrectinib clinical trials (NCT02576431 [NAVIGATE] and NCT02637687 [SCOUT]) were included in this analysis.
 - Patients were considered treatment-naïve if they had not received systemic therapy (excluding prior radioactive iodine) in the metastatic and/or unresectable settings.
- Patients received larotrectinib 100 mg/m^2 (maximum dose of 100 mg) twice daily.
- The primary endpoint was overall response rate (ORR) as assessed by independent review committee (IRC) per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1.
- In a post-hoc analysis, patients in the SCOUT study were permitted to stop larotrectinib in the absence of on-treatment disease progression ('wait-and-see'); patients who stopped treatment for ≥28 days were actively followed for progression according to protocol.
 - Both off-treatment progression and response to treatment were re-assessed by investigator per RECIST v1.1.
- Data cut-off date was 20 July 2022.

RESULTS

Patients

- At data cut-off, a total of 37 paediatric patients with locally advanced or metastatic TRK fusion cancer had received larotrectinib in the first-line setting (**Table 1**).
- *NTRK* gene fusions were identified locally by next-generation sequencing (NGS), fluorescence in situ hybridisation and polymerase chain reaction in 24 (65%), 7 (19%) and 6 (16%) patients, respectively.
- There were 9 unique fusion partners, with *ETV6*::*NTRK3* being the most common (n=19; 51%; **Figure 1**).

Table 1. Baseline characteristics

Characteristic	N=37
Age, median (range), years	2.1 (0–17)
Sex, n (%)	
Male	22 (59)
Female	15 (41)
CNS metastases at baseline, n (%)	
Yes	1 (3)
No	36 (97)
Disease status at enrolment, n (%)	
Locally advanced	27 (73)
Metastatic	10 (27)
Prior therapies, n (%)†,‡	
Surgery	12 (32)
Radioactive iodine	2 (5)
Radiotherapy	1 (3)
***NTRK* gene fusion**, n (%)	
NTRK1	17 (46)
NTRK3	20 (54)
ECOG or equivalent Lansky/Karnofsky performance status, n (%)	
0	32 (86)
1	4 (11)
2	1 (3)
Tumour types, n (%)	
Infantile fibrosarcoma	18 (49)
Soft tissue sarcoma	14 (38)
Congenital mesoblastic nephroma	2 (5)
Thyroid	2 (5)
Breast	1 (3)

†Patients were considered treatment-naïve if they had not received systemic therapy (excluding prior radioactive iodine) in the metastatic and/or unresectable settings (2 patients each had 1 prior line and 1 patient had 2 prior lines in the neoadjuvant, adjuvant or locally advanced settings). ‡Patients may be counted in more than 1 row. CNS, central nervous system; ECOG, Eastern Cooperative Oncology Group.

Figure 1. *NTRK* fusion partner distribution†

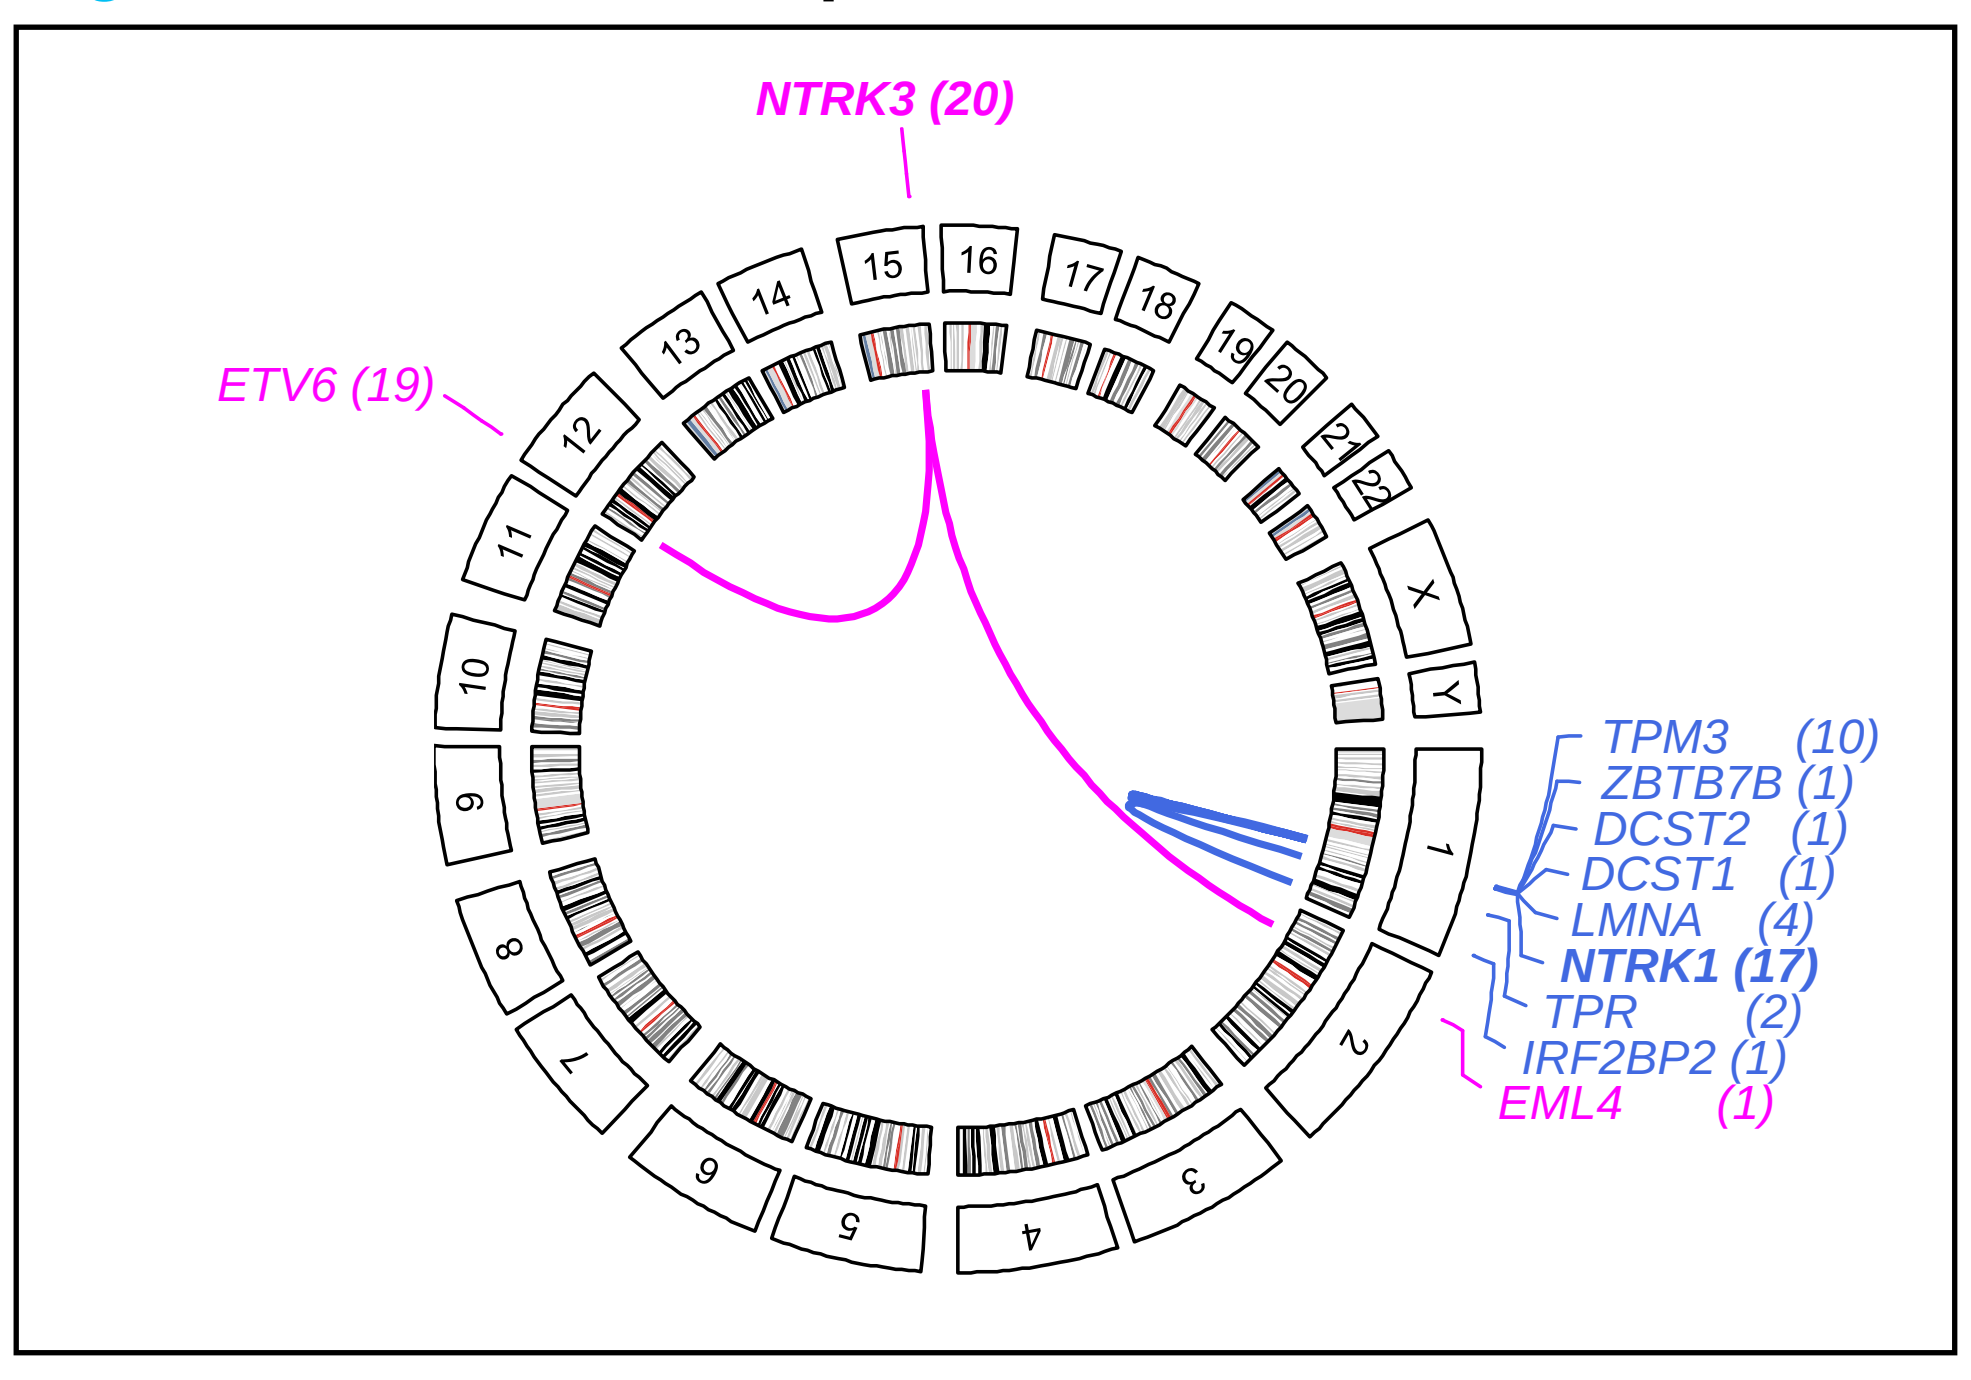
NTRK3 (20)
ETV6 (19)
TPM3 (10)
ZBTB7B (1)
DCST2 (1)
DCST1 (1)
LMNA (4)
NTRK1 (17)
TPR (2)
IRF2BP2 (1)
EML4 (1)

†One patient had 4 different gene fusions *(DCST1::NTRK1, DCST2::NTRK1, TPM3::NTRK1* and *ZBTB7B::NTRK1).*
The number of patients with each fusion is indicated in the parentheses.

Efficacy

- Tumour responses for all patients are shown in **Figure 2**.
- Median duration of response (DoR), progression-free survival (PFS) and overall survival (OS) are reported in **Figure 3**.
- Treatment duration ranged from 1 to 64+ months (median 31.2) (**Figure 4**). The median time to response in all patients was 1.8 months (range 0.9–5.4).
- Ten (27%) patients permanently discontinued treatment, with 4 discontinuing due to tumour progression.

References

1. Amatu A et al. *Ann Oncol*. 2019;30:viii5–viii15.
2. Bayer. VITRAKVI US PI. 2021. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/210861s006lbl.pdf. Accessed 01 March 2024.
3. Bayer. VITRAKVI SmPC. 2021 Available at: https://www.ema.europa.eu/en/documents/product-information/vitrakvi-epar-product-information_en.pdf. Accessed 01 March 2024.

Acknowledgements

We thank the patients and their families, many of whom travelled long distances to participate in these studies. Medical writing assistance was provided by Patricia Badia Folgado, MSc, and editorial and typesetting assistance were provided by Melissa Ward, BA, both of Scion (a division of Prime, London, UK), supported by Bayer Healthcare Pharmaceuticals, Inc. Programming was provided by Stefan Hindel (Chrestos Concept GmbH & Co. KG) and Wenjun Xin (Bayer Healthcare Pharmaceuticals, Inc.).

Figure 2. Maximum change in target lesion size in patients with TRK fusion cancer (N=35)†

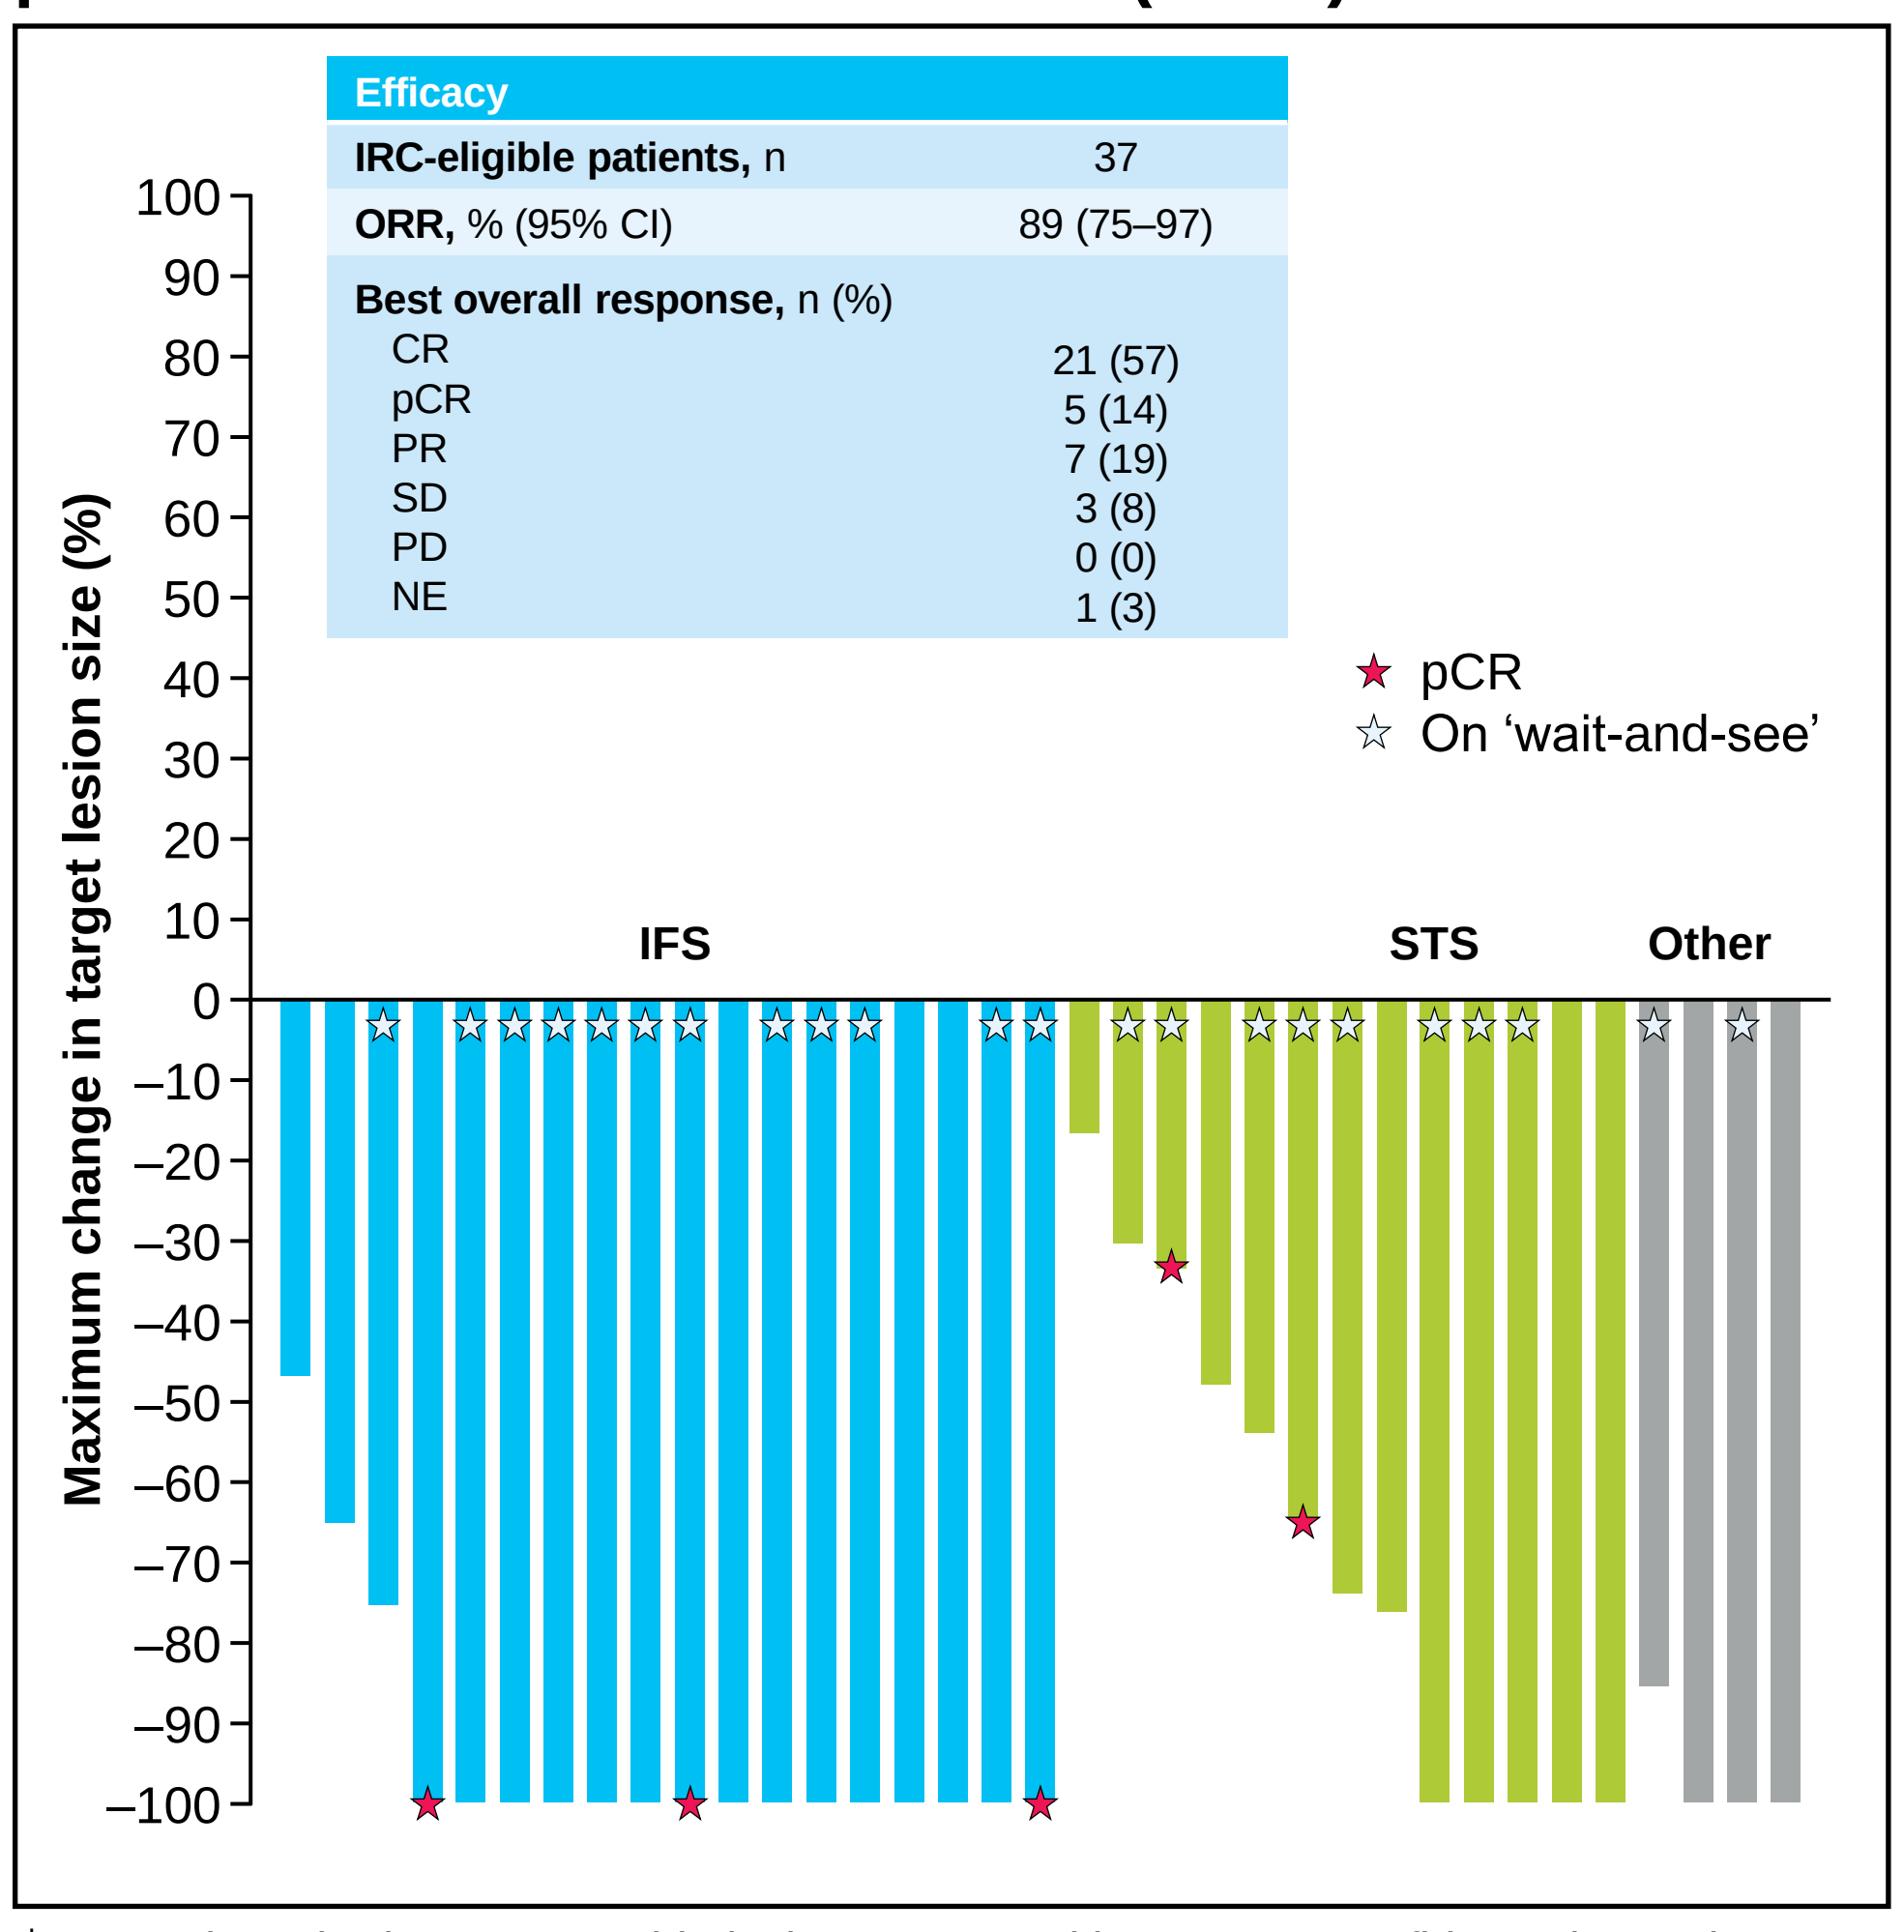
Efficacy
IRC-eligible patients, n 37
ORR, % (95% CI) 89 (75–97)
Best overall response, n (%)
CR 21 (57)
pCR 5 (14)
PR 7 (19)
SD 3 (8)
PD 0 (0)
NE 1 (3)
pCR
On 'wait-and-see'
Maximum change in target lesion size (%)
IFS
STS
Other

†Two patients had no measurable lesions assessed by IRC. CI, confidence interval; CR, complete response; IFS, infantile fibrosarcoma; IRC, independent review committee; NE, not evaluable; ORR, overall response rate; pCR, pathological complete response; PD, progressive disease; PR, partial response; SD, stable disease; STS, soft tissue sarcoma.

CONCLUSIONS

- In this analysis, paediatric patients who were treated with larotrectinib in the first-line setting demonstrated rapid and durable responses, extended survival and a favourable safety profile.
- These results support the wider adoption of NGS panels that include *NTRK* gene fusions to identify patients who may benefit from treatment.

In treatment-naïve paediatric patients with TRK fusion cancer

89% ORR by IRC | 38.4 mo Median DoR | 40.2 mo Median PFS | 95% 48-mo OS rate

PLAIN LANGUAGE SUMMARY

- Larotrectinib is a targeted cancer treatment for patients with TRK fusion cancer.
- This study looked at how paediatric patients with TRK fusion cancer responded to larotrectinib when given as the first treatment.
- A total of 37 patients with TRK fusion cancer across 5 different tumour types were included in this analysis.
- All of the patients experienced tumour shrinkage with larotrectinib.
- Overall, most common side effects with larotrectinib were manageable.
- These results demonstrate that larotrectinib is a fast-acting effective treatment option for patients with TRK fusion cancer without prior systemic cancer treatment.
- Testing patients for *NTRK* gene fusions is important for early identification of those who can benefit from this targeted therapy.

Post-hoc analysis

- For the post-hoc analysis, 22 patients stopped larotrectinib in the absence of on-treatment progression and entered into the 'wait-and-see' analysis.
- Nine patients had surgery prior to stopping larotrectinib; 11 did not have surgery, and the surgical status of 2 patients was unknown.
- At the time of data cut-off, the 'wait-and-see' periods ranged from 0 to 44+ months.
- Best responses before or at the time of stopping larotrectinib, median time to stopping larotrectinib and median time from stopping to subsequent progression are shown in **Table 2**.
- Median PFS of the 22 patients was not reached 95% CI (25.5–NE) at a follow-up of 41.3 months.
- Of the 22 patients, 5 (23%) had documented progression and resumed treatment.
- Of these 5 patients, 4 showed re-response to treatment (3 complete responses and 1 partial response; the fifth patient had stable disease).
- All 22 patients were alive at data cut-off.

Table 2. Baseline 'wait-and-see' post-hoc analysis (N=22)

Best response before or at the time of stopping larotrectinib	CR (n=15)	PR (n=5)	SD (n=2)	Total (N=22)
Median time on treatment prior to stopping larotrectinib, months (range)	20.2 (5–42)	8.6 (3–15)	7.8 (5–10)	14.3 (3–42)
Progressed off treatment, n	4	1	0	5
Median time from stopping larotrectinib to progression, months (range)	20.2 (0–44)	18.3 (6–41)	27.1 (27–27)	21.6 (0–44)

CR, complete response; PR, partial response; SD, stable disease.

Disclosures

These studies were funded by Bayer Healthcare Pharmaceuticals, Inc. The presenting author discloses consultancy: Bayer and Roche; and research funding: Bayer.

Scan QR code to download the poster.
Copies of this e-poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Figure 3. DoR, PFS and OS in patients with TRK fusion cancer

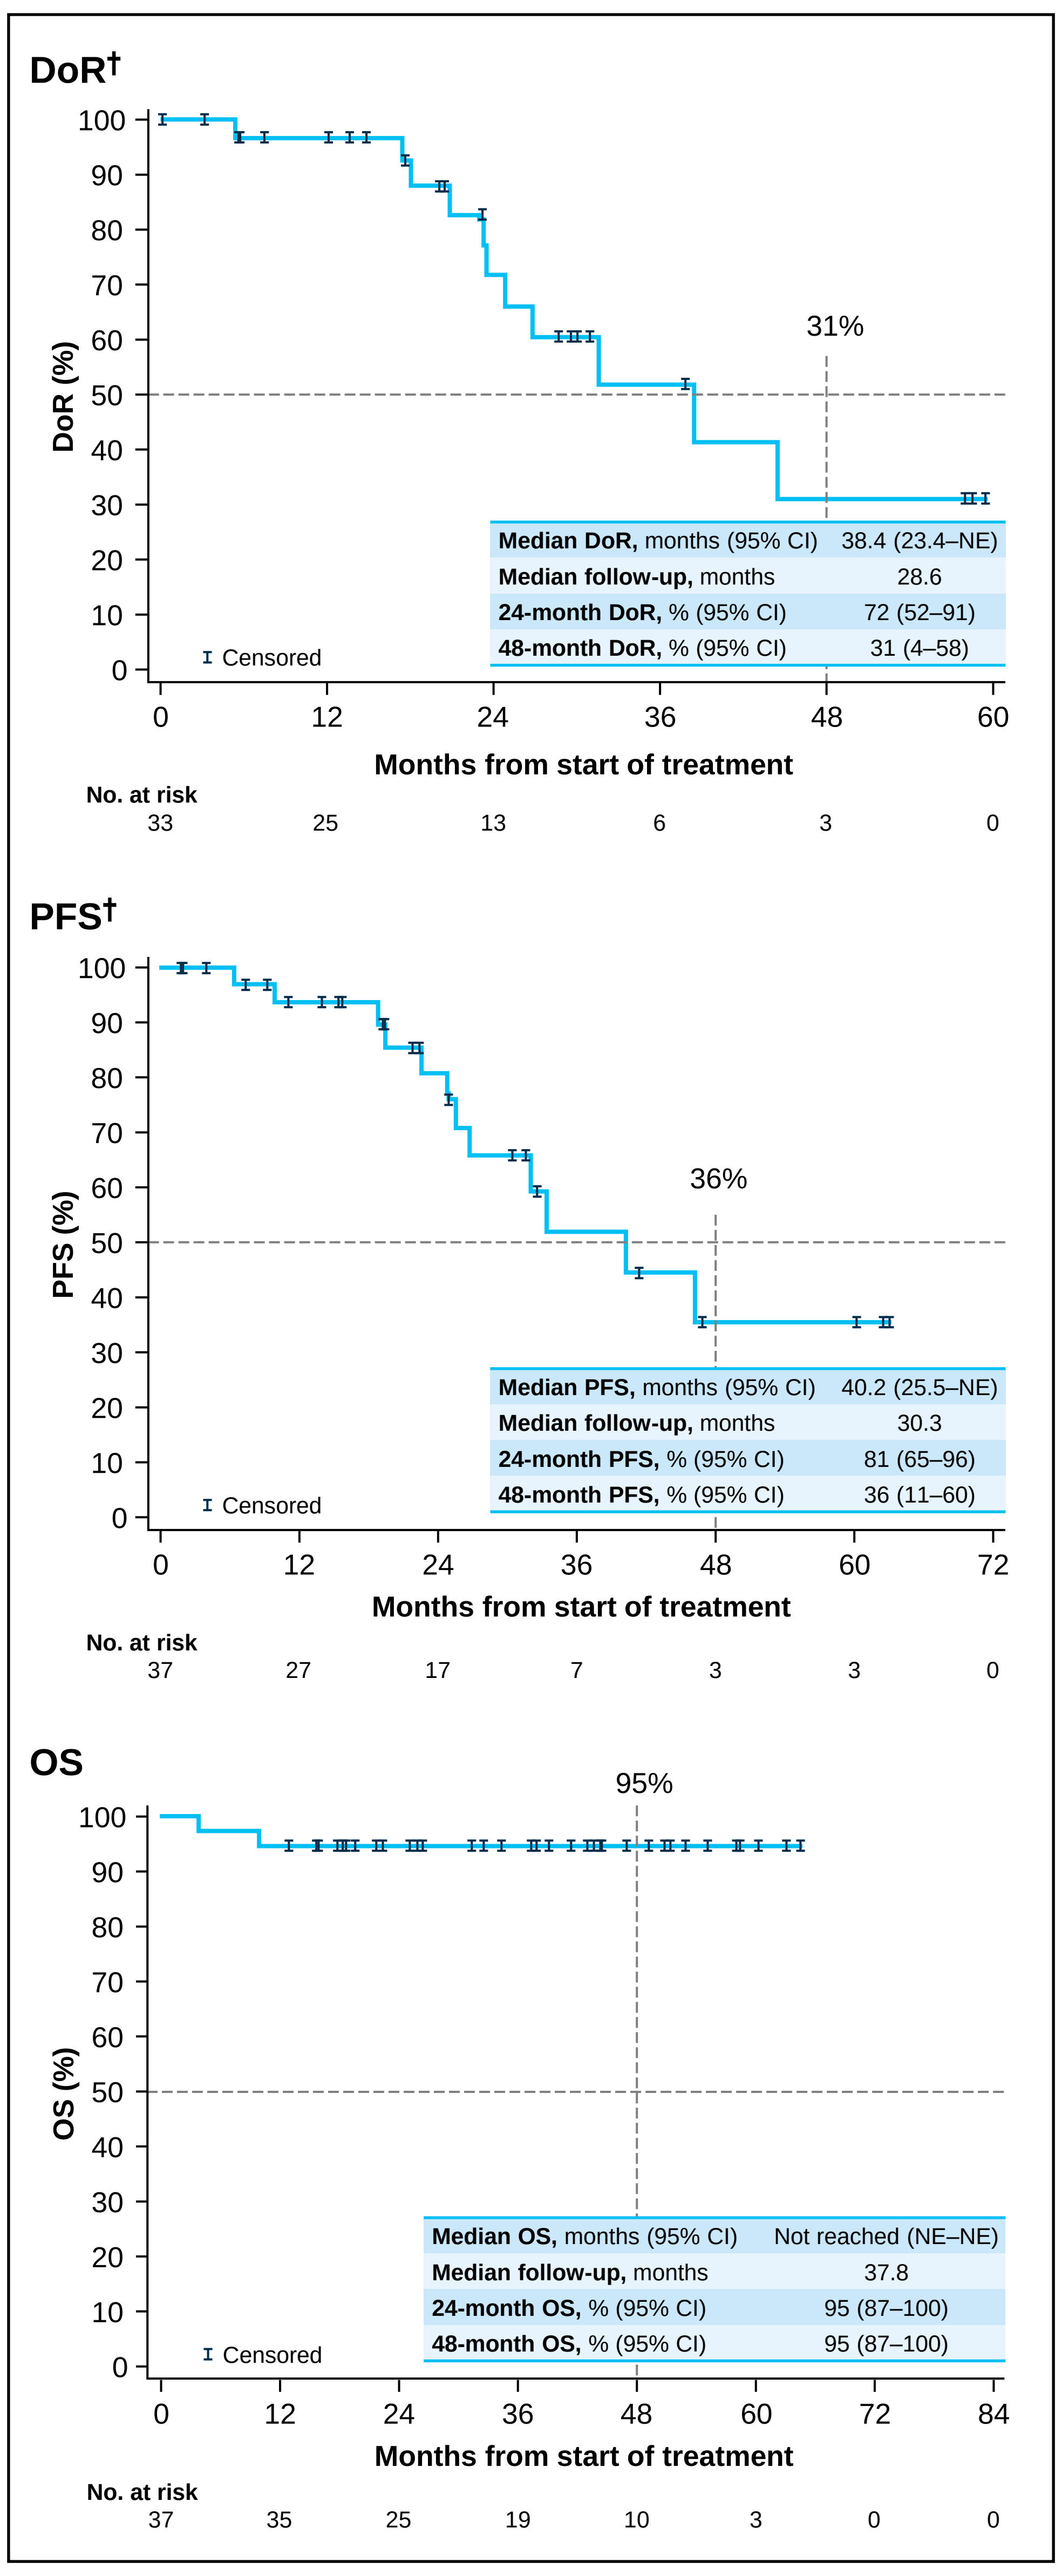
DoR†
DoR (%)
31%
Censored
Median DoR, months (95% CI) 38.4 (23.4–NE)
Median follow-up, months 28.6
24-month DoR, % (95% CI) 72 (52–91)
48-month DoR, % (95% CI) 31 (4–58)
Months from start of treatment
No. at risk 33 25 13 6 3 0
PFS†
PFS (%)
36%
Censored
Median PFS, months (95% CI) 40.2 (25.5–NE)
Median follow-up, months 30.3
24-month PFS, % (95% CI) 81 (65–96)
48-month PFS, % (95% CI) 36 (11–60)
Months from start of treatment
No. at risk 37 27 17 7 3 3 0
OS
OS (%)
95%
Censored
Median OS, months (95% CI) Not reached (NE–NE)
Median follow-up, months 37.8
24-month OS, % (95% CI) 95 (87–100)
48-month OS, % (95% CI) 95 (87–100)
Months from start of treatment
No. at risk 37 35 25 19 10 3 0 0

†DoR and PFS analyses included first progression or death for patients in 'wait-and-see', if applicable. CI, confidence interval; DoR, duration of response; NE, not estimable; OS, overall survival; PFS, progression-free survival.

Figure 4. Patients with TRK fusion cancer on study (N=37)†

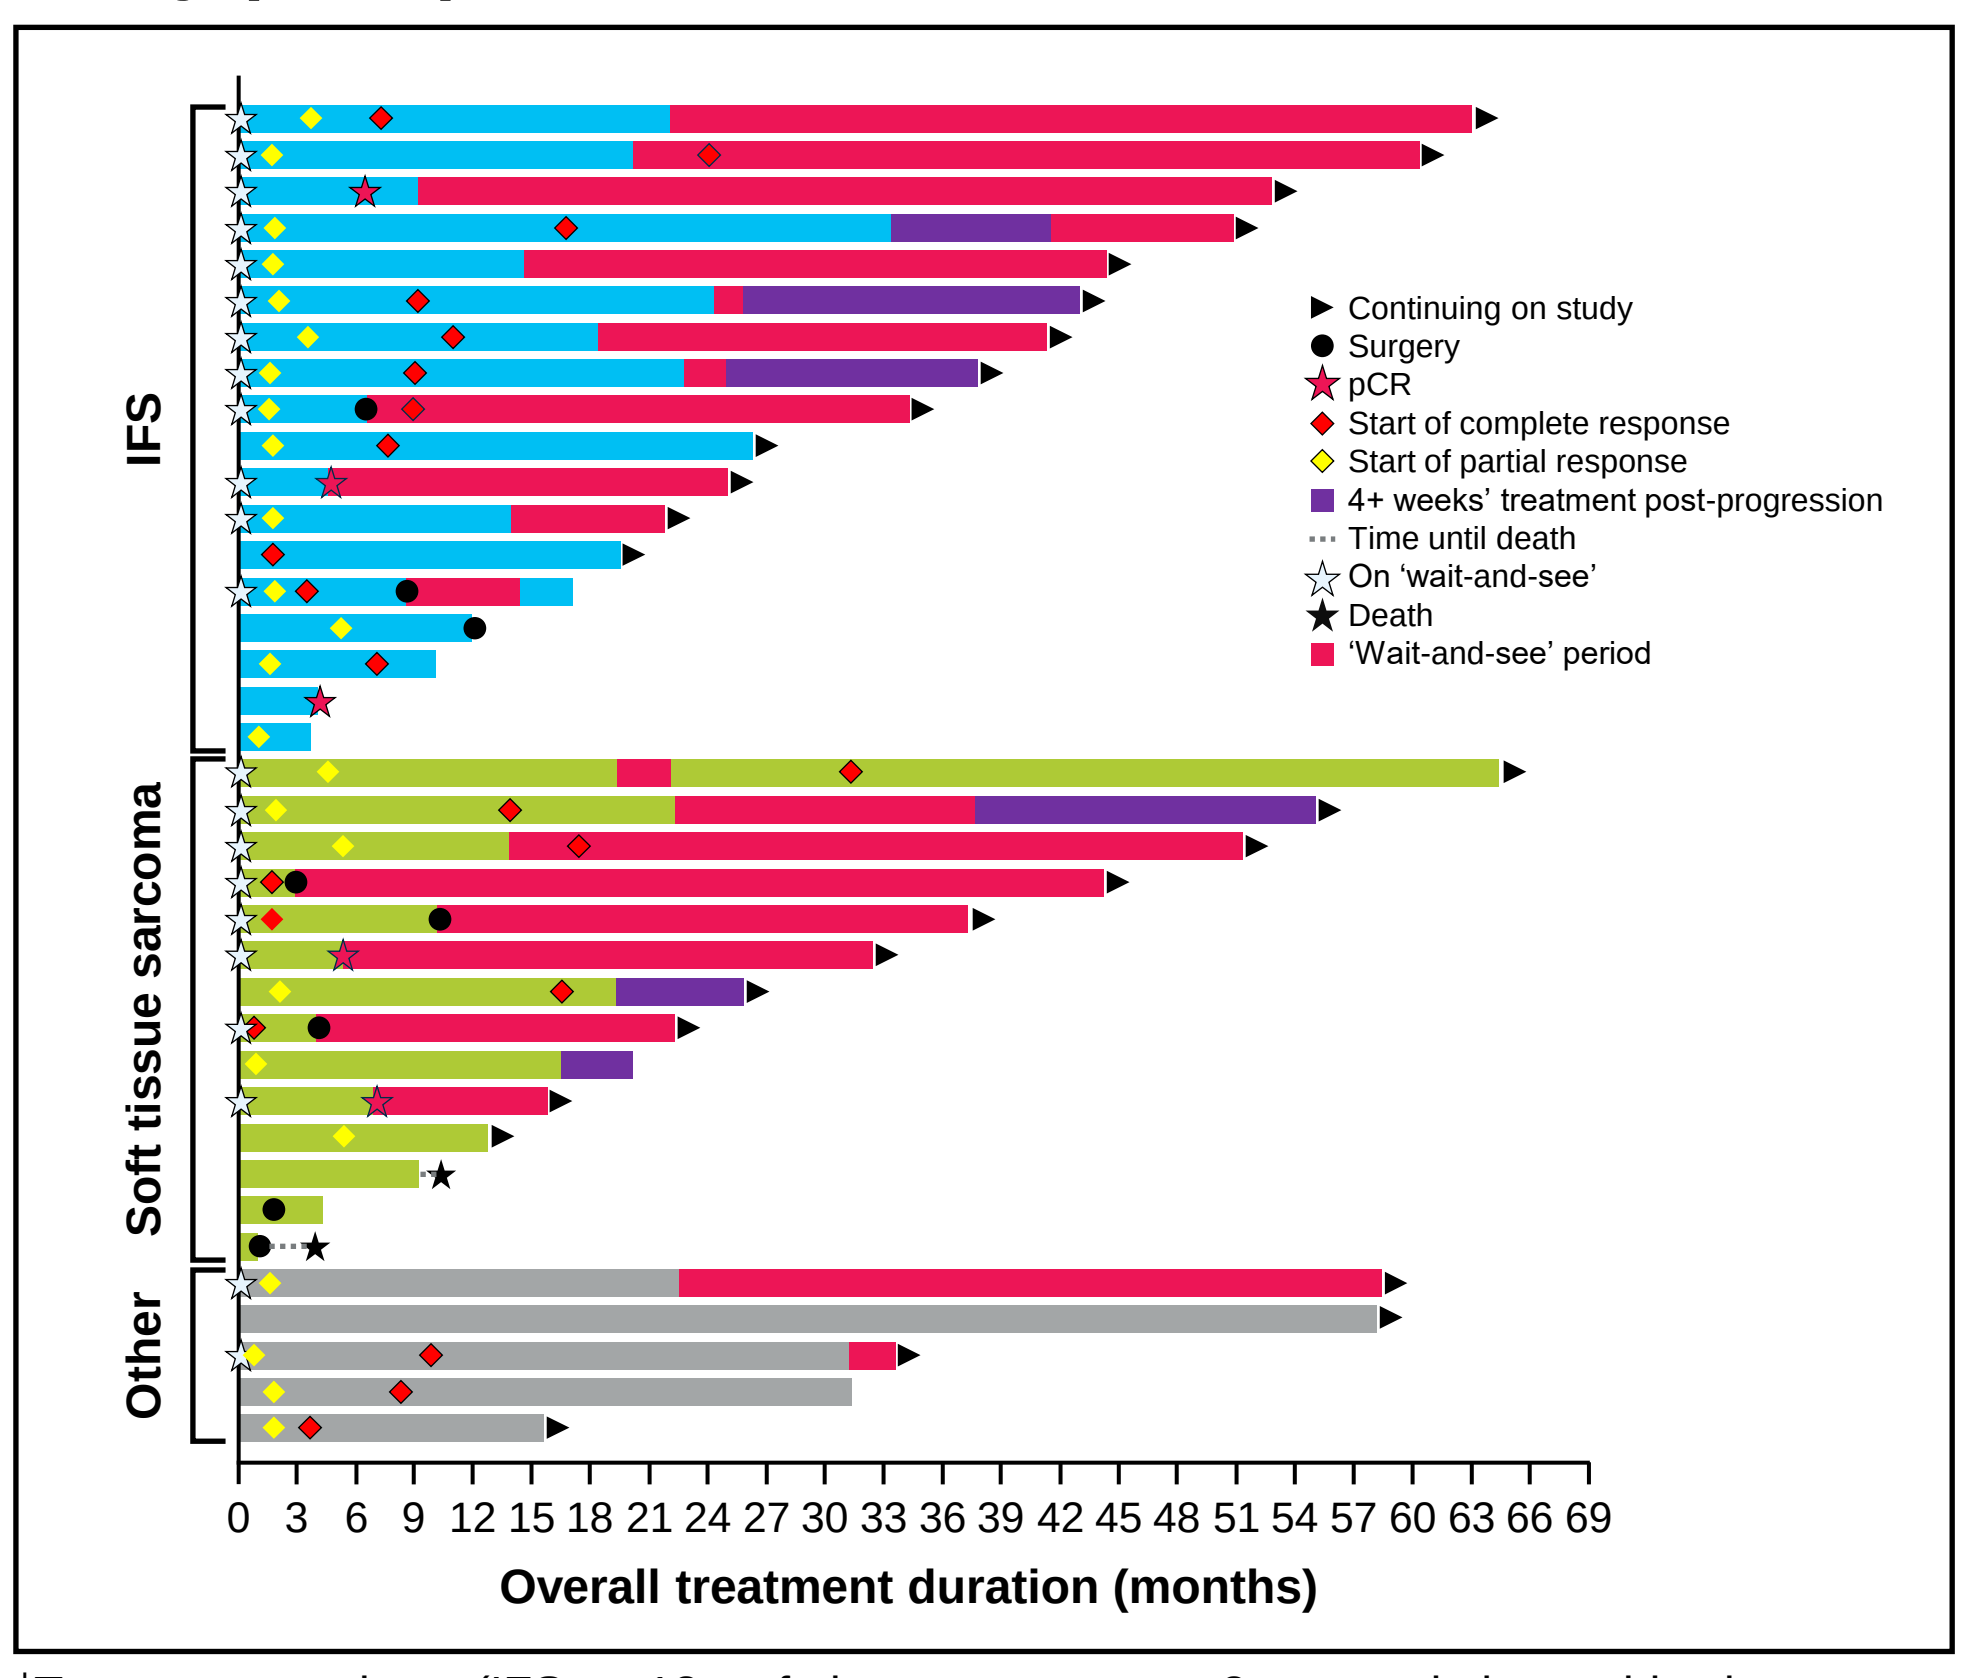
IFS
Soft tissue sarcoma
Other
Continuing on study
Surgery
pCR
Start of complete response
Start of partial response
4+ weeks' treatment post-progression
Time until death
On 'wait-and-see'
Death
'Wait-and-see' period
Overall treatment duration (months)

†Twenty-two patients (IFS: n=12; soft tissue sarcoma: n=8; congenital mesoblastic nephroma: n=2) were enrolled in a 'wait-and-see' analysis (patients with a response were taken off drug and still enrolled in the trial).
IFS, infantile fibrosarcoma; pCR, pathological complete response.

Safety

- Treatment-related adverse events (TRAEs) occurred in 33 (89%) patients and were mainly Grade 1 or 2 (**Figure 5**).
- Grade 3 and 4 TRAEs were reported in 15 (41%) patients. The most common was neutrophil count decrease (8%).
- Two patients discontinued treatment due to a TRAE (malaise and hypoventilation occurred in 1 patient each).
- There were no deaths due to TRAEs. One patient died due to a treatment-emergent adverse event (respiratory failure).

Figure 5. AEs occurring in ≥20% of patients with TRK fusion cancer (N=37)

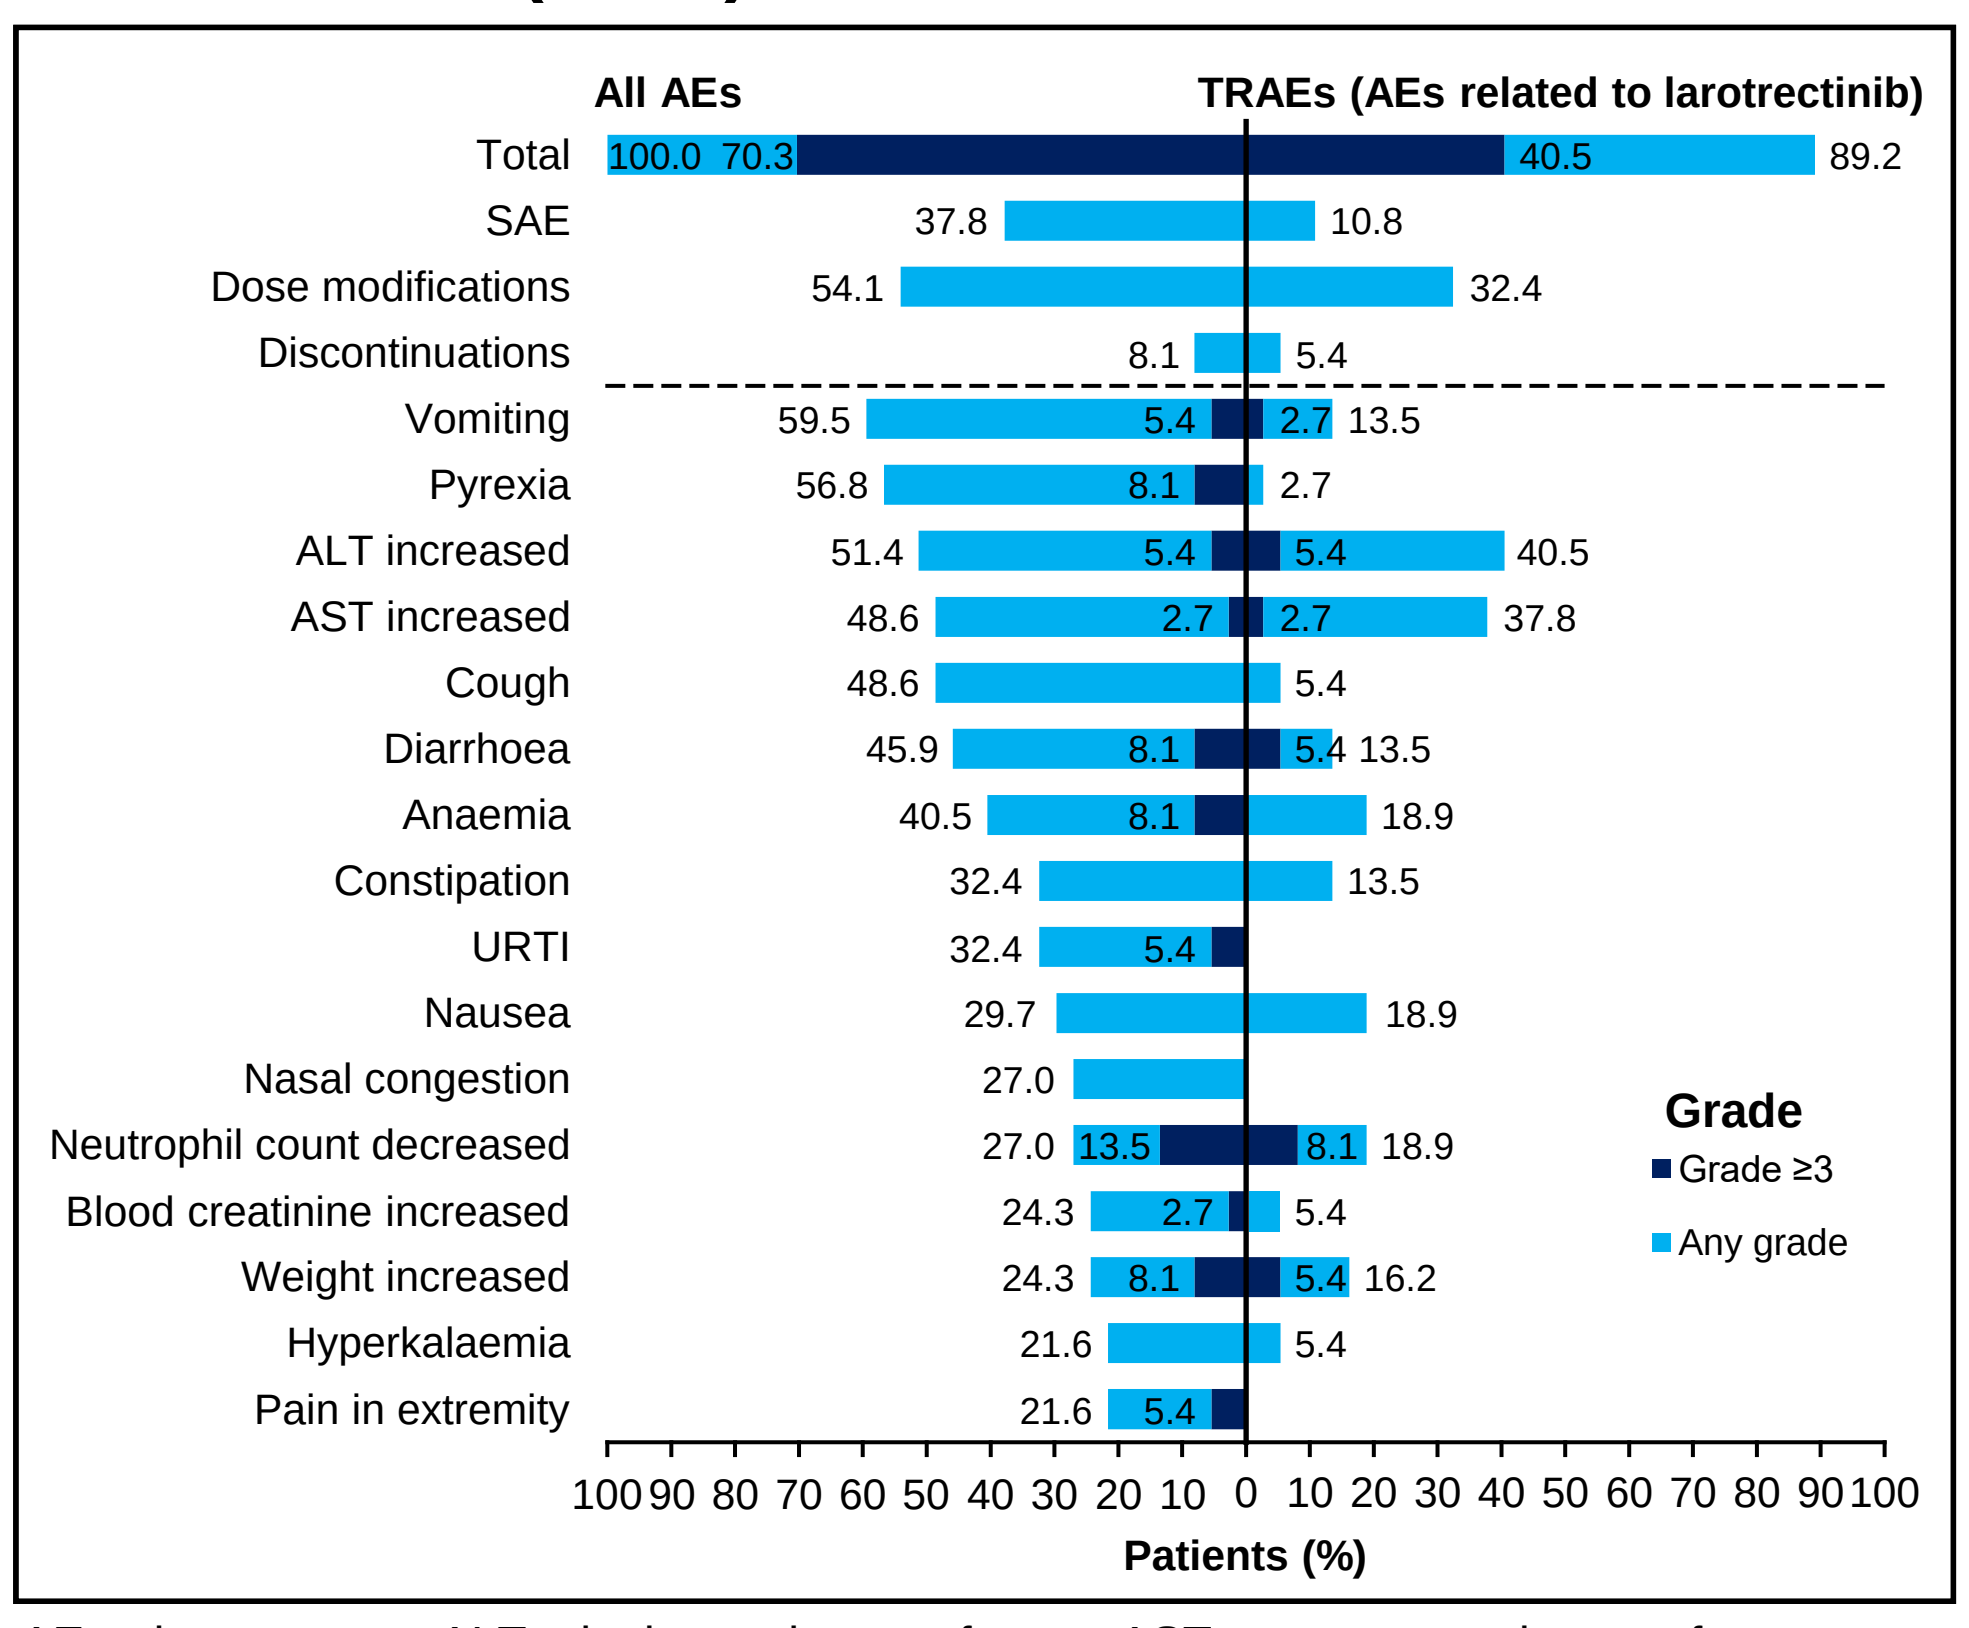
All AEs
TRAEs (AEs related to larotrectinib)
Total 100.0 70.3 40.5 89.2
SAE 37.8 10.8
Dose modifications 54.1 32.4
Discontinuations 8.1 5.4
Vomiting 59.5 5.4 2.7 13.5
Pyrexia 56.8 8.1 2.7
ALT increased 51.4 5.4 5.4 40.5
AST increased 48.6 2.7 2.7 37.8
Cough 48.6 5.4
Diarrhoea 45.9 8.1 5.4 13.5
Anaemia 40.5 8.1 18.9
Constipation 32.4 13.5
URTI 32.4 5.4
Nausea 29.7 18.9
Nasal congestion 27.0
Neutrophil count decreased 27.0 13.5 8.1 18.9
Blood creatinine increased 24.3 2.7 5.4
Weight increased 24.3 8.1 5.4 16.2
Hyperkalaemia 21.6 5.4
Pain in extremity 21.6 5.4
Grade
Grade ≥3
Any grade
Patients (%)

AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; SAE, serious adverse event; TRAE, treatment-related adverse event; URTI, upper respiratory tract infection.

Presented at: ESMO Sarcoma and Rare Cancers, 14–16 March 2024, Lugano, Switzerland

Presented by: Daniel Orbach, daniel.orbach@curie.fr