Impact of Baseline Central Retinal Thickness on Vision Among Patients with Diabetic Macular Edema: Post Hoc Analysis of the Phase 2/3 PHOTON Trial

6219-B0129

Deepak Sambhara, MD, on behalf of the PHOTON study investigators

Eye Clinic of Wisconsin, Wausau, Wisconsin

BACKGROUND & PURPOSE

- Aflibercept is a fully human recombinant fusion protein that binds vascular endothelial growth factor (VEGF)-A, VEGF-B, and placental growth factor, thereby inhibiting the activation of cognate VEGF receptors[1,2]
- Aflibercept 8 mg, a novel intravitreal formulation that delivers a 4-times higher molar dose than aflibercept 2 mg, has demonstrated improved functional and anatomic outcomes at dosing intervals of ≥12 weeks[3-5]
- In the PHOTON study, aflibercept 8 mg demonstrated non-inferiority in the mean change in best-corrected visual acuity (BCVA) at Week 48 and a comparable safety profile versus aflibercept 2 mg with fewer injections at Week 48 in patients with DME, including those with more severe disease at baseline[5]
- This post hoc analysis evaluated the effect of aflibercept 8 mg versus 2 mg on clinical outcomes in patients with DME through Week 48 based on disease severity, as defined by baseline central retinal thickness (CRT)

METHODS

- In PHOTON, eligible patients with DME were randomized 1:2:1 to receive aflibercept 8 mg every 12 or 16 weeks after 3 initial monthly doses (8q12 or 8q16) or aflibercept 2 mg every 8 weeks after 5 initial monthly doses (2q8) (**Figure 1**)
- Starting at Week 16, patients in the aflibercept 8q12 and 8q16 groups had their randomized dosing intervals shortened if dose regimen modification (DRM) criteria were met

Figure 1. PHOTON Study Design

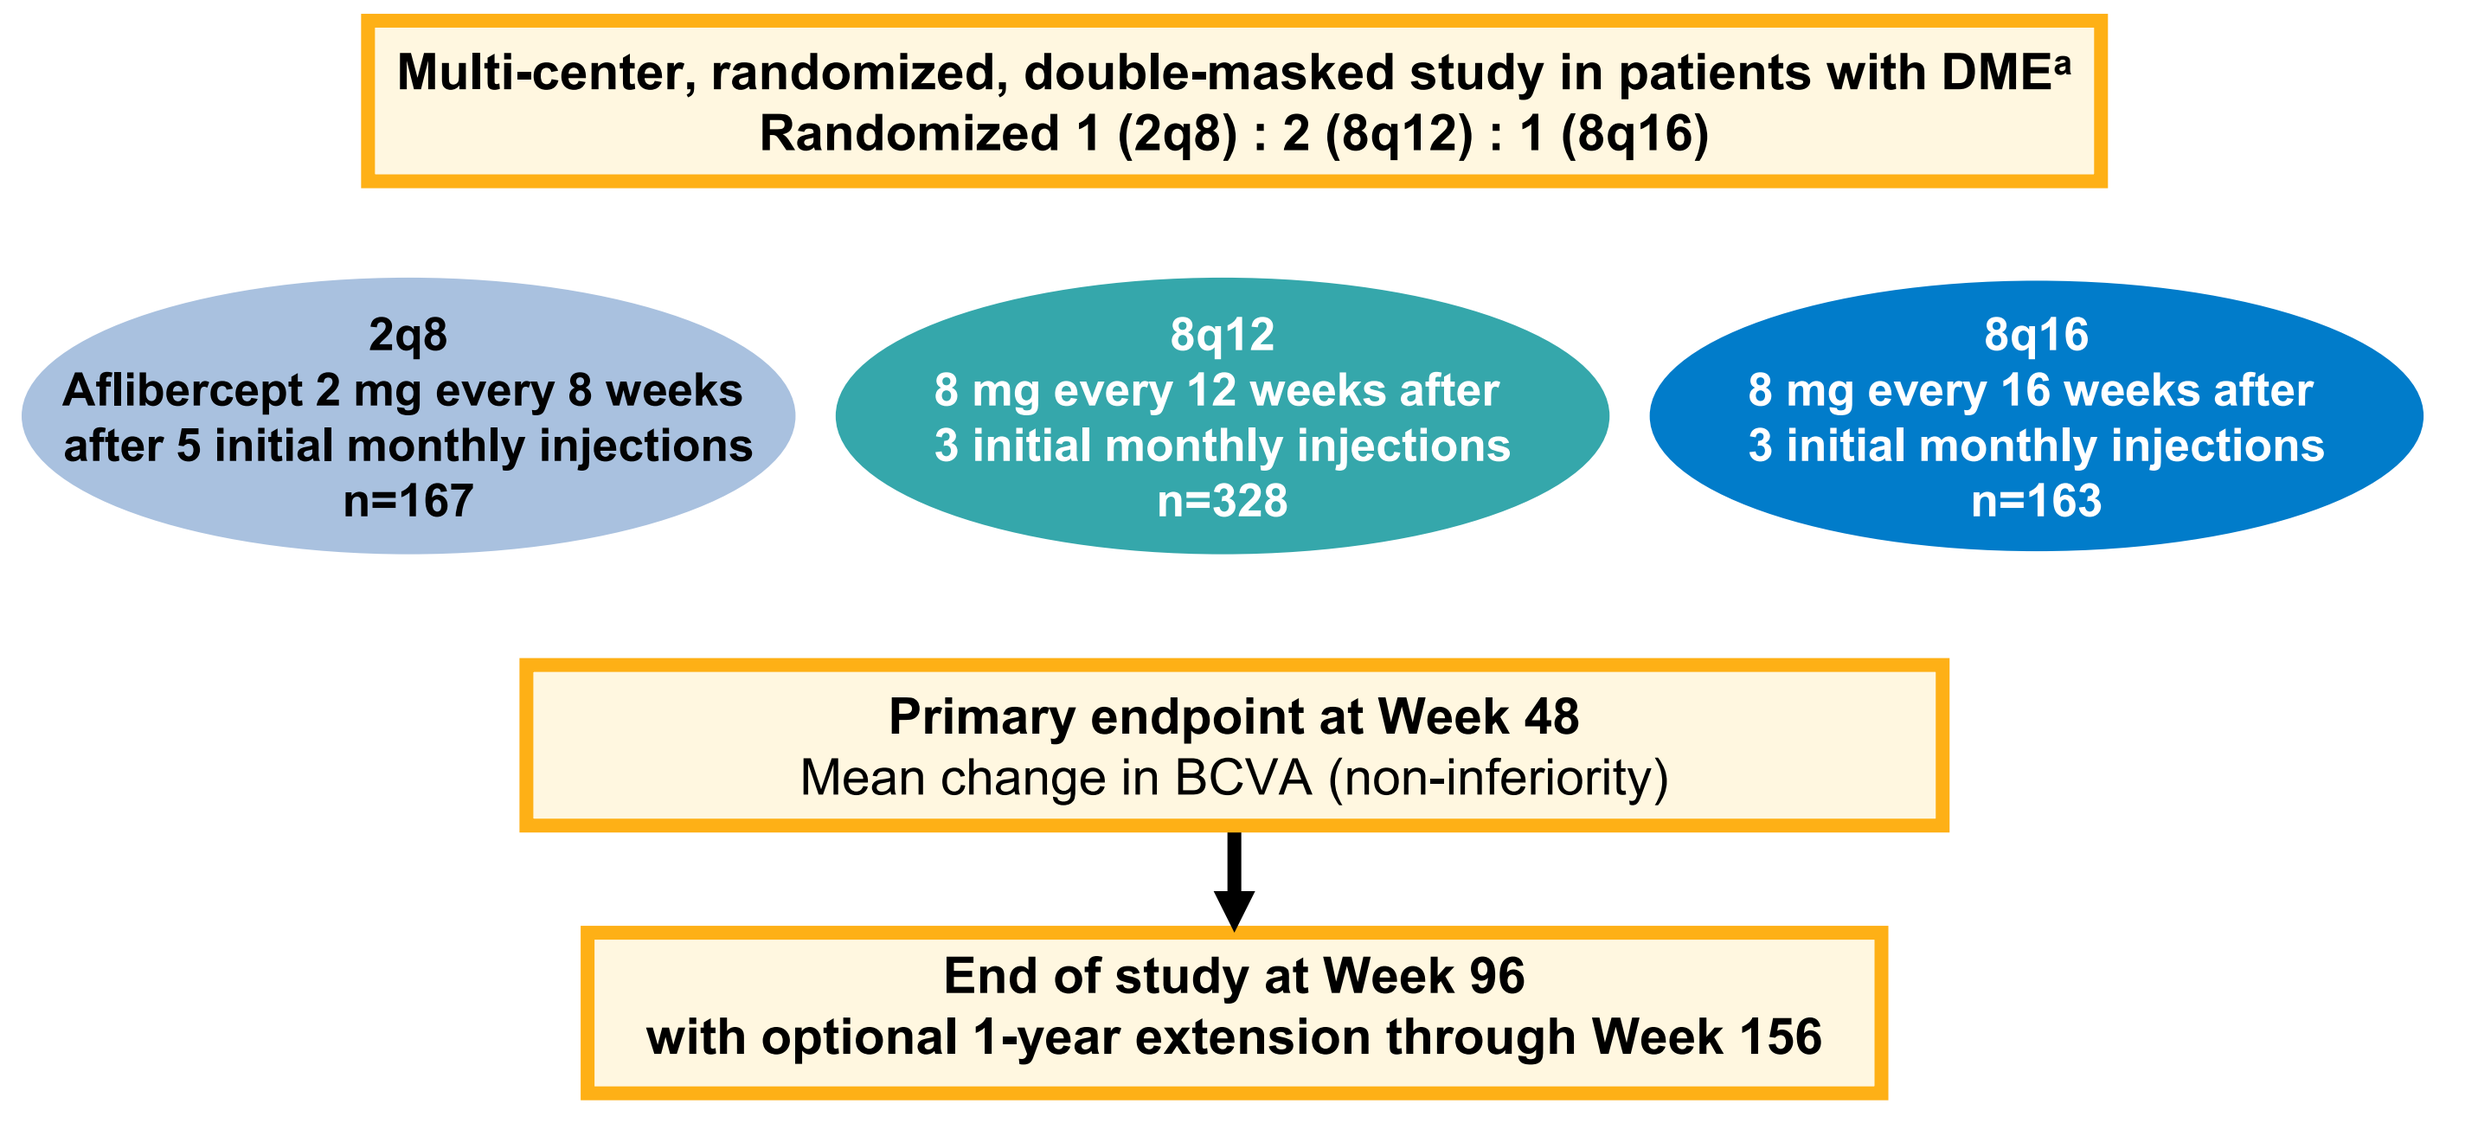
Multi-center, randomized, double-masked study in patients with DME[a]
Randomized 1 (2q8) : 2 (8q12) : 1 (8q16)
2q8
Aflibercept 2 mg every 8 weeks after 5 initial monthly injections
n=167
8q12
8 mg every 12 weeks after 3 initial monthly injections
n=328
8q16
8 mg every 16 weeks after 3 initial monthly injections
n=163
Primary endpoint at Week 48
Mean change in BCVA (non-inferiority)
End of study at Week 96
with optional 1-year extension through Week 156

Note: The 2-mg arm received 5 initial monthly injections vs 8-mg arms which received only 3 initial monthly injections.
[a]Treatment naive and previously treated.

Post hoc analysis

- This post hoc analysis evaluated the effect of 8q12 and 8q16 versus 2q8 on BCVA and CRT through Week 48 by disease severity, as defined by baseline CRT quartiles (quartile [Q]1: ≤360 μm; Q2: ≥361-≤430 μm; Q3: ≥431-≤528 μm; Q4: >528 μm)
- All analyses were descriptive, and 1 patient was excluded due to missing baseline CRT
- Outcomes assessed were mean change in BCVA and CRT through Week 48, and the proportion of patients who maintained their original randomized dosing intervals through Week 48

RESULTS

Table 1. Baseline Characteristics and Treatment Exposure to Week 48 by Baseline CRT Quartiles

	Q1: ≤360 μm (n=167)			Q2: ≥361-≤430 μm (n=163)			Q3: ≥431-≤528 μm (n=163)			Q4: >528 μm (n=164)		
	2q8 (n=47)	8q12 (n=85)	8q16 (n=35)	2q8 (n=39)	8q12 (n=78)	8q16 (n=46)	2q8 (n=36)	8q12 (n=92)	8q16 (n=35)	2q8 (n=45)	8q12 (n=72)	8q16 (n=47)
Age, years	63.3 (10.7)	61.7 (10.8)	62.9 (9.5)	64.1 (8.7)	63.9 (10.8)	62.5 (9.1)	63.9 (8.5)	62.0 (9.9)	60.4 (9.8)	61.2 (10.6)	60.8 (13.2)	61.4 (9.8)
Male, n (%)	28 (59.6)	56 (65.9)	21 (60.0)	17 (43.6)	47 (60.3)	26 (56.5)	18 (50.0)	51 (55.4)	22 (62.9)	29 (64.4)	55 (76.4)	30 (63.8)
Duration of diabetes, years	18.2 (11.6)	15.3 (9.6)	18.9 (12.5)	16.8 (9.8)	16.6 (11.1)	14.4 (10.1)	14.1 (9.31)	14.3 (9.4)	14.9 (9.0)	14.3 (8.8)	14.2 (9.7)	15.1 (10.7)
BCVA, ETDRS letters	64.8 (9.9)	66.6 (7.8)	68.4 (7.1)	63.1 (10.6)	66.1 (10.1)	64.0 (11.3)	61.3 (9.8)	64.0 (8.2)	62.4 (11.3)	56.7 (12.8)	57.4 (11.5)	53.1 (10.8)
CRT, μm	320.0 (22.1)	318.7 (26.4)	326.1 (23.9)	390.3 (18.6)	391.6 (21.3)	394.2 (19.4)	475.3 (32.5)	475.0 (29.1)	479.3 (28.5)	644.2 (128.2)	632.4 (114.8)	610.9 (77.5)
Mean number of injections to Week 48	7.6	5.7	5.0	7.6	5.7	4.9	7.9	5.7	5.0	7.8	5.8	4.9

FAS, observed cases.
Unless otherwise specified, values shown represent mean (SD).
ETDRS, Early Treatment of Diabetic Retinopathy Study; FAS, full analysis set; SD, standard deviation.

- Across increasing baseline CRT quartiles, mean age and proportion of males were similar, while mean BCVA decreased (**Table 1**)
- Patients treated with aflibercept 8 mg received, on average, up to 3 fewer injections versus those treated with aflibercept 2 mg in all quartiles, with similar exposure across all quartiles

Figure 2. Mean BCVA and CRT Through Week 48 by Baseline CRT Quartiles

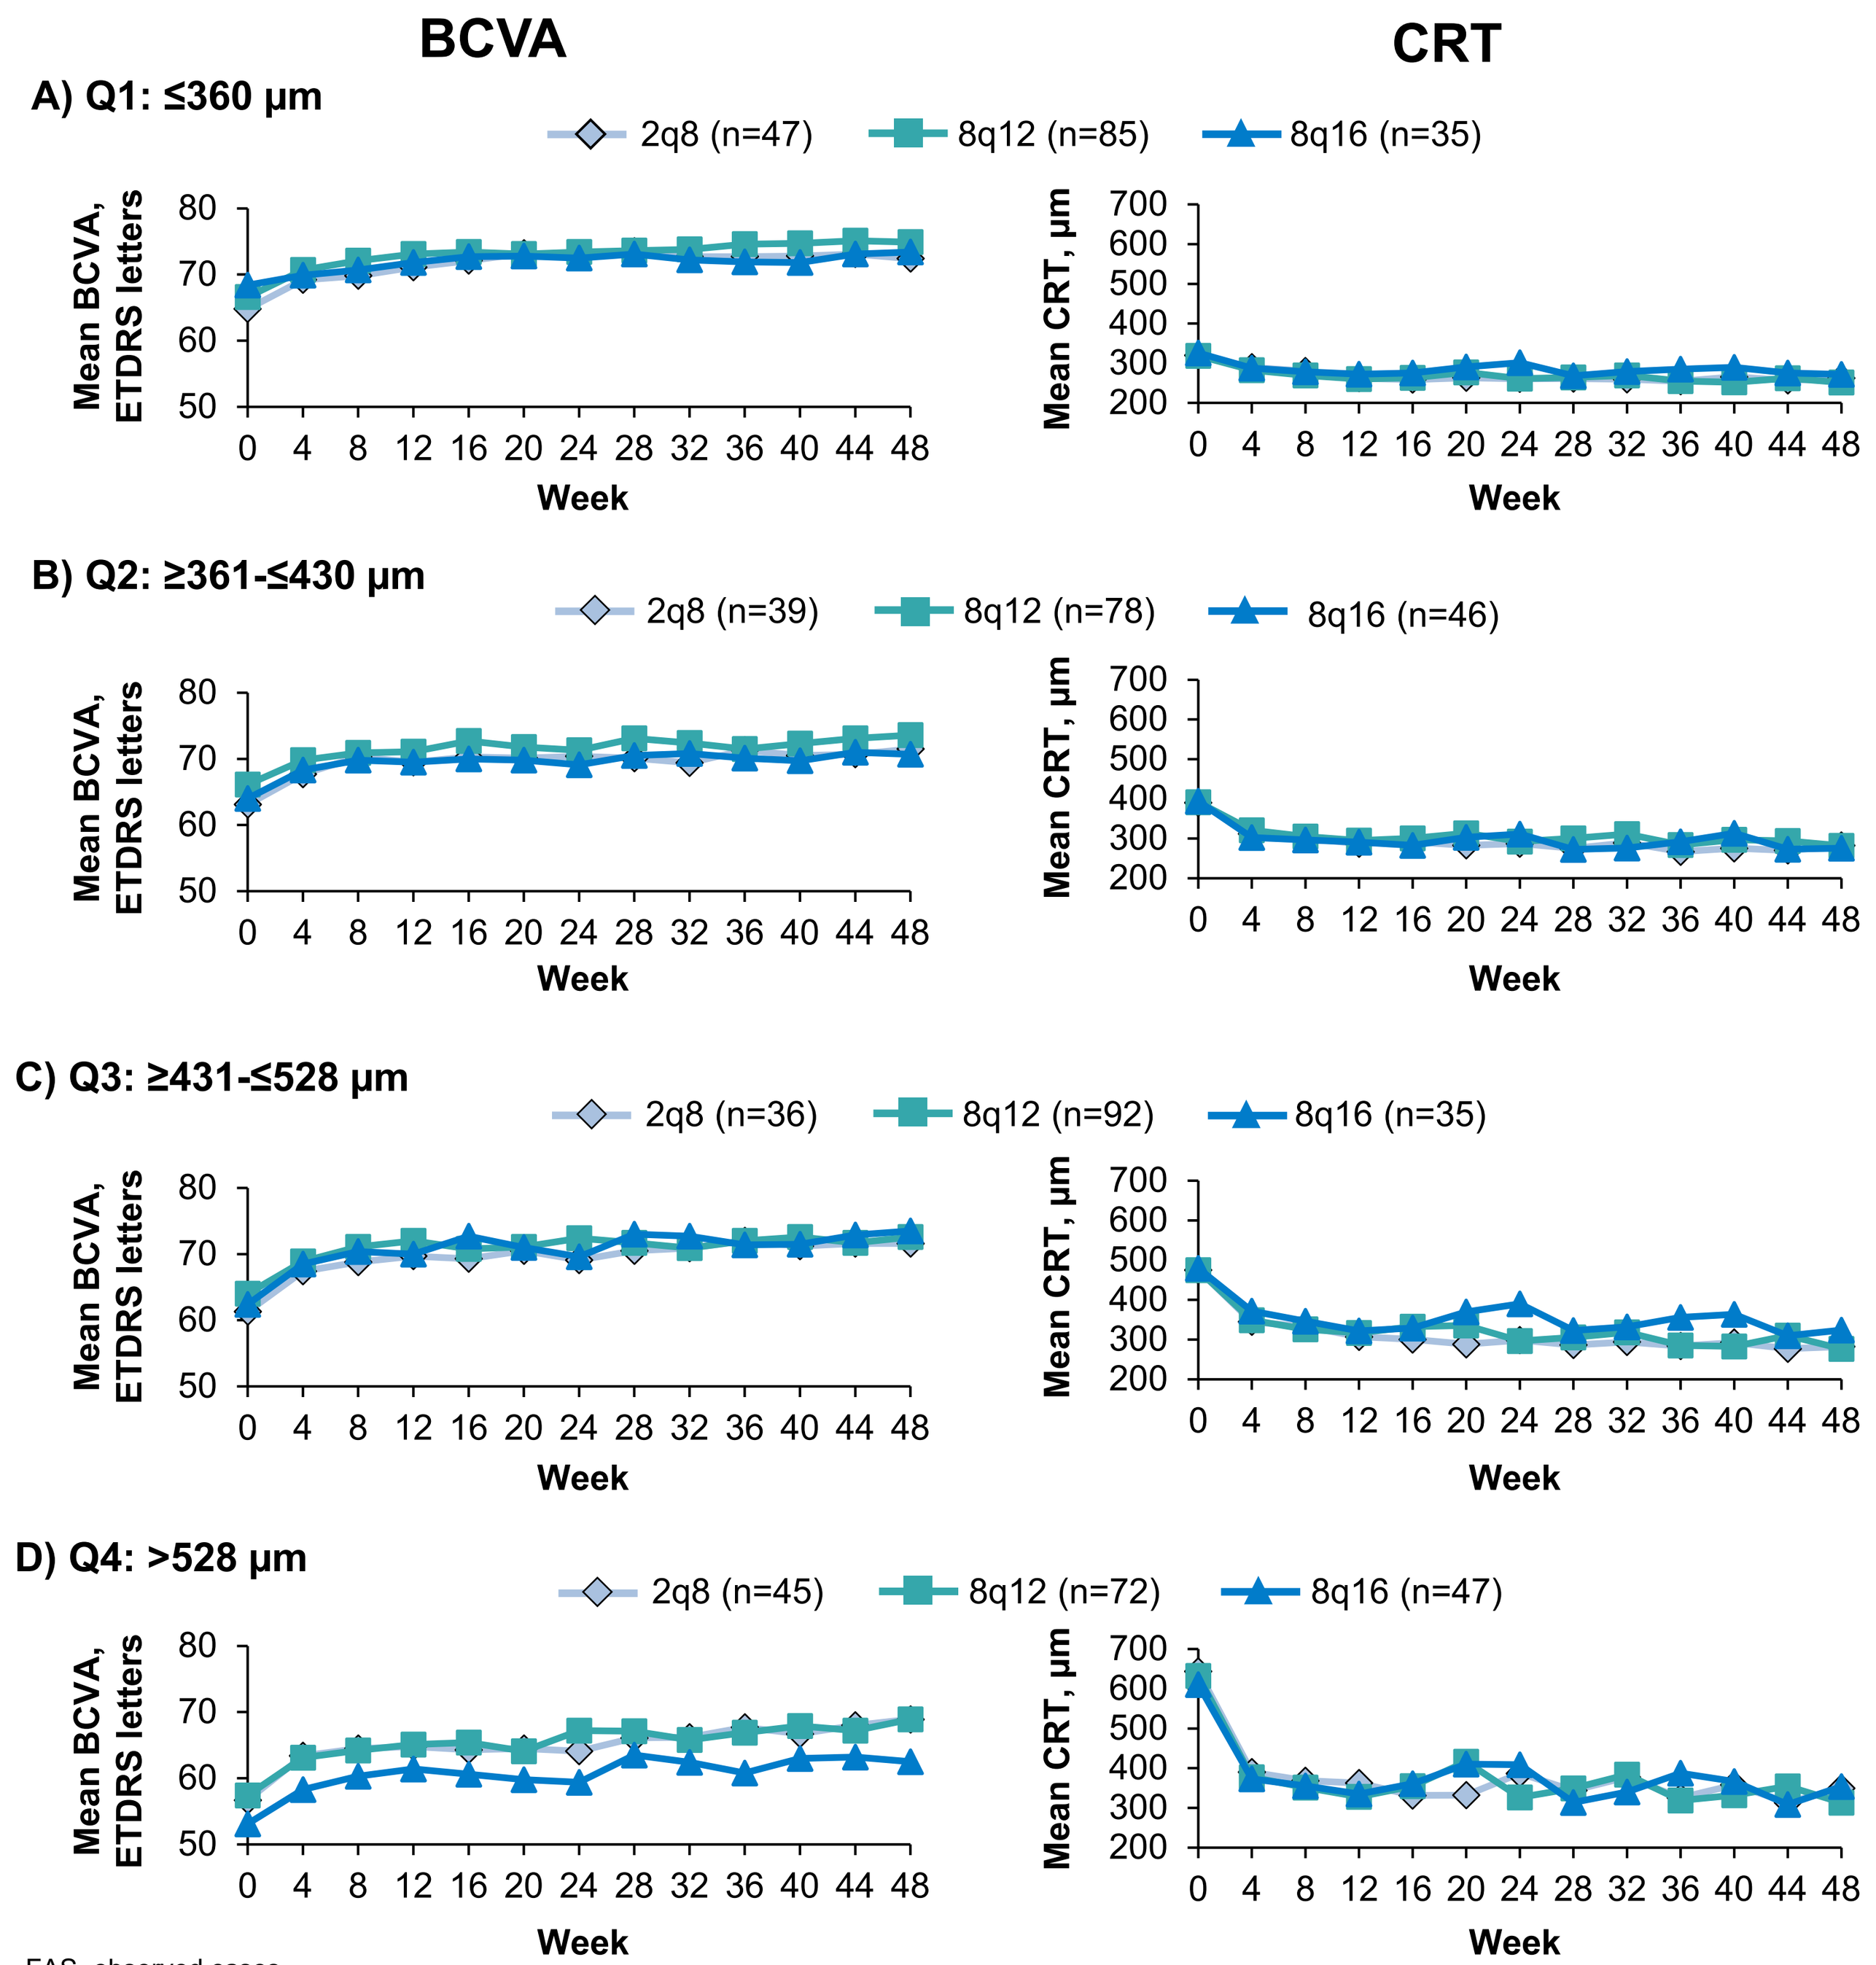
BCVA
CRT
A) Q1: ≤360 μm
2q8 (n=47)
8q12 (n=85)
8q16 (n=35)
B) Q2: ≥361-≤430 μm
2q8 (n=39)
8q12 (n=78)
8q16 (n=46)
C) Q3: ≥431-≤528 μm
2q8 (n=36)
8q12 (n=92)
8q16 (n=35)
D) Q4: >528 μm
2q8 (n=45)
8q12 (n=72)
8q16 (n=47)
Mean BCVA, ETDRS letters
Mean CRT, μm
Week

FAS, observed cases.

- In Q1 and Q2, patients in the 8q12 and 8q16 groups achieved similar BCVA and CRT through Week 48 as those in the 2q8 group, despite receiving, on average, up to 3 fewer injections (**Figure 2A and B**)
- In Q3 and Q4, comparable anatomic outcomes were observed in the 8q12 and 2q8 groups whereas some fluctuations were observed in the 8q16 group (**Figure 2C and D**)
- In Q4, patients in the 8q12 versus 2q8 group achieved comparable mean BCVA through Week 48, despite receiving an average of 2 fewer injections (**Figure 2D**)
- Patients in Q4 treated with 8q16 had a numerically lower BCVA at baseline than those treated with 8q12 and 2q8, and BCVA remained relatively lower at each visit
 - However, the change in BCVA through Week 48 with 8q16 was comparable to what was observed with 2q8

Figure 3. Fluid Reaccumulation with Aflibercept 8 mg Versus 2 mg in Eyes with Baseline CRT >528 μm 8 Weeks Following the Last Initial Monthly Dose

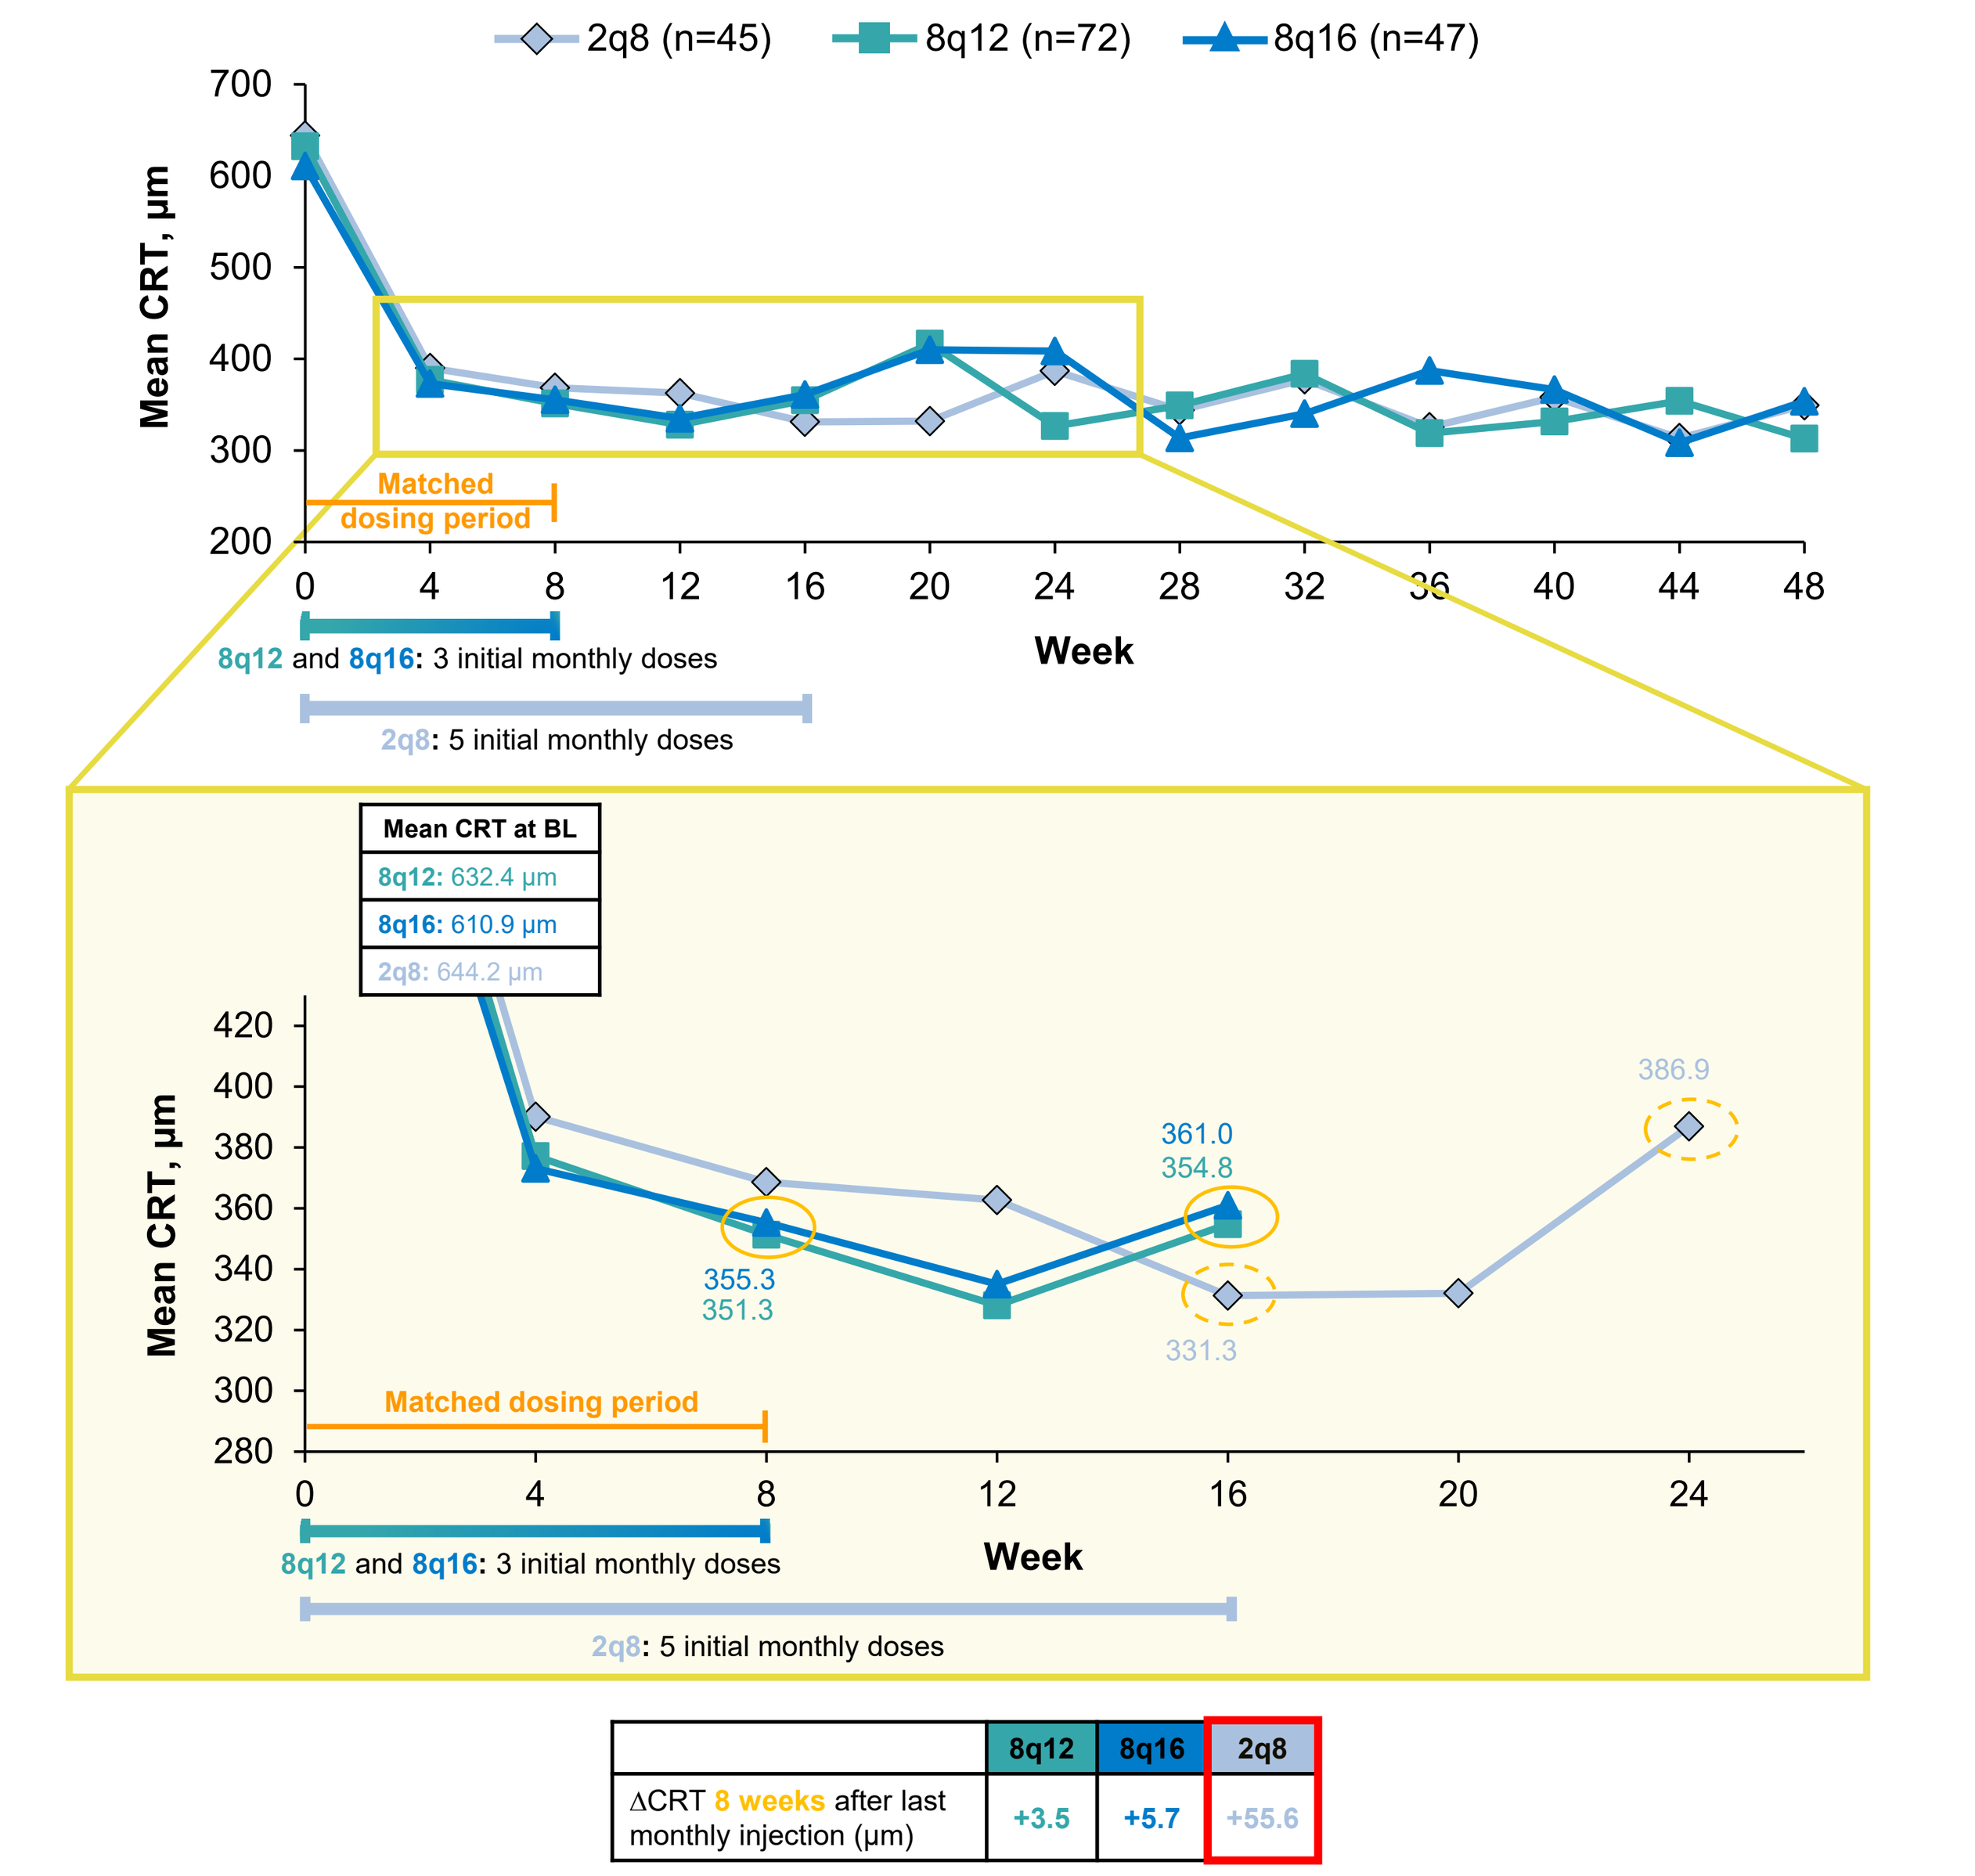
2q8 (n=45)
8q12 (n=72)
8q16 (n=47)
Mean CRT, μm
Week
Matched dosing period
8q12 and 8q16: 3 initial monthly doses
2q8: 5 initial monthly doses
Mean CRT at BL
8q12: 632.4 μm
8q16: 610.9 μm
2q8: 644.2 μm
355.3
351.3
361.0
354.8
331.3
386.9

	8q12	8q16	2q8
ΔCRT 8 weeks after last monthly injection (μm)	+3.5	+5.7	+55.6

The yellow box on the upper panel indicates the magnified region of the graph shown in the lower panel. During the matched dosing period, patients in all groups received monthly dosing with aflibercept as part of the initial dosing period. Orange circles highlight CRT values at the last monthly dose, and 8 weeks after the last initial monthly dose in the 8q12 and 8q16 groups (solid circles) and 2q8 group (dashed circles).
FAS, observed cases.
BL, baseline.

- In Q4, mean CRT for eyes in the 8q12 and 8q16 groups after 3 initial monthly doses was 351.3 μm and 355.3 μm, respectively; mean CRT for eyes in the 2q8 group after 5 initial monthly doses was 331.3 μm (**Figure 3**)
- In Q4, 8 weeks after the last initial monthly dose, a sharp increase in CRT was observed with 2q8 with a difference from the last initial monthly dose of 55.6 μm versus 3.5 μm and 5.7 μm with 8q12 and 8q16, respectively, suggesting slower reaccumulation with aflibercept 8 mg versus 2 mg in this subgroup

Figure 4. Randomized Dosing Intervals with Aflibercept 8 mg Through Week 48 by Baseline CRT Quartiles

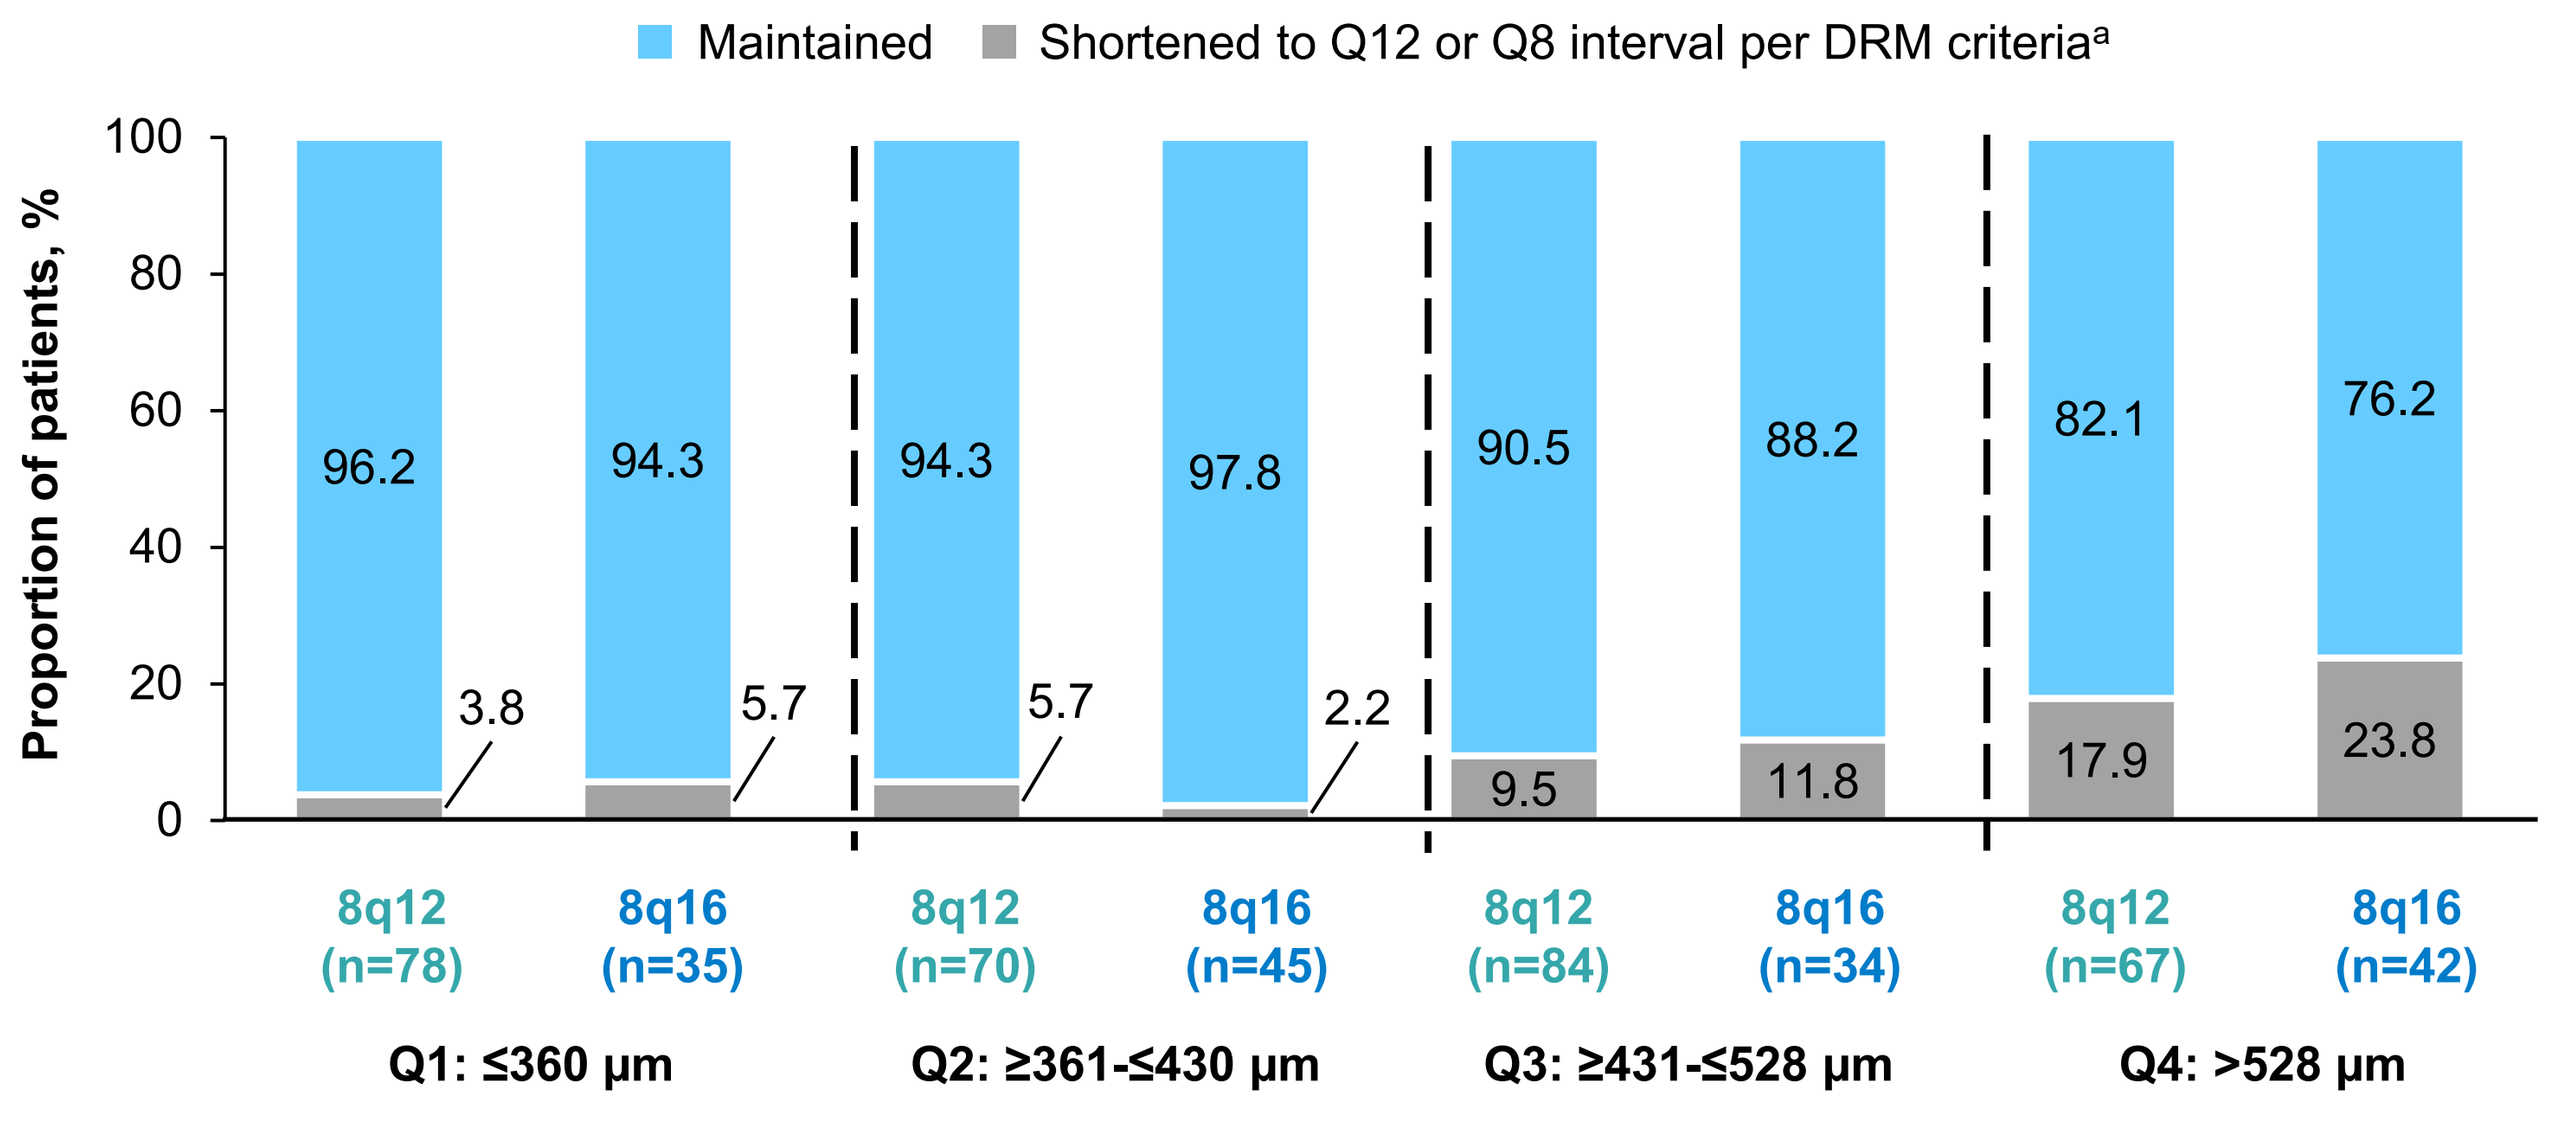
Maintained
Shortened to Q12 or Q8 interval per DRM criteria[a]
Proportion of patients, %
96.2
3.8
94.3
5.7
94.3
5.7
97.8
2.2
90.5
9.5
88.2
11.8
82.1
17.9
76.2
23.8
8q12 (n=78)
8q16 (n=35)
8q12 (n=70)
8q16 (n=45)
8q12 (n=84)
8q16 (n=34)
8q12 (n=67)
8q16 (n=42)
Q1: ≤360 μm
Q2: ≥361-≤430 μm
Q3: ≥431-≤528 μm
Q4: >528 μm

FAS, patients who completed Week 48.
[a]Dosing intervals of patients who met study-specified DRM criteria for interval shortening (loss of >10 letters from Week 12 due to persistent or worsening DME and >50-μm increase in CRT from Week 12) at prespecified timepoints were shortened to either Q12 or Q8 through Week 48.

- Consistent with the results for the overall population,[5] most patients in the 8-mg groups across the quartiles maintained their randomized dosing intervals through Week 48 irrespective of baseline CRT (**Figure 4**)
 - Although the number of patients in each treatment group within each quartile was relatively low, an increasing proportion of patients with shortened dosing intervals was observed with increasing disease severity

CONCLUSIONS

- Aflibercept 8 mg demonstrated meaningful visual and anatomic improvements in patients with DME at Week 48 across a wide range of baseline CRT values, with, on average, up to 3 fewer injections compared with aflibercept 2 mg
- In eyes with the most severe disease (Q4), fluid reaccumulation was numerically less 8 weeks after the third initial monthly dose with aflibercept 8 mg versus 2 mg after 5 initial monthly doses, suggesting a more durable treatment effect

ACKNOWLEDGMENTS & DISCLOSURES

- Dr Sambhara has served as an advisor for Alimera Sciences, Coherus Biosciences, Genentech, and Regeneron Pharmaceuticals, Inc.; participated in Speakers' Bureau for Apellis Pharmaceuticals, Regeneron Pharmaceuticals, Inc., and IvericBio; has served as a speaker for Heidelberg Engineering; and is a stockholder of Eclipse Life Sciences
- The PHOTON trial was sponsored by Regeneron Pharmaceuticals, Inc. (Tarrytown, NY) and Bayer AG (Leverkusen, Germany). This analysis was funded by Regeneron Pharmaceuticals, Inc. (Tarrytown, NY). The sponsor participated in the design and conduct of the study, analysis of the data, and preparation of this poster
- Medical writing support was provided by Disha Patel, PhD, and Kaitlyn Scacalossi, PhD, of Regeneron Pharmaceuticals, Inc., and Mahalia Gilmartin, PhD, of Core (a division of Prime, London, UK) funded by Regeneron Pharmaceuticals, Inc, in accordance with Good Publication Practice guidelines

REFERENCES

1. Holash J et al. *Proc Natl Acad Sci.* 2002;99(17):11393–11398.
2. Papadopoulos N et al. *Angiogenesis.* 2012;15(2):171–185.
3. Wykoff CC et al. *JAMA Ophthalmol.* 2023;141(9):834–842.
4. Lanzetta P et al. *Lancet.* 2024;403:1141–1152.
5. Brown DM et al. *Lancet.* 2024;403;1153–1163.
6. EYLEA® HD (aflibercept) injection, for intravitreal use. Highlights of prescribing information. Regeneron Pharmaceuticals, Inc.; 2023. Accessed September 14, 2023. https://www.regeneron.com/downloads/eyleahd_fpi.

Poster presented by Deepak Sambhara, MD, email: deepaksambhara@gmail.com, at the ARVO 2024 Meeting, May 5-9, 2024