Delphi study on the management of patients with Heart Failure reduced Ejection Fraction (HFrEF) in Spain: post- discharge follow-up of a patient with an episode of Worsening Heart Failure (WHF)

Authors: JI. Morgado Garcia-Polavieja[1] , A. Garcia Quintana[2] , J. Alvarez-Garcia[3], S. Ruiz-Bustillo[4]

Affiliations: [1] University Hospital of Virgen Macarena, Department of Cardiology, Virgen Macarena Hospital , Seville, Spain;[2] University Hospital Dr Negrín, Heart Failure Unit Department of Cardiology at Hospital Universitario de Gran Canaria Doctor Negrín., Las Palmas De Gran Canaria, Spain
[3] CIBER of Cardiovascular Diseases, Department of Cardiology at Ramón y Cajal University Hospital, Madrid, Spain; [4] Hospital del Mar, Cardiology. Heart Diseases Biomedical Research Group, Hospital del Mar Research Institute, Barcelona, Spain

Introduction and Purpose

- The clinical progression of HFrEF is characterized by periods of worsening symptoms and signs. These episodes are associated to a higher risk of hospitalizations, leading to a major burden on the healthcare system due to their frequency and prognostic impact. The period following hospital discharge is critical for the patient.
- This study aimed to evaluate the consensus among cardiologists to better understand their perception and management of HFrEF, and to identify the care process for such management.

Methodology

- A Scientific Committee (SC) of 4 experts in the management of HFrEF created and validated a Delphi questionnaire comprised of 257 statements, which was completed in two rounds by 79 cardiologists (Figure 1).
- Each statement was assessed by a Likert ordinal scale of 1 to 9 points (1 = Completely Disagree to 9 = Completely Agree).
- In this communication, it will be presented the results of post- discharge patient care management

Figure 1. Study design: Delphi methodology adapted in 2 rounds

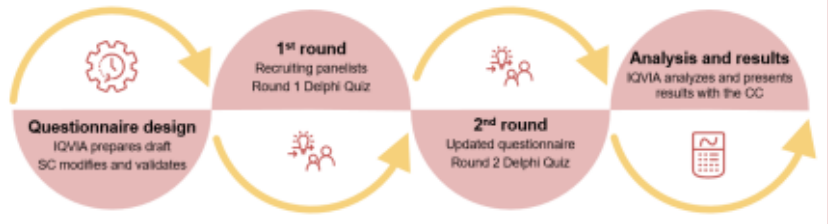
Questionnaire design
IQVIA prepares draft
SC modifies and validates
1st round
Recruiting panelists
Round 1 Delphi Quiz
2nd round
Updated questionnaire
Round 2 Delphi Quiz
Analysis and results
IQVIA analyzes and presents results with the CC

Results

- Responses were collected over two rounds, achieving consensus on 75% of the statements.
- In the first Delphi round, the defined level of agreement was reached for the 76 statements (63.3%), and in second round, 44 statements that did not reach consensus were revisited, with agreement achieved on 31.8% (Table 1).

Table 1. Results of the patient management (care and treatment) blocks of Round 1 and 2 of the Delphi Study

Blocks	Round 1			Round 2		
	Number of statements	Consensus Assessment		Number of statements	Consensus Assessment	
		Consensus	No consensus		Consensus	No consensus
Patient management (care)	120	76 (63.3%)	44 (24.4%)	44	14 (31.8%)	30 (68.2%)
Patient management (treatment)	87	40 (45.9%)	47 (54.0%)	40*	11 (27.5%)	29 (72.5%)

* The number of statements is reduced from 47 to 40 by the reformulation of some statements in round 2 with the aim of gaining greater understanding from the panelists.

- Regarding hospital discharge and patient follow-up, patients should attend a follow-up visit with Primary Care within 72 hours (at least via telephone) (8; 7-9), patient should also attend an appointment within 7-15 days with a cardiologist and specialized care nurse for clinical evaluation (9; 8-9) and another one within 15-30 days to reassess the clinical situation (8;7-9) (Figure 2).
- A blood test should be performed at 2-3 months and at 6 months follow-up (9;7-9 and 8;8-9, respectively) (Figure 2).
- Regarding treatment titration/monitoring after discharge, patient should be followed up after 7-15 days (9;8-9), after 15-30 days (9; 7,5-9) and after 2-3 months (8;8-9) (Figure 2).
- Additionally, areas of improvement were identified, such as the importance of using more PROs to assess the patient's quality of life and psychological well-being (8;7-9) and the promotion of specialized nursing profiles in Heart Failure Unit (HFU) (9;8-9) (Figure 2).

Figure 2. Consensus and no consensus in Round 1 and Round 2 statements in the Delphi study

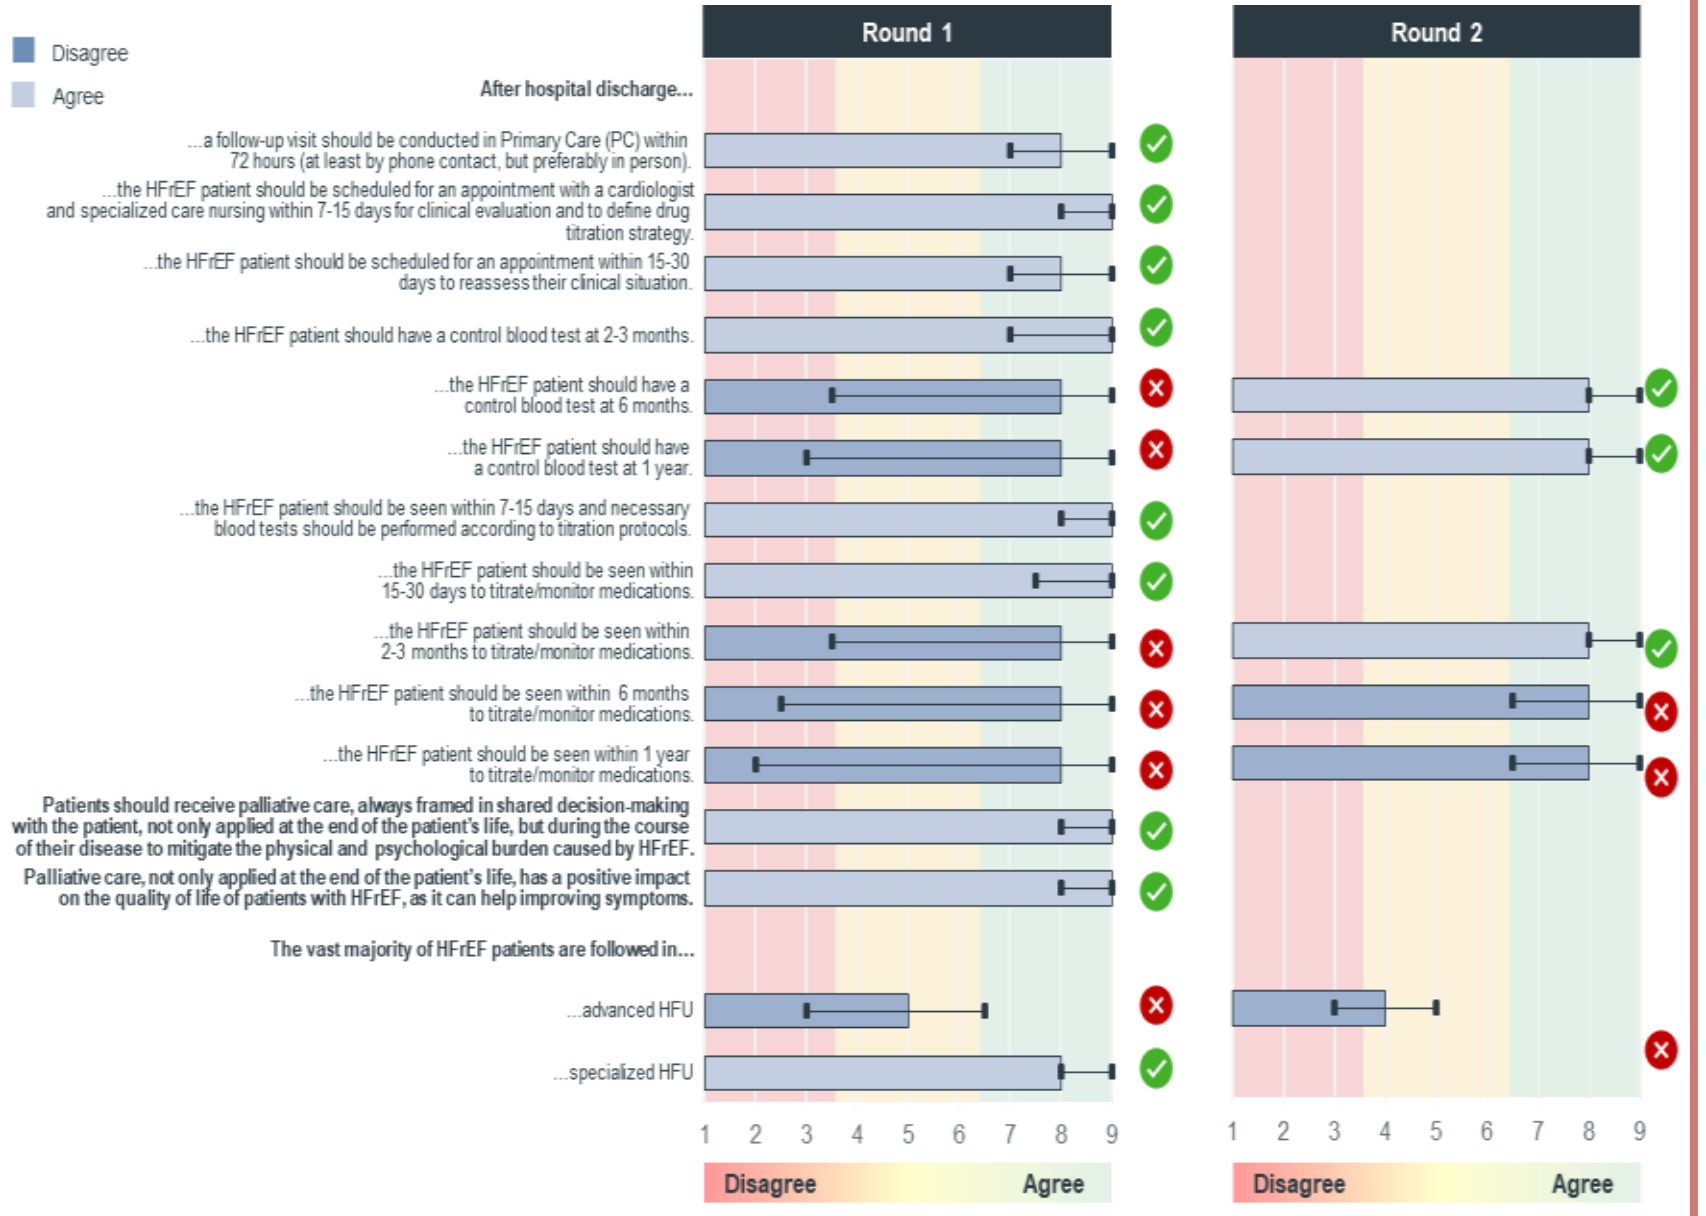
Disagree
Agree
Round 1
Round 2
After hospital discharge...
...a follow-up visit should be conducted in Primary Care (PC) within 72 hours (at least by phone contact, but preferably in person).
...the HFrEF patient should be scheduled for an appointment with a cardiologist and specialized care nursing within 7-15 days for clinical evaluation and to define drug titration strategy.
...the HFrEF patient should be scheduled for an appointment within 15-30 days to reassess their clinical situation.
...the HFrEF patient should have a control blood test at 2-3 months.
...the HFrEF patient should have a control blood test at 6 months.
...the HFrEF patient should have a control blood test at 1 year.
...the HFrEF patient should be seen within 7-15 days and necessary blood tests should be performed according to titration protocols.
...the HFrEF patient should be seen within 15-30 days to titrate/monitor medications.
...the HFrEF patient should be seen within 2-3 months to titrate/monitor medications.
...the HFrEF patient should be seen within 6 months to titrate/monitor medications.
...the HFrEF patient should be seen within 1 year to titrate/monitor medications.
Patients should receive palliative care, always framed in shared decision-making with the patient, not only applied at the end of the patient's life, but during the course of their disease to mitigate the physical and psychological burden caused by HFrEF.
Palliative care, not only applied at the end of the patient's life, has a positive impact on the quality of life of patients with HFrEF, as it can help improving symptoms.
The vast majority of HFrEF patients are followed in...
...advanced HFU
...specialized HFU
1 2 3 4 5 6 7 8 9
Disagree
Agree
1 2 3 4 5 6 7 8 9
Disagree
Agree

Conclusions

This study highlights the necessity of comprehensive follow-up for HFrEF patients after WHF discharge, considering both clinical and psychological aspects. Fluent communication among health care professionals and sites in Spain is crucial during the early post-discharge period.

Heart Failure 2025 & World Congress on Acute Heart Failure

Abbreviations: HFrEF: Heart failure with reduced ejection fraction; WHF: Worsening Heart Failure; HFU: Heart Failure Unit; PROs: Patient Reported Outcomes.

Disclaimer: The authors thank Pau Vesa, Isabel Soto and Núria Perulero, IQVIA workers, for their support in the design of the study, data analysis and in the writing of the poster. These services were funded by Bayer Hispania, S.L. The study was carried out with funding from Bayer Spain. Bayer was involved in the design, data analysis, and drafting of the poster. Authors have not received authorship fees.
Presented at the 2025 Heart Failure Congress of the European Society of Cardiology (ESC), May 17th – 20th, 2025, Belgrade, Serbia. Abstract #61113

RESTRICTED