Outcomes of Patients With Diabetic Macular Edema and Baseline Best-corrected Visual Acuity of 20/50 or Worse or 20/40 or Better Who Were Treated With Aflibercept 8 mg and 2 mg: a Post Hoc Analysis of the Phase 2/3 PHOTON Trial

6238-B0148

Sean Adrean, MD, on behalf of the PHOTON study investigators
Retina Consultants of Orange County, Fullerton, California

BACKGROUND & PURPOSE

- In the PHOTON trial, intravitreal aflibercept 8 mg demonstrated non-inferior visual outcomes to aflibercept 2 mg at Week 48 with fewer injections in patients with diabetic macular edema (DME).[1] However, the impact of baseline best-corrected visual acuity (BCVA) on clinical outcomes following treatment with aflibercept 8 mg is not well characterized
- A post hoc analysis was conducted to evaluate visual and anatomic outcomes with aflibercept 8 mg and 2 mg in patients with DME by baseline BCVA (20/50 or worse and 20/40 or better)

METHODS

- PHOTON enrolled patients aged ≥18 years with type 1 or type 2 diabetes, DME with central involvement and central retinal thickness (CRT) ≥300 µm in the study eye, and BCVA of 78 to 24 Early Treatment Diabetic Retinopathy Study (ETDRS) letters (Snellen equivalent 20/32 to 20/320) with decreased vision due to DME[1]
- Eligible patients with DME were randomized 1:2:1 to receive aflibercept 2 mg every 8 weeks after 5 monthly doses (2q8) or aflibercept 8 mg every 12 or 16 weeks after 3 monthly doses (8q12 or 8q16) (**Figure 1A**)
 - Starting at Week 16, dosing intervals of patients in the aflibercept 8q12 and 8q16 groups were shortened if dose regimen modification (DRM) criteria were met (**Figure 1B**)

Figure 1. (A) PHOTON Study Design and (B) Dosing Schedule

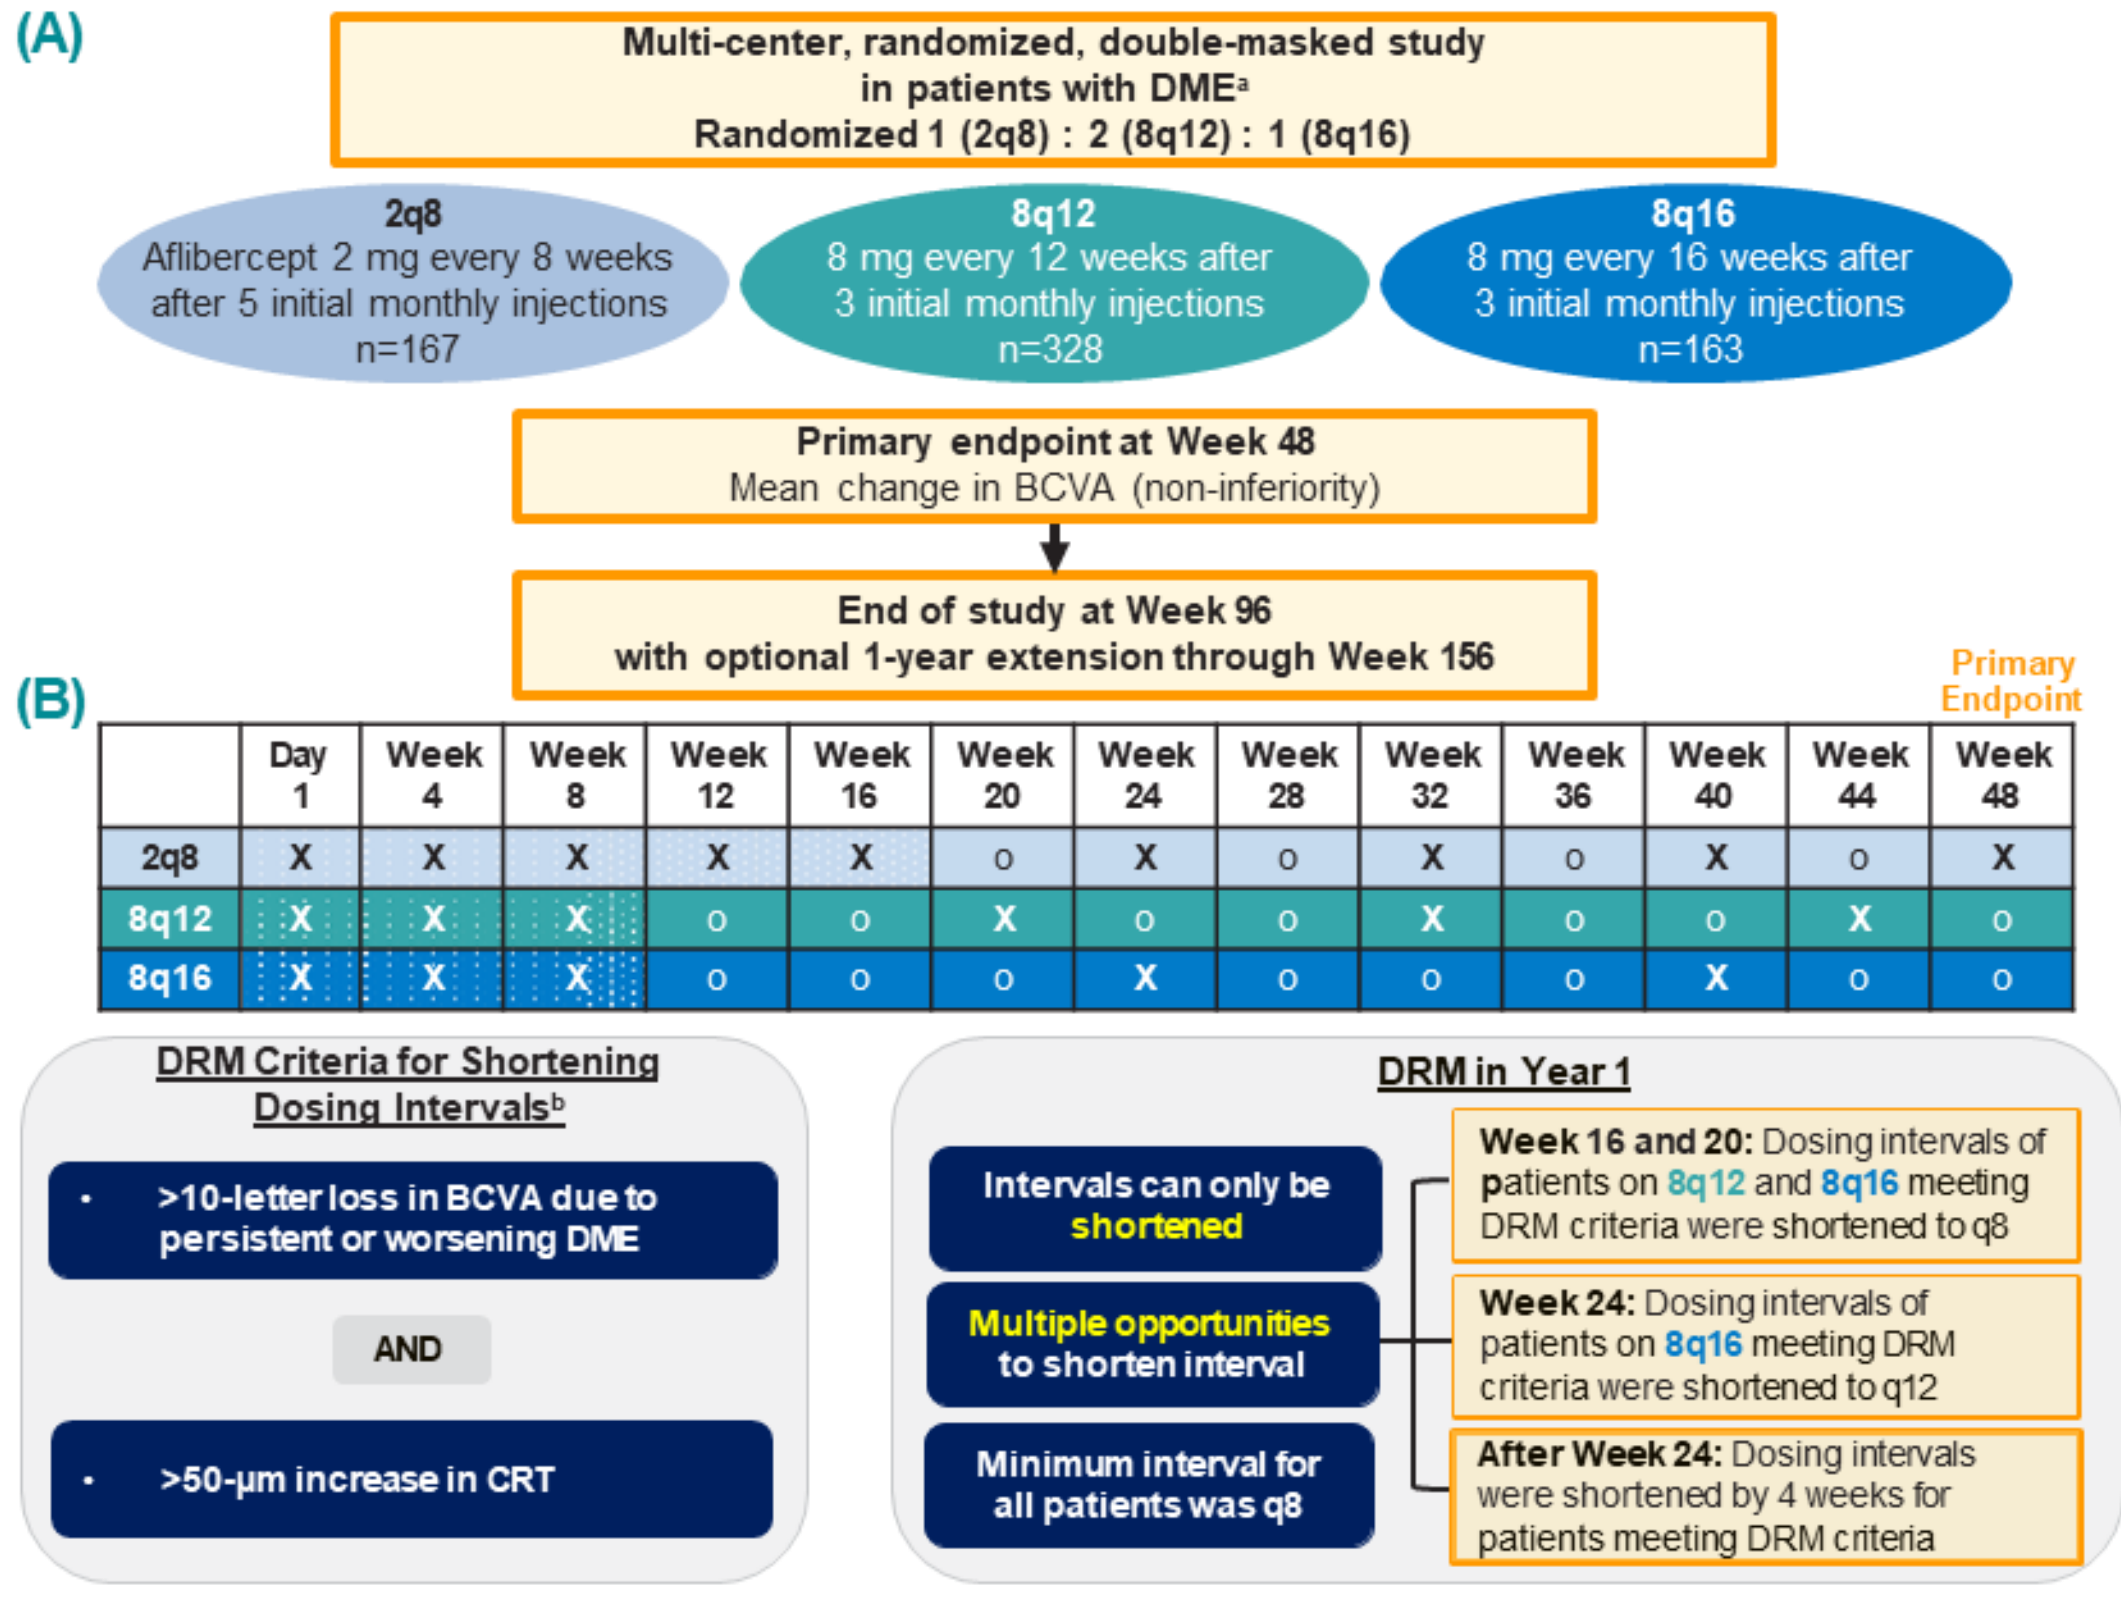
(A)
Multi-center, randomized, double-masked study in patients with DME[a]
Randomized 1 (2q8) : 2 (8q12) : 1 (8q16)
2q8
Aflibercept 2 mg every 8 weeks after 5 initial monthly injections
n=167
8q12
8 mg every 12 weeks after 3 initial monthly injections
n=328
8q16
8 mg every 16 weeks after 3 initial monthly injections
n=163
Primary endpoint at Week 48
Mean change in BCVA (non-inferiority)
End of study at Week 96
with optional 1-year extension through Week 156
(B)
Primary Endpoint
Day 1 | Week 4 | Week 8 | Week 12 | Week 16 | Week 20 | Week 24 | Week 28 | Week 32 | Week 36 | Week 40 | Week 44 | Week 48
2q8 X X X X X o X o X o X o X
8q12 X X X o o X o o X o o X o
8q16 X X X o o o X o o o X o o
DRM Criteria for Shortening Dosing Intervals[b]
>10-letter loss in BCVA due to persistent or worsening DME
AND
>50-µm increase in CRT
DRM in Year 1
Intervals can only be shortened
Multiple opportunities to shorten interval
Minimum interval for all patients was q8
Week 16 and 20: Dosing intervals of patients on 8q12 and 8q16 meeting DRM criteria were shortened to q8
Week 24: Dosing intervals of patients on 8q16 meeting DRM criteria were shortened to q12
After Week 24: Dosing intervals were shortened by 4 weeks for patients meeting DRM criteria

Stippled boxes=initial treatment phase; X=active injection; o=sham injection.
Note: Figure does not reflect all dosing options once treatment intervals are shortened.
[a]Treatment naïve and previously treated. [b]All assessments compared to Week 12.

Post Hoc Analysis

- This analysis was conducted using data through Week 48 from patients in the full analysis set (FAS), defined as patients who were randomized and treated with aflibercept 8 mg or 2 mg
- Patients were grouped by baseline BCVA of 20/40 or worse (defined as <69 ETDRS letters) or 20/50 or better (defined as ≥69 ETDRS letters)
- All analyses were descriptive

RESULTS

- Patients with baseline BCVA 20/50 or worse had lower mean BCVA and greater mean CRT at baseline than those with baseline BCVA 20/40 or better (**Table 1**)

Table 1. Baseline Characteristics by Baseline BCVA

	Baseline BCVA 20/50 or Worse			Baseline BCVA 20/40 or Better		
	2q8 (n=115)	8q12 (n=203)	8q16 (n=104)	2q8 (n=52)	8q12 (n=125)	8q16 (n=59)
BCVA, mean (SD), ETDRS letters	56.5 (9.9)	57.8 (8.3)	55.1 (10.1)	72.6 (2.9)	73.2 (2.7)	72.6 (2.6)
CRT, mean (SD), µm	482.9 (154.2)	472.7 (136.4)	491.5 (120.4)	400.6 (97.8)	411.1 (100.7)	405.3 (90.8)
Prior DME treatment, n (%)	51 (44.3)	95 (46.8)	53 (51.0)	23 (44.2)	48 (38.4)	18 (30.5)
DRSS score, n (%)						
DRSS 47 or worse	40 (34.8)	73 (36.0)	33 (31.7)	13 (25.0)	40 (32.0)	13 (22.0)
DRSS 43 or better	66 (57.4)	115 (56.7)	62 (59.6)	39 (75.0)	82 (65.6)	45 (76.3)
Non-gradable	9 (7.8)	15 (7.4)	9 (8.7)	0	3 (2.4)	1 (1.7)

FAS.
DRSS, Diabetic Retinopathy Severity Scale.

- Improvements in BCVA and CRT through Week 48 were comparable across all 3 treatment groups, regardless of baseline BCVA (**Figures 2 and 3**)
 - However, the magnitude of improvement was numerically greater in patients with baseline BCVA 20/50 or worse compared with those with baseline BCVA 20/40 or better

Figure 2. Mean Change From Baseline in BCVA Through Week 48 by Baseline BCVA

(A) Baseline BCVA 20/50 or Worse

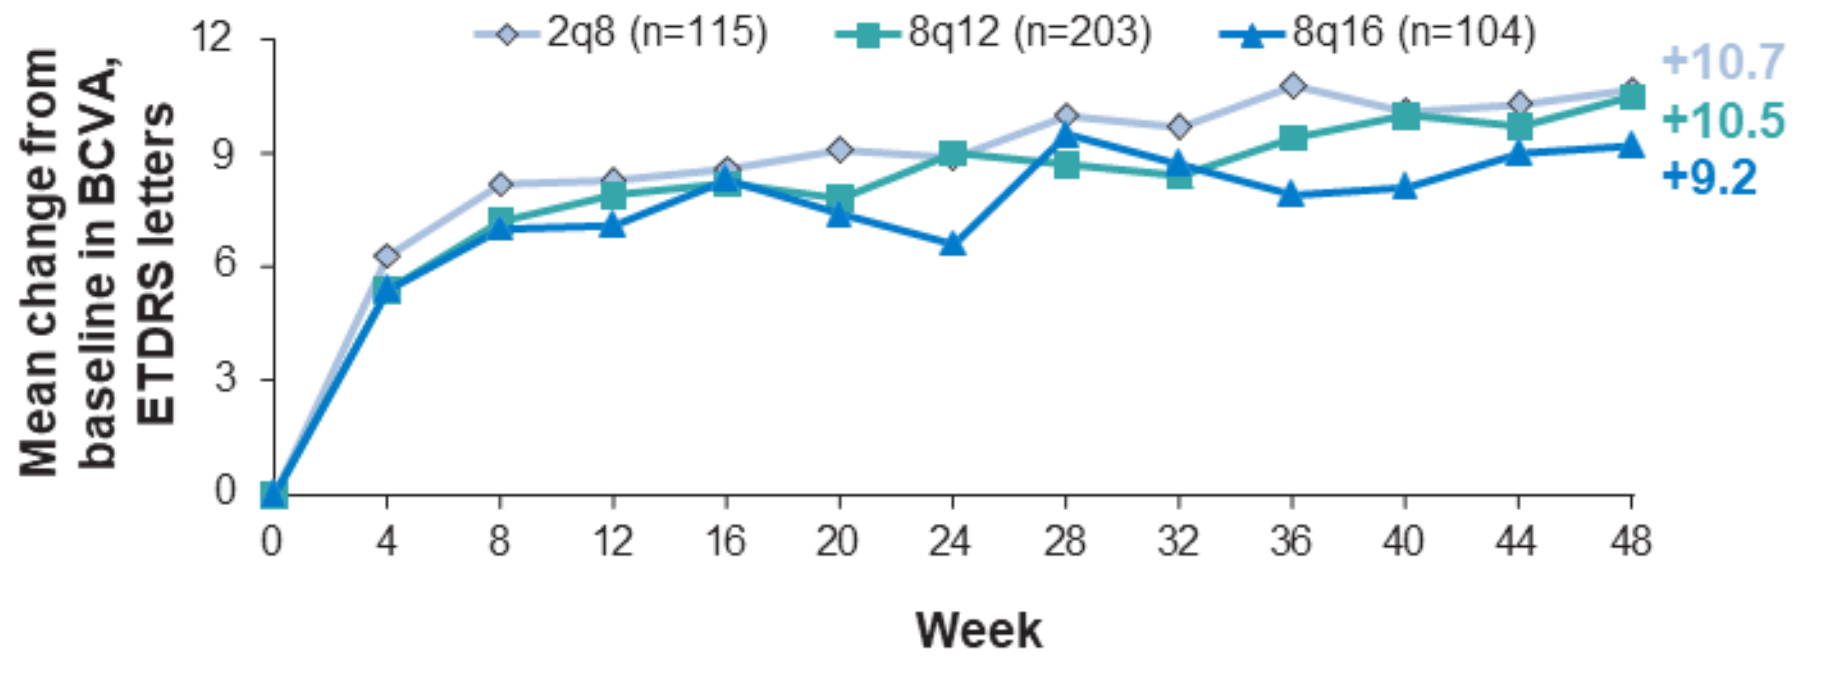
2q8 (n=115)
8q12 (n=203)
8q16 (n=104)
Mean change from baseline in BCVA, ETDRS letters
Week
+10.7
+10.5
+9.2

(B) Baseline BCVA 20/40 or Better

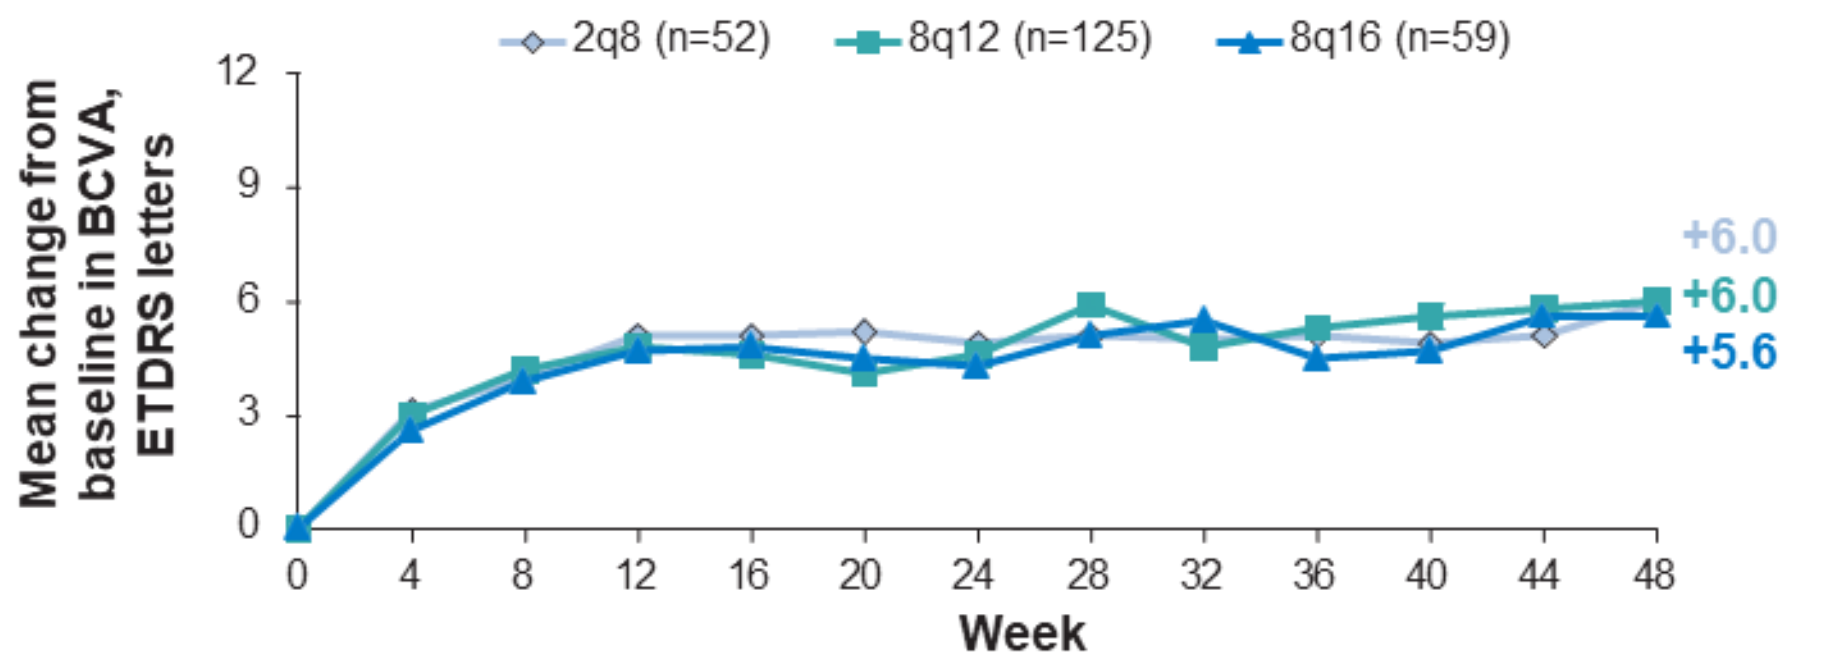
2q8 (n=52)
8q12 (n=125)
8q16 (n=59)
Mean change from baseline in BCVA, ETDRS letters
Week
+6.0
+6.0
+5.6

FAS, observed cases.

Figure 3. Mean Change From Baseline in CRT Through Week 48 by Baseline BCVA

(A) Baseline BCVA 20/50 or Worse

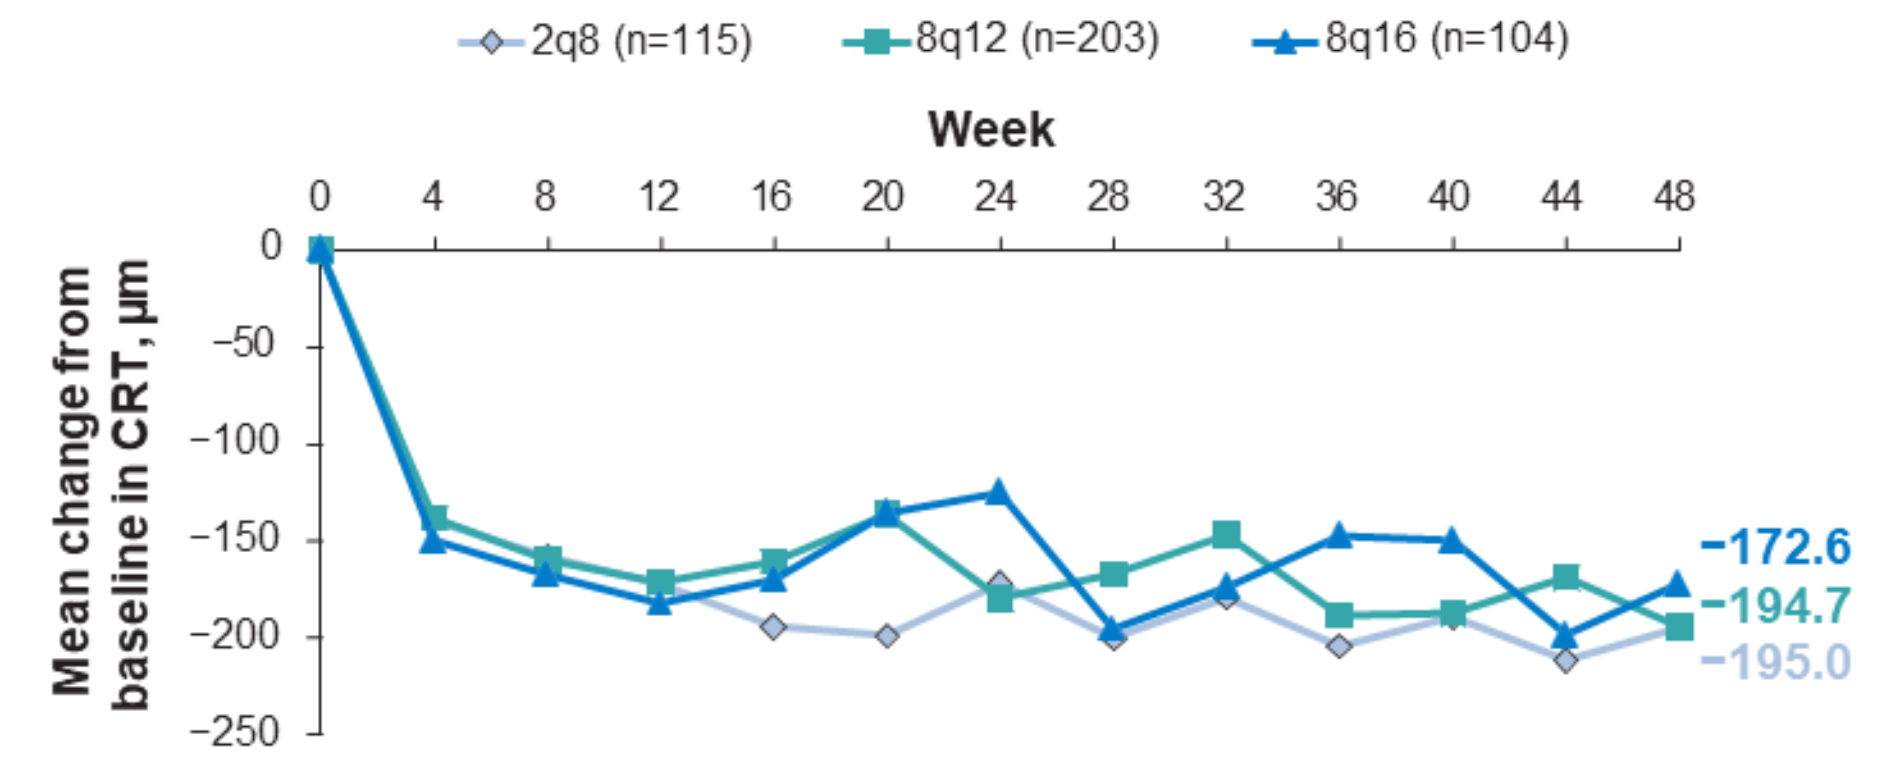
2q8 (n=115)
8q12 (n=203)
8q16 (n=104)
Week
Mean change from baseline in CRT, µm
−172.6
−194.7
−195.0

(B) Baseline BCVA 20/40 or Better

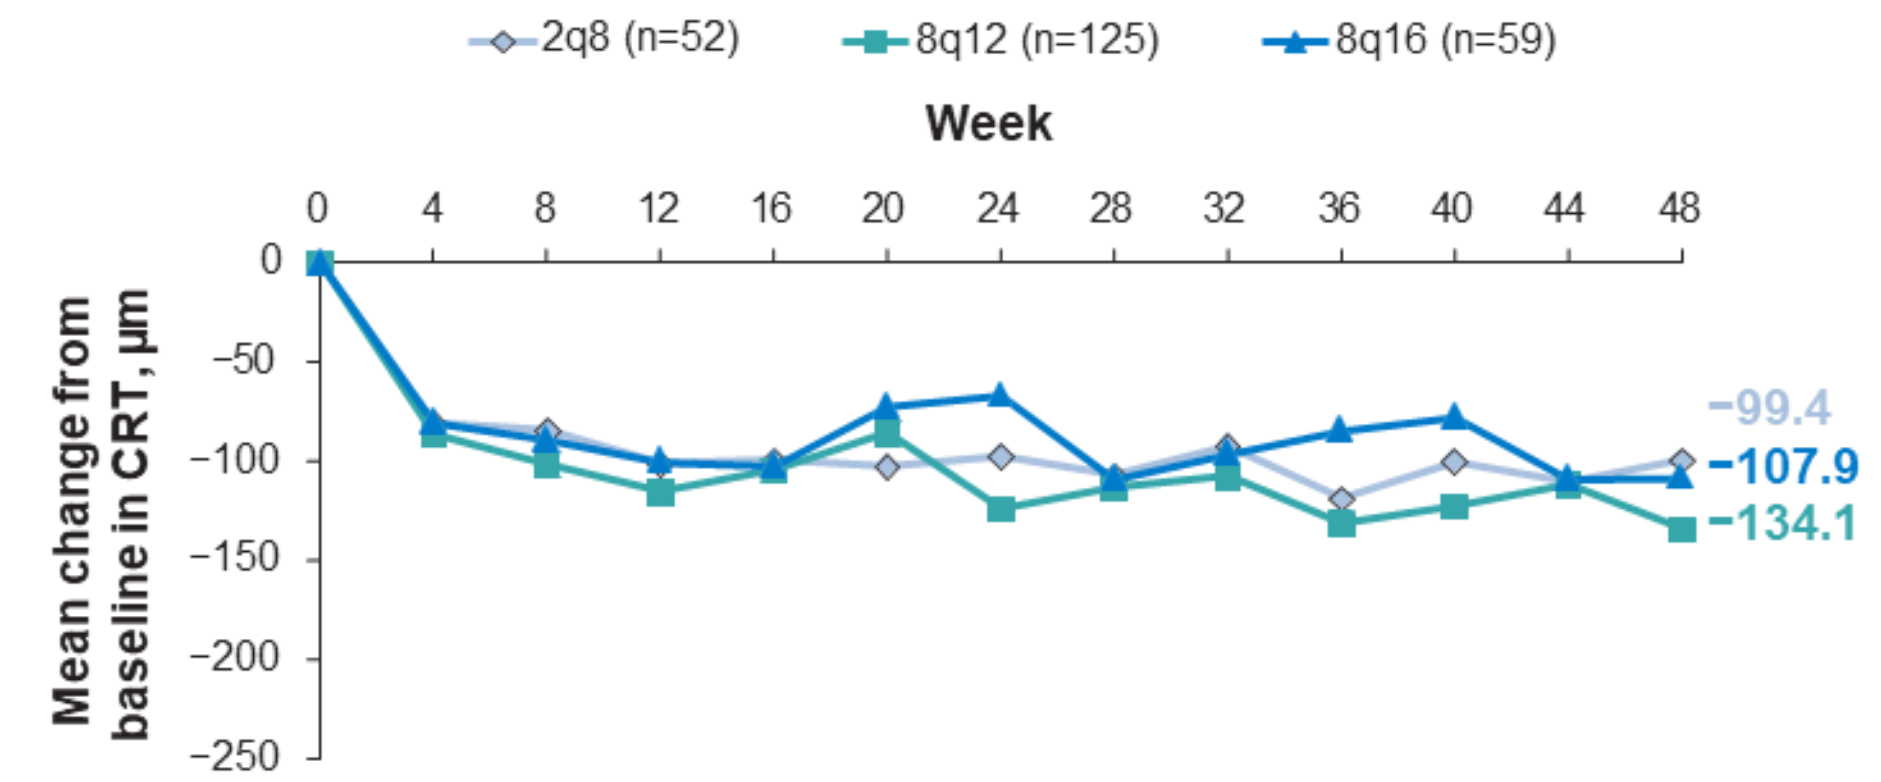
2q8 (n=52)
8q12 (n=125)
8q16 (n=59)
Week
Mean change from baseline in CRT, µm
−99.4
−107.9
−134.1

FAS, observed cases.

Figure 4. Treatment Exposure Through Week 48 by Baseline BCVA

(A) Baseline BCVA 20/50 or Worse

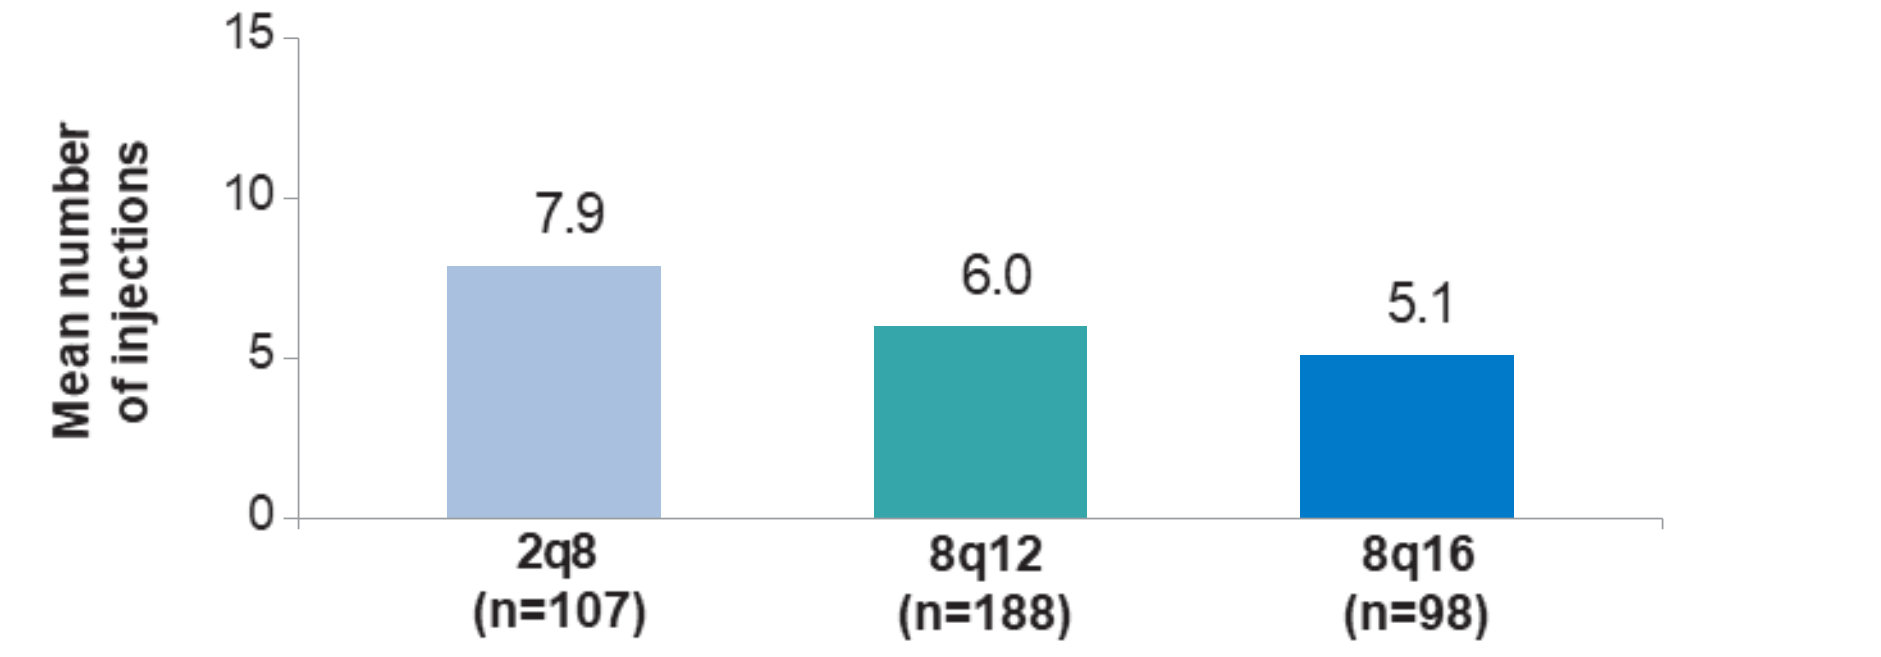
Mean number of injections
7.9
6.0
5.1
2q8 (n=107)
8q12 (n=188)
8q16 (n=98)

(B) Baseline BCVA 20/40 or Better

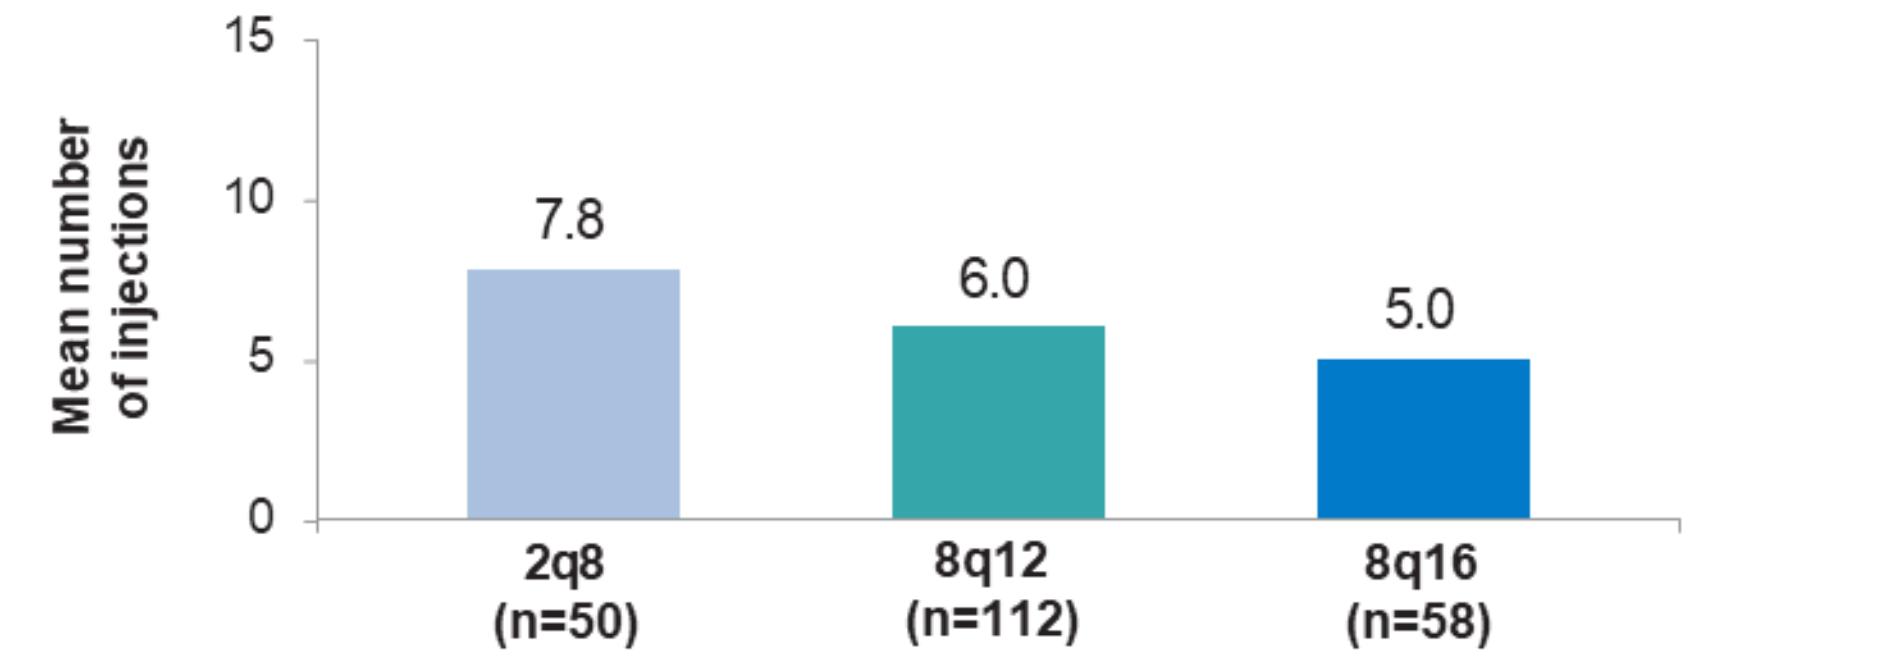
Mean number of injections
7.8
6.0
5.0
2q8 (n=50)
8q12 (n=112)
8q16 (n=58)

FAS, patients who completed Week 48.

- A large majority of aflibercept 8 mg-treated patients in each subgroup maintained their randomized dosing intervals through Week 48 with fewer injections than aflibercept 2 mg-treated patients (**Figure 4** and **Figure 5**)

Figure 5. Proportion of Aflibercept 8 mg-treated Patients Who Maintained Their Randomized Dosing Intervals by Baseline BCVA

(A) Baseline BCVA 20/50 or Worse

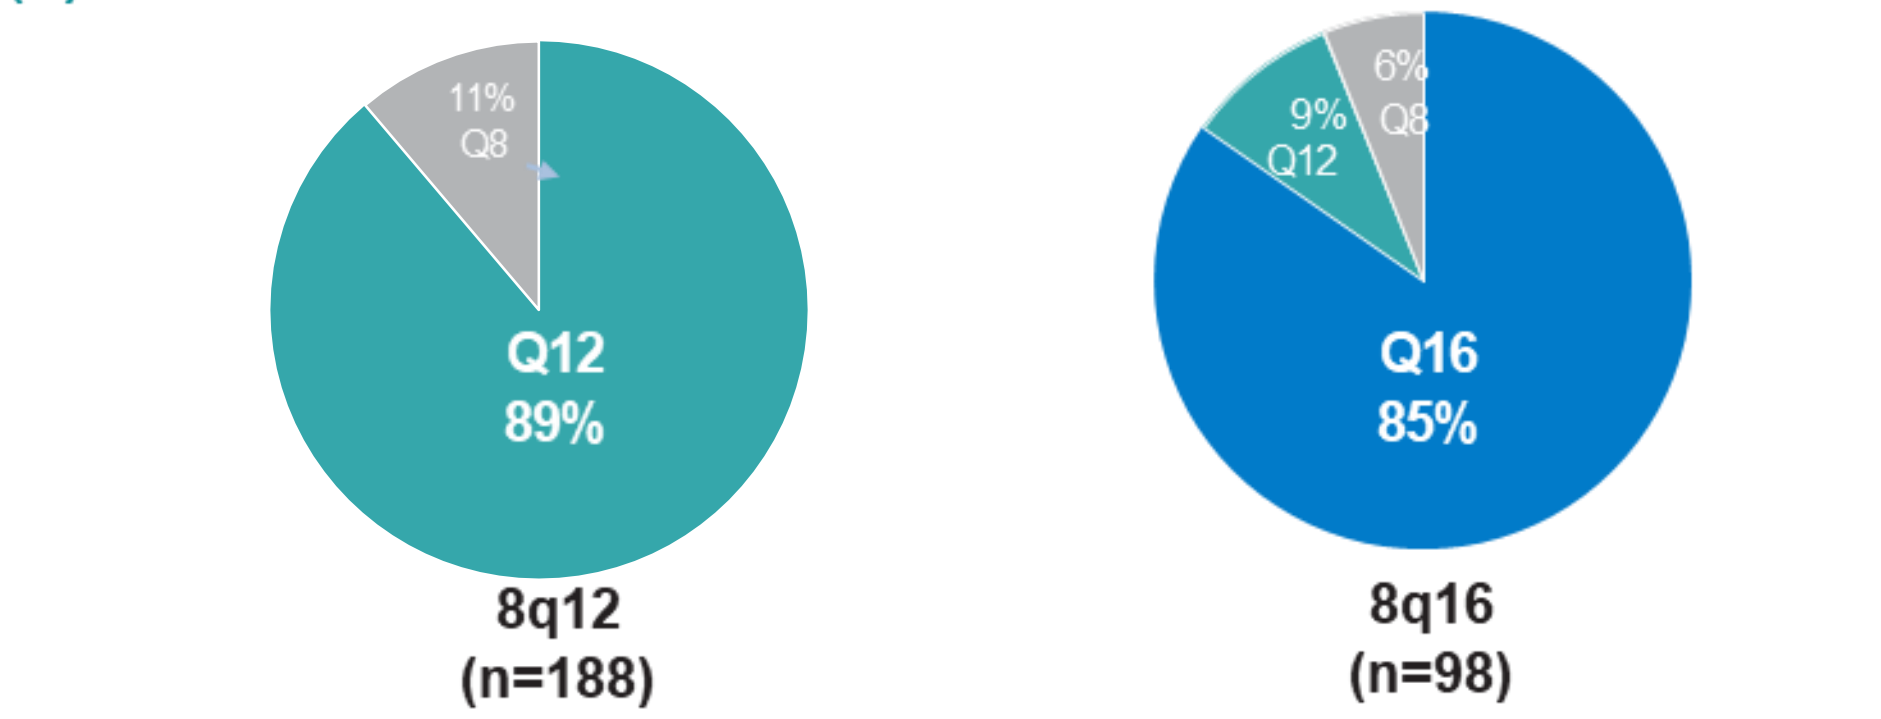
11% Q8
Q12 89%
8q12 (n=188)
6% Q8
9% Q12
Q16 85%
8q16 (n=98)

(B) Baseline BCVA 20/40 or Better

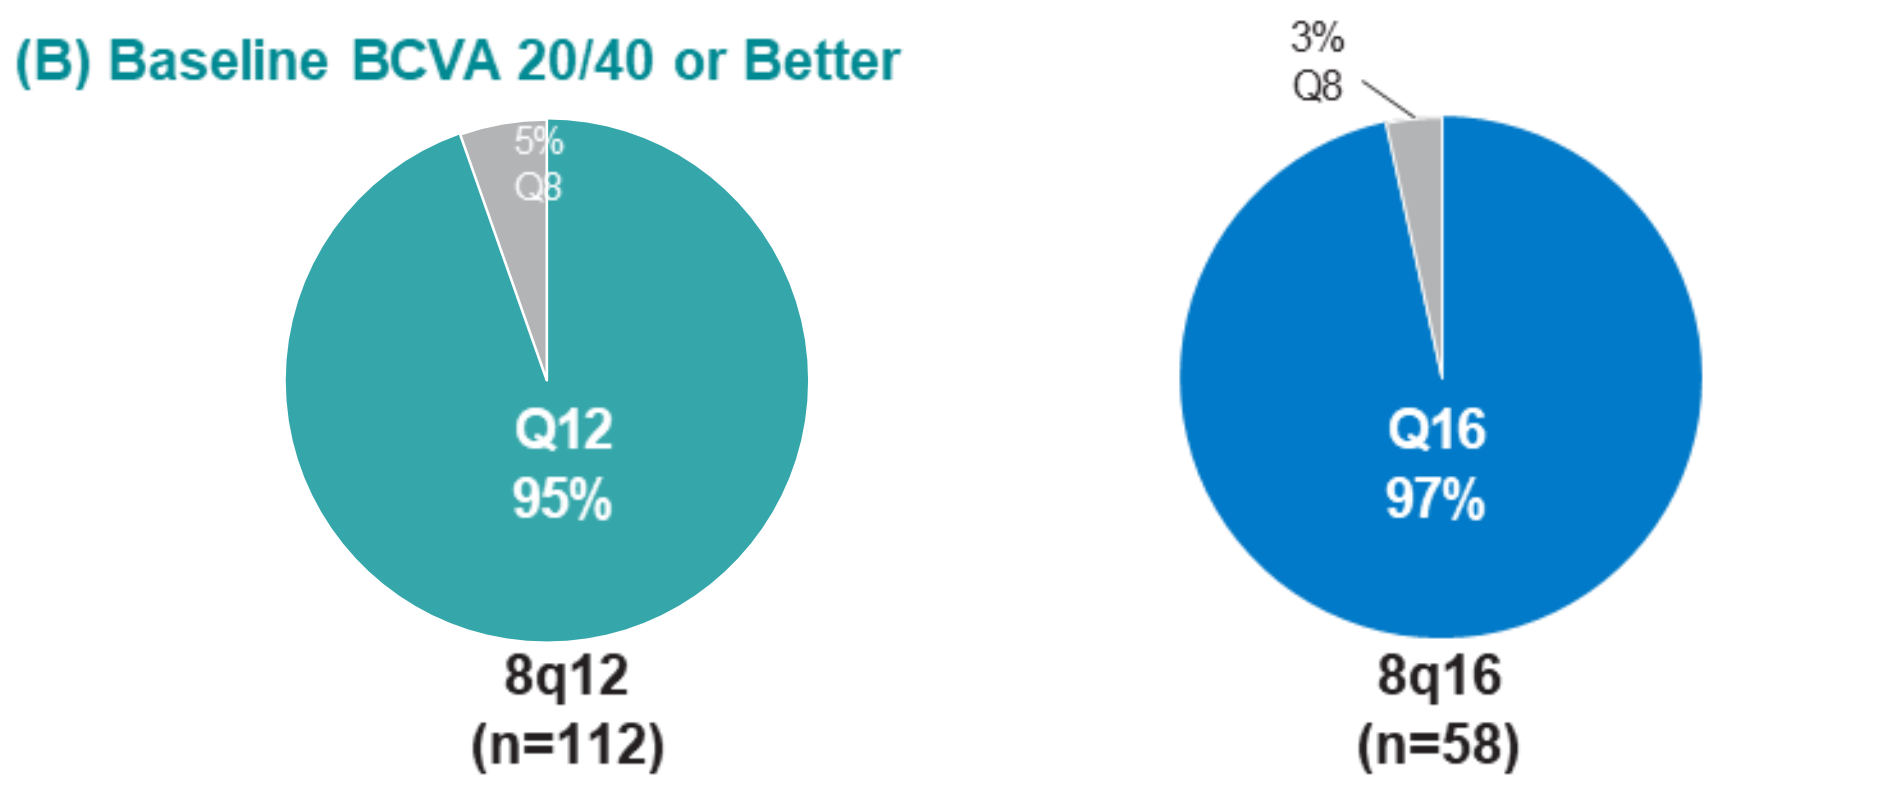
5% Q8
Q12 95%
8q12 (n=112)
3% Q8
Q16 97%
8q16 (n=58)

FAS, patients who completed Week 48.

CONCLUSIONS

- Aflibercept 8 mg demonstrated meaningful visual and anatomic improvements from baseline to Week 48 in patients with DME with fewer injections than aflibercept 2 mg, irrespective of baseline BCVA
 - As expected, patients with baseline BCVA 20/50 or worse achieved numerically greater improvements from baseline in BCVA and CRT than those with baseline BCVA 20/40 or better with both aflibercept 8 mg and 2 mg
- Greater proportions of aflibercept 8 mg-treated patients with baseline BCVA 20/40 or better versus 20/50 or worse maintained their randomized dosing intervals through Week 48

ACKNOWLEDGMENTS & DISCLOSURES

- Dr. Adrean received grant support from Adverum, Alexion, Amgen, Apellis, Genentech, Iveric Bio, Kodiak Life Sciences, NGM Pharmaceuticals, Opthea, Regeneron Pharmaceuticals, Inc., REGENXBIO, and Rezolute; served as a consultant/advisor for Adverum, Alimera, Genentech, and REGENXBIO; and holds stock/equity in Apellis
- The PHOTON study was sponsored by Regeneron Pharmaceuticals, Inc. (Tarrytown, NY) and co-funded by Bayer AG (Leverkusen, Germany). This analysis was funded by Regeneron Pharmaceuticals, Inc. (Tarrytown, NY). The sponsor participated in the design and conduct of the analysis, interpretation of the data, and preparation of this presentation
- Medical writing support was provided by Disha Patel, PhD, and Stephanie, Agbu, PhD, Regeneron Pharmaceuticals, Inc. Editorial support was provided by Core (a division of Prime, London, UK), funded by Regeneron Pharmaceuticals, Inc. according to Good Publication Practice guidelines

REFERENCE

1. Brown DM et al. *Lancet.* 2024;403:1153–1163.

Poster presented by Sean Adrean, MD, email: seadrean@yahoo.com, at the ARVO 2024 Meeting, May 5-9, 2024