The Potential Long-term Comparative Effectiveness of Larotrectinib vs. Repotrectinib for Treatment of *NTRK* Gene Fusion-positive Cancers

Kangho Suh, PharmD, PhD[1], Ashley Kang, MPH[2], Jamie Grossman, PhD, MBA[3], Sreevalsa Appukkuttan, MPH[3], Sean D. Sullivan, PhD[2,4]

[1]University of Pittsburgh, Pittsburgh, PA, USA; [2]Curta Inc., Seattle, WA, USA; [3]Bayer U.S. LLC, Whippany, NJ, USA; [4]CHOICE Institute, University of Washington, Seattle, WA, USA

Introduction

- Research and clinical findings suggest that tropomyosin receptor kinase (TRK) fusions promote tumor dependence on oncogenic signaling, regardless of tissue type, and may account for up to 1% of all solid malignancies.[1,2]
- Larotrectinib, the first approved therapy for *NTRK* gene fusion positive solid cancers, has continued to demonstrate extended durable responses and survival benefit.[3-5]
- A second generation TRK tyrosine kinase inhibitor (TKI), repotrectinib, assessed progression free survival in TRK TKI-naïve patients with *NTRK* gene fusion-positive locally advanced or metastatic solid tumors from a Phase I/II study.[6]

Objective

- This study aimed to estimate and compare long term expected life-years (LYs) and quality-adjusted life-years (QALYs) for adult patients with locally advanced or metastatic *NTRK* gene fusion-positive cancers with larotrectinib or repotrectinib.

Methods

Modeling Approach

- Partitioned survival models were developed to project long-term comparative effectiveness of larotrectinib vs. repotrectinib.
- PFS and OS were estimated from parametric survival distributions (Exponential, Weibull, Log-logistic, and Log-normal).
- OS curves were adjusted as needed so that the study-based mortality rates could not be lower than observed mortality rates for the general public based on U.S. life tables.[7]
- Probabilistic sensitivity analysis with 5,000 simulations were run to obtain 95% credible intervals (CrI).
- A lifetime horizon was used, and outcomes (LYs, QALYs) were discounted at 3%.

Larotrectinib Data Source

- Larotrectinib survival data were derived from the July 2023 analysis of 209 adult patients with *NTRK* gene-fusion positive cancers from the larotrectinib clinical trials program (NCT02637687 and NCT02576431) and included patients with primary central nervous system (CNS) tumors.[8]

Repotrectinib Data Source

- Repotrectinib survival data were derived from a Phase I/II study (NCT03093116) of 40 adult patients with *NTRK* gene fusion-positive cancers with no prior use of TRK inhibitors.[6,9]
- Since repotrectinib OS data were not available for this study population, we imputed repotrectinib OS by applying the OS to PFS ratio observed in the larotrectinib adult population to the repotrectinib PFS in the study population, following the approach used in a previous study.[10]

Methods

Health state utility values

- There were limited publicly available utility data for all tumor types assessed in larotrectinib and repotrectinib
- Utility values were available for four of the most prevalent tumor types across clinical trials for both interventions: Non-small cell lung cancer (NSCLC), soft tissue sarcoma (STS), thyroid, and colorectal cancer (CRC) (Table 1).
- A weighted average utility was calculated based on the prevalence of NSCLC, STS, thyroid, and CRC in both larotrectinib and repotrectinib trials, excluding less common tumor types (Table 2).
- QALYs were estimated by adjusting the time spent in the pre-progression and post-progression health states by utility values derived from publicly available literature (Table 3).

Table 1. Tumor Type Prevalence by Treatment

	Larotrectinib, n (%)[8]	Repotrectinib, n (%)[6]
NSCLC	29 (13.88)	21 (56.76)
STS	29 (13.88)	2 (5.41)
Thyroid	25 (11.96)	5 (13.51)
CRC	26 (12.44)	1 (2.70)
Other	100 (47.85)	8 (21.62)

Table 2. Utilities by Tumor Type

	Pre-progression utility	Responsive Utility	Progressive Utility
NSCLC[11]	0.653	0.019	0.473
STS[12]	0.43	0.125	0.3
Thyroid[13]	0.8	0.06	0.5
CRC[14]	0.82	0.03	0.643
Prevalence-weighted average*	0.67	0.05	0.47

*Weighted by prevalence from Table 1

Table 3. On-Treatment Health State Utility Values and Response Rates*

	Larotrectinib	Repotrectinib
NTRK gene fusion-positive cancers (response rate±)	0.697 (0.47§)	0.701 (0.58)

*On-treatment utilities were calculated as a weighted average of the utility for those in pre-progression and recurrent disease based on the response rate for each treatment.
±Complete or partial response assessed by independent review committee (IRC)
§IRC assessment of both primary CNS and non-CNS TRK fusion tumors

Results

- Exponential curve fits were used based on goodness-of-fit and clinical plausibility for PFS and OS (Figures 1 and 2).

Figure 1. Extrapolated Progression Free Survival

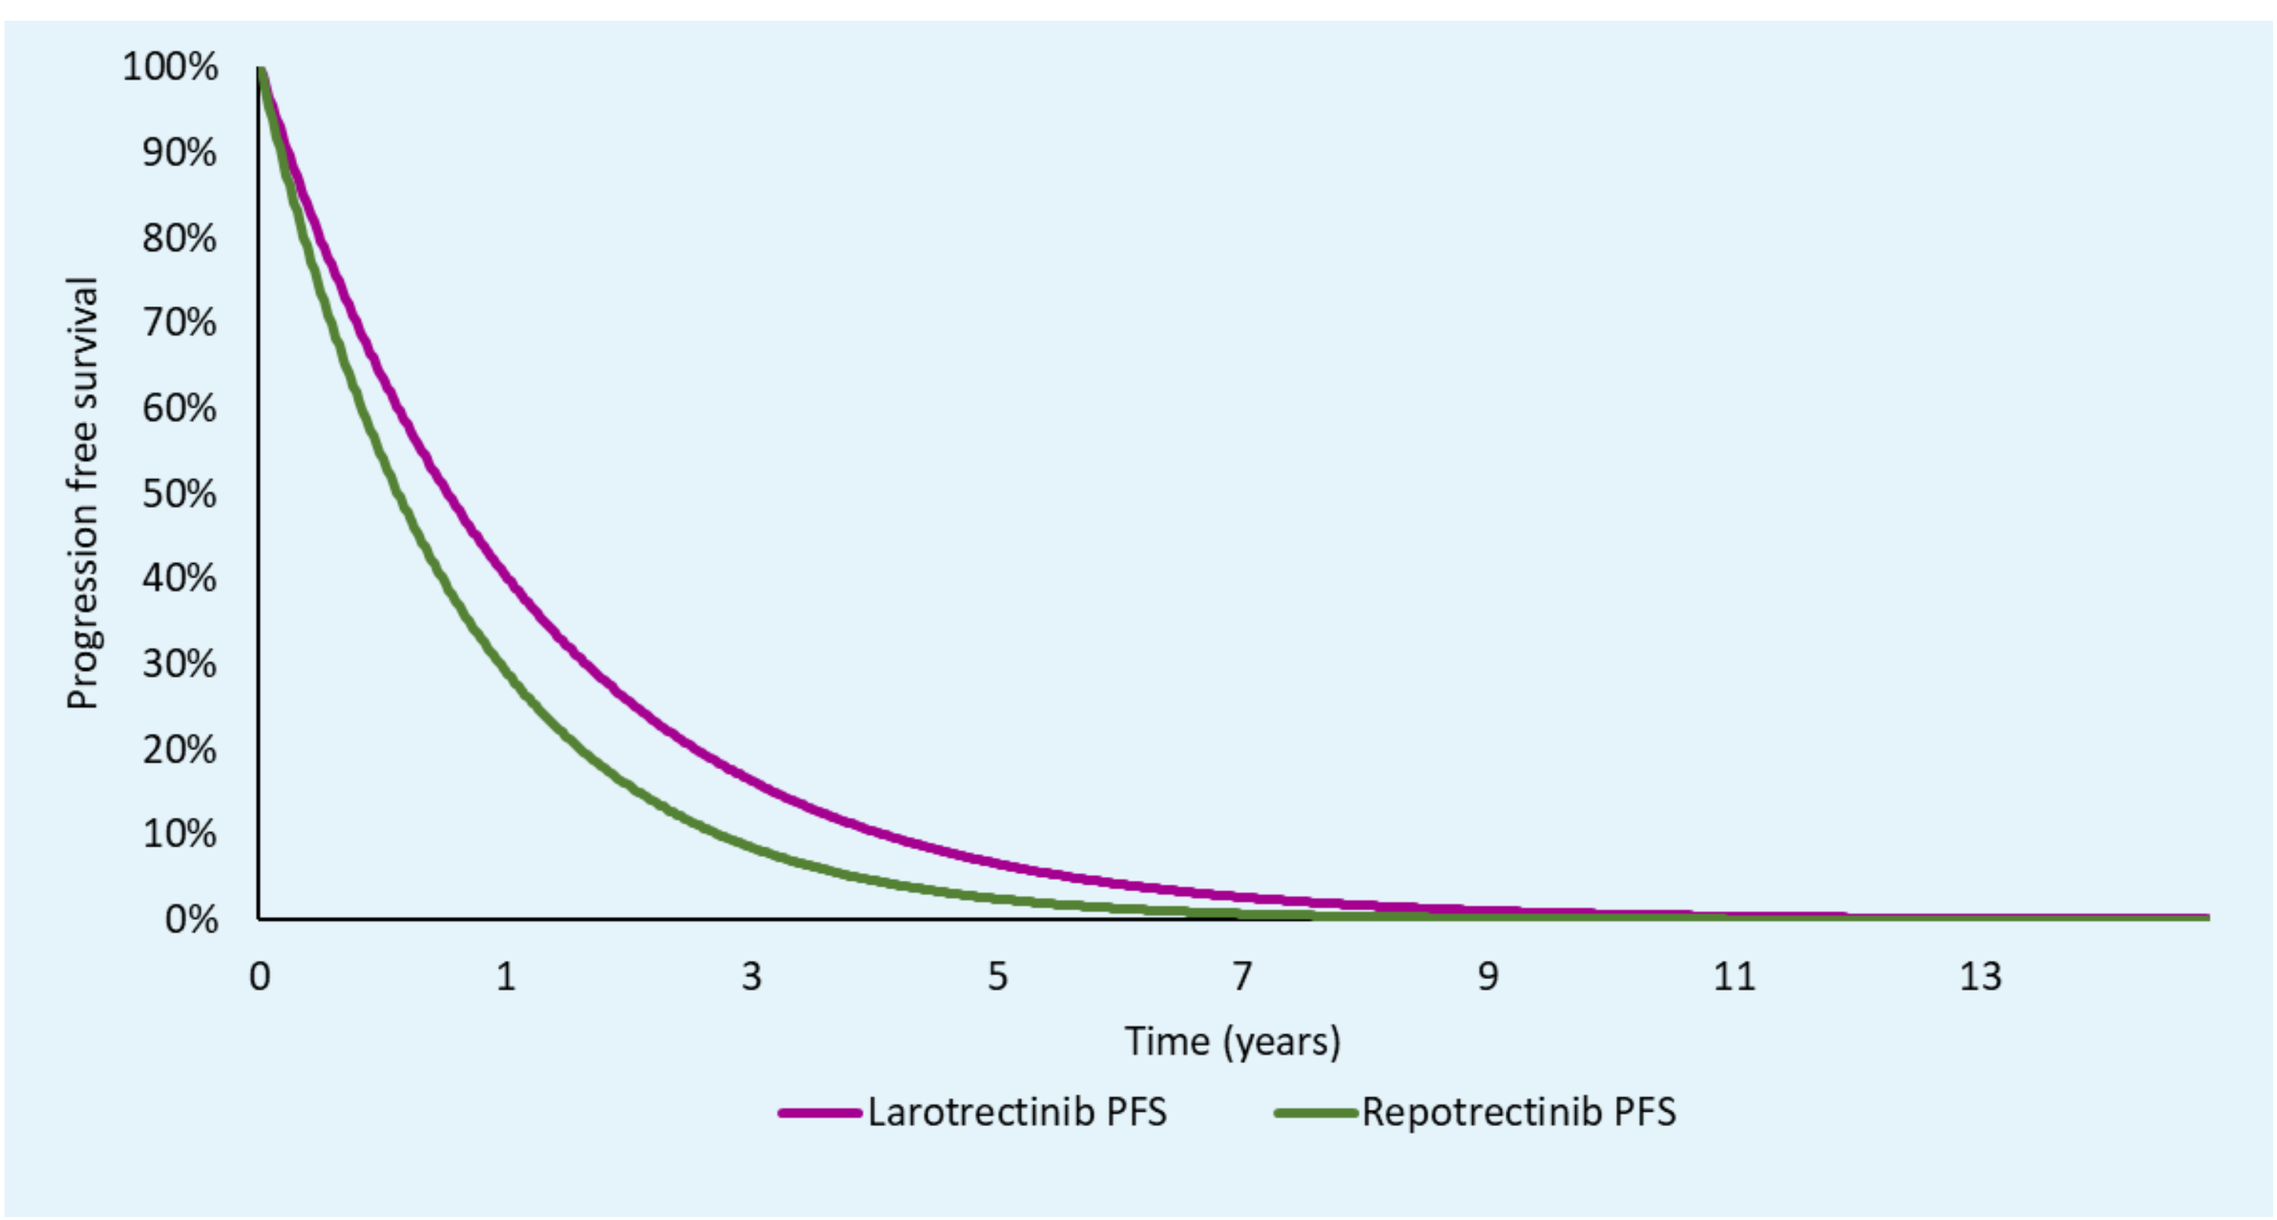
Progression free survival
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0 1 3 5 7 9 11 13
Time (years)
Larotrectinib PFS
Repotrectinib PFS

Figure 2. Extrapolated Overall Survival

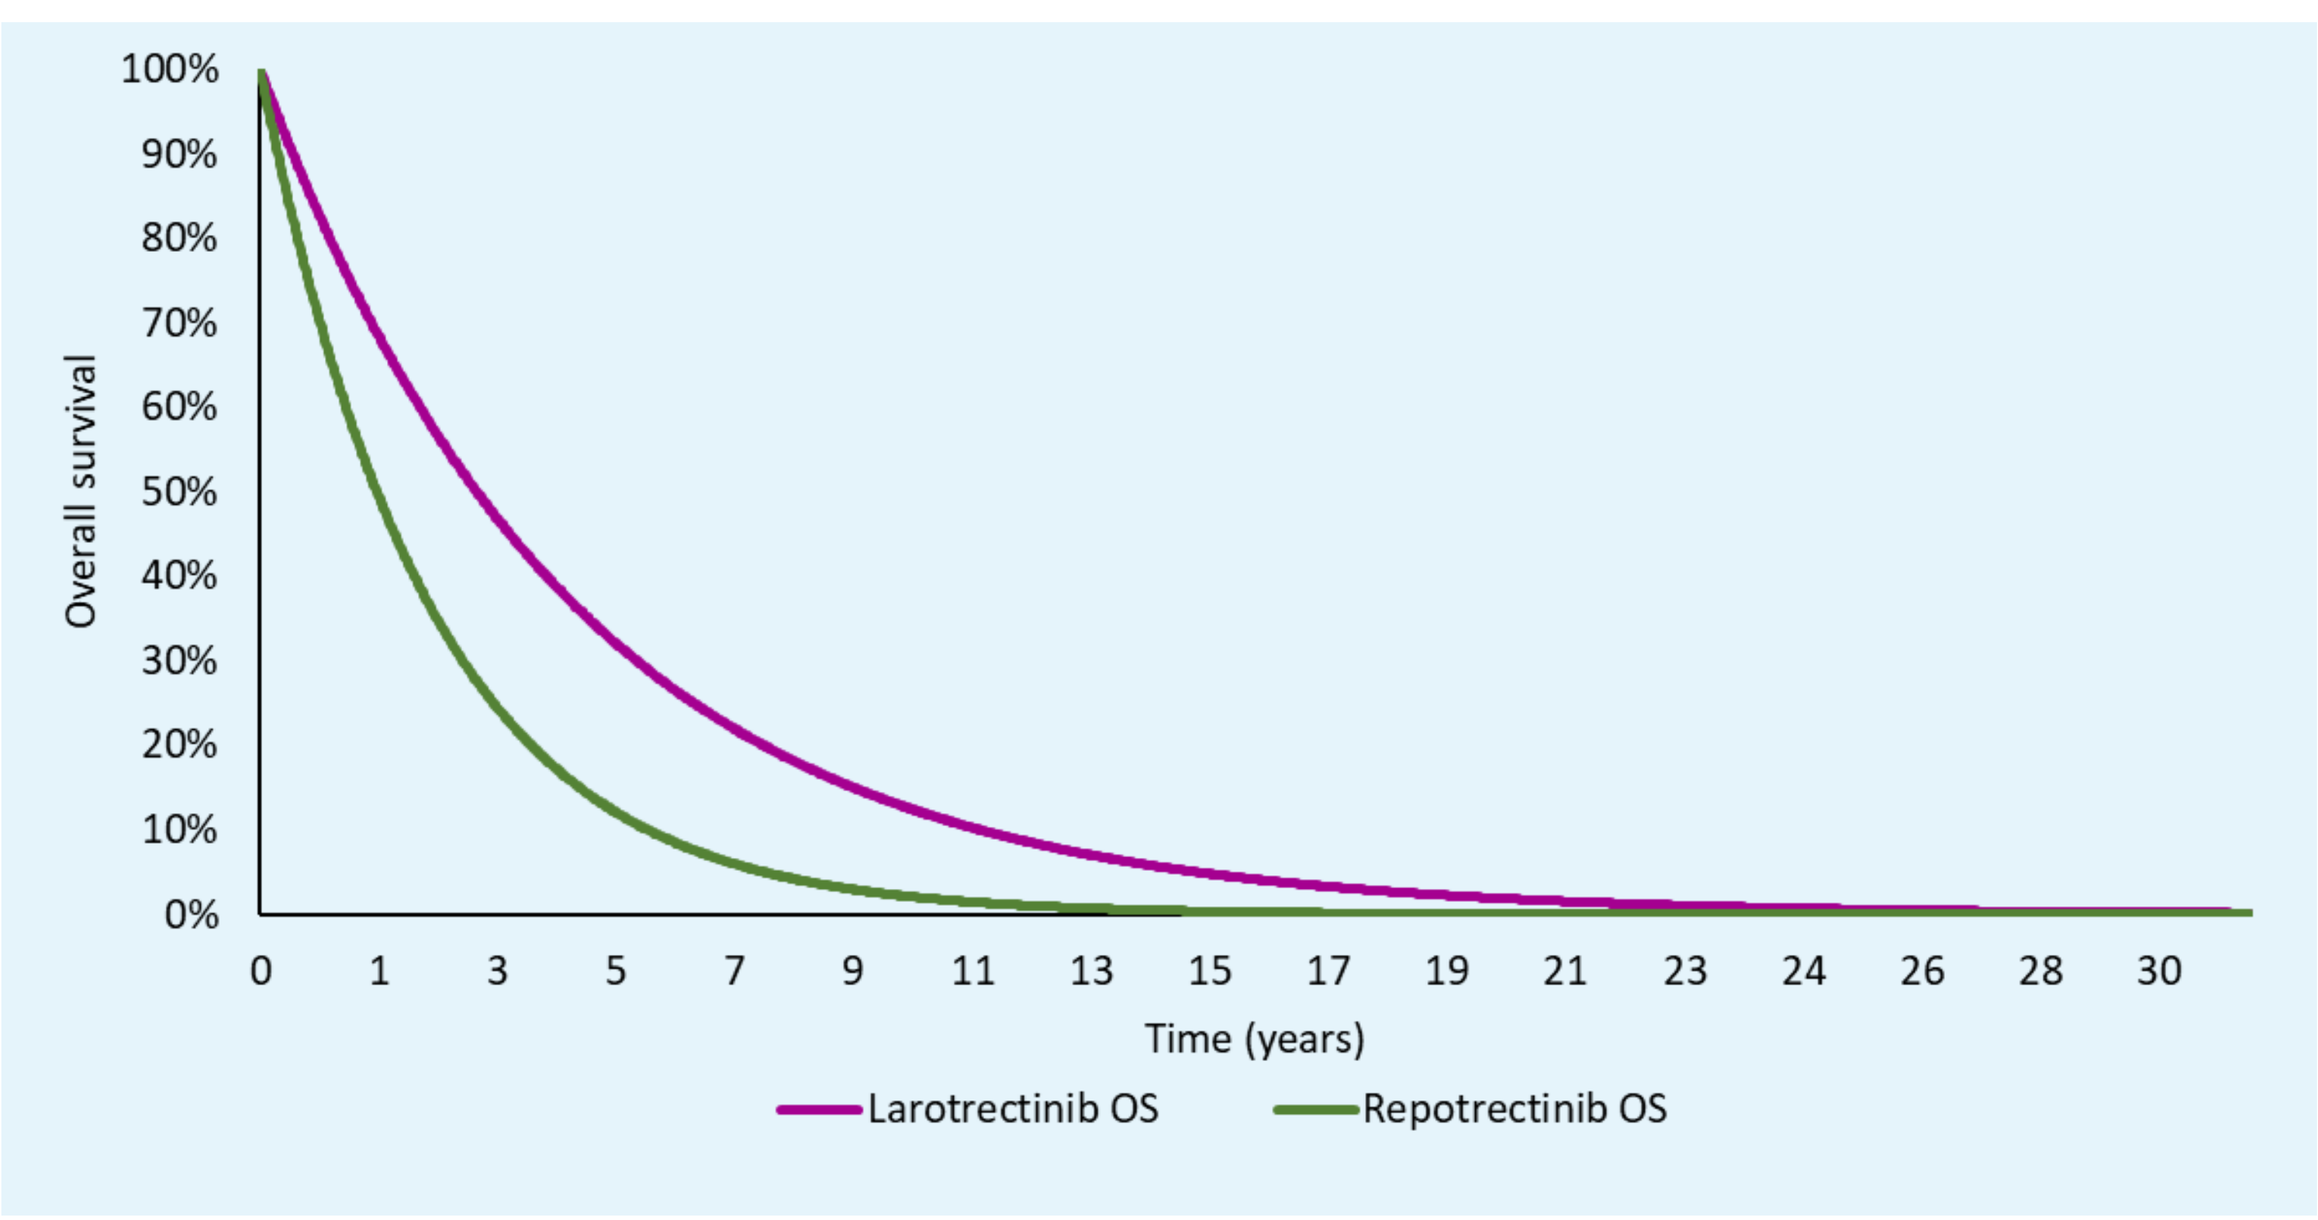
Overall survival
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0 1 3 5 7 9 11 13 15 17 19 21 23 24 26 28 30
Time (years)
Larotrectinib OS
Repotrectinib OS

Results

- In the treatment of *NTRK* gene fusion-positive cancers in adults, larotrectinib resulted in 4.83 LYs (95% Credible Interval [CrI]: 3.93, 5.89) and 2.80 QALYs (95% CrI: 1.46, 4.62) over the time horizon (Table 4).
- These led to gains of 2.47 total LYs compared to repotrectinib, which translated to gains of 1.33 total QALYs.

Table 4. Survival and Quality-Adjusted Survival Outcomes

(95% CrI)	Larotrectinib	Repotrectinib
Pre-Progression LYs	2.36 (1.99, 2.83)	1.56 (0.99, 2.47)
Post-Progression LYs	2.46 (1.41, 3.60)	0.80 (0.22, 2.97)
Total LYs	4.83 (3.93, 5.89)	2.36 (1.23, 5.31)
Pre-Progression QALYs	1.64 (1.15, 2.19)	1.09 (0.63, 1.83)
Post-Progression QALYs	1.17 (0.01, 2.98)	0.38 (0.00, 1.84)
Total QALYs	2.80 (1.46, 4.62)	1.47 (0.71, 3.41)

CrI: credible interval, NE: not estimable

Conclusions

- In adult patients with locally advanced or metastatic *NTRK* gene fusion-positive cancers, larotrectinib may produce substantial life expectancy and QALY gains compared to repotrectinib.
- Additional data with more mature data and larger sample size and additional real-world studies would further inform our results.

Limitations

- We used an unadjusted naïve direct comparison in the absence of direct comparative data.
- Due to the lack of publicly available data on repotrectinib in adult patients treated for *NTRK* gene fusion-positive cancers, OS was imputed.
- Utility values were approximated using a prevalence-weighted average of the most frequently observed tumor types across both clinical trials, as publicly available data were not available for all assessed tumor types.
- Tumor response criteria differed between treatments, with larotrectinib using both RECIST and RANO due to inclusion of CNS tumors, whereas repotrectinib used RECIST alone.

References

1. Cocco E, et al. Nat Rev Clin Oncol 2018;15:731-747.
2. Amatu A, et al. Ann Oncol 2019;30:vii5-viii15.
3. VITRAKVI [package insert]. Bayer AG. 2018.
4. Drilon AE, et al. ASCO Annual 2022;Abstract 3100.
5. Drilon AE, et al. ASCO Annual 2024:Abstract 8570.
6. Solomon B, et al. ESMO 2023:Poster 1372P.
7. US Life Tables, 2021. *NVSR* 2023;72(12):1-64.
8. Bayer US LLC. Data on File. 2024.
9. Besse B, et al. ENA Symposium 2022;Abstract 209.
10. Roth JA, et al. J Manag Care Spec Pharm 2020:981-986.
11. Nafees B, et al. Health Qual Life Outcomes 2008;84.
12. Guest JF, et al. Sarcoma 2013;863056.
13. Fordham BA, et al. Patient Prefer Adherence 2015;9:1561-72.
14. Farkkila N, et al. Colorectal Dis. 2013;e215-22.

QR Code Here

Disclosures

This study was sponsored by Bayer AG. The sponsor was involved in the study design and writing of the report.

ISPOR 2025, Montreal, QC, Canada: May 13-16, 2025