Real-World Effectiveness and Safety of Damoctocog Alfa Pegol in Canadian Patients with Hemophilia A: Interim Results from the HEM-POWR Study

Publication No. PB0661

Davide Matino[1], Anthony KC Chan[2], Nasrin Samji[2], Alfonso Iorio[1]

[1]McMaster University, Department of Medicine, Hamilton, Canada; [2]McMaster Children's Hospital, Hamilton, Canada.

Contact details for presenting author: Davide Matino, matinod@mcmaster.ca

CONCLUSIONS

- **This third interim analysis of the HEM-POWR study includes a subgroup of patients from Canada, and demonstrates the real-world effectiveness and acceptable safety profile of prophylactic damoctocog alfa pegol in previously treated patients (PTPs) with hemophilia A.**
- **The findings provide valuable insights into real-world clinical practice in Canada.**

AIMS

- **To explore effectiveness and safety of damoctocog alfa pegol in a subgroup of PTPs from Canada with hemophilia A from the third interim analysis of the HEM-POWR study.**

INTRODUCTION

- Damoctocog alfa pegol (BAY 94-9027, Jivi®) is an extended half-life PEGylated recombinant Factor VIII product approved in Canada for treatment of PTPs aged ≥12 years with hemophilia A.[1]
- Damoctocog alfa pegol has demonstrated real-world effectiveness and safety in earlier interim analyses of the HEM-POWR study.[2,3]
- HEM-POWR (NCT03932201) is an ongoing, observational, multicenter, open-label, prospective Phase 4 study assessing the effectiveness and safety of damoctocog alfa pegol in PTPs with hemophilia A in clinical practice.[4]

METHODS

- This third interim analysis of the HEM-POWR study included all patients currently enrolled from Canada. The primary endpoint was annualized bleeding rate (ABR), and secondary endpoints included joint health and treatment-emergent adverse events (TEAEs).
- PTPs with hemophilia A receiving damoctocog alfa pegol with any treatment modality were eligible for enrollment to the study.
- Data were collated from patient e-diaries and physician records. Statistics were descriptive and exploratory. Ethical approval was obtained for all study sites.
- Patients were included in the safety analysis set (SAF) if they had ≥1 study dose in the observation period and provided informed consent; patients included in the full analysis set (FAS) fulfilled all inclusion criteria with a documented first study drug dose and ≥1 infusion during the observation period.

RESULTS

- At data cut-off (August 17, 2022), the FAS for this interim subgroup analysis included 19 patients, including 1 (5.3%) pediatric patient. Patients were observed for a mean (SD) of 345.9 (131.0) days.
- The majority of patients (n=18/19; 94.7%) had severe hemophilia A at study enrollment **(Table 1)**.

Table 1: DEMOGRAPHICS AND BASELINE CHARACTERISTICS (FAS)

Characteristics for subgroup of patients from Canadian study sites	FAS, n (%) (N=19)
Observation period, days, median (Q1, Q3); mean (SD)	386.0 (208.0, 466.0); 345.9 (131.0)
Sex, male, n (%)	19 (100.0)
Race, n (%)	
White	16 (84.2)
Black or African American	3 (15.8)
Age at enrollment, years, median (Q1, Q3); mean (SD)	33.0 (24.0, 41.0); 35.3 (12.8)
Age at enrollment, years, n (%)	
≥12 to <18	1 (5.3)
≥18 to <65	18 (94.7)
Weight, kg, median (Q1, Q3); mean (SD)*	84.0 (82.0, 86.0); 87.4 (18.6)
Severity of hemophilia A at initial diagnosis, n (%)	
Mild	0 (0.0)
Moderate	1 (5.3)
Severe	18 (94.7)
Patient history of inhibitors, no, n (%)	19 (100.0)
Prophylactic treatment prior to enrollment, yes, n (%)†	18 (94.7)
Prescribed dose per infusion per kg of damoctocog alfa pegol at first visit during the observation period, IU/kg, median (Q1, Q3); mean (SD)‡	24.4 (23.5, 34.9); 28.7 (7.4)

*Data missing for 9 (47.4%) patients; †Data missing for 1 (5.3%) patient; ‡Data missing for 5 (26.3%) patients.
FAS, full analysis set; Q1, first quartile; Q3, third quartile; SD, standard deviation.

- A total of 18/19 patients received prophylactic treatment prior to the study (1 missing) and 14/19 (73.7%) patients were pre-treated with damoctocog alfa pegol.
- Most patients (n=11/19; 57.9%) were treated with damoctocog alfa pegol every 3–4 days at baseline **(Figure 1)**.
- Within 12 months prior to damoctocog alfa pegol initiation, the median (mean, SD) number of total bleeds was 2.0 (3.3, 6.2). During the observation period, the median (mean, SD) ABR for total bleeds was 0.0 (0.7, 1.0) **(Figure 2)**.

Figure 1: PRESCRIBED PROPHYLAXIS REGIMEN OF DAMOCTOCOG ALFA PEGOL AT BASELINE (FAS)

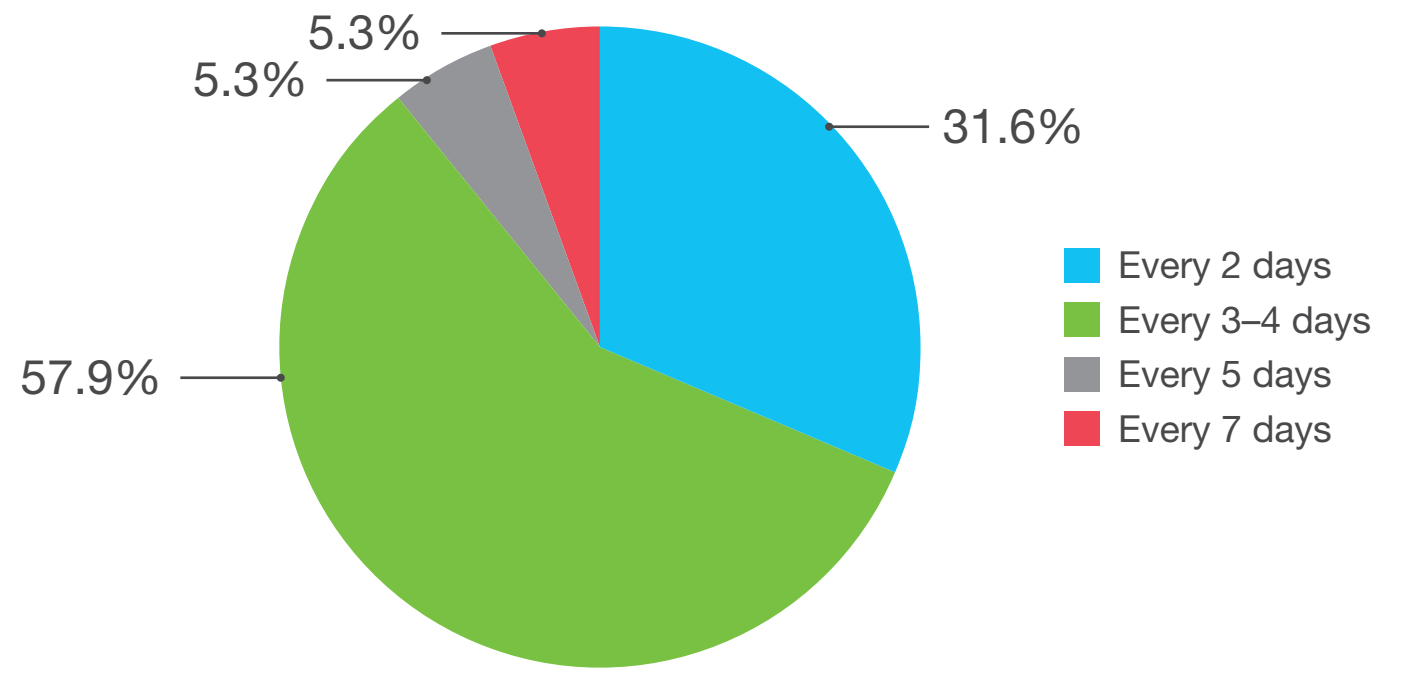
5.3%
5.3%
31.6%
57.9%
Every 2 days
Every 3–4 days
Every 5 days
Every 7 days

Figure 2: ABR PRIOR TO INITIATION OF DAMOCTOCOG ALFA PEGOL AND DURING THE OBSERVATION PERIOD

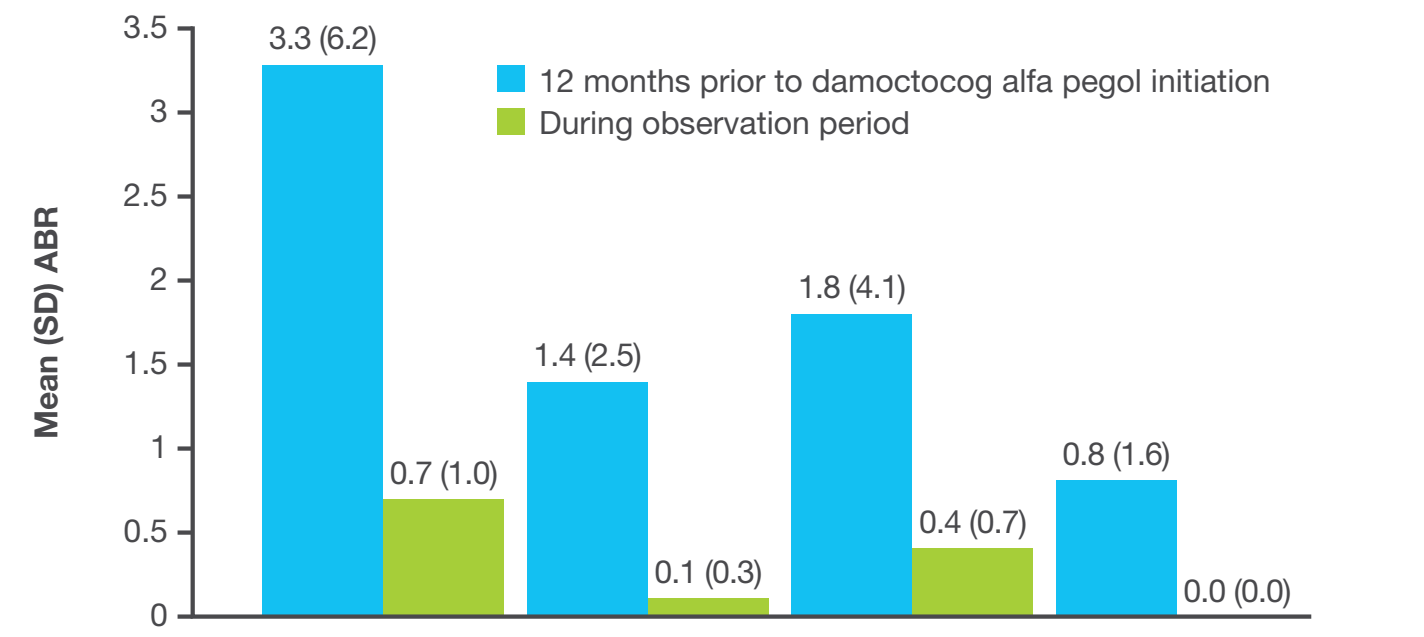
Mean (SD) ABR
3.5
3
2.5
2
1.5
1
0.5
0
3.3 (6.2)
0.7 (1.0)
1.4 (2.5)
0.1 (0.3)
1.8 (4.1)
0.4 (0.7)
0.8 (1.6)
0.0 (0.0)
12 months prior to damoctocog alfa pegol initiation
During observation period

	Total	Spontaneous	Joint	Spontaneous joint
Median (Q1, Q3) number of bleeds prior to damoctocog alfa pegol initiation	2.0 (0.0, 4.0)	0.0 (0.0, 2.0)	0.0 (0.0, 2.0)	0.0 (0.0, 1.0)
Median (Q1, Q3) ABR during observation period	0.0 (0.0, 1.0)	0.0 (0.0, 0.0)	0.0 (0.0, 0.8)	0.0 (0.0, 0.0)

Data from 12 months prior to initiation were the average number of bleeds over 12 months; data during the observation period were calculated based on an annualized rate.
ABR, annualized bleeding rate; FAS, full analysis set; Q1, first quartile; Q3, third quartile; SD, standard deviation.

- Overall median (mean, SD) change in ABR during the observation period from prior to damoctocog alfa pegol initiation was –1.0 (–2.6, 6.0). The median (mean, SD) overall change in ABR during the observation period from prior to damoctocog alfa pegol was 0.0 (–1.3, 2.6) for spontaneous bleeds, 0.0 (–1.4, 3.8) for joint bleeds, and 0.0 (–0.8, 1.6) for spontaneous joint bleeds **(Figure 3)**.
- In the SAF, TEAEs occurred in 8/19 (42.1%) patients; 3/19 (15.8%) patients had serious TEAEs. There was 1 serious TEAE that led to a change of treatment regimen **(Table 2)**.
- No study-drug-related TEAEs or discontinuations were reported.

Figure 3: DIFFERENCE IN ABR DURING OBSERVATION PERIOD FROM 12 MONTHS PRIOR TO DAMOCTOCOG ALFA PEGOL INITIATION (FAS, N=19)

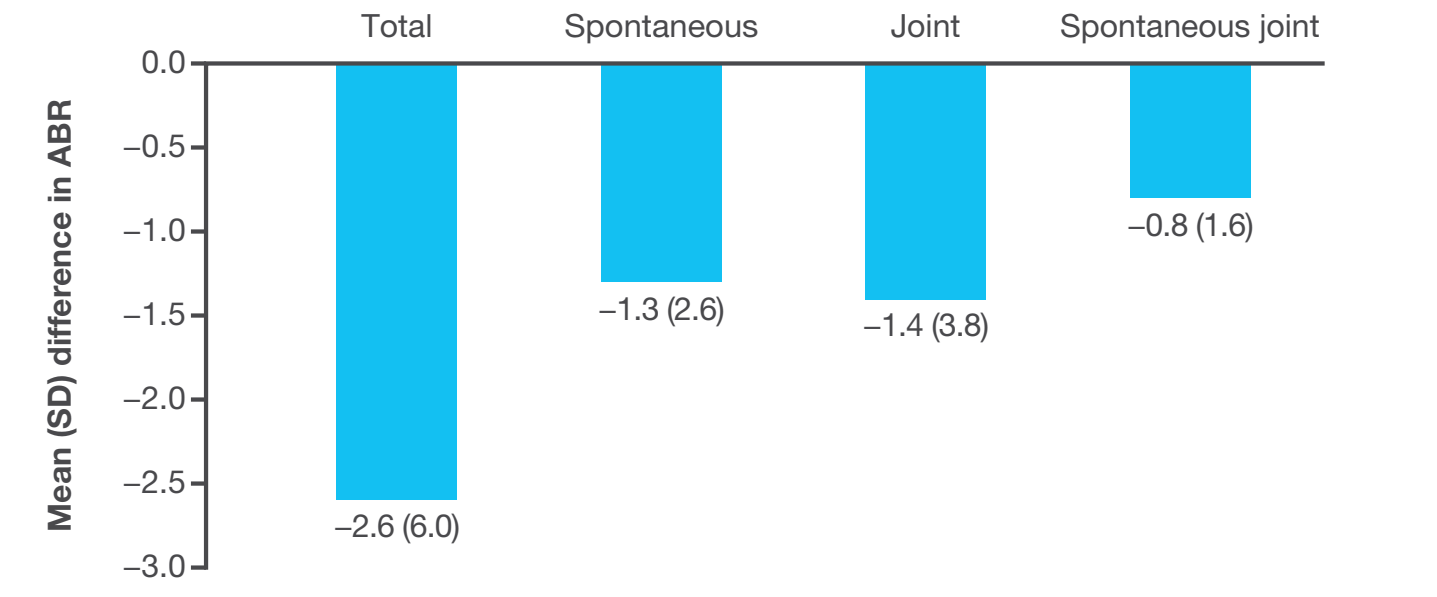
Total
Spontaneous
Joint
Spontaneous joint
Mean (SD) difference in ABR
0.0
–0.5
–1.0
–1.5
–2.0
–2.5
–3.0
–2.6 (6.0)
–1.3 (2.6)
–1.4 (3.8)
–0.8 (1.6)

	Total	Spontaneous	Joint	Spontaneous joint
Median (Q1, Q3) difference in ABR during observation period from 12 months prior to damoctocog alfa pegol initiation	–1.0 (–3.0, 0.0)	0.0 (–2.0, 0.0)	0.0 (–1.2, 0.0)	0.0 (–1.0, 0.0)

ABR, annualized bleeding rate; FAS, full analysis set; Q1, first quartile; Q3, third quartile; SD, standard deviation.

Table 2: SUMMARY OF TEAES (SAF)

Characteristic	FAS, n (%) (N=19)
Any TEAE, n (%)	8 (42.1)
Any TEAE of special interest	0 (0.0)
Any study-drug-related TEAE	0 (0.0)
TEAE-related death	0 (0.0)
Any TEAE leading to change of treatment regimen of damoctocog alfa pegol	1 (5.3)
Any TEAE leading to discontinuation of damoctocog alfa pegol treatment	0 (0.0)
Any TEAE related to inhibitor development	0 (0.0)
Any serious TEAE, n (%)	3 (15.8)
Any serious TEAE of special interest	0 (0.0)
Any study-drug-related serious TEAE	0 (0.0)
Any serious TEAE leading to change of treatment regimen of damoctocog alfa pegol	1 (5.3)
Any serious TEAE leading to discontinuation of damoctocog alfa pegol treatment	0 (0.0)
Any serious TEAE related to inhibitor development	0 (0.0

SAF, safety analysis set; TEAE, treatment-emergent adverse event.

This is an interim analysis and data are presented as observed until that time point. Information about bleeds/infusions are provided by the patients at a visit and therefore enter the database retrospectively. Data tables for this analysis were updated following submission of the abstract and we are presenting the updated data here. The conclusions and interpretation of results are consistent with the abstract.

Acknowledgments

The authors would like to acknowledge Martin Sanabria of Bayer, for his contributions to the HEM-POWR study. Editorial support (in the form of writing assistance, including development of the initial draft based on author direction, collating authors' comments and grammatical editing) was provided by Natalie Mitchell, MSc, of Fishawack Communications Limited, part of Fishawack Health, UK, and was funded by Bayer. This study was funded by Bayer.

Conflict of interest

DM has received research support from Bayer, Bioverativ/Sanofi, and Pfizer; and honoraria for speaking/participating in advisory boards from Bayer, Bioverativ/Sanofi, BIOVIIIx, Pfizer, and Sigilon.
AC has received support with clinical trials from Bayer, Centessa Pharmaceuticals, Novo, Pfizer, Takeda
NS is Principal Investigator for several clinical trials funded by Bayer, and an investigator for clinical trials supported by Novo Nordisk, Pfizer, Centessa Pharmaceuticals, and Sanofi.
AI's institution has received project-based funding via research or service agreements with Bayer, CSL, Grifols, Novo Nordisk, Octapharma, Pfizer, Roche, Sanofi, Sobi, and Takeda.

References

1. Bayer Inc. Damoctocog alfa pegol (Jivi®). Product Monograph. Last updated September 2021. Available at: https://www.bayer.com/sites/default/files/2020-11/Jivi-pm-en.pdf [Accessed: April 2023]
2. Reding M (2022). ISTH 2022 Congress. PB1123.
3. Reding M, et al. PO124 poster presentation. *Haemophilia.* 28:25-126. Available at: https://doi.org/10.1111/hae.14479.
4. Sanabria M, et al. *BMJ Open.* 2021;11(9):e044997.

Presented at ISTH 2023 congress, 24-28 June