Risk of all-cause mortality and HF hospitalizations in patients treated with vericiguat in real-world in Germany

Fabian Kerwagen[1], Christoph Ohlmeier[2], Gerald S Laux[3], Philip Wenzel[3], Marco Alibone[4], Dennis Häckl[5], Martin Schulz[6], Rolf Wachter[7], Michael Böhm[8], Stefan Störk[1]

1 Comprehensive Heart Failure Center & Dept. of Medicine I, Univ. Hospital Würzburg, Germany **2** Bayer AG, Berlin, Germany **3** Dept. of Cardiology, Univ. Medical Center Mainz, Germany **4** InGef, Berlin, Germany **5** WIG2, Leipzig, Germany **6** DAPI & Institute of Pharmacy, Freie Univ. Berlin, Germany **7** Dept. of Cardiology, Univ. Hospital Leipzig, Germany **8** Dept. of Internal Medicine III, Saarland Univ. Hospital, Homburg/Saar, Germany

Background & Purpose

- Vericiguat reduced clinical endpoints in patients with worsening heart failure (HF) in randomized trials, but evidence from routine care remains limited.
- To investigate the risk of all-cause mortality (ACM), heart failure hospitalizations (HFH), and hypotension in patients treated with vericiguat in routine care, in Germany.

Methods

- This retrospective cohort study utilized the InGef Health Research Database, encompassing anonymized claims data such as prescriptions and ICD-10-documented diagnoses from about 14% of Germany's 73 million statutory health insurance beneficiaries.
- All patients with a new prescription of vericiguat from September 2021 to September 2023 were included. We calculated incidence rates and Kaplan-Meier curves for ACM and HFH.
- The factors predicting outcomes were identified using multivariable Cox proportional hazard regression. Occurrence of hypotension and syncope was assessed in the three months following treatment initiation.

Results

- Vericiguat was initiated in 491 patients (mean age 70 ± 13 years; 22% women). In 63% of these patients, a worsening HF (wHF) event had occurred within the six months prior to initiation of vericiguat. Of note, 73% of these events had occurred 1–29 days prior to initiation. Compared to the pivotal VICTORIA trial, comorbidities were more frequent: renal impairment (65% vs 25%), atrial fibrillation (61% vs 44%), diabetes mellitus (60% vs 49%), and NYHA class III/IV (92% vs 41%). Prior to initiation, hypotension and hyperkalemia had been documented in 17% and 21%, respectively.
- During a median follow-up period of 207 days (IQR 108–374 days), mortality was 5.9%, translating into a death rate of 8.7 per 100 patient-years (95%CI 5.8–12.5), with a stable risk over time. HFH occurred in 33.0%, with a rate of 64.7 per 100 patient-years (55.1–75.5). A recent wHF event prior to vericiguat initiation was identified as the strongest predictor for HFH (OR 2.8 [1.8-4.1]). Incident hypotension or syncope after vericiguat initiation was recorded in 8.0% within three months, whereas syncope alone was observed in 2.1%.

Conclusion

- In routine care, in Germany, vericiguat typically was initiated shortly after a wHF event. Compared to the VICTORIA landmark trial, patients had a notably more pronounced cardiorenal comorbidity burden indicating the use of vericiguat in a high-risk population.

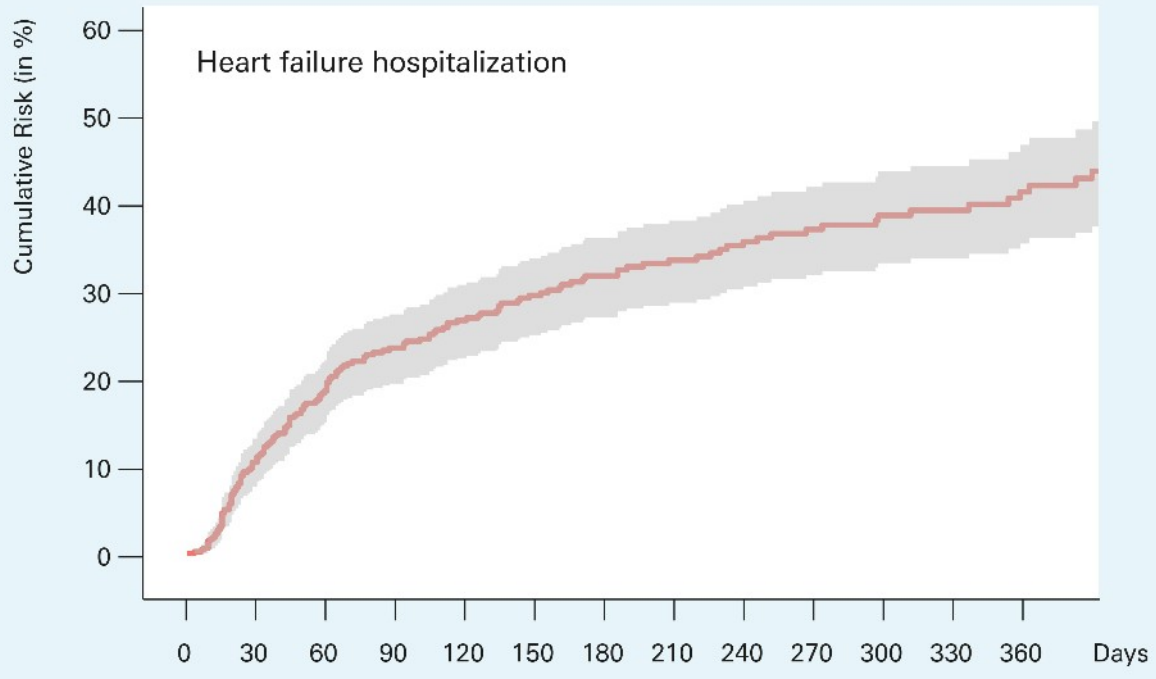
Cumulative Risk (in %)
60
50
40
30
20
10
0
Heart failure hospitalization
0 30 60 90 120 150 180 210 240 270 300 330 360 Days

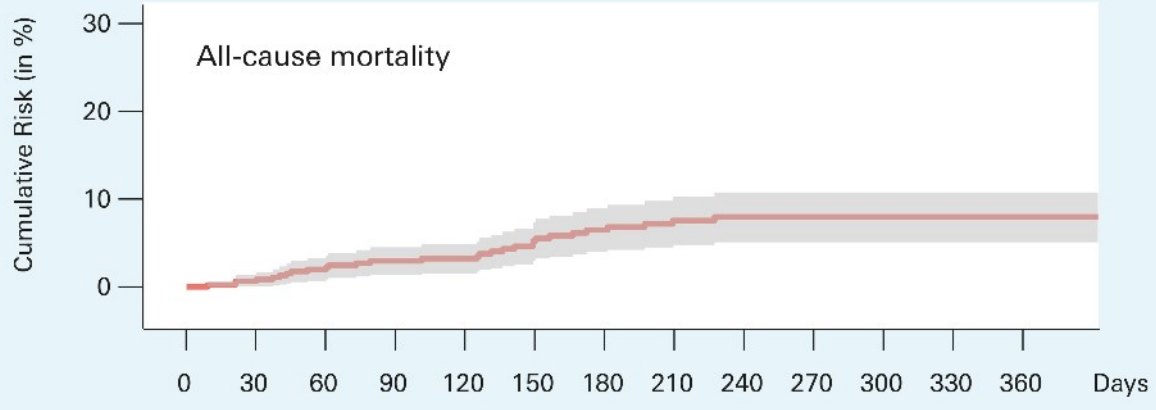
Cumulative Risk (in %)
30
20
10
0
All-cause mortality
0 30 60 90 120 150 180 210 240 270 300 330 360 Days

Figure: Kaplan-Meier-curves for heart failure hospitalization and all-cause mortality

Contact: Fabian Kerwagen
E-Mail: Kerwagen_F@ukw.de
Funding: This study is supported by Bayer AG