POSTER 166

Impact of sleep disturbances on health-related quality of life in postmenopausal women

Claudio N. Soares[1], Malek Bajbouj[2], Nils Schoof[3], Amit Kishore[4], Cecilia Caetano[5]

[1]Queen's University School of Medicine, Kingston, Ontario, Canada; [2]Charité – Universitätsmedizin Berlin, Berlin, Germany; [3]Bayer AG, Berlin, Germany; [4]Accuscript Consultancy, Ludhiana, Punjab, India; [5]Bayer Consumer Care, Basel, Switzerland

IMS 19 WORLD CONGRESS ON MENOPAUSE
MELBOURNE
19–22 OCTOBER 2024

INTRODUCTION

Sleep disturbances are common during and after the menopausal transition.[1–4] Menopause-associated sleep disturbances include night-time awakenings, longer, more frequent periods of wakefulness after sleep onset, poor-quality, insufficient, or nonrestorative sleep, difficulty in sleep initiation, and early waking.[5] Despite having potential effects on health and quality of life (QoL),[6–8] sleep disturbances are often underdiagnosed and undertreated.[5] **The present systematic literature review (SLR) was conducted to understand the current evidence on the prevalence of menopause-associated sleep disturbances and their impact on women's health-related quality of life (HRQoL) throughout the stages of menopause.**

METHODS

Seven consensus questions were developed to determine: (i) the **proportion of women in menopause with sleep disturbances**; (ii) the **proportion of women with poor sleep quality who have vasomotor symptoms (VMS)**; (iii) the **occurrence of sleep disturbances in menopausal women without VMS**; (iv) **risk factors or protective factors** responsible for worsening or improving sleep quality in menopause; (v) the **impact of depressive symptoms on HRQoL**; (vi) the **impact of sleep disturbances on HRQoL**; and (vii) the **impact of sleep disturbances on work productivity.** Searches were conducted in PubMed and Excerpta Medica Database to identify articles published between 2013 and 2023 containing **evidence for the impact of sleep quality on HRQoL and the epidemiology of sleep disturbances in menopausal women.** The protocol for this SLR was registered in the International Prospective Register of Systematic Reviews (www.crd.york.ac.uk/prospero; registration number: CRD42023448601) and conducted in line with Preferred Reporting Items for Systematic Review and Meta-Analyses (PRISMA) guidelines. Predefined eligibility criteria based on Population, Intervention, Control, Outcomes, Study design, and Timeframe (PICOST) were used to identify relevant studies based on the aims and objectives of the review. A **targeted approach** was employed to study the **prevalence and risk factors of sleep disturbances associated with menopause,** while a **systematic approach** was employed to evaluate the **impact of sleep quality on HRQoL in women during menopause.**

RESULTS

Figure 1. Prevalence of sleep disturbances in menopausal women by geographical region

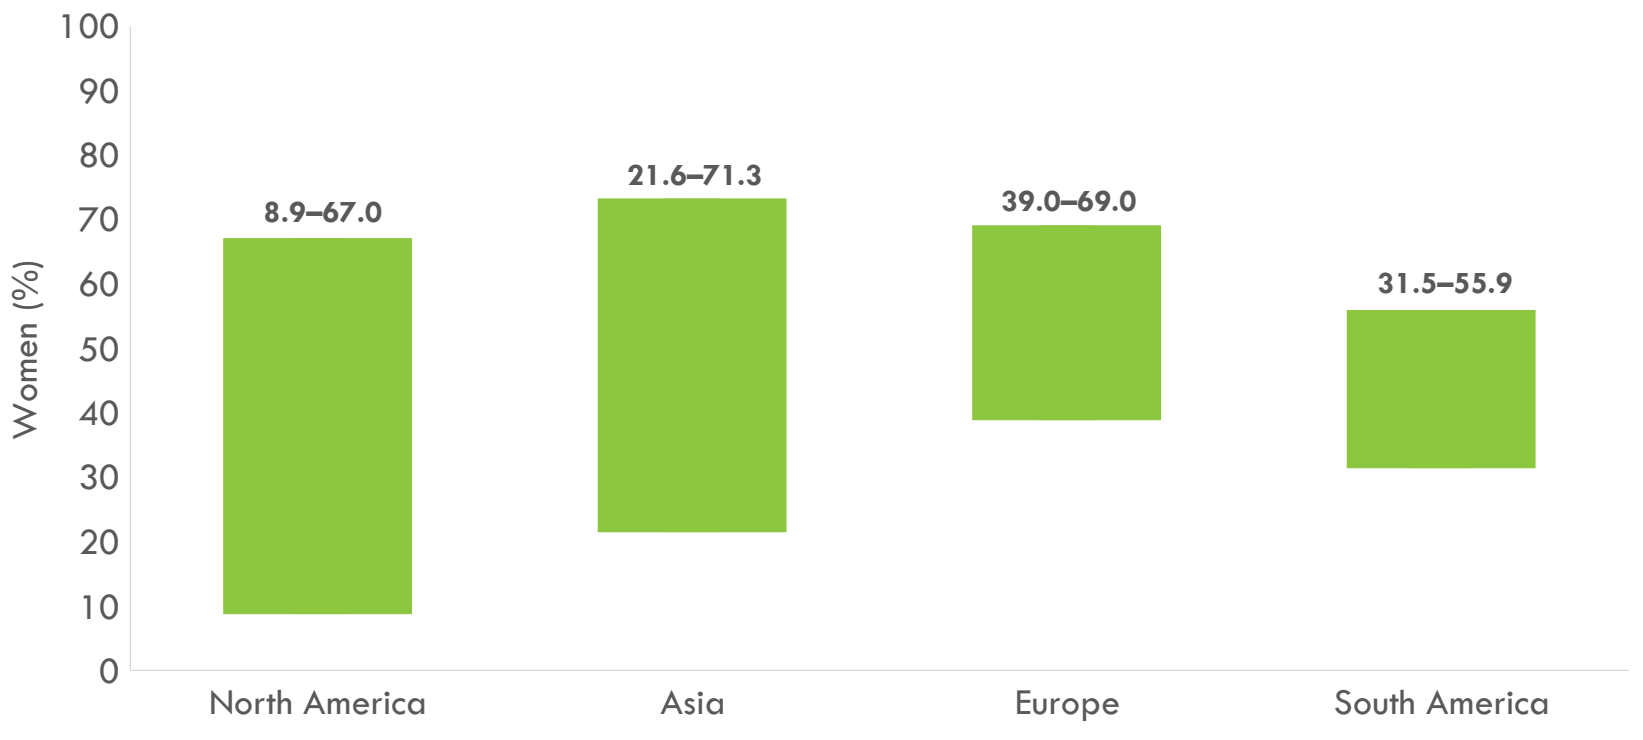
Women (%)
100
90
80
70
60
50
40
30
20
10
0
8.9–67.0
21.6–71.3
39.0–69.0
31.5–55.9
North America
Asia
Europe
South America

Figure 2. Percentage of reported sleep disturbances that occurred in women not experiencing VMS[9]

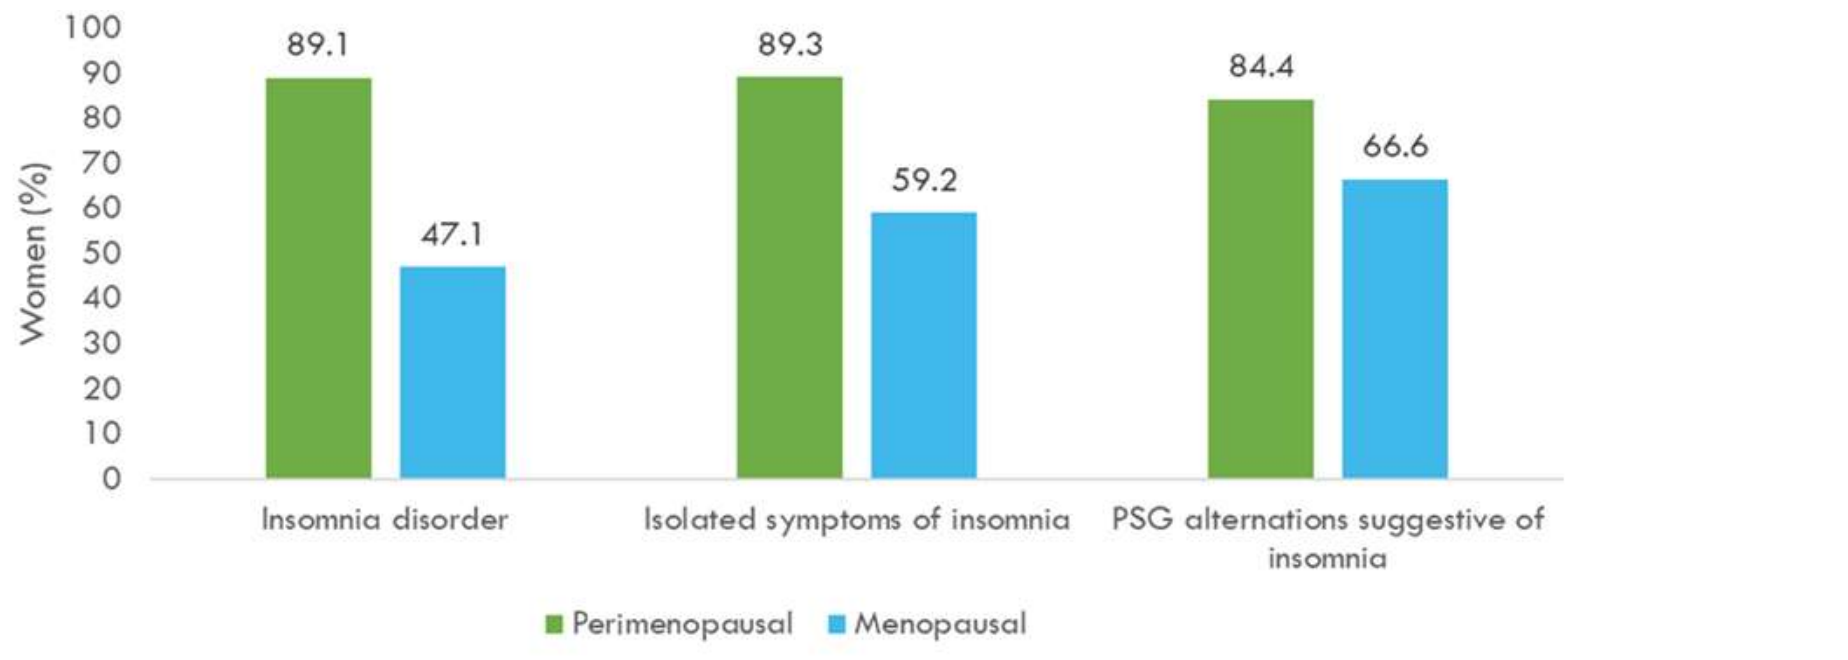
Women (%)
100
90
80
70
60
50
40
30
20
10
0
89.1
47.1
89.3
59.2
84.4
66.6
Insomnia disorder
Isolated symptoms of insomnia
PSG alternations suggestive of insomnia
Perimenopausal
Menopausal

PSG, polysomnography; VMS, vasomotor symptoms

In total, 30 publications focusing on the epidemiological outcomes of sleep disturbances and 28 studies focusing on the impact of sleep quality on HRQoL were identified. These studies confirmed the high prevalence of sleep disturbances in menopausal women (**Figure 1**), including those without VMS (**Figure 2**).

Risk factors for sleep disturbances included menopausal status, depression, menopausal symptoms such as VMS, high glycemic index diets, and age.

In nine cross-sectional studies reporting the impact of sleep quality on QoL using menopause-specific tools, significant correlations were observed between **poor sleep and lower QoL.**

Nine studies assessed the relationship between sleep disturbances and QoL using a wide range of HRQoL tools. Overall, **poor sleep** was negatively correlated with both **physical and mental health** components.

Eleven studies reported the impact of sleep disturbances on **emotional status,** showing an association between sleep problems and depression/anxiety symptoms in menopausal women. Sleep efficiency, quality, latency, and duration were reduced as the degree of depressive symptomology increased.

In two cross-sectional studies, **musculoskeletal pain** was significantly associated with sleep disorders and sleep quality.

Five studies reported a significant impact of sleep quality on **work productivity and occupational QoL.**

CONCLUSIONS

Sleep disturbances are highly prevalent in menopausal women, even in those without VMS, and can severely impact women's health, well-being, work productivity, and HRQoL

Sleep disturbances are significantly associated with impaired HRQoL, mood/anxiety symptoms, musculoskeletal pain, and impaired work productivity

Better recognition of menopause-related sleep disturbances as well as their risk factors and comorbidities is essential to improve management

Clinicians are encouraged to counsel menopausal women on sleep disturbance to improve recognition and identify those who could be offered treatment

ACKNOWLEDGEMENTS

The authors would like to acknowledge the original study authors and their participating patients for their work underlying this review, and Highfield (Oxford, UK) for their assistance with producing this abstract and poster.

REFERENCES

1. Nappi RE, et al. Menopause 2021;28:875–82; **2.** Amyx M, et al. Matern Child Health J 2017;21:306–14; **3.** Tandon VR, et al. J Midlife Health 2022;13:26–33; **4.** Thurston R, Joffe H. Obstet Gynecol Clin 2011;38:489–501; **5.** Maki PM, et al. Menopause. 2024;31:724–33; **6.** Korhonen K, et al. J Affect Disord 2021;295:831–8; **7.** Fernandez-Mendoza J, et al. Hypertension 2012;60:929–35; **8.** Meng L, et al. Hypertens Res 2013;36:985–95; **9.** Hachul H, et al. J Clin Sleep Med. 2021;17:2257–67.

DISCLOSURES

This study was funded by Bayer AG, Berlin, Germany. CNS has acted as a consultant for Otsuka, Bayer, Eisai, and Diamond Therapeutics, and has received grants from Ontario Brain Institute, Clairvoyant, and Diamond Therapeutics. MB has received research funding from the German Research Foundation, the German Federal Ministry of Education and Research, the Foreign Office, and the Federal Ministry of Economic Development and Cooperation. He has served on advisory boards or as an external consultant for Bayer, Flying Health, GH Research, Johnson and Johnson, Parexel, and the Shohan Foundation. NS and CC are employees of Bayer. AK is an employee of Accuscript Consultancy.