Biomarkers, Proteomics and Echocardiography Lend Insights into Cardiovascular Death in HFrEF: A VICTORIA Substudy

Palak Shah[1], Yinggan Zheng[2], Burkert Pieske[3], Vojtech Melenovsky[4], Carolyn S.P. Lam[5], Karen S. Sliwa[6], Javed Butler[7], Justin A. Ezekowitz[2], Christopher deFilippi[1], Christopher M. O'Connor[1], Lothar Roessig[8], Jasper Tromp[9], Cynthia M. Westerhout[2], Adriaan A. Voors[10], Paul W. Armstrong[2] for the VICTORIA Study Group

Affiliations: (1) The Inova Center of Outcomes Research, Inova Heart and Vascular Institute, Falls Church, Virginia, USA (2) University of Alberta, Canadian VIGOUR Centre, Edmonton, Canada (3) Charité - University Medicine Berlin, Berlin, Germany (4) Department of Cardiology, Institute for Clinical and Experimental Medicine, Prague, Czech Republic (5) National Heart Centre Singapore (6) Hatter Institute for Cardiovascular Research in Africa, Faculty of Health Sciences, Department of Medicine and Cardiology, University of Cape Town, Cape Town, South Africa (7) Baylor University, Houston Texas, USA (8) Bayer, Wuppertal, Germany (9) Saw Swee Hock School of Public Health National University of Singapore (10) University Medical Centre Groningen, the Netherlands

Background

Using baseline data from the Vericiguat Global Study in Subjects With Heart Failure With Reduced Ejection Fraction (VICTORIA) ECHO substudy, we evaluated the potential complementary role of cardiac function, biomarkers, and proteomics in association with cardiovascular death (CVD), a key component of the primary composite (HF hospitalization or CVD) in high-risk patients with heart failure with reduced ejection fraction (HFrEF).

Hypothesis

Measurement of cardiac biomarkers, proteins, and ECHO parameters may provide insights into the mechanism of CVD in HFrEF.

Methods

At baseline, left ventricular ejection fraction (LVEF), LV end-systolic volume index (LVESVI) and diastolic volume index (LVEDVI) on ECHO, six quantitative biomarkers (high sensitivity cardiac troponin T, n-terminal pro-b-type natriuretic peptide, interleukin-6, high sensitivity C-reactive protein, growth differentiation factor-15, cystatin C), and 92 proteins (Olink CVD III panel) were assessed by blinded core labs. The association between the variables and CVD was assessed after adjustment for the Meta-Analysis Global Group in Chronic Heart Failure (MAGGIC) risk score. HRs (95% CIs) are presented. Proteomics associations were adjusted for false discovery.

Canadian **VIGOUR** Centre
Bridging Hearts and Minds

Results

- Of 583 patients, 96 (16.5%) had CVD (21.6/100 pt-years) at a median time of 8 months (IQR 6-11) from randomization.
- The MAGGIC-adjusted HRs for LVEF, LVESVI, LVEDVI and biomarkers are presented in the Table. NT-proBNP was most strongly associated with CVD (HR per doubling 1.53 [1.32-1.77]).
- Proteins significantly associated with CVD were cathepsin D (2.32 [1.62-3.32]), matrix metalloproteinase-2 (2.31 [1.45-3.68]), IL-2 receptor (1.74 [1.40-2.14]), transferrin receptor (1.64 [1.31-2.04]) and tumor necrosis superfamily (1.88 [1.41-2.50]) (Fig).

Table MAGGIC-adjusted associations of echocardiographic and biomarkers with CV death

	Mean (SD)		HR (95% CI)	p-value
	No CVD (n=487)	CVD (n=96)		
Echocardiographic parameters				
LVEF, per 5% increase	32.6 (8.9)	29.9 (8.7)	0.89(0.78-1.01)	0.064
LVESVI, per 10mL/m^2 increase	63.8 (28.1)	73.3 (33.3)	1.08(1.02-1.15)	0.009
LVEDVI, per 10mL/m^2 increase	92.6 (32.4)	103.2 (40.8)	1.07 (1.01-1.13)	0.014
Biomarkers			**(per doubling for HR)**	
hs-cTnT, ng/L	38.6 (49.0)	61.5 (61.7)	1.38(1.19-1.61)	<.001
NT-proBNP, pg/mL	4471 (5744)	9915 (10740)	1.53(1.32-1.77)	<.001
IL-6, pg/mL	10.5 (23.1)	27.4 (106.6)	1.46(1.26-1.68)	<.001
hsCRP, mg/L	8.5 (14.8)	17.2 (29.9)	1.27(1.13-1.42)	<.001
GDF-15, pg/mL	3898 (4144)	7382 (9808)	1.46(1.23-1.73)	<.001
Cystatin C, mg/L	1.36 (0.55)	1.60 (0.69)	1.07(0.68-1.68)	0.765

Figure MAGGIC-adjusted associations of individual proteins with CVD

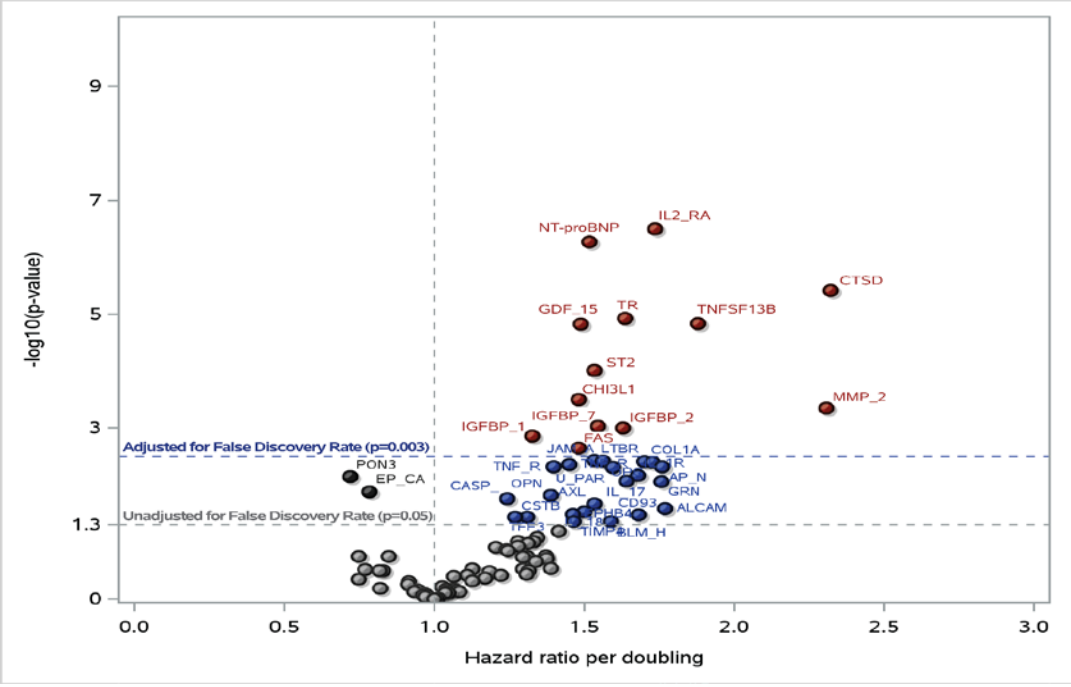
-log10(p-value)
Hazard ratio per doubling
0
1.3
3
5
7
9
0.0
0.5
1.0
1.5
2.0
2.5
3.0
Adjusted for False Discovery Rate (p=0.003)
Unadjusted for False Discovery Rate (p=0.05)
NT-proBNP
IL2_RA
CTSD
GDF_15
TR
TNFSF13B
ST2
CHI3L1
MMP_2
IGFBP_1
IGFBP_7
IGFBP_2
FAS
LTBR
COL1A
TNF_R
U_PAR
AP_N
GRN
CASP
OPN
AXL
IL_17
CD93
ALCAM
CSTB
TIMP
BLM_H
PON3
EP_CA

Volcano plot of protein changes at randomization associated with CVD during follow-up after adjustment for the MAGGIC risk score. Increasing protein levels were associated with increased risk of CVD (right) and reduced risk of CVD (left). Markers above the blue line are statistically significant after adjustment for the false discovery rate.

Conclusions

After MAGGIC adjustment, LVESVI and LVEDVI but not LVEF, were associated with CVD. Upregulation of selected biomarkers/proteins related to myocardial wall stress, extracellular matrix turnover, inflammation, and oxidative stress were also associated with CVD and may serve as novel future therapeutic targets in HFrEF.

References
1. Pieske B, et al. Effect of vericiguat on left ventricular structure and function in patients with heart failure with reduced ejection fraction: the VICTORIA echocardiographic substudy. *European Journal of Heart Failure* **n/a**.

Duke Clinical Research Institute