Early Insights From Real-World Use of Aflibercept 8 mg Among Eyes With Diabetic Macular Edema (DME) Switching From Other Anti-VEGF Agents

2396-B0083

Michael Javaheri, MD,[1] Steven Sherman, MPH,[2] Keran Moll, PhD,[2] Dana Murdock, PhD,[2] Nick Boucher, BS,[3] Rishi P. Singh, MD,[4] Ferhina S. Ali, MD, MPH,[5] Durga Borkar, MD,[6] Theodore Leng, MD,[7] Nitish Mehta, MD[8]

[1]Retina Specialists of Beverly Hills, Beverly Hills, California; [2]Regeneron Pharmaceuticals, Inc., Tarrytown, New York; [3]Vestrum Health, Naperville, Illinois; [4]Cleveland Clinic Martin Hospitals, Cleveland Clinic Florida, Stuart, Florida; [5]New York Medical College, Valhalla, New York; [6]Duke University Eye Center, Durham, North Carolina; [7]Byers Eye Institute, Stanford University School of Medicine, Palo Alto, California; [8]Department of Ophthalmology, NYU Langone Health, New York, New York

BACKGROUND & PURPOSE

- In the PHOTON trial, aflibercept 8 mg with extended dosing achieved non-inferior visual acuity (VA) outcomes with fewer injections compared to aflibercept 2 mg in patients with diabetic macular edema (DME) through 96 weeks[1,2]
- Real-world evidence for use of aflibercept 8 mg in previously treated patients with DME could be informative for clinical practice
- This analysis aimed to describe real-world treatment patterns in patients with DME who were previously treated with other anti-vascular endothelial growth factor (VEGF) agents before switching to aflibercept 8 mg

METHODS

- Two cohorts of eyes with DME that were previously treated with anti-VEGF agent(s) and had initiated aflibercept 8 mg were identified from electronic health records in the Intelligent Research in Sight (IRIS®) Registry and Vestrum Health Retina database, respectively (**Figure 1**)
 - Eyes initiating aflibercept 8 mg between August 18, 2023, to June 30, 2024, for the IRIS cohort, or August 18, 2023, to July 31, 2024, for the Vestrum cohort (indexing period), were followed from initiation (index date) until last visit or censoring event (treatment switch or missing information on treatment laterality), whichever occurred first
 - Data were available through December 31, 2024, for the IRIS cohort, and January 31, 2025, for the Vestrum cohort
- Injection intervals were evaluated for eyes that were consistently treated with anti-VEGF (defined as ≥6 months of treatment with an average injection interval of ≤8 weeks for the most recent anti-VEGF agent) and ≥1 post-initial dosing phase injection
- The last observed injection interval in the pre-switch phase (6-12 months prior to the index date) and after the initial dosing phase (defined as the first 3 injections or 90 days, whichever occurred first) were assessed, stratified by mean injection interval before switching (4-<6 or ≥6-8 weeks)

Figure 1. Inclusion Criteria and Attrition

Inclusion criteria	IRIS	Vestrum
Eyes with ≥1 diagnosis of DME on the index date receiving aflibercept 8 mg and no other anti-VEGF agent or other treatments[a]	n=30,266	n=7845
Adult patients (aged ≥18 years) with no diagnosis of nAMD or RVO during the 12 months prior to/on the index date	n=28,854	n=7410
Eyes treated with anti-VEGF therapy or other treatments[a] during the 12 months prior to the index date	n=24,808	n=7026
≥1 visit ≥6 months prior to the index date	n=23,436	n=6166
For patients with both eyes eligible for inclusion, 1 eye was randomly selected per patient	n=17,938	n=4017
Eyes that were switched from an anti-VEGF agent to aflibercept 8 mg during the indexing period	n=10,092	n=3706
Eyes that were consistently treated pre-switch[b]	n=3963	n=2162
Eyes with an average injection interval of 4-8 weeks before switching and ≥1 post-initial dosing phase injection of aflibercept 8 mg	n=2280	n=653

Where n is the number of eyes. Criteria in the lower green box apply to the injection interval analyses. [a]Other treatments included intravitreal steroids and laser therapy. [b]Treated with an anti-VEGF agent for ≥6 months and an average injection interval of ≤8 weeks for the most recent anti-VEGF agent. nAMD, neovascular age-related macular degeneration; RVO, retinal vein occlusion.

RESULTS

Table 1. Patient Characteristics at Index Date

	IRIS (n=10,092)	Vestrum (n=3706)
Mean age (SD), years	66.5 (10.5)	66.3 (10.9)
Male, n (%)	5576 (55)	2033 (55)
Race/ethnicity, n (%)		
Hispanic or Latino	866 (10)	NA
White	5548 (62)	NA
Black or African American	1025 (12)	NA
Asian or Pacific Islander	276 (3)	NA
Other	1174 (13)	NA
Bilateral disease, n (%)	9264 (92)	3210 (87)
Fellow eye treated with aflibercept 8 mg on the index date, n (%)	3671 (36)	1064 (29)
Mean VA (SD), ETDRS letters	65.5 (18.2)	70 (10.7)

ETDRS, Early Treatment of Diabetic Retinopathy Study; NA, not available (due to limitation of the Vestrum database); SD, standard deviation.

- Most eyes initiating aflibercept 8 mg were switched from aflibercept 2 mg (**Figure 2**)

Figure 2. Anti-VEGF Agent Used Before Switching to Aflibercept 8 mg

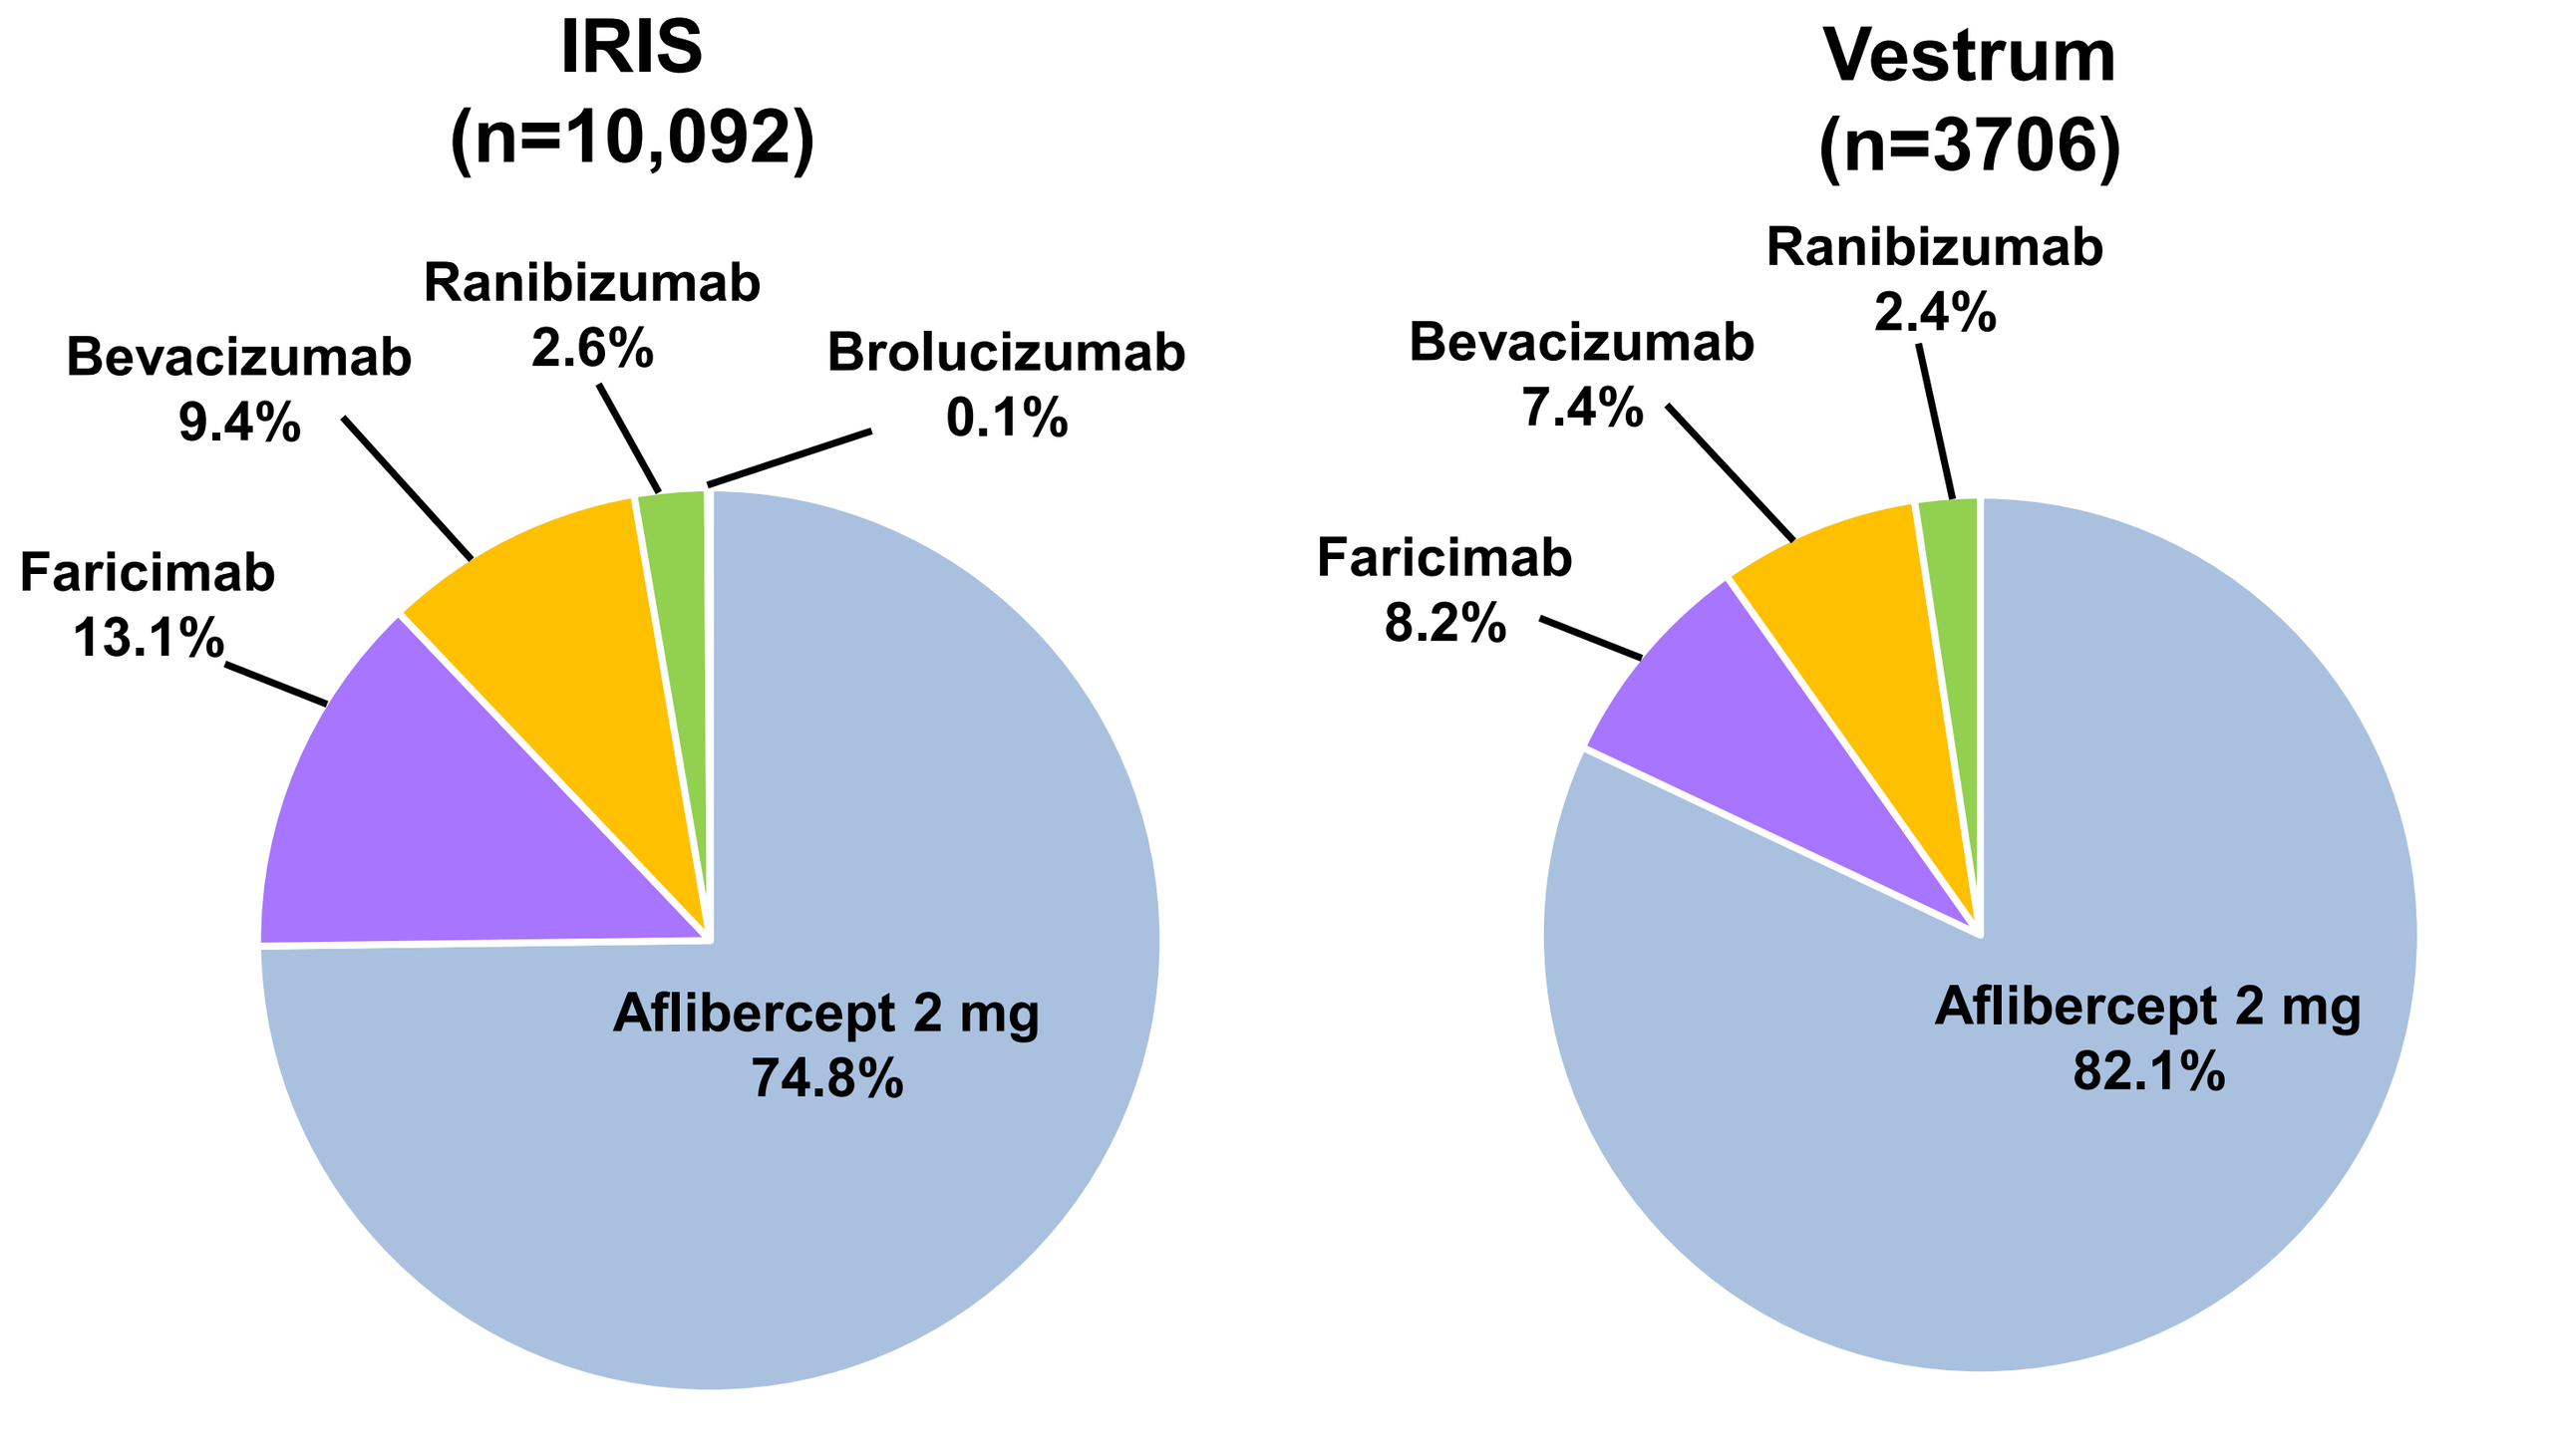
IRIS
(n=10,092)
Ranibizumab 2.6%
Bevacizumab 9.4%
Brolucizumab 0.1%
Faricimab 13.1%
Aflibercept 2 mg 74.8%
Vestrum
(n=3706)
Ranibizumab 2.4%
Bevacizumab 7.4%
Faricimab 8.2%
Aflibercept 2 mg 82.1%

Data represent the proportion of eyes receiving each anti-VEGF agent. Values may not add up to 100% due to rounding. Ranibizumab comprised ranibizumab-eqrn (1.4%), ranibizumab (1.1%), and ranibizumab-nuna (0.1%) in the IRIS cohort; and ranibizumab-eqrn (1.8%), ranibizumab (0.5%), and ranibizumab-nuna (0.1%) in the Vestrum cohort.

Dosing Patterns

- Mean (SD) post-switch follow-up was 167 (109) and 246 (110) days in the IRIS and Vestrum cohorts, respectively (median [quartile {Q}1, Q3]: 162 [71, 245] and 252 [175, 329] days, respectively)
- Mean (SD) number of injections of aflibercept 8 mg administered during follow-up (including index date) was 3 (2) and 3 (1) in the IRIS and Vestrum cohorts, respectively (median [Q1, Q3]: 3 [1, 4] and 3 [2, 4], respectively)

Table 2. Injection Interval Pre- and Post-Switch to Aflibercept 8 mg[a]

	Eyes with pre-switch injection interval 4-<6 weeks		Eyes with pre-switch injection interval ≥6-8 weeks	
	IRIS (n=959)	Vestrum (n=224)	IRIS (n=1321)	Vestrum (n=429)
Last observed pre-switch injection interval, days				
Mean (SD)	37 (8)	39 (10)	51 (13)	52 (14)
Median (Q1, Q3)	35 (29, 42)	35 (31, 42)	49 (42, 56)	49 (42, 56)
Last observed post-initial dosing phase injection interval, days				
Mean (SD)	59 (24)	65 (18)	67 (24)	71 (25)
Median (Q1, Q3)	56 (49, 63)	62 (56, 70)	63 (56, 77)	63 (56, 84)
Change in last pre-switch to last post-initial dosing phase injection interval				
Mean change (SD)	23 (25)	26 (19)	16 (25)	19 (27)
Median (Q1, Q3)	21 (12, 28)	21 (14, 35)	14 (1, 26)	14 (5, 28)

[a]Included eyes that were consistently treated: ≥6 months of treatment prior to switch with an average pre-switch injection interval of 4-8 weeks and ≥1 post-initial dosing phase aflibercept 8 mg injection.

- Mean changes (SD) from last pre-switch to last post-initial dosing phase injection intervals are shown for eyes with a pre-switch injection interval of 4-<6 weeks (**Figure 3**) or a pre-switch injection interval of ≥6-8 weeks (**Figure 4**)

Figure 3. Injection Interval Extension of ~3-4 Weeks After Switching to Aflibercept 8 mg in Eyes With Average Pre-Switch Injection Interval 4-<6 Weeks[a]

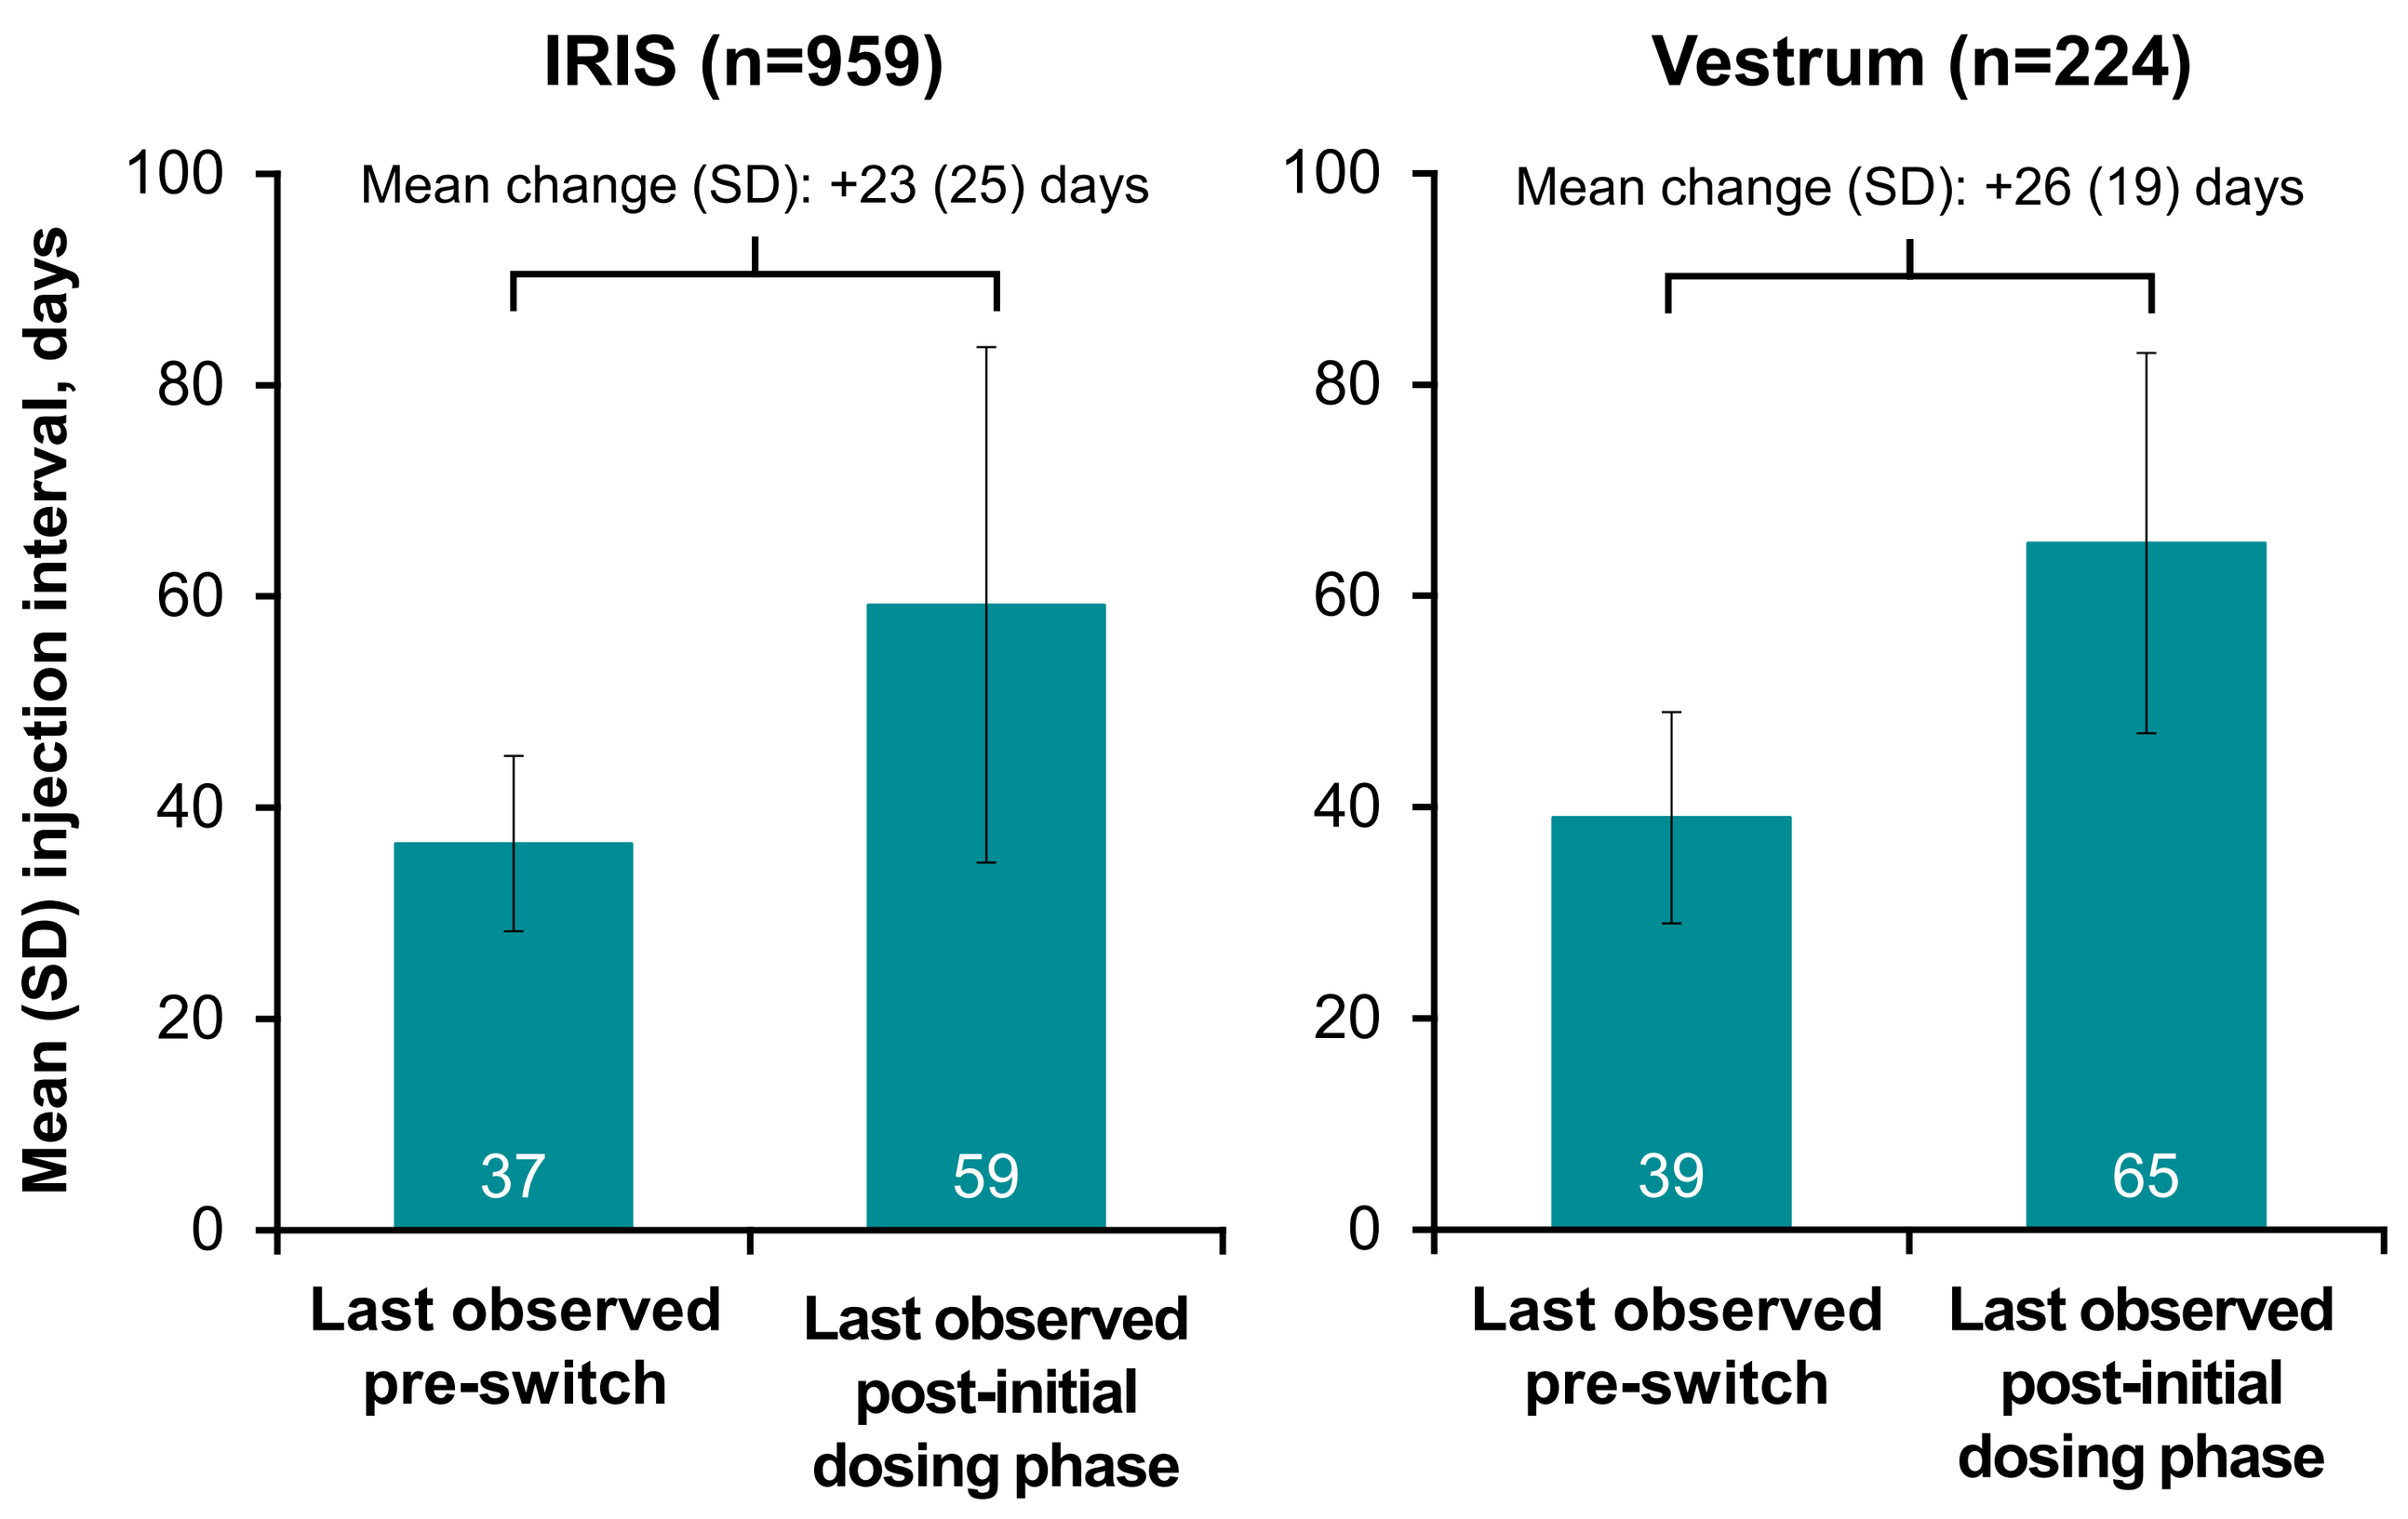
IRIS (n=959)
Mean change (SD): +23 (25) days
Vestrum (n=224)
Mean change (SD): +26 (19) days
Mean (SD) injection interval, days
0
20
40
60
80
100
37
59
39
65
Last observed pre-switch
Last observed post-initial dosing phase

[a]Included eyes that were consistently treated: ≥6 months of treatment prior to switch with an average pre-switch injection interval of 4-<6 weeks and ≥1 post-initial dosing phase aflibercept 8-mg injection.

Figure 4. Injection Interval Extension of ~2-3 Weeks After Switching to Aflibercept 8 mg in Eyes With Average Pre-Switch Injection Interval ≥6-8 Weeks[a]

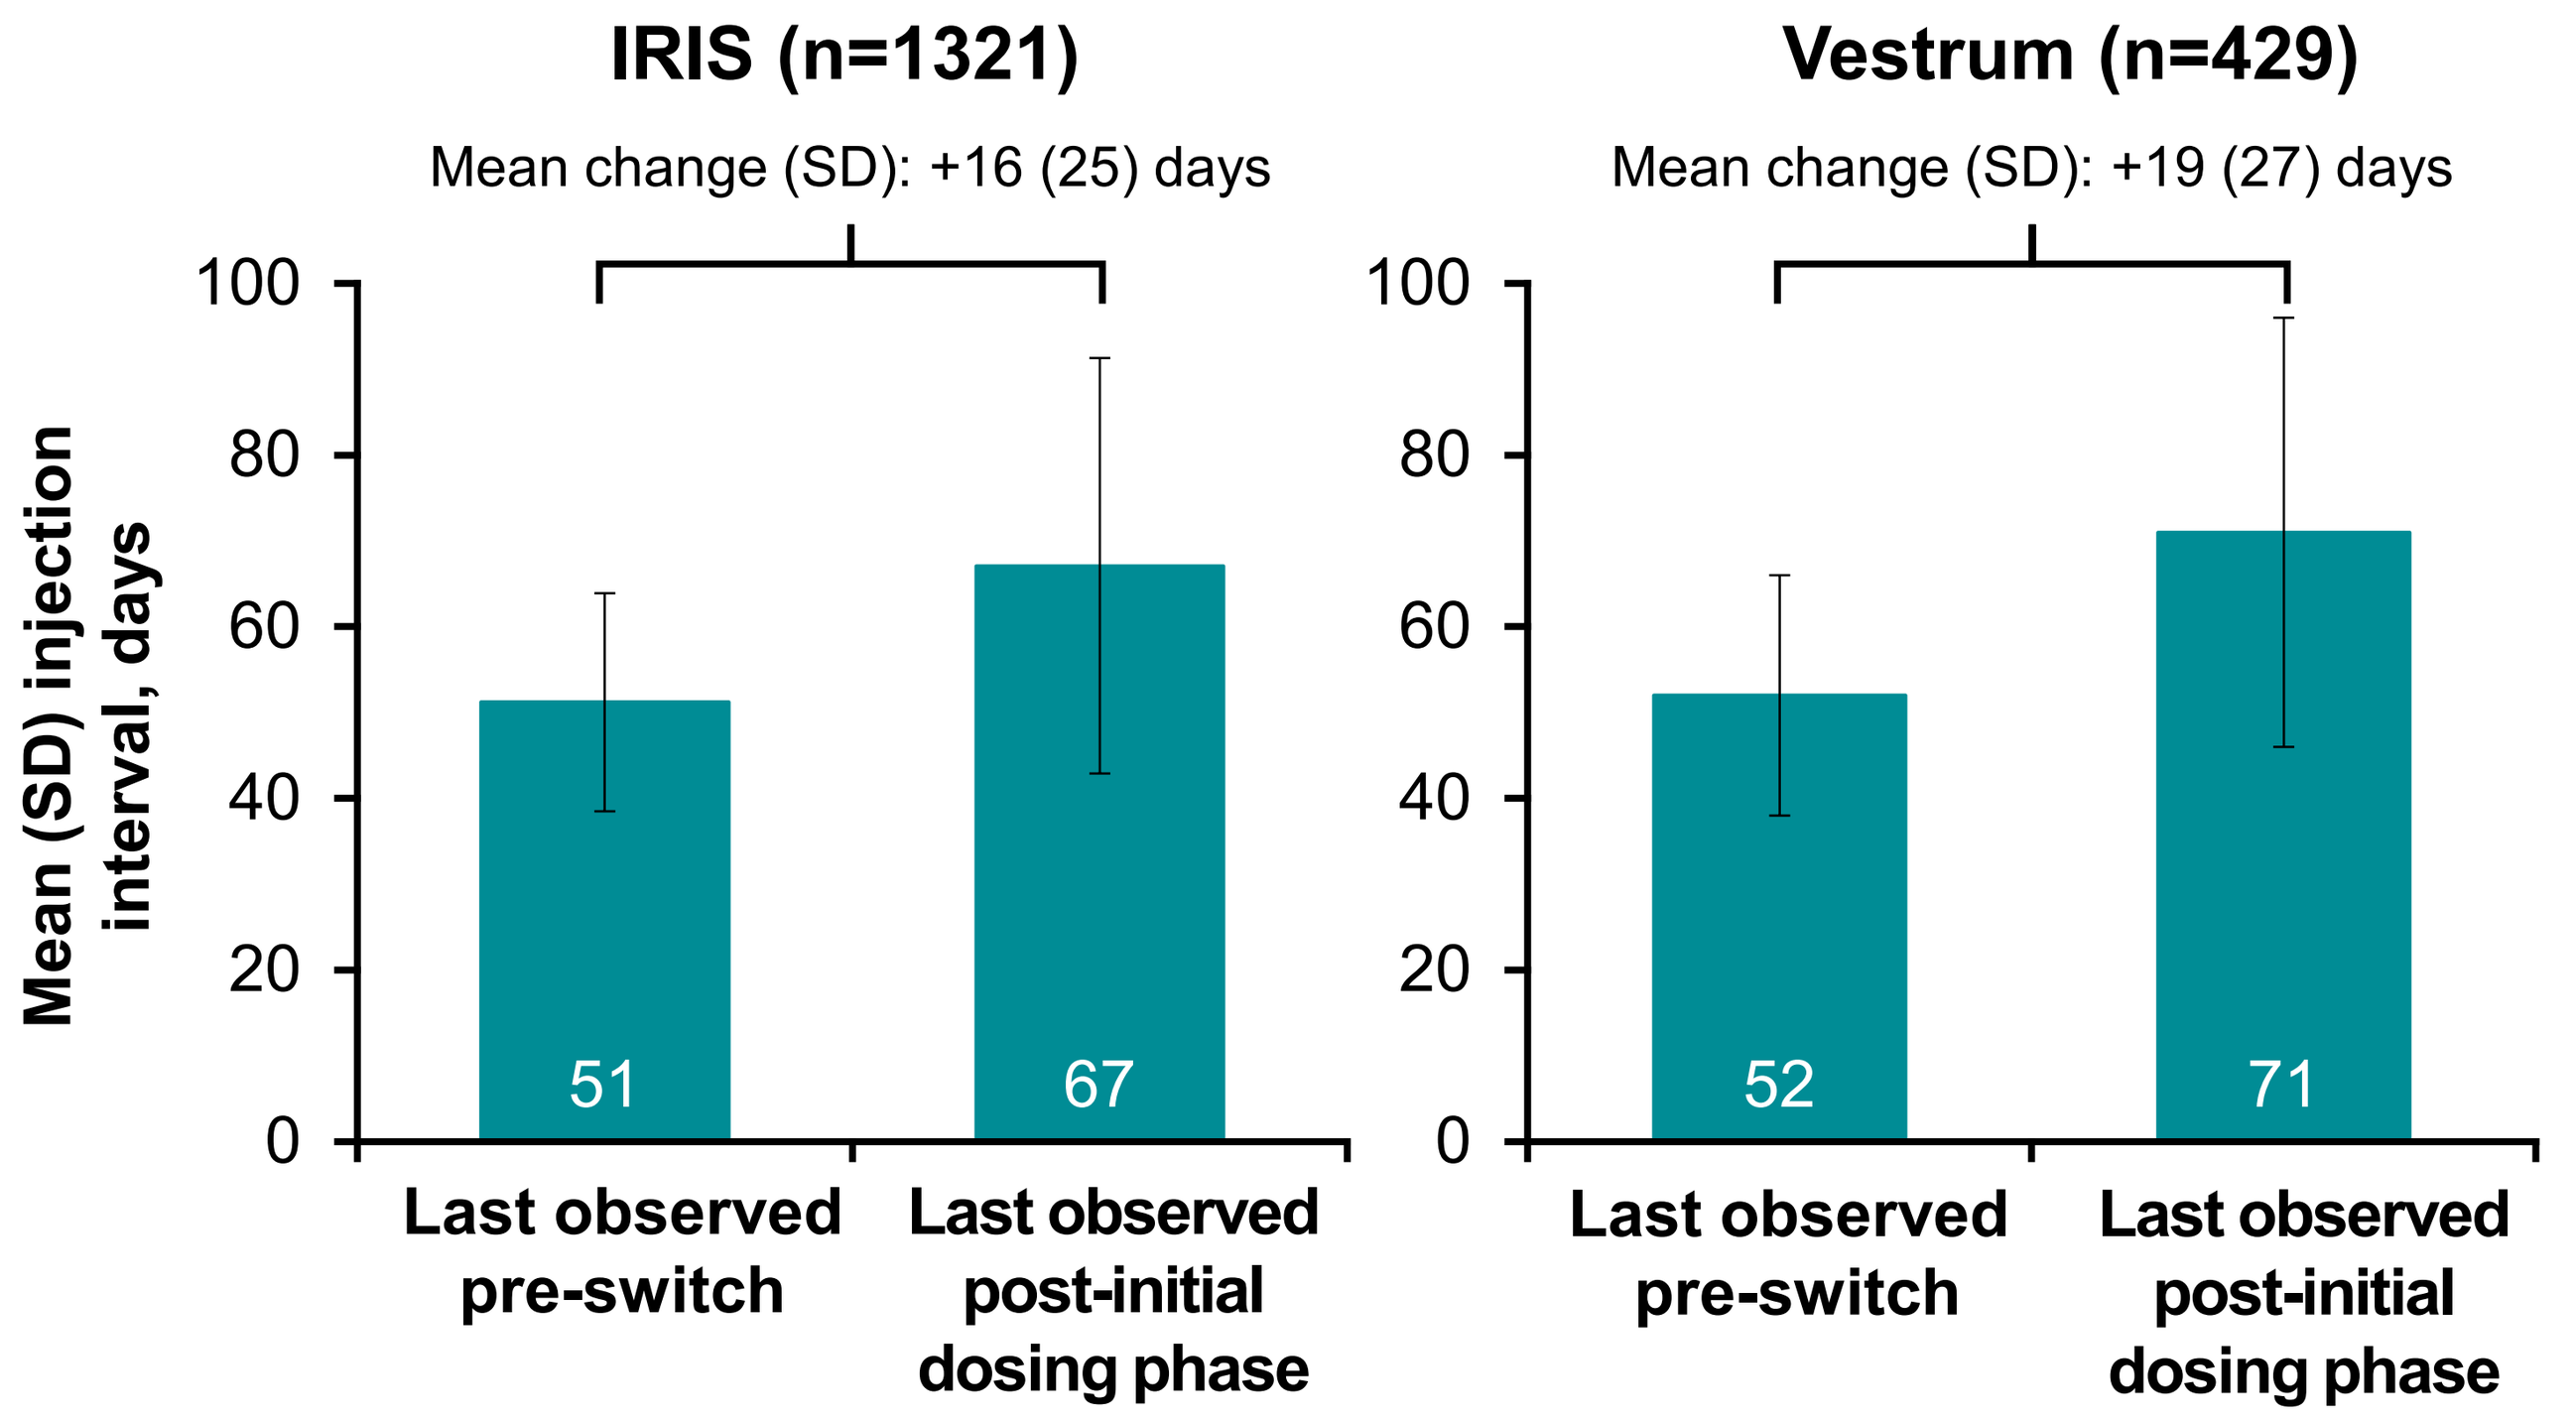
IRIS (n=1321)
Mean change (SD): +16 (25) days
Vestrum (n=429)
Mean change (SD): +19 (27) days
Mean (SD) injection interval, days
0
20
40
60
80
100
51
67
52
71
Last observed pre-switch
Last observed post-initial dosing phase

[a]Included eyes that were consistently treated: ≥6 months of treatment prior to switch with an average pre-switch injection interval of ≥6-8 weeks and ≥1 post-initial dosing phase aflibercept 8-mg injection.

Limitations

- This study was based on data from electronic medical records, which may not reflect patients' full medical history, including prior treatment history
- This study included a subset of patients who switched to aflibercept 8 mg from other anti-VEGF therapies and may not represent the wider population
- This study represents early real-world experience with aflibercept 8 mg and had a limited follow-up period

CONCLUSIONS

- Most eyes were consistently treated with aflibercept 2 mg prior to initiating aflibercept 8-mg treatment
- Eyes switching to aflibercept 8 mg after consistent treatment with a previous anti-VEGF agent had an average injection interval extension of:
 - Approximately 3-4 weeks for eyes with an average pre-switch injection interval of 4-<6 weeks
 - Approximately 2-3 weeks for eyes with an average pre-switch injection interval of ≥6-8 weeks
- Additional analyses with longer-term follow-up are ongoing to assess real-world durability and outcomes of aflibercept 8 mg in previously treated patients with DME

REFERENCES

1. Brown DM et al. *Lancet*. 2024;403:1153–1163.
2. Do D. Presented at the American Academy of Ophthalmology Meeting; November 3-6, 2023; San Francisco, CA.

ACKNOWLEDGMENTS & DISCLOSURES

- Michael Javaheri has acted as a speaker and consultant, and partaken in advisory boards, for Genentech and Regeneron Pharmaceuticals, Inc. Steven Sherman, Dana Murdock, and Keran Moll are employees of, and stockholders in, Regeneron Pharmaceuticals, Inc. Nick Boucher is an employee of Vestrum Health. Rishi P. Singh has received personal fees from Apellis, Iveric Bio, EyePoint, REGENXBIO, Genentech, Bausch + Lomb, Zeiss, Alcon, and Regeneron Pharmaceuticals, Inc., and has received research grants from Janssen. Ferhina S. Ali has received honoraria and served on speaker bureaus for 4DMT, Allergan, Apellis, EyePoint, Genentech, Iveric Bio, OcuTerra, Optomed, Ocuphire, Outlook Therapeutics, and Regeneron Pharmaceuticals, Inc. Durga Borkar has acted as a consultant for 4DMT, Alimera Sciences, Astellas, Apellis, AbbVie/Allergan, EyePoint, Genentech, Glaukos, ONL Therapeutics, Regeneron Pharmaceuticals, Inc., and Verana Health, and as a speaker for Astellas and Genentech. Theodore Leng has received funding from Astellas, and acted as a consultant for Astellas, Boehringer Ingelheim, Regeneron Pharmaceuticals, Inc., Roche/Genentech, Topcon, and Virtual Field. Nitish Mehta has no disclosures to report
- This analysis was sponsored by Regeneron Pharmaceuticals, Inc. (Tarrytown, NY). The sponsor participated in the design and conduct of the study, analysis of the data, and preparation of this poster
- Medical writing support was provided by Matthew Young, DPhil, and editorial support by Jess Fawcett, BSc, of Core (a division of Prime, London, UK), funded by Regeneron Pharmaceuticals, Inc. according to Good Publication Practice guidelines

Poster presented by Michael Javaheri, MD, email: mikejavaheri@gmail.com, at the ARVO 2025 Meeting, May 4-8, 2025