Geographic Healthcare Disparities and Diagnostic Trends Among Patients With Transthyretin Amyloid Cardiomyopathy

Joshua Mitchell,[1] Hanh Dung Dao,[2] Elizabeth Nagelhout,[2] Neil Lamarre,[2] Margarita Udall[3]

[1]Washington University School of Medicine, St. Louis, MO, USA; [2]Genesis Research Group, Hoboken, NJ, USA; [3]BridgeBio Pharma, Inc., San Francisco, CA, USA

OBJECTIVE

- **To assess the diagnostic trends and the geographic distribution of amyloid centers and cardiology services, with the goal of elucidating diagnostic disparities of ATTR-CM in the US**

BACKGROUND

- ATTR-CM is a rare, underdiagnosed, life-threatening systemic disease that presents as a restrictive cardiomyopathy with cardiac and/or neurologic symptoms[1]
 - ATTR-CM includes both wild type (ATTRwt-CM) and pathogenic variant (ATTRv-CM) forms of the disease[1-3]
- The true incidence and prevalence of ATTR-CM is unknown; ATTR-CM has historically been considered a rare disease, though identification has become more common with recent advances in noninvasive cardiac imaging, genetic testing, and diagnostics[2]
- Timely and accurate diagnosis of ATTR-CM remains a challenge due to disease heterogeneity, limited awareness, and a clinical presentation that overlaps with more common conditions[2]
- Access to cardiology services and specialized amyloid centers may play a role in the diagnosis of ATTR-CM; however, the relationship between proximity to these services and ATTR-CM diagnosis is not well understood

METHODS

- This was a cross-sectional geospatial analysis of a retrospective cohort of patients with ATTR-CM from the Komodo Healthcare Map® from July 1, 2016, to June 30, 2024, in relation to amyloid centers and cardiology providers in the US (**Figure 1**)

FIGURE 1. Data Source and Inclusion/Exclusion Criteria

Data source

Cross-sectional geospatial analysis of a retrospective cohort of patients with ATTR-CM from the Komodo Healthcare Map® from July 1, 2016, to June 30, 2024, in relation to amyloid centers and cardiology providers in the US

ATTR-CM diagnosis

The type of ATTR-CM was determined based on the ATTR-CM diagnosis codes on claims occurring during the identification period

- To be considered ATTRv-CM, a patient was required to have ≥2 ATTRv-CM codes (on separate days) during the identification period
- If a patient only had ATTRwt-CM codes and/or <2 ATTRv-CM codes during the identification period, the patient was considered to be ATTRwt-CM
- If a patient only had a tafamidis claim and no relevant ATTR-CM codes during the identification period, then the patient was labeled as "unknown"
- If a patient had 1 ATTRv-CM code and no ATTRwt-CM codes (with tafamidis claim), the patient was labeled as "unknown"

Inclusion criteria

- Two or more claims with an amyloidosis diagnosis code (E85.0, E85.1, E85.2, E85.4, or E85.82) occurring on separate days AND ≥2 claims for a cardiac-related ICD-10-CM code; OR ≥1 claim for tafamidis based on National Drug Codes
- ≥50 years of age on the index date (earliest date of either ATTR-CM diagnosis or tafamidis claim)
- ≥6 months (182 days) of continuous enrollment in a closed claims source before the index date

Exclusion criteria

- >1 ICD-10-CM diagnosis code of light-chain amyloidosis (E85.81) on different dates during the study period
- >1 claim (on separate days) for multiple myeloma at any time during the study period
- Received hematopoietic stem cell transplant at any time during the study period
- Resided in territories not in the 50 US states

- Outcomes included:
 - Baseline demographics and clinical characteristics among patients with ATTR-CM
 - Geospatial distribution of amyloid centers in the US
 - Amyloid centers in the US were classified by the International Society of Amyloidosis, Amyloidosis Foundation, and/or Amyloid Research Consortium[4-6]
 - Map of cardiologist deserts in the US
 - Cardiologists were identified using the specialization variables in the Komodo Healthcare Map[7]
 - Patient location was calculated using proxy variables as follows: the Komodo Healthcare Map is limited to a 3-digit zip code for patients; as a proxy for their location, a 5-digit zip code was imputed from either the patient's outpatient primary care provider claims or pharmacy (mail-order pharmacies excluded)
 - Distance was calculated from patient location proxy to the nearest amyloid center and cardiologist zip code
 - Diagnosed prevalence of ATTR-CM overall and over time, stratified by race
- Statistical analyses were descriptive in nature; R statistical software, R version 4.2.1, was used for analytics[8]

CONCLUSIONS

- **The diagnosed prevalence of ATTR-CM increased approximately 7-fold from 2017 to 2023 in the setting of available treatment, improved awareness, and less invasive diagnostics**
- **There remain geographic disparities and racial differences in ATTR-CM diagnosed prevalence**
- **As the majority of patients with amyloidosis were located near an amyloid center, there remain large areas of the country with significantly reduced diagnostic rates and poor access to care**
- **Strategies to address these regional disparities need to be identified, as there likely remains a substantial population of undiagnosed patients outside of the immediate radius of amyloid centers**

RESULTS

DEMOGRAPHICS

- A total of 14,980 patients were identified who met inclusion/exclusion criteria (**Table 1**)
- Mean age was 74.8 years (SD, 9.7), 62.3% were male, 54.1% were White, and 28.8% were Black/African American
- Regional representation was highest from the Northeast (n=5893 [39.3%]), followed by the Midwest (n=3571 [23.8%])

TABLE 1. Demographics by Region in the US

Characteristic	Overall (N=14,980)	Subgroup: geographic region			
		Northeast (n=5893)	Midwest (n=3571)	West (n=2023)	South (n=3493)
Age at index, years					
Mean (SD)	74.8 (9.7)	76.6 (8.9)	74.5 (9.7)	73.6 (10.3)	72.7 (9.9)
Median (Q1, Q3)	77.0 (68.0, 83.0)	79.0 (72.0, 84.0)	77.0 (68.0, 83.0)	76.0 (66.0, 82.0)	74.0 (65.0, 81.0)
Min, Max	50.0, 89.0	50.0, 89.0	50.0, 89.0	50.0, 89.0	50.0, 89.0
Sex, n (%)					
Male	9330 (62.3)	3691 (62.6)	2286 (64.0)	1278 (63.2)	2075 (59.4)
Female	5207 (34.8)	2003 (34.0)	1208 (33.8)	711 (35.1)	1285 (36.8)
Missing	443 (3.0)	199 (3.4)	77 (2.2)	34 (1.7)	133 (3.8)
Race/ethnicity, n (%)					
White	8102 (54.1)	3523 (59.8)	2009 (56.3)	1131 (55.9)	1439 (41.2)
Black/African American	4313 (28.8)	1468 (24.9)	1188 (33.3)	246 (12.2)	1411 (40.4)
Other	1915 (12.8)	694 (11.8)	206 (5.8)	562 (27.8)	453 (13.0)
Missing	650 (4.3)	208 (3.5)	168 (4.7)	84 (4.2)	190 (5.4)
ATTR-CM type, n (%)					
Variant	1492 (10.0)	568 (9.6)	312 (8.7)	228 (11.3)	384 (11.0)
Wild type	13,266 (88.6)	5239 (88.9)	3216 (90.1)	1770 (87.5)	3041 (87.1)
Unknown	222 (1.5)	86 (1.5)	43 (1.2)	25 (1.2)	68 (2.0)
Distance to amyloid center, miles					
Mean (SD)	48.5 (59.5)	32.3 (39.3)	44.6 (52.2)	57.2 (77.2)	75.0 (71.5)
Median (Q1, Q3)	22.7 (7.9, 71.2)	15.9 (5.5, 46.0)	25.2 (8.4, 71.0)	21.6 (8.7, 62.2)	51.8 (12.9, 122.0)
Min, Max	0.0, 542.3	0.0, 356.0	0.0, 542.3	0.0, 399.1	0.0, 359.8
Distance to cardiology provider, miles					
Mean (SD)	4.3 (10.4)	2.8 (6.2)	7.1 (15.0)	4.7 (13.5)	3.7 (7.0)
Median (Q1, Q3)	0.0 (0.0, 4.1)	0.0 (0.0, 3.2)	1.3 (0.0, 6.2)	0.0 (0.0, 3.4)	0.0 (0.0, 4.4)
Min, Max	0.0, 159.1	0.0, 159.1	0.0, 143.4	0.0, 118.0	0.0, 52.3

GEOSPATIAL DISTRIBUTION OF PATIENTS RELATIVE TO TREATMENT PROVIDERS AND CENTERS

- The mean distance of patients to a cardiology provider was 4.3 miles (SD, 10.4) and to an amyloid center was 48.5 miles (SD, 59.5) (**Table 1**)
- The distance to an amyloid center was shortest in the Northeast (mean, 32.3 miles; SD, 39.3), where diagnosed prevalence was highest, and longest in the South (mean, 75.0 miles; SD, 71.5), where prevalence was next to lowest
- There were 80 amyloid centers identified (Northeast, 17; Midwest, 17; South, 26; West, 20); the mean number of amyloid centers per capita was 0.24 per 1,000,000 (Northeast, 0.30; Midwest, 0.25; West, 0.26; South, 0.21)
- The number of amyloid centers by state is shown in **Figure 2**

ATTR-CM PREVALENCE

- Overall period prevalence (cases/100,000) during the time of the study (2016-2024) was 4.54 and highest among patients who were Black/African American (10.75), followed by patients who were White (4.16) (**Table 2**)
- Prevalence (cases/100,000) was highest in the Northeast (10.28) and lowest in the West (2.61)

FIGURE 2. Number of Amyloid Centers by State

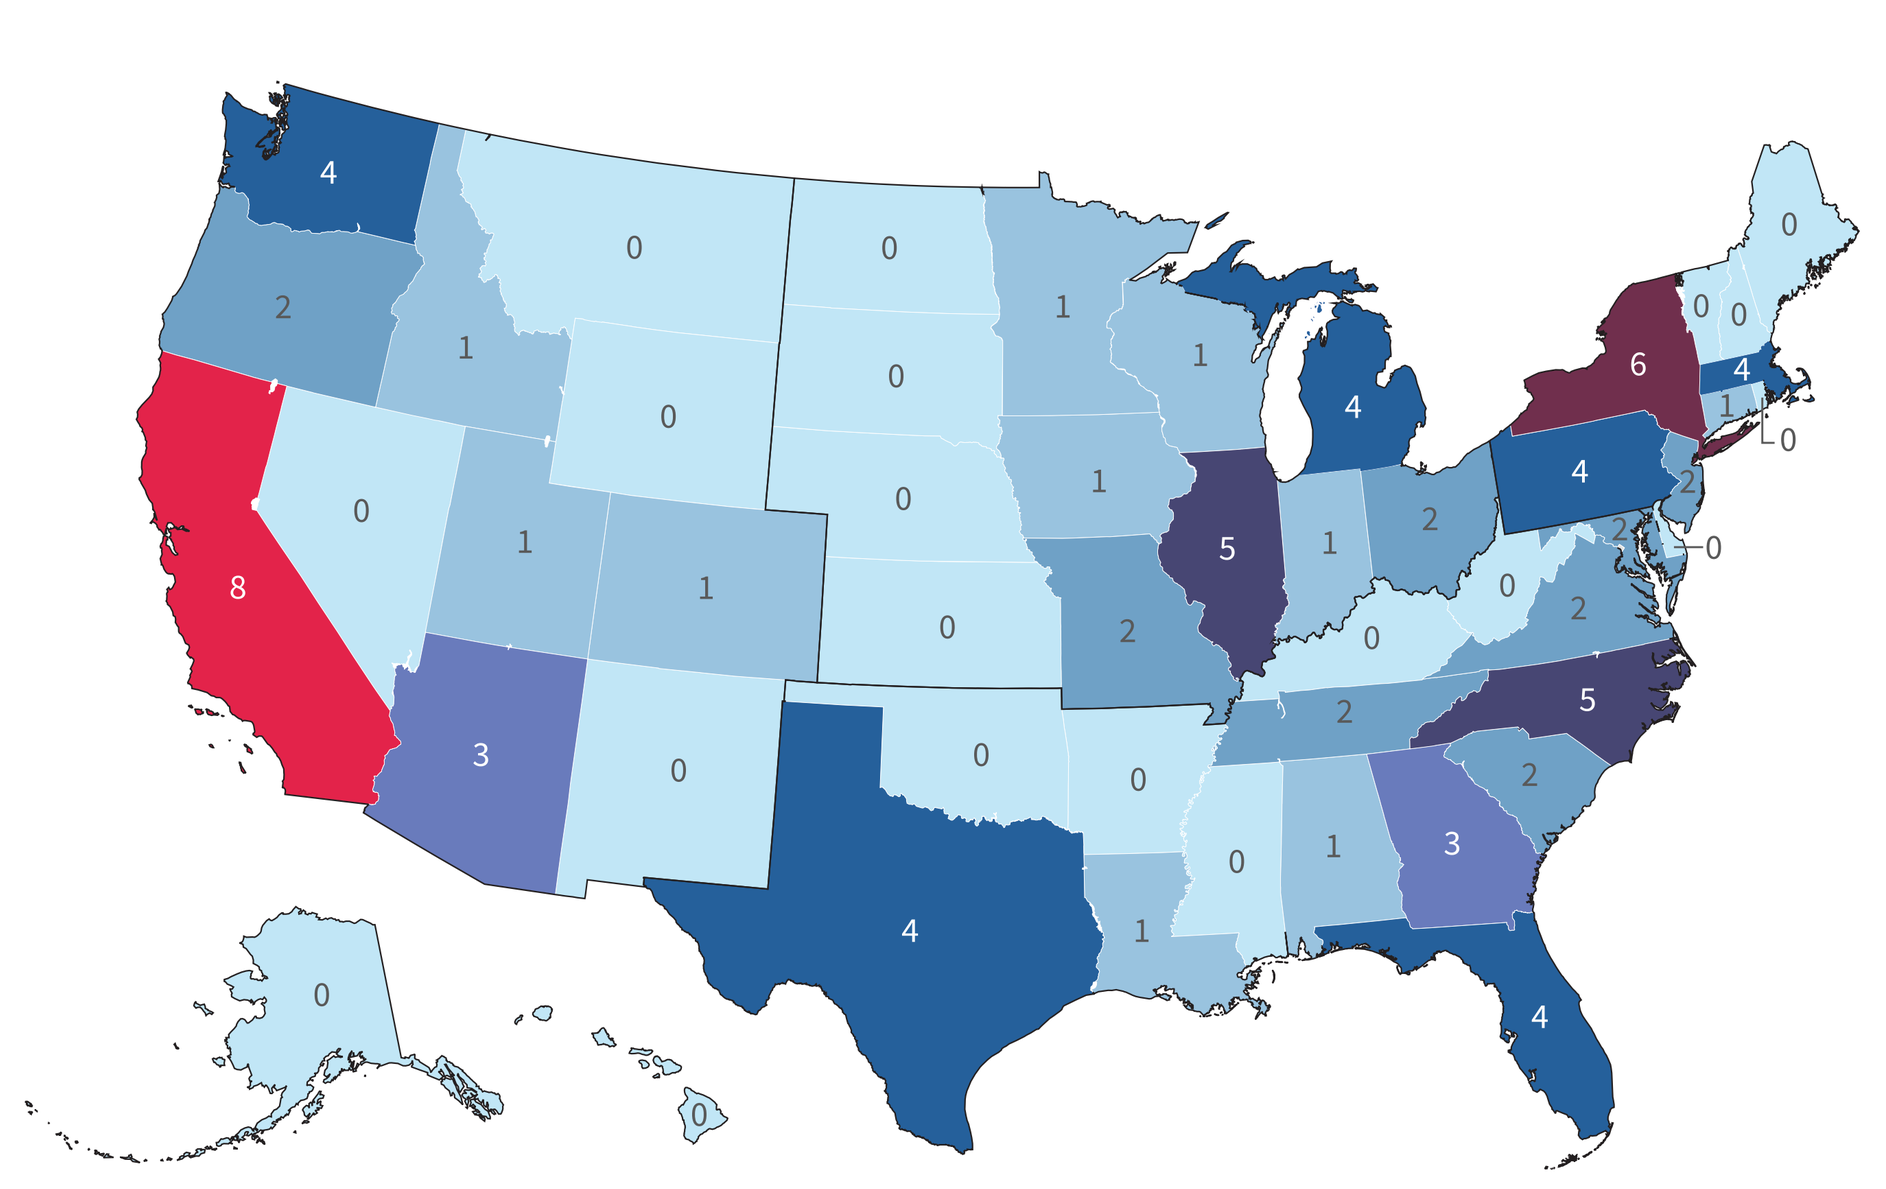

TABLE 2. ATTR-CM Prevalence by Race/Ethnicity and Region

Population	Prevalence (cases/100,000)
Overall	4.54
Race/ethnicity	
White	4.16
Black/African American	10.75
Other	2.01
Region	
Northeast	10.28
Midwest	5.19
West	2.61
South	2.76

- The prevalence of ATTR-CM increased over time, from 0.50 in 2017 to 3.12 in 2023, and varied by race (**Figure 3**)
 - The largest increase in prevalence over time was seen in patients who were Black/African American
- The overall distribution of ATTR-CM prevalence is shown in **Figure 4A**
 - The highest prevalence rates were seen in areas where there are amyloid centers, particularly in the Northeast
 - There are large areas of the country, particularly in the Midwest, where there are cardiology deserts with limited cardiologists and amyloid centers, resulting in fewer patients with ATTR-CM being diagnosed than would be expected
 - The distribution of ATTR-CM prevalence stratified by race/ethnicity is shown in **Figure 4B-D**
 - There are clusters of patients with ATTR-CM who are White (eg, Montana, Nebraska, New Mexico, North Dakota, South Dakota, and Wyoming) who are getting diagnosed despite further distance from an amyloid center (**Figure 4B**)
 - In patients with ATTR-CM who are Black/African American, we do not see similar clusters inside cardiology deserts, with prevalence being limited to areas with amyloid centers (**Figure 4C**)

FIGURE 3. Annual Prevalence of ATTR-CM by Race/Ethnicity in the US[a]

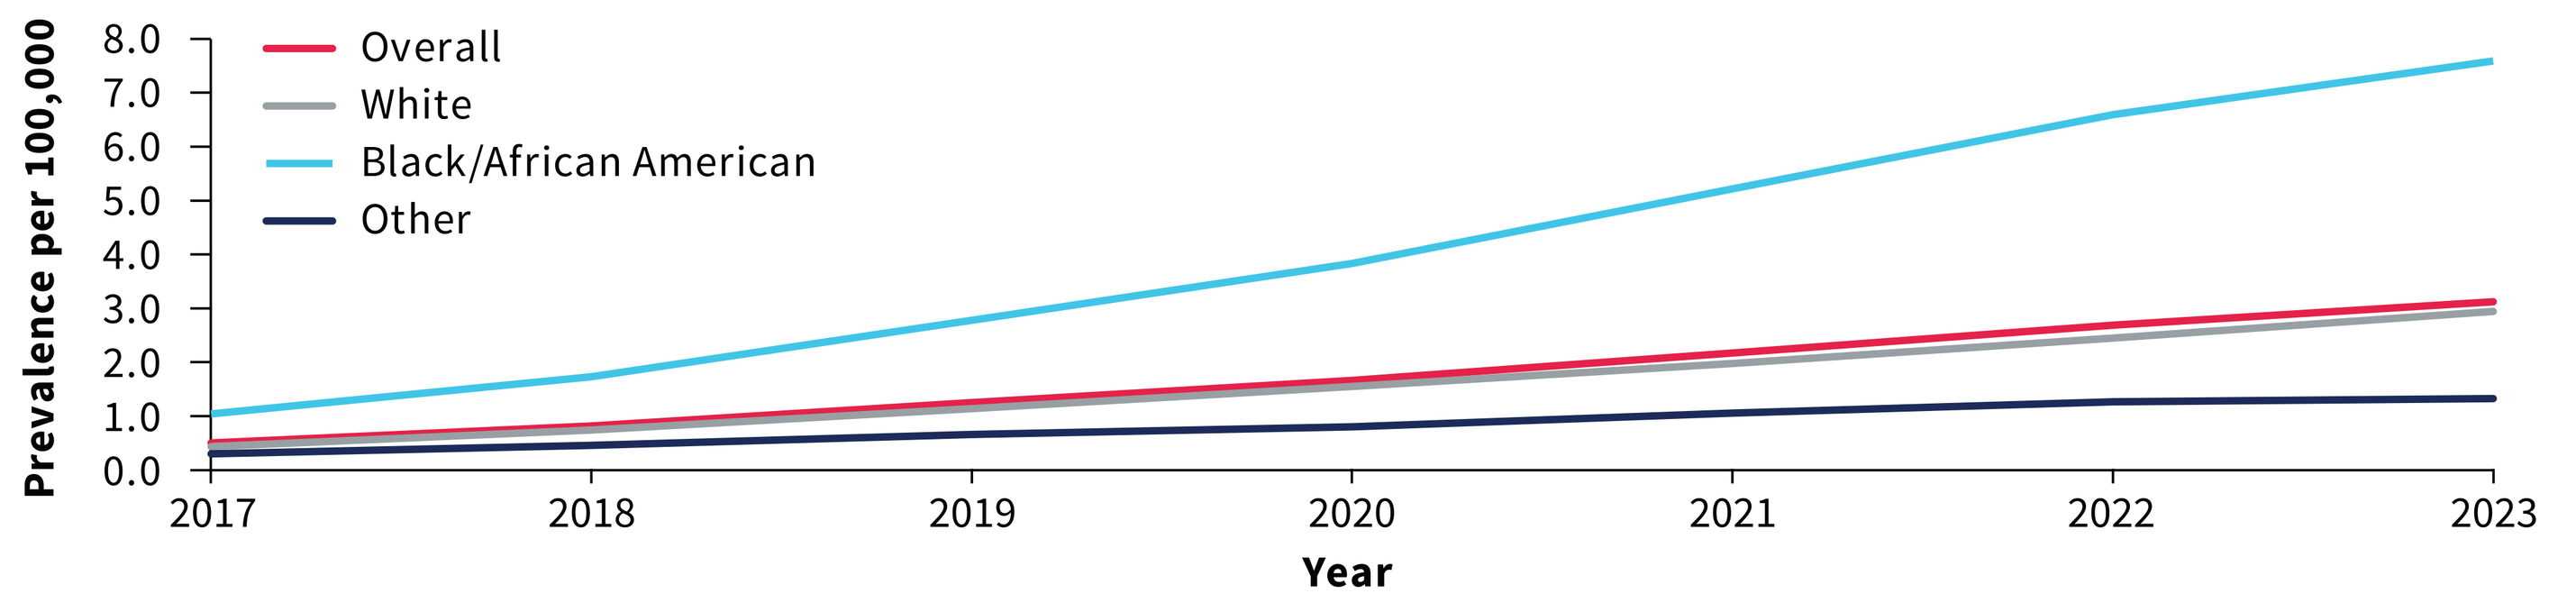
Prevalence per 100,000
8.0
7.0
6.0
5.0
4.0
3.0
2.0
1.0
0.0
2017
2018
2019
2020
2021
2022
2023
Year
Overall
White
Black/African American
Other

[a]Prevalence data were included for years in which complete study data were available.

FIGURE 4. Distribution of ATTR-CM Prevalence by Race/Ethnicity[a]

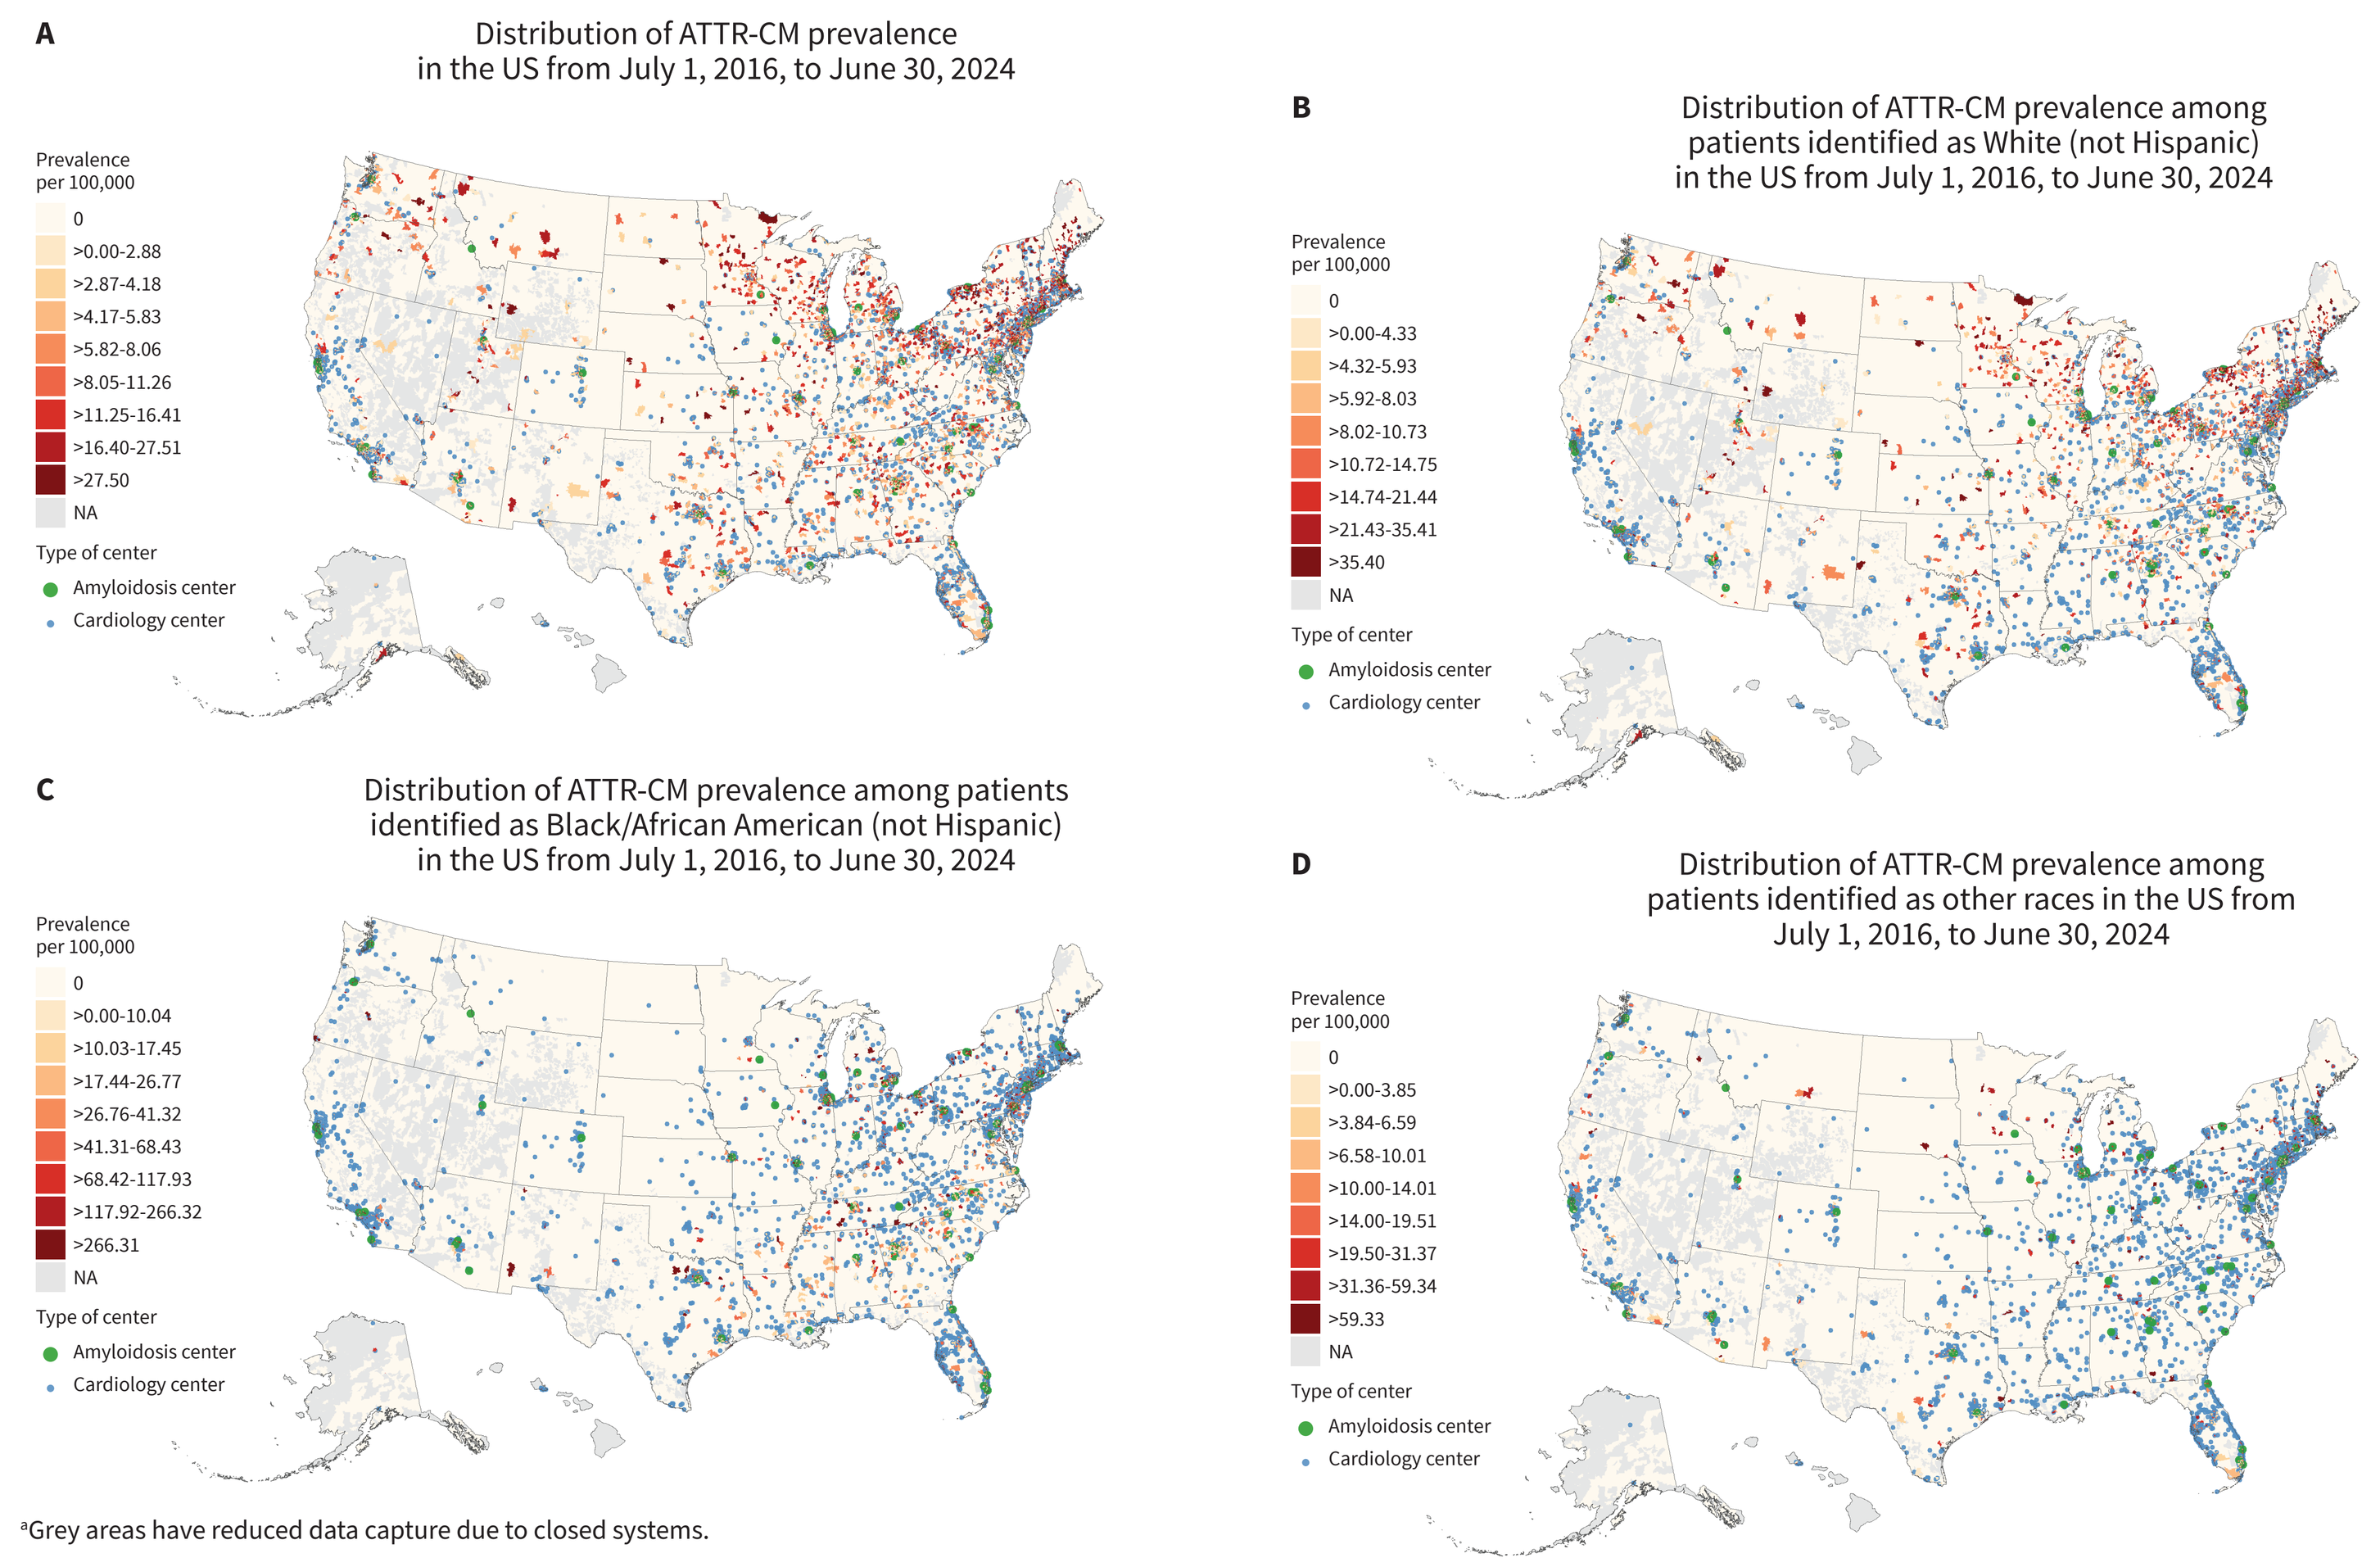
A
Distribution of ATTR-CM prevalence in the US from July 1, 2016, to June 30, 2024
Prevalence per 100,000
0
>0.00-2.88
>2.87-4.18
>4.17-5.83
>5.82-8.06
>8.05-11.26
>11.25-16.41
>16.40-27.51
>27.50
NA
Type of center
Amyloidosis center
Cardiology center
B
Distribution of ATTR-CM prevalence among patients identified as White (not Hispanic) in the US from July 1, 2016, to June 30, 2024
Prevalence per 100,000
0
>0.00-4.33
>4.32-5.93
>5.92-8.03
>8.02-10.73
>10.72-14.75
>14.74-21.44
>21.43-35.41
>35.40
NA
Type of center
Amyloidosis center
Cardiology center
C
Distribution of ATTR-CM prevalence among patients identified as Black/African American (not Hispanic) in the US from July 1, 2016, to June 30, 2024
Prevalence per 100,000
0
>0.00-10.04
>10.03-17.45
>17.44-26.77
>26.76-41.32
>41.31-68.43
>68.42-117.93
>117.92-266.32
>266.31
NA
Type of center
Amyloidosis center
Cardiology center
D
Distribution of ATTR-CM prevalence among patients identified as other races in the US from July 1, 2016, to June 30, 2024
Prevalence per 100,000
0
>0.00-3.85
>3.84-6.59
>6.58-10.01
>10.00-14.01
>14.00-19.51
>19.50-31.37
>31.36-59.34
>59.33
NA
Type of center
Amyloidosis center
Cardiology center

[a]Grey areas have reduced data capture due to closed systems.

LIMITATIONS

- Prevalence of ATTR-CM was calculated using an ICD-10-CM coding algorithm, which may result in missing or misidentifying true patients with ATTR-CM; therefore, the estimates provided here are a diagnosed prevalence, not a true prevalence
- Only the first 3 digits of patients' zip codes were available, requiring proxy calculations to be used to determine their location
- Patient data was limited in parts of the West (eg, Alaska and California) due to lack of Komodo Healthcare Map data capture in closed systems

DATA AVAILABILITY: The data that support the findings of this study were used under license from Komodo Health® and derived from the Komodo Healthcare Map®. Due to data use agreements and its proprietary nature, restrictions apply regarding the availability of the data. Further information is available from the sponsor of the study, BridgeBio Pharma, Inc., San Francisco, CA, USA.

FUNDING: The study was funded by BridgeBio Pharma, Inc. (San Francisco, CA, USA).

ABBREVIATIONS: ATTR-CM, transthyretin amyloid cardiomyopathy; ATTRv-CM, variant transthyretin amyloid cardiomyopathy; ATTRwt-CM, wild-type transthyretin amyloid cardiomyopathy; ICD-10-CM, International Classification of Diseases, Tenth Revision, Clinical Modification; Max, maximum; Min, minimum; NA, not available; Q1, first quartile; Q3, third quartile; SD, standard deviation; US, United States.

ACKNOWLEDGMENTS: All authors contributed to and approved the poster; editorial support was provided by Kavitha Kuppusamy, PhD, and Shweta Rane, PhD, CMPP, of BridgeBio Pharma, Inc. Writing and editorial assistance were provided by Robert Schupp, PharmD, CMPP, and Lauren Connor, ELS, of The Lockwood Group (Stamford, CT, USA), funded by BridgeBio Pharma, Inc.

REFERENCES: **1.** Ruberg FL, et al. *J Am Coll Cardiol*. 2019;73(22):2872-2891. **2.** Brito D, et al. *Glob Heart*. 2023;18(1):59. **3.** Rozenbaum MH, et al. *Cardiol Ther*. 2021;10(1):141-159. **4.** The Amyloidosis Foundation. The Patient Resources; Treatment Centers. Accessed February 3, 2025. https://www.amyloidosis.org/resources#treatment-centers. **5.** International Society of Amyloidosis. Amyloidosis Centers by Region. Accessed February 3, 2025. https://www.isaamyloidosis.org/resources. **6.** Amyloidosis Research Consortium. Treatment Centers & Clinical Trials. Accessed February 3, 2025. https://arci.org/patients-and-caregivers/treatment-centers-clinical-trials/. **7.** Motairek I, et al. *Circ Cardiovasc Qual Outcomes*. 2023;16(10):e010133. **8.** R Development Core Team. Vienna, Austria: R Foundation for Statistical Computing; 2010. R: A language and environment for statistical computing. ISBN 3-900051-07-0. Accessed February 3, 2025. http://www.R-project.org.

DISCLOSURES: **JM:** Researcher for Abbott Laboratories and Myocardial Solutions; consultant, advisor, and speaker for Alnylam, BridgeBio Pharma, Inc., Jazz Pharmaceuticals, Pfizer, and Race Oncology; **HDD, EN,** and **NL**: Contractor for BridgeBio Pharma, Inc.; **MU:** Employee and stockholder of BridgeBio Pharma, Inc.; stockholder of Pfizer.

PRESENTED AT THE 74TH ANNUAL SCIENTIFIC SESSION OF THE AMERICAN COLLEGE OF CARDIOLOGY; MARCH 29-31, 2025; CHICAGO, IL, USA.