N-terminal pro-B-type natriuretic peptide concentrations, testing and associations with outcomes
An analysis of the Swedish Heart Failure Registry and the Stockholm CREAtinine Measurements Cohorts

Giulia Ferrannini[1], Lina Benson[1], Dominik Lautsch[2], Ulf Dahlström[3], Lars H Lund[1,4], Gianluigi Savarese[1,4*], Juan Jesus Carrero[5*]

[1] Division of Cardiology, Department of Medicine, Karolinska Institutet, Stockholm, Sweden; [2] Merck & Co., Inc., Rahway, NJ, USA; [3] Department of Cardiology and Department of Medical and Health Sciences, Linköping University, Linköping, Sweden; [4] Heart and Vascular Theme, Karolinska University Hospital, Stockholm, Sweden; [5] Department of Medical Epidemiology and Biostatistics, Karolinska Institutet, Stockholm, Sweden. * These authors equally contributed

Background

The measurement of N-terminal pro-B-type natriuretic peptide (NT-proBNP) is recommended by international Heart failure (HF) guidelines for diagnosis, classification, assessing disease severity, and prognostic stratification.
Data on the implementation of NT-proBNP testing in clinical practice is sparse, especially as regards patients who suffered a worsening heart failure event (WHFE).

Aims

To assess, in a real-world HF population:

1) the prevalence of and the patient characteristics associated with the likelihood of NT-proBNP testing in patients with HF with vs. without a worsening HF event (WHFE vs. NWHFE);
2) the distribution and patterns of changes in NT-proBNP levels in patients with vs. without a WHFE;
3) their associations with outcomes.

Materials and Methods

Data from the Swedish Heart failure Registry (SwedeHF) and the Stockholm CREAtinine Measurements project, linked with The Regional Healthcare Utilization Database (VAL), The National Board of Health and the longitudinal integrated database for health insurance and labour market studies (LISA).

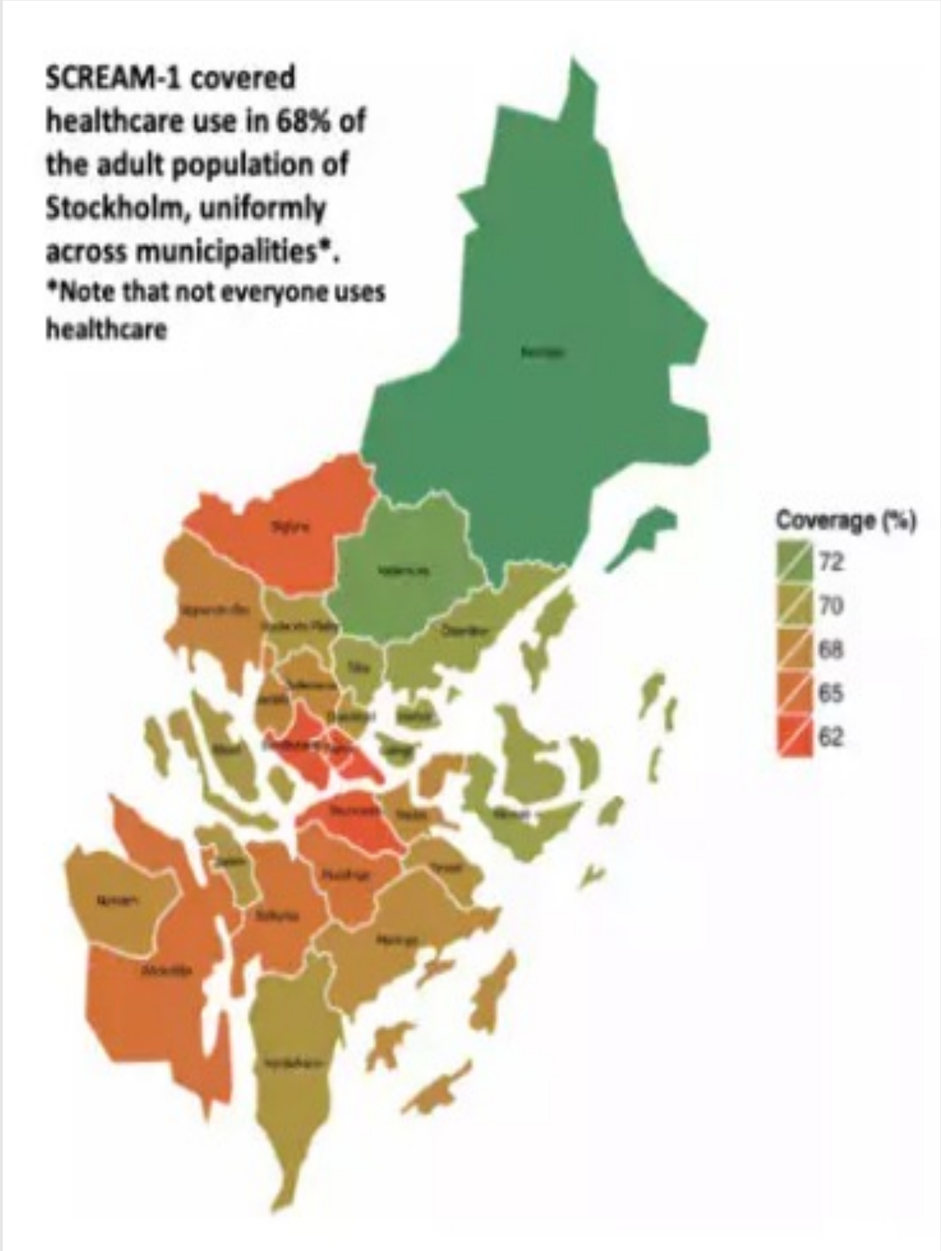

SCREAM-1 covered healthcare use in 68% of the adult population of Stockholm, uniformly across municipalities*.
*Note that not everyone uses healthcare
Coverage (%)
72
70
68
65
62

Changes in NT-proBNP concentrations:

- **Stable low**: <3000 ng/L at index and follow-up
- **Increased**: <3000 ng/L at the index and ≥3000 ng/L at the follow-up
- **Decreased**: ≥3000 ng/L at the baseline and <3000 ng/L at the follow-up
- **Stable high**: ≥3000 ng/L at the baseline and follow-up

Inclusion:
January 2011 - December 2018

Index date: discharge from the hospital for HF

WHFE:
Registration as in-patient at the index date and HF verified as being the cause for hospitalization in VAL

Outcomes:

- Number of NTproBNP testing over time
- Associations of clinical characteristics with testing
- Distribution of NTproBNP levels
- Associations between changes in NTproBNP and risk of all-cause and cardiovascular death

Funding

This study received support from Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA through a grant to Karolinska Institutet.

Giulia Ferrannini MD - giulia.ferrannini@ki.se
Cardiology Unit - Department of Medicine
Karolinska Institutet, Stockholm, Sweden

Results

Figure 1. NT-proBNP testing overtime in the overall population and in HFrEF (A) and time from the index date to the first NT-proBNP testing in the overall population and in HFrEF (B).

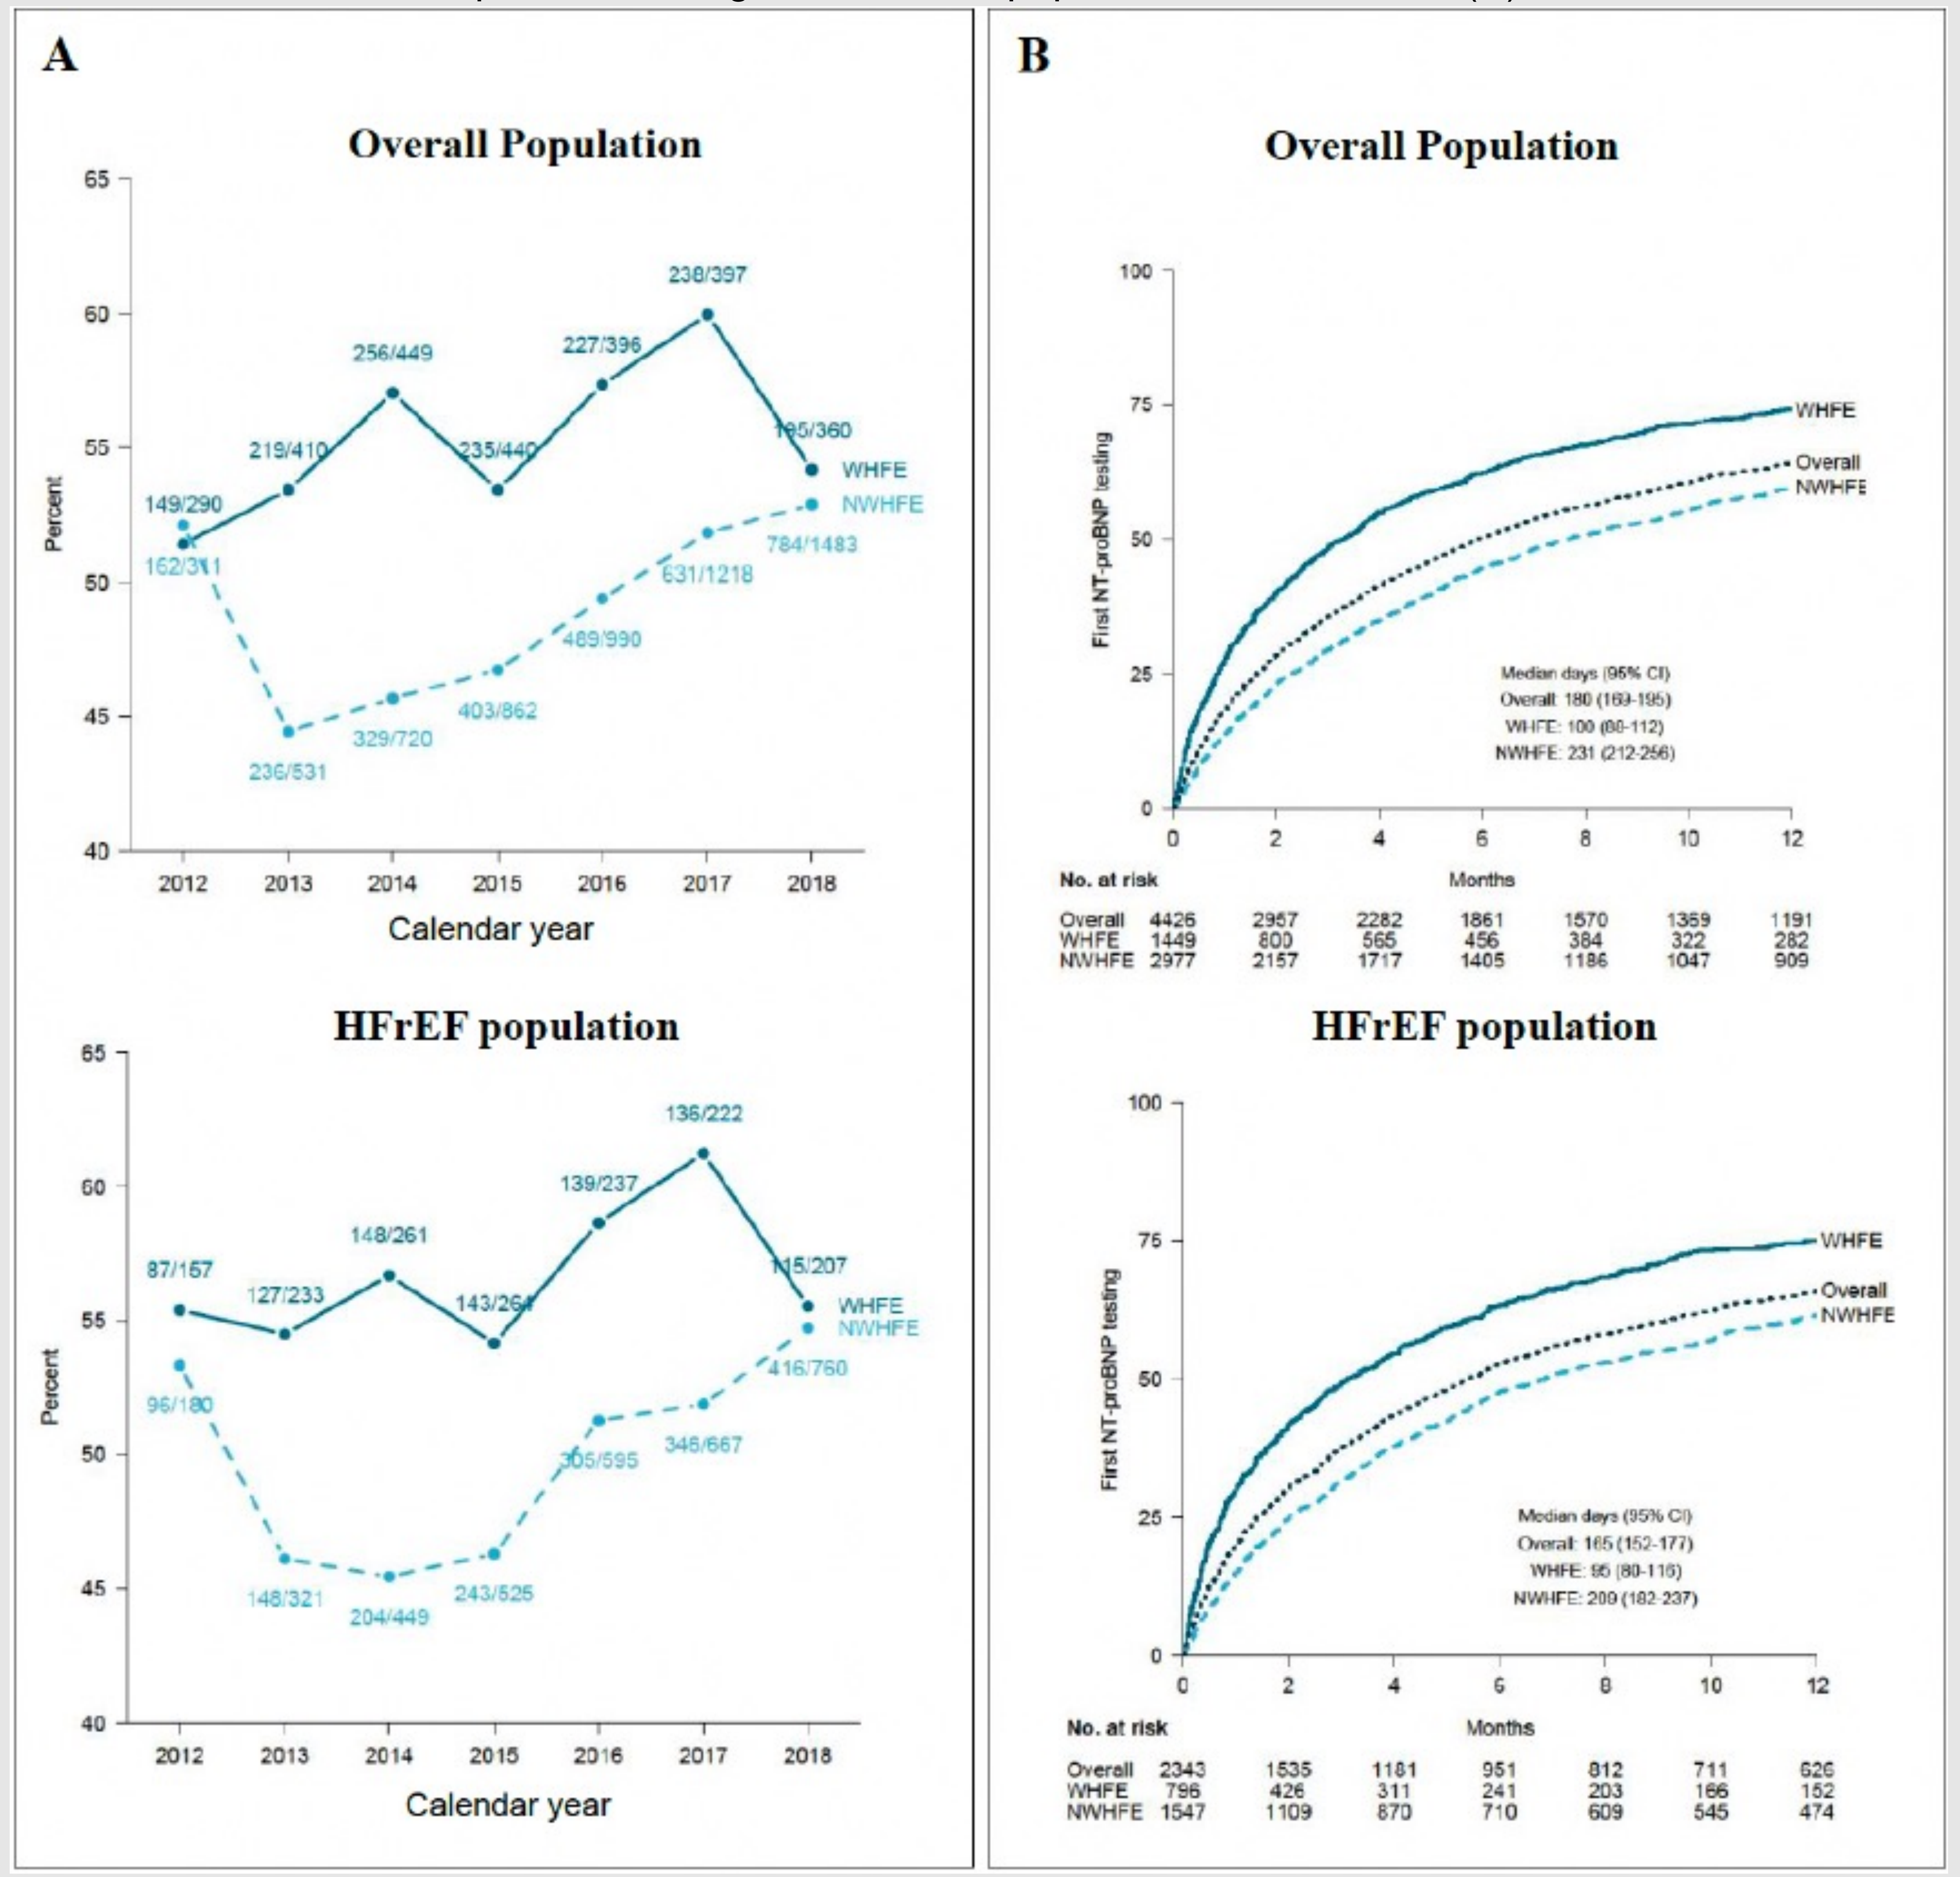

A
Overall Population
149/290
162/311
219/410
256/449
235/440
227/396
238/397
195/360
236/531
329/720
403/862
489/990
631/1218
784/1483
WHFE
NWHFE
Percent
Calendar year
HFrEF population
87/157
127/233
148/261
143/264
139/237
136/222
115/207
96/180
146/321
204/449
243/525
305/595
346/667
416/760
B
Overall Population
First NT-proBNP testing
Median days (95% CI)
Overall: 180 (169-195)
WHFE: 100 (88-112)
NWHFE: 231 (212-256)
Months
No. at risk
Overall 4426 2957 2282 1861 1570 1369 1191
WHFE 1449 800 565 456 384 322 282
NWHFE 2977 2157 1717 1405 1186 1047 909
HFrEF population
Median days (95% CI)
Overall: 165 (152-177)
WHFE: 95 (80-116)
NWHFE: 209 (182-237)
No. at risk
Overall 2343 1535 1181 951 812 711 626
WHFE 796 426 311 241 203 166 152
NWHFE 1547 1109 870 710 609 545 474

Figure 2. Multiadjusted associations between patient characteristics and NT-proBNP testing.

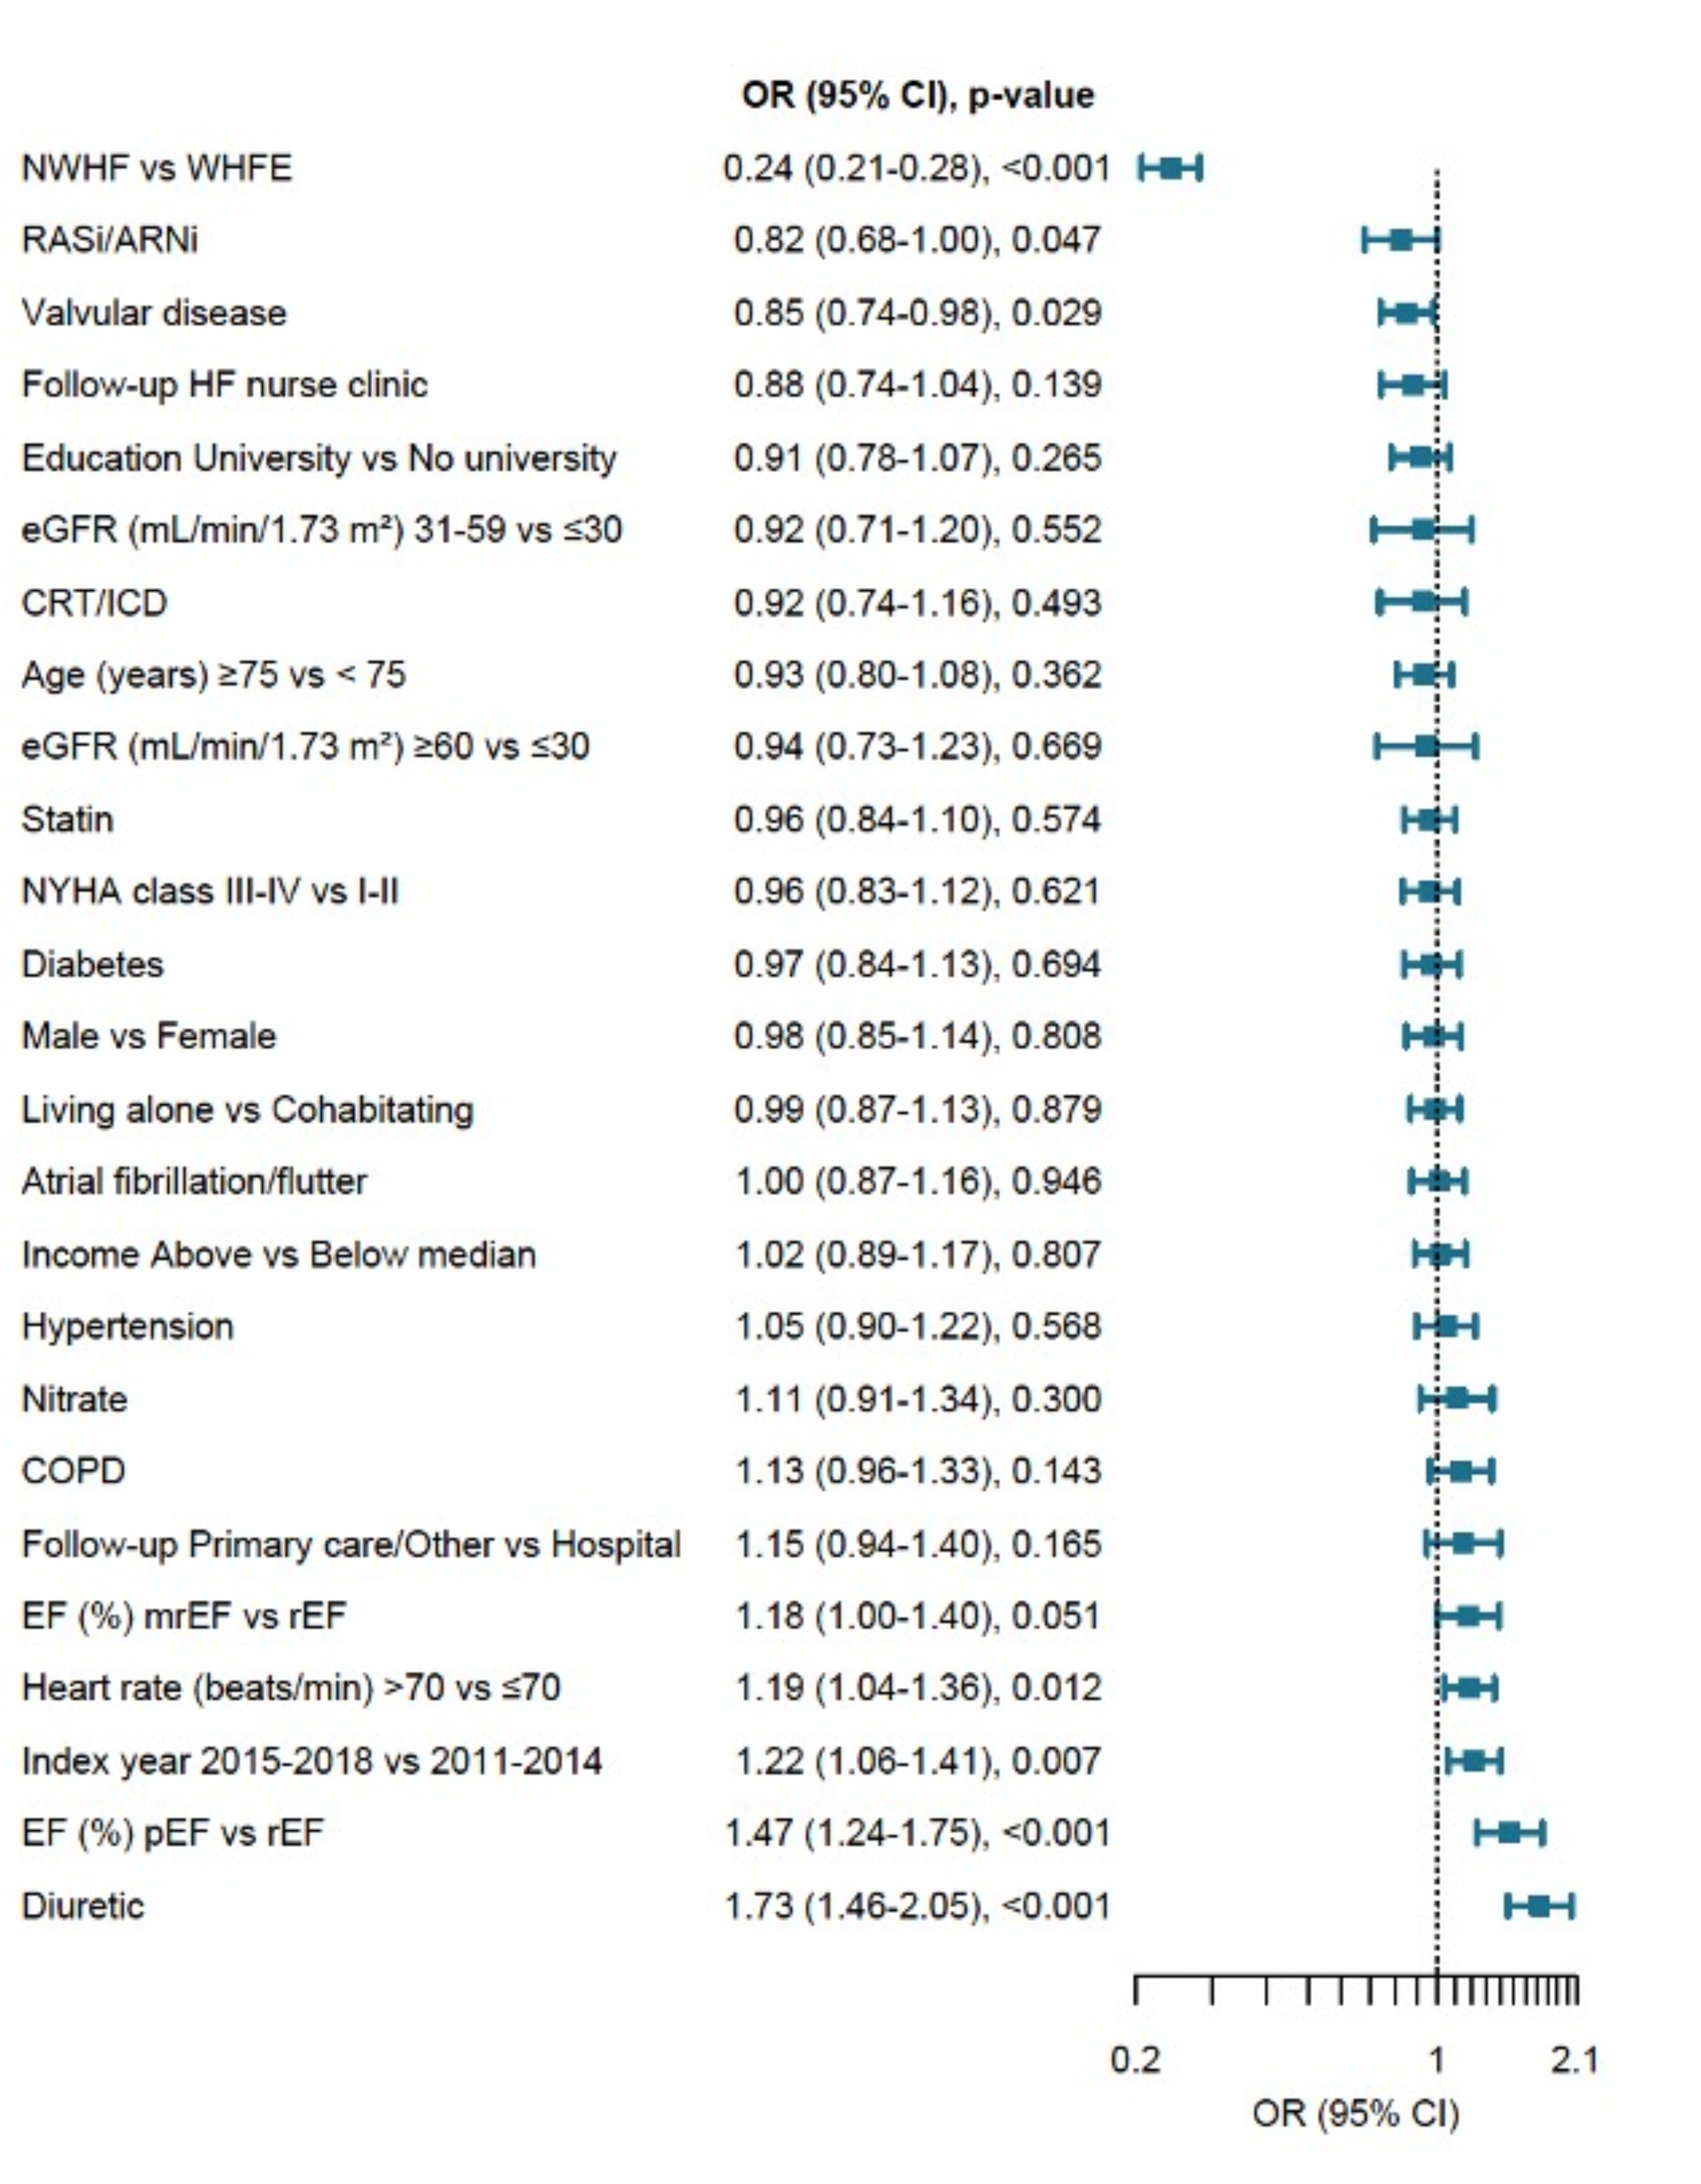

OR (95% CI), p-value
NWHF vs WHFE 0.24 (0.21-0.28), <0.001
RASi/ARNi 0.82 (0.68-1.00), 0.047
Valvular disease 0.85 (0.74-0.98), 0.029
Follow-up HF nurse clinic 0.88 (0.74-1.04), 0.139
Education University vs No university 0.91 (0.78-1.07), 0.265
eGFR (mL/min/1.73 m²) 31-59 vs ≤30 0.92 (0.71-1.20), 0.552
CRT/ICD 0.92 (0.74-1.16), 0.493
Age (years) ≥75 vs < 75 0.93 (0.80-1.08), 0.362
eGFR (mL/min/1.73 m²) ≥60 vs ≤30 0.94 (0.73-1.23), 0.669
Statin 0.96 (0.84-1.10), 0.574
NYHA class III-IV vs I-II 0.96 (0.83-1.12), 0.621
Diabetes 0.97 (0.84-1.13), 0.694
Male vs Female 0.98 (0.85-1.14), 0.808
Living alone vs Cohabitating 0.99 (0.87-1.13), 0.879
Atrial fibrillation/flutter 1.00 (0.87-1.16), 0.946
Income Above vs Below median 1.02 (0.89-1.17), 0.807
Hypertension 1.05 (0.90-1.22), 0.568
Nitrate 1.11 (0.91-1.34), 0.300
COPD 1.13 (0.96-1.33), 0.143
Follow-up Primary care/Other vs Hospital 1.15 (0.94-1.40), 0.165
EF (%) mrEF vs rEF 1.18 (1.00-1.40), 0.051
Heart rate (beats/min) >70 vs ≤70 1.19 (1.04-1.36), 0.012
Index year 2015-2018 vs 2011-2014 1.22 (1.06-1.41), 0.007
EF (%) pEF vs rEF 1.47 (1.24-1.75), <0.001
Diuretic 1.73 (1.46-2.05), <0.001
OR (95% CI)

Figure 3. Concentrations of NT-proBNP at the index date, in the overall population and according to WHFE.

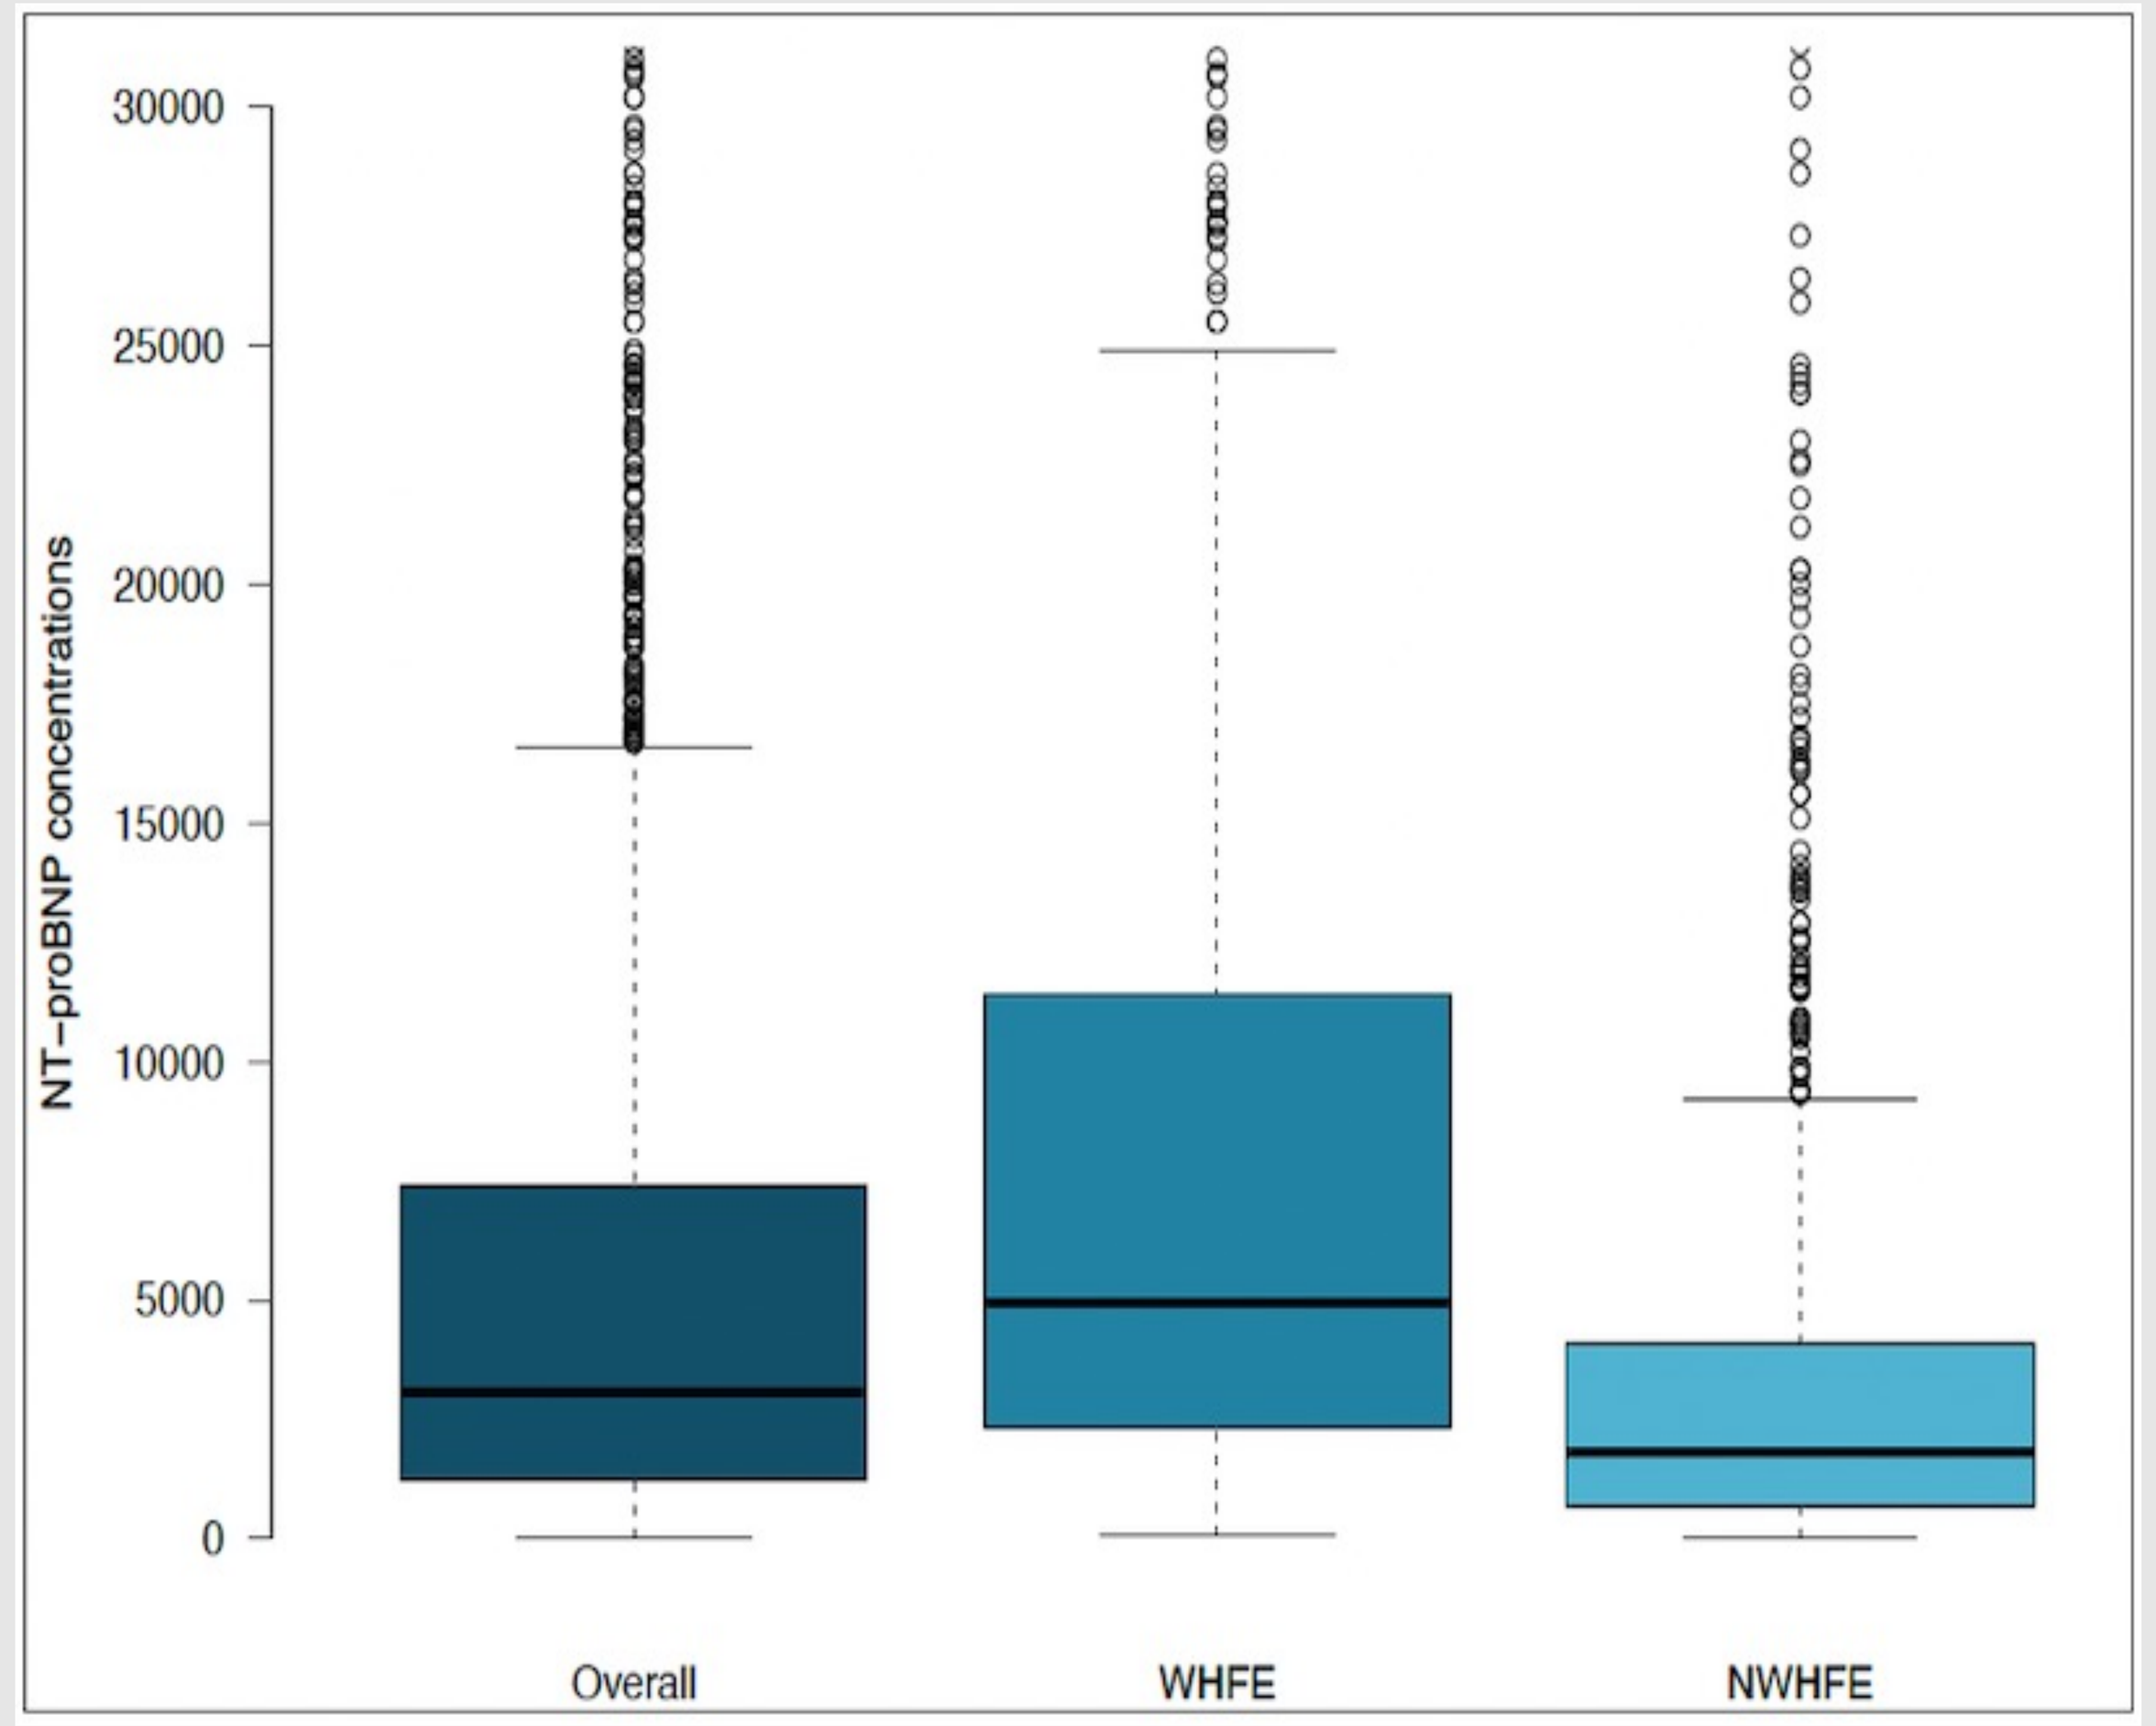

NT-proBNP concentrations
Overall
WHFE
NWHFE

NTproBNP ≤1000 ng/L: 8% WHFE, 33% NWHFE
NT-proBNP >8000 ng/L: 35% WHFE, 12% NWHFE
NT-proBNP >14000 ng/L: 20% WHFE, 6% NWHFE

In HFrEF:
Median NT-proBNP levels 4150 ng/L
(IQR 1760, 10900)

Figure 4. Kaplan-Meier curves for the associations between NTproBNP changes and outcomes.

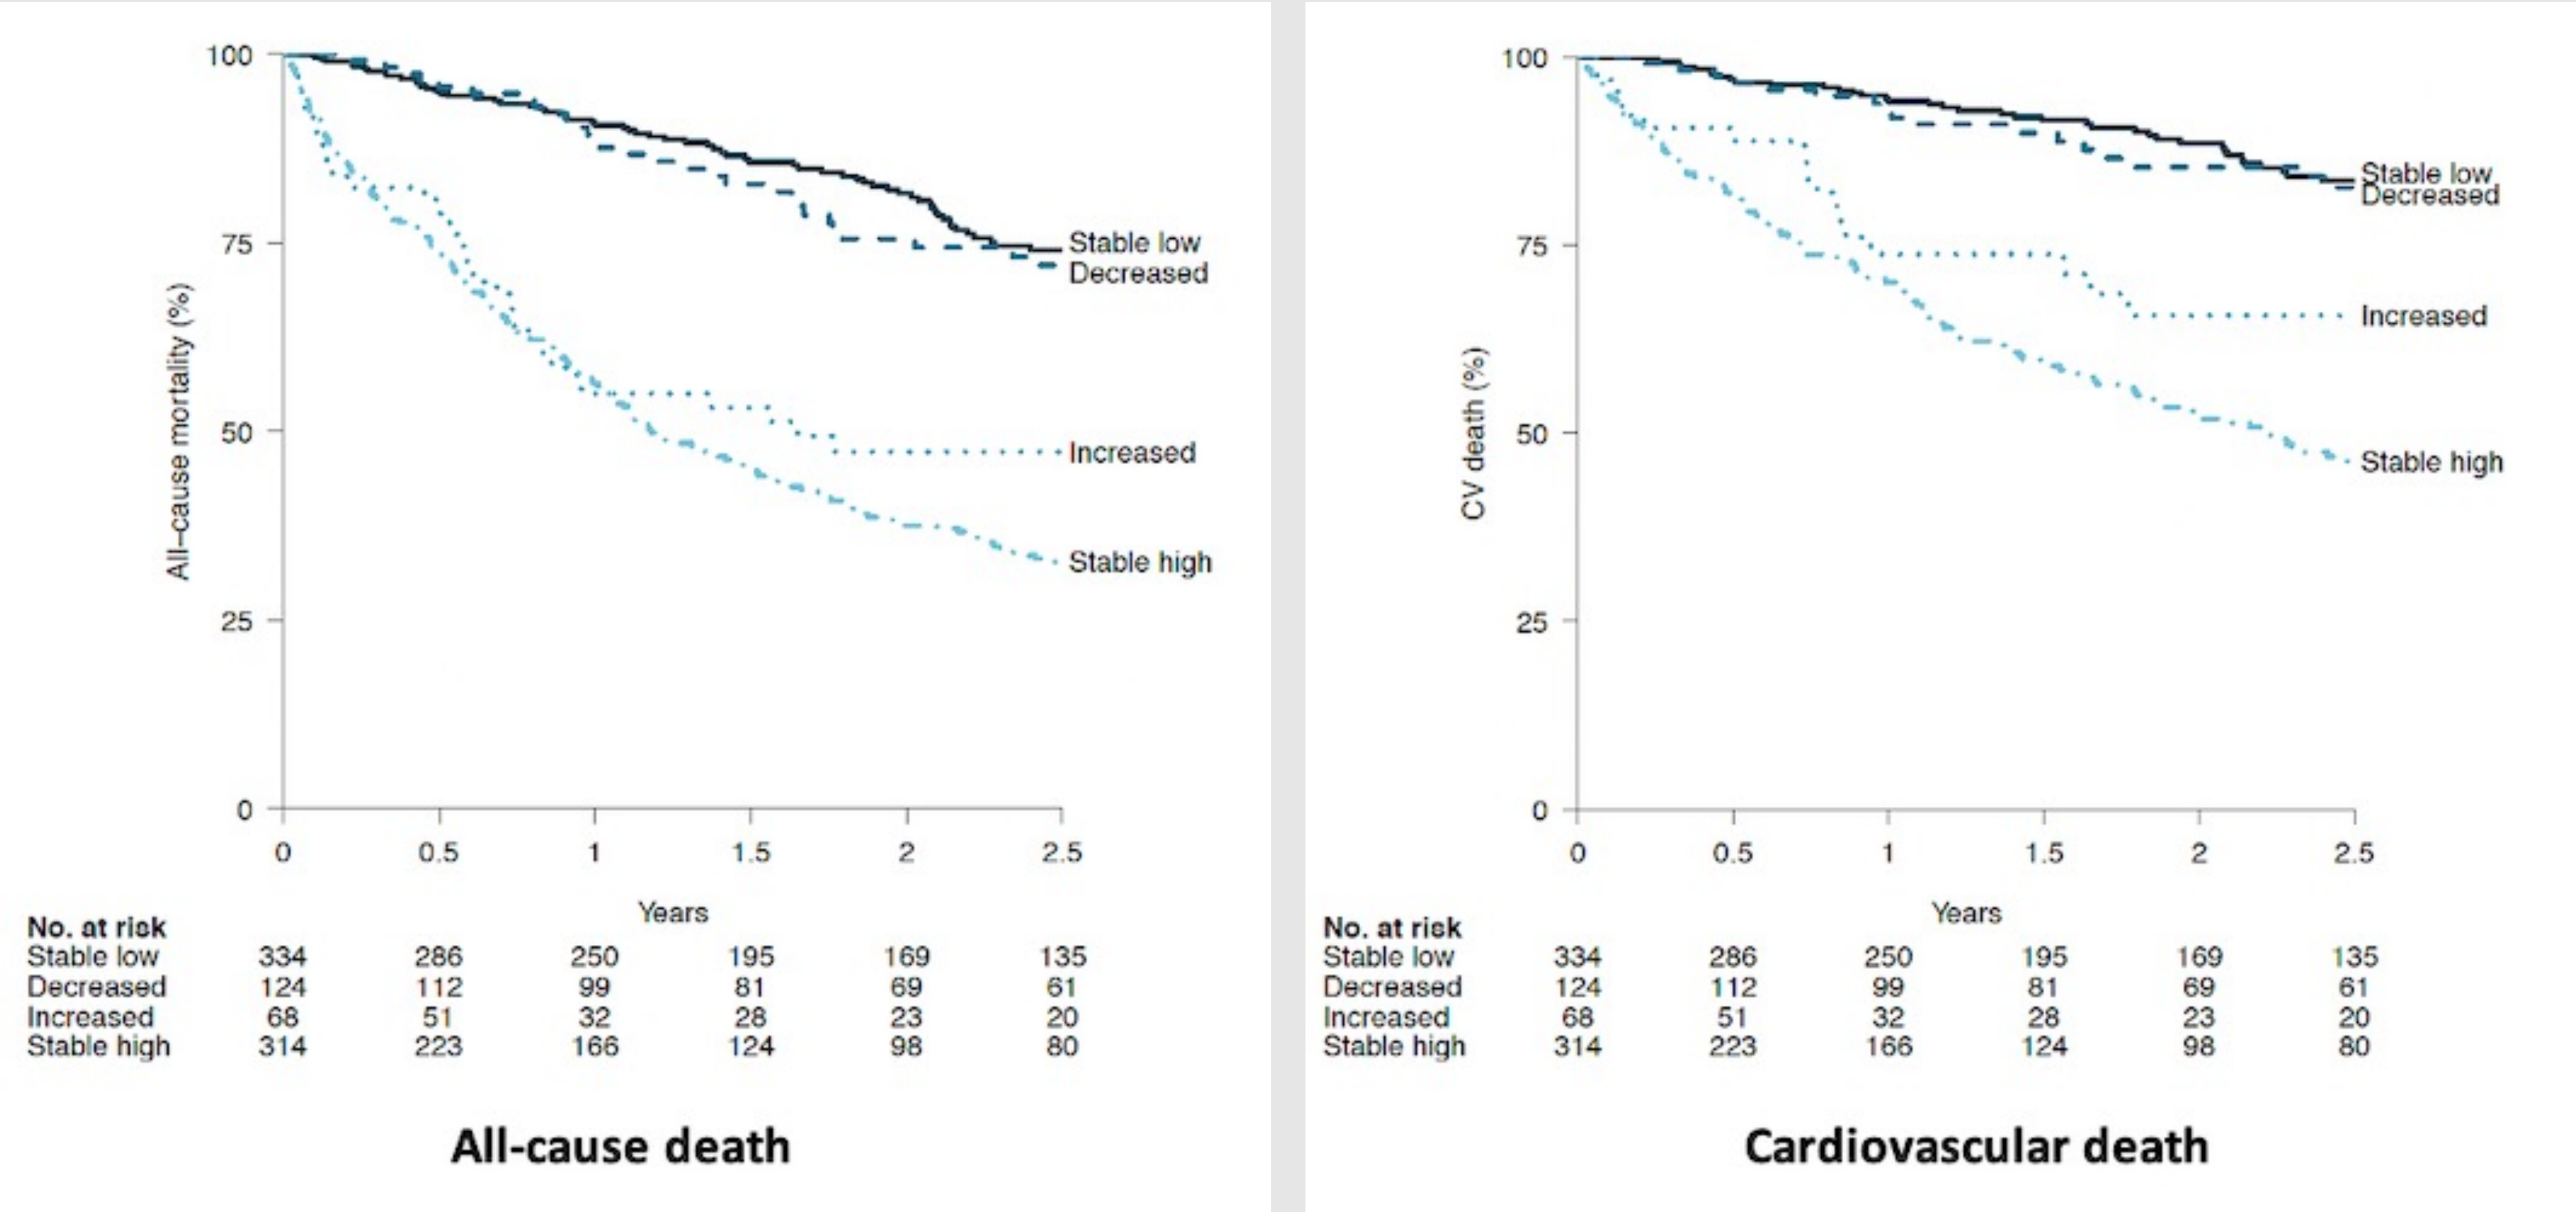

All-cause mortality (%)
Stable low
Decreased
Increased
Stable high
Years
No. at risk
Stable low 334 286 250 195 169 135
Decreased 124 112 99 81 69 61
Increased 68 51 32 28 23 20
Stable high 314 223 166 124 98 80
All-cause death
CV death (%)
Cardiovascular death

Conclusions

- NT-proBNP testing has increased over time in the Stockholm region, up to 55% in 2018; still it is underperformed
- Testing was performed twice as often, with time to testing less than a half, in patients with a WHFE compared to those without
- Independent predictors of testing were surrogates of a more severe HF or need for differential diagnosis
- NT-proBNP levels were high, underlining the need for guideline-directed medical therapy optimisation in patients with HF
- Increasing and stable high levels of NT-proBNP were associated with higher mortality

Overall, our data highlight the benefits of carrying further implementation of NT-proBNP testing in clinical practice