Delphi study on the management of patients with Heart Failure reduced Ejection Fraction (HFrEF) in Spain: The diagnosis, a multidisciplinary approach.

Ruiz-Bustillo Sonia[1], Alvarez-Garcia Jesús[2], Morgado Garcia-Polavieja José Ignacio[3], Garcia Quintana Antonio [4],

[1] Hospital del Mar, Cardiology. Heart Diseases Biomedical Research Group, Hospital del Mar Research Institute, Barcelona, Spain; [2] CIBER of Cardiovascular Diseases, Department of Cardiology at Ramón y Cajal University Hospital, Madrid, Spain; [3] University Hospital of Virgen Macarena, Department of Cardiology, Virgen Macarena Hospital , Seville, Spain;[4] University Hospital Dr Negrín, Heart Failure Unit Department of Cardiology at Hospital Universitario de Gran Canaria Doctor Negrín., Las Palmas De Gran Canaria, Spain

Introduction and Purpose

- Despite the diagnosis of Heart Failure (HF) being primarily clinical, its management can be complex due to multiple factors, such as symptoms or signs that may be difficult to assess in some patients. Close monitoring between different levels of care, especially between cardiologists and primary care physicians, is recommended as Heart Failure with Reduced Ejection Fraction (HFrEF) requires a multidisciplinary approach that improves patient outcomes.
- This study aimed to evaluate the consensus among cardiologists to better understand their perception and management of HFrEF, and to identify the care process for such management.

Methodology

- A Scientific Committee (SC) of 4 experts in the management of HFrEF created and validated a Delphi questionnaire comprised of 257 statements, which was completed in two rounds by 79 cardiologists (Figure 1).
- Each statement was assessed by a Likert ordinal scale of 1 to 9 points (1 = Completely Disagree to 9 = Completely Agree).
- We present the results from the patient care management section related to the diagnosis

Figure 1. Study design: Delphi methodology adapted in 2 rounds

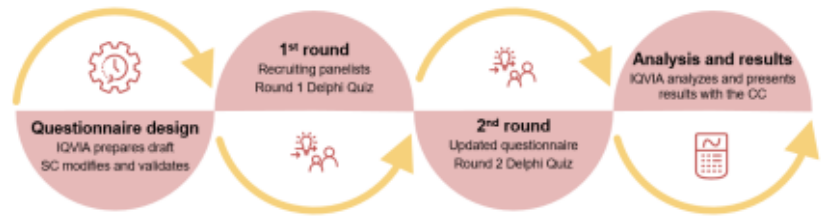
1st round
Recruiting panelists
Round 1 Delphi Quiz
Questionnaire design
IQVIA prepares draft
SC modifies and validates
2nd round
Updated questionnaire
Round 2 Delphi Quiz
Analysis and results
IQVIA analyzes and presents
results with the CC

Results

- Responses were collected over two rounds, achieving consensus on 75% of the statements.
- In the first Delphi round, the defined level of agreement was reached for the 76 statements (63.3%), and in second round, 44 statements that did not reach consensus were revisited, with agreement achieved on 31.8% (Table 1).

Table 1. Results of the patient management (care and treatment) blocks of Round 1 and 2 of the Delphi Study

Blocks	Round 1			Round 2		
	Number of statements	Consensus Assessment		Number of statements	Consensus Assessment	
		Consensus	No consensus		Consensus	No consensus
Patient management (care)	120	76 (63.3%)	44 (24.4%)	44	14 (31.8%)	30 (68.2%)
Patient management (treatment)	87	40 (45.9%)	47 (54.0%)	40*	11 (27.5%)	29 (72.5%)

* The number of statements is reduced from 47 to 40 by the reformulation of some statements in round 2 with the aim of gaining greater understanding from the panelists.

- Regarding the diagnosis of HFrEF, there was broad consensus based on symptoms, signs, and an LVEF ≤ 40% (median; interquartile range: 9; 8-9), confirmed by history, physical examination, electrocardiogram, echocardiography, and natriuretic peptides (BNP or NT-proBNP) (: 9; 9-9 for all items). The evaluation should be clinical (9; 9-9), etiological, and functional (9; 8-9; and 9; 9-9, respectively). (Figure 2).
- Patients with HFrEF should receive comprehensive care from a multidisciplinary team including cardiologists (9; 9-9), primary care physicians (9; 8-9), specialized nursing (9; 9-9), internal medicine (8; 7-9), geriatrics (8; 7-9), emergency services (8; 7-9), and nephrology (8; 7-9). (Figure 2).
- It was agreed by almost all panelists that nurses in the unit should act as the link between specialties (including primary care) and as the contact person for patients (9; 8-9). (Figure 2).

- Barriers to effective communication in primary care include the absence of more integrated and multidisciplinary models (8; 7-9), specific protocols coordinated between primary care and specialist care (8; 7-8), specialized nursing staff responsible for communication between primary care and cardiology (8; 8-9), optimal IT tools for clinical management (8; 7-9), and training in primary care to understand the referral or consultation processes for patients with HF (8; 7-8.5).

Figure 2. Consensus and no consensus in Round 1 and Round 2 statements in the Delphi study

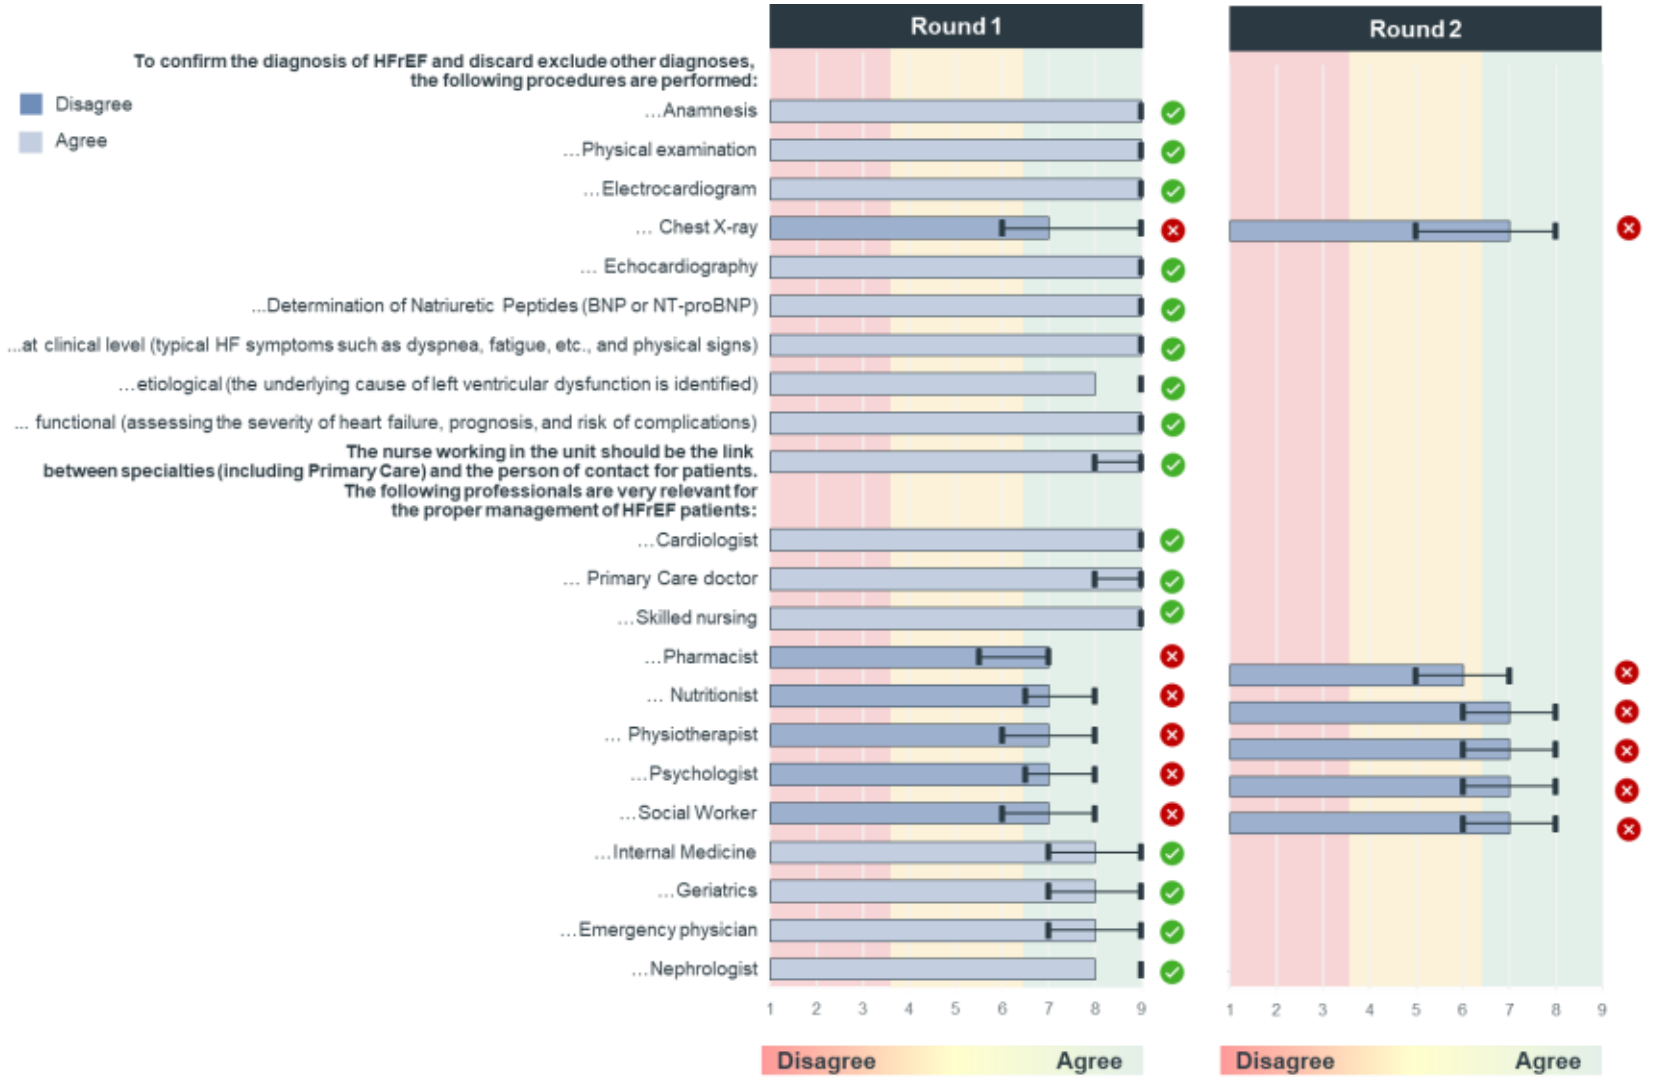
Round 1
Round 2
Disagree
Agree
To confirm the diagnosis of HFrEF and discard exclude other diagnoses, the following procedures are performed:
...Anamnesis
...Physical examination
...Electrocardiogram
... Chest X-ray
... Echocardiography
...Determination of Natriuretic Peptides (BNP or NT-proBNP)
...at clinical level (typical HF symptoms such as dyspnea, fatigue, etc., and physical signs)
...etiological (the underlying cause of left ventricular dysfunction is identified)
... functional (assessing the severity of heart failure, prognosis, and risk of complications)
The nurse working in the unit should be the link between specialties (including Primary Care) and the person of contact for patients.
The following professionals are very relevant for the proper management of HFrEF patients:
...Cardiologist
... Primary Care doctor
...Skilled nursing
...Pharmacist
... Nutritionist
... Physiotherapist
...Psychologist
...Social Worker
...Internal Medicine
...Geriatrics
...Emergency physician
...Nephrologist
1 2 3 4 5 6 7 8 9
1 2 3 4 5 6 7 8 9
Disagree
Agree
Disagree
Agree

Conclusions

The findings of our study reflected that specialists agree on HFrEF diagnosis and management but identify resource gaps for effective communication and monitoring. Future efforts should focus on providing resources and awareness for optimal HFrEF management in Spanish public hospitals.

Heart Failure 2025 & World Congress on Acute Heart Failure

Disclaimer: The authors thank Pau Vesa, Isabel Soto and Núria Perulero, IQVIA workers, for their support in the design of the study, data analysis and in the writing of the poster. These services were funded by Bayer Hispania, S.L. The study was carried out with funding from Bayer Spain. Bayer was involved in the design, data analysis, and drafting of the poster. Authors have not received authorship fees.
Presented at the 2025 Heart Failure Congress of the European Society of Cardiology (ESC), May 17th – 20th, 2025, Belgrade, Serbia. Abstract #61109