Association of alpha-fetoprotein (AFP) and clinical outcomes in patients with unresectable hepatocellular carcinoma (uHCC) treated with camrelizumab + rivoceranib vs sorafenib (CARES-310)

FPN# 177P
Poster presented at ESMO GI 2025

Copies of this poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Richard Kim,[1] Wei Shi,[2] Xiuzhi Wu,[2] Xianzhang Meng,[3] Peter Lu,[3] Chris Galloway,[3] Kristin Ryan,[3] Laura Alexander,[3] Arndt Vogel[4]

[1]Moffitt Cancer Center, Tampa, FL, USA; [2]Jiangsu Hengrui Pharmaceuticals, Shanghai, China; [3]Elevar Therapeutics, Fort Lee, NJ, USA; [4]Princess Margaret Cancer Centre, Toronto, ON, Canada

BACKGROUND

- CARES-310 (NCT03764293) compared the combination of the anti-programmed death-1 (PD-1) antibody camrelizumab plus the vascular endothelial growth factor receptor 2 (VEGFR2)-targeted tyrosine kinase inhibitor (TKI) rivoceranib versus sorafenib for the treatment of unresectable hepatocellular carcinoma (uHCC).[1]
- In the CARES-310 study, camrelizumab plus rivoceranib showed significantly improved median overall survival (mOS) and median progression-free survival (mPFS) compared with sorafenib (mOS 23.8 months vs 15.2 months; HR 0.64 [95% CI, 0.52-0.79]; mPFS 5.6 months vs 3.7 months; HR 0.54 [95% CI, 0.44-0.67]).[2]
- The most common ≥5% grade 3-4 treatment-related adverse events for camrelizumab plus rivoceranib were hypertension (38%) and increased aspartate aminotransferase (AST; 17%) vs palmar-plantar erythrodysesthesia syndrome (16%) for sorafenib.[2]
- We performed a post-hoc exploratory analysis of the CARES-310 study evaluating the impact of baseline serum alpha fetoprotein (AFP) levels (<400 ng/mL vs ≥400 ng/mL)[3] on efficacy and safety outcomes.

METHODS

CARES-310 Study Design and Endpoints[1]

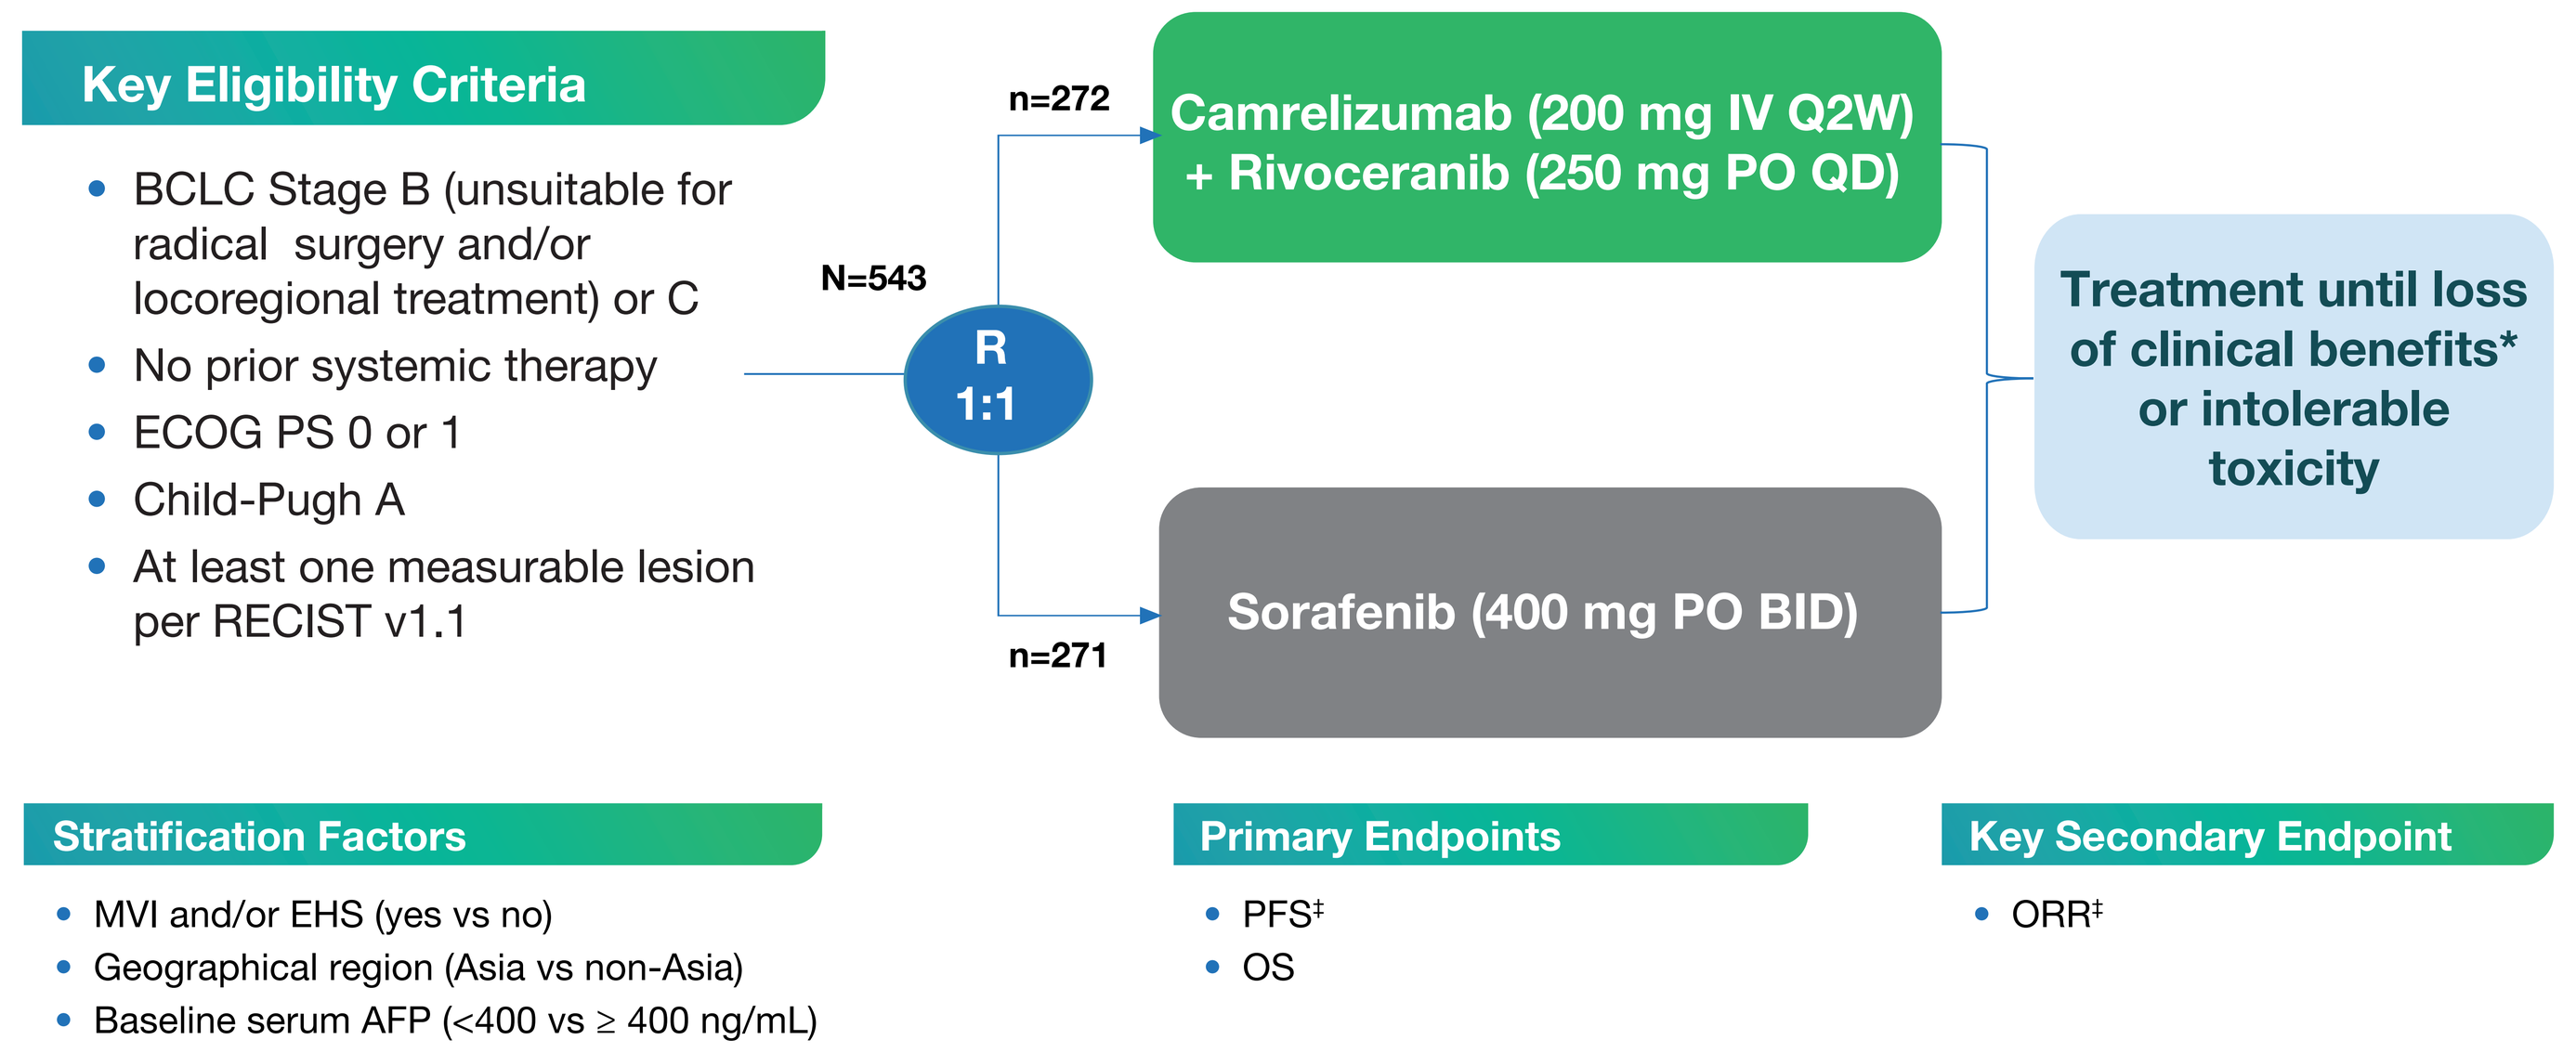
Key Eligibility Criteria
BCLC Stage B (unsuitable for radical surgery and/or locoregional treatment) or C
No prior systemic therapy
ECOG PS 0 or 1
Child-Pugh A
At least one measurable lesion per RECIST v1.1
N=543
R 1:1
n=272
Camrelizumab (200 mg IV Q2W) + Rivoceranib (250 mg PO QD)
n=271
Sorafenib (400 mg PO BID)
Treatment until loss of clinical benefits* or intolerable toxicity
Stratification Factors
MVI and/or EHS (yes vs no)
Geographical region (Asia vs non-Asia)
Baseline serum AFP (<400 vs ≥ 400 ng/mL)
Primary Endpoints
PFS‡
OS
Key Secondary Endpoint
ORR‡

*Treatment beyond progression allowed if there was evidence of clinical benefits per investigator. ‡By BIRC per RECIST v1.1.
AFP, alpha-fetoprotein; BCLC, Barcelona Clinic Liver Cancer; BID, twice daily; BIRC, blinded independent review committee; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; IV, intravenous; MVI, macrovascular invasion; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PO, by mouth; Q2W, every 2 weeks; QD, once daily; RECIST v1.1, Response Evaluation Criteria for Solid Tumors version 1.1.

Post-hoc Analysis

- This post-hoc exploratory analysis assessed post-treatment outcomes by baseline serum AFP levels (<400 ng/mL vs ≥400 ng/mL), where mOS and mPFS were estimated using the Kaplan-Meier method.
- Separate analyses were performed for the intention-to-treat (ITT) population and for the safety population.

RESULTS

- In this post-hoc exploratory analysis, most patients were male, Barcelona Clinic Liver Cancer stage C, Child-Pugh Class A5, and had received previous local therapy (**Table 1**).
- Objective response rate per blinded independent review committee (BIRC) assessment favored the camrelizumab plus rivoceranib arm regardless of baseline AFP level. Disease control rate per BIRC assessment also favored the camrelizumab plus rivoceranib arm regardless of baseline AFP level (**Table 2**).
- Median overall survival was longer with camrelizumab plus rivoceranib compared with sorafenib in patients with baseline AFP <400 ng/mL (**Figure 1**) and ≥400 ng/mL (**Figure 2**).
- Median progression-free survival was longer with camrelizumab plus rivoceranib compared with sorafenib in patients with baseline AFP <400 ng/mL (**Figure 3**) and ≥400 ng/mL (**Figure 4**).
- The most common treatment-related adverse events (TRAEs) were similar for patients who received camrelizumab plus rivoceranib or sorafenib, regardless of baseline serum AFP level (**Tables 3-4**).

RESULTS

Table 1: Baseline Characteristics by AFP Subgroup (ITT Population)

	AFP <400 ng/mL		AFP ≥400 ng/mL	
	Rivoceranib + Camrelizumab (N=176)	Sorafenib (N=171)	Rivoceranib + Camrelizumab (N=96)	Sorafenib (N=100)
Median age, years (range)	60 (23-83)	57 (28-82)	54 (27-76)	52 (23-82)
Male sex, n (%)	148 (84.1)	148 (86.5)	79 (82.3)	82 (82.0)
Geographic region, n (%)				
Asia	142 (80.7)	139 (81.3)	83 (86.5)	85 (85.0)
Non-Asia	34 (19.3)	32 (18.7)	13 (13.5)	15 (15.0)
Race, n (%)				
Asian	143 (81.3)	139 (81.3)	83 (86.5)	85 (85.0)
Caucasian (non-Asian)	33 (18.8)	32 (18.7)	13 (13.5)	15 (15.0)
ECOG PS, n (%)				
0	80 (45.5)	77 (45.0)	40 (41.7)	39 (39.0)
1	96 (54.5)	94 (55.0)	56 (58.3)	61 (61.0)
BCLC stage, n (%)				
B (middle stage)	27 (15.3)	28 (16.4)	11 (11.5)	12 (12.0)
C (advanced stage)	149 (84.7)	143 (83.6)	85 (88.5)	88 (88.0)
Child-Pugh score, n (%)				
A5	154 (87.5)	144 (84.2)	82 (85.4)	86 (86.0)
A6	22 (12.5)	27 (15.8)	14 (14.6)	14 (14.0)
MVI and/or EHS, n (%)				
Absence	50 (28.4)	49 (28.7)	22 (22.9)	22 (22.0)
Presence	126 (71.6)	122 (71.3)	74 (77.1)	78 (78.0)
EHS, n (%)				
Presence	116 (65.9)	115 (67.3)	59 (61.5)	65 (65.0)
Absence	60 (34.1)	56 (32.7)	37 (38.5)	35 (35.0)
Previous local therapy, n (%)				
Yes	103 (58.5)	102 (59.6)	58 (60.4)	48 (48.0)
No	73 (41.5)	69 (40.4)	38 (39.6)	52 (52.0)
HCC etiology, n (%)				
HBV	129 (73.3)	120 (70.2)	79 (82.3)	77 (77.0)
HCV	15 (8.5)	20 (11.7)	7 (7.3)	9 (9.0)
Non-viral	32 (18.2)	31 (18.1)	10 (10.4)	14 (14.0)
BMI, n (%)				
<30 kg/m²	168 (95.5)	160 (93.6)	92 (95.8)	97 (97.0)
≥30 kg/m²	8 (4.5)	10 (5.8)	4 (4.2)	3 (3.0)
Missing	0	1 (0.6)	0	0

BCLC, Barcelona Clinic Liver Cancer; BMI, body mass index; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; HBV, hepatitis B virus; HCC, hepatocellular carcinoma; HCV, hepatitis C virus; ITT, intent-to-treat; MVI, macrovascular invasion.

Table 2: Summary of Response Rate by BIRC Assessment (RECIST v1.1) (ITT Population)

	AFP <400 ng/mL		AFP ≥400 ng/mL	
	Rivoceranib+ Camrelizumab (N=176)	Sorafenib (N=171)	Rivoceranib + Camrelizumab (N=96)	Sorafenib (N=100)
Best overall response, n (%)				
Complete response	5 (2.8)	2 (1.2)	0	0
Partial response	55 (31.3)	11 (6.4)	13 (13.5)	3 (3.0)
Stable disease	84 (47.7)	90 (52.6)	55 (57.3)	39 (39.0)
Progressive disease	26 (14.8)	54 (31.6)	19 (19.8)	46 (46.0)
Not evaluable	6 (3.4)	14 (8.2)	9 (9.4)	12 (12.0)
Objective response rate, n (%)	60 (34.1)	13 (7.6)	13 (13.5)	3 (3.0)
[95% CI]	[27.1, 41.6]	[4.1, 12.6]	[7.4, 22.0]	[0.6, 8.5]
Disease control rate[a], n (%)	144 (81.8)	103 (60.2)	68 (70.8)	42 (42.0)
[95% CI]	[75.3, 87.2]	[52.5, 67.6]	[60.7, 79.7]	[32.2, 52.3]
Median follow-up, months	25	19	16	10
Median time to response, months	2	4	2	2

[a]Disease control rate is defined as the percentage of patients with complete response, partial response, or stable disease ≥7 weeks.
BIRC, blinded independent review committee; ITT, intent-to-treat; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

Figure 1: Overall Survival for Patients with Baseline AFP < 400 ng/mL

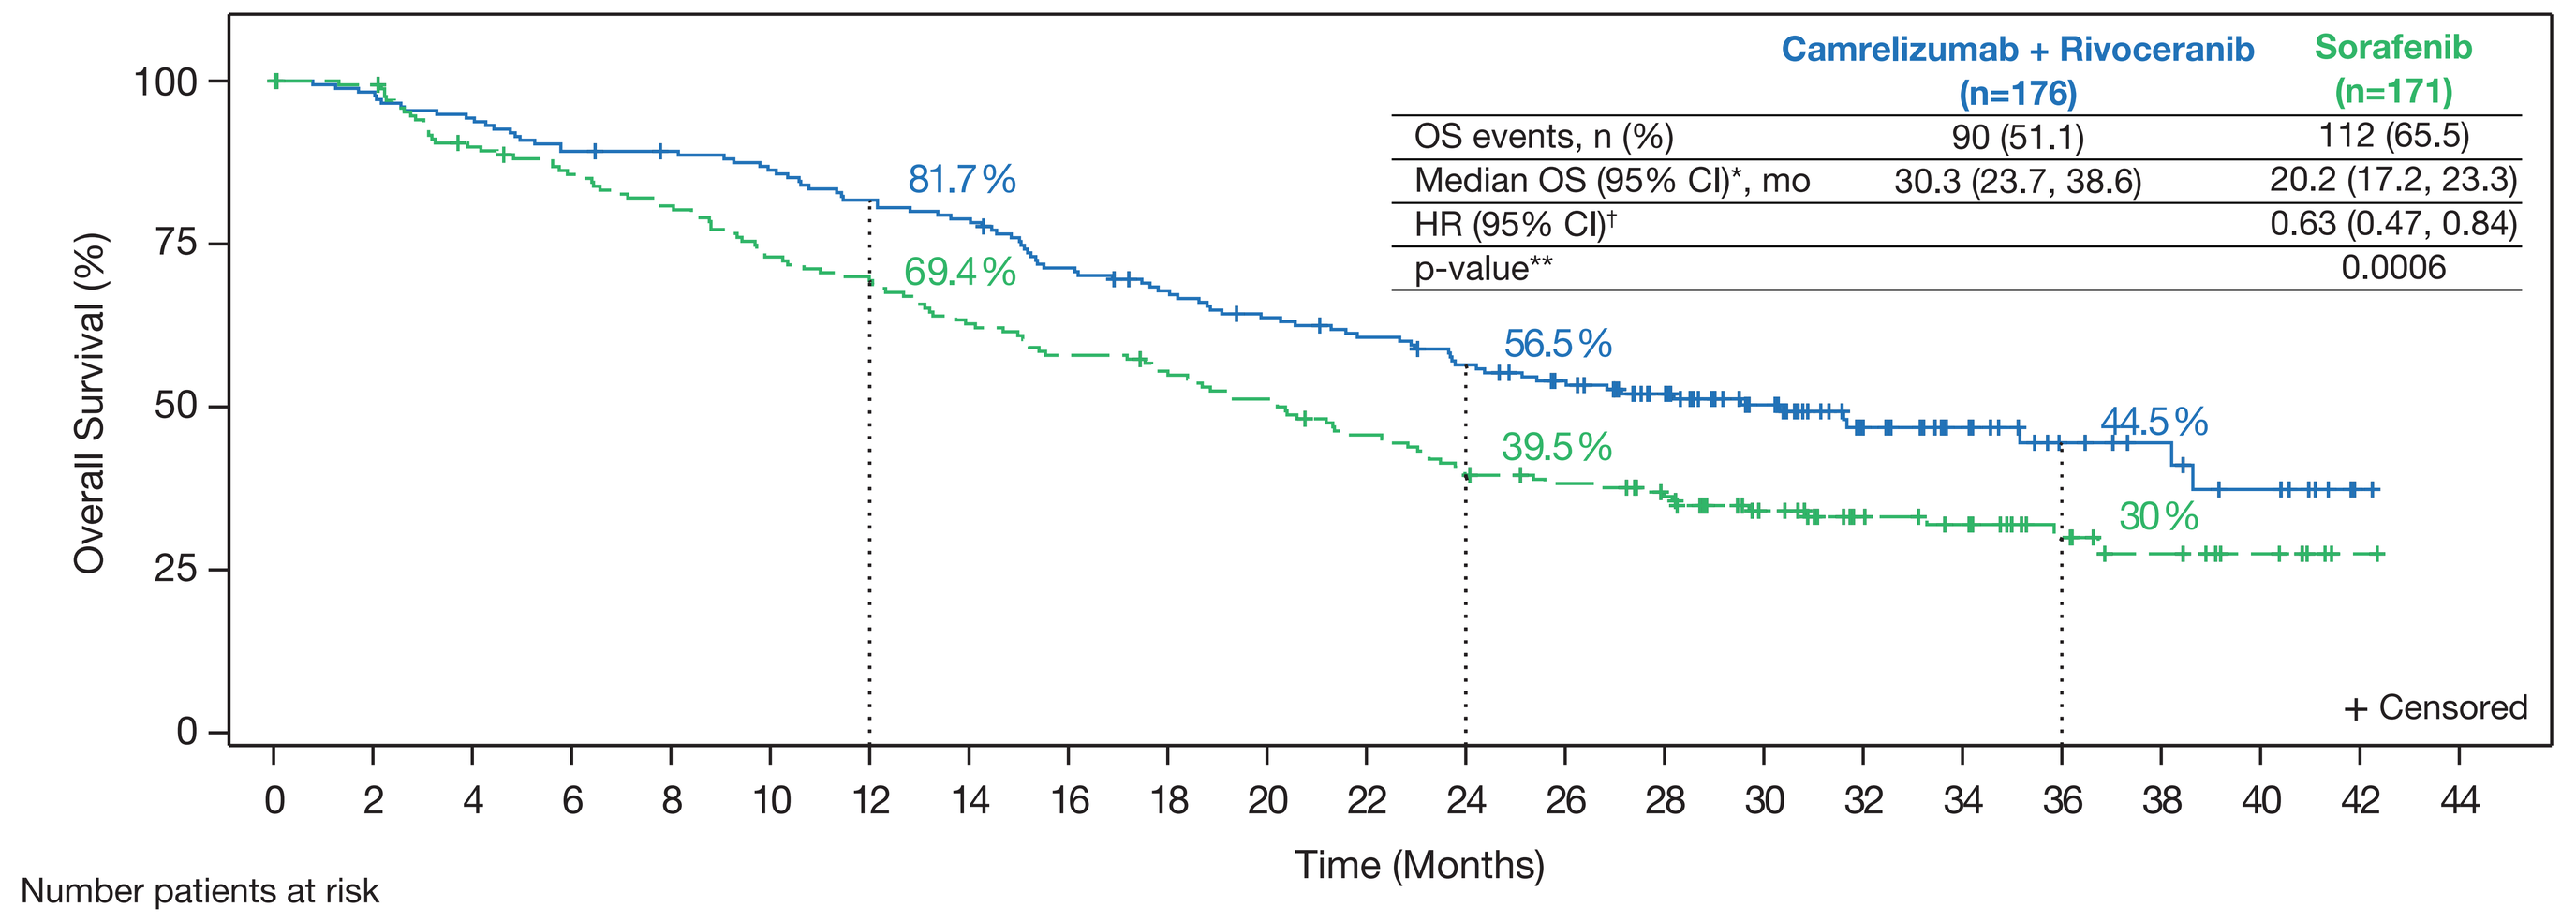
Camrelizumab + Rivoceranib (n=176)
Sorafenib (n=171)
OS events, n (%) 90 (51.1) 112 (65.5)
Median OS (95% CI)*, mo 30.3 (23.7, 38.6) 20.2 (17.2, 23.3)
HR (95% CI)† 0.63 (0.47, 0.84)
p-value** 0.0006
Overall Survival (%)
81.7%
69.4%
56.5%
39.5%
44.5%
30%
+ Censored
Time (Months)

Number patients at risk

Time (Months)	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32	34	36	38	40	42	44
Cam + Rivo	176	173	166	157	155	150	142	137	123	115	107	101	93	84	71	53	35	25	16	13	9	1	0
Sorafenib	171	168	150	142	134	121	115	104	96	91	84	74	64	60	53	40	30	26	15	10	6	1	0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence) and geographical region (Asia vs countries outside of Asia) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Figure 2: Overall Survival for Patients with Baseline AFP ≥400 ng/mL

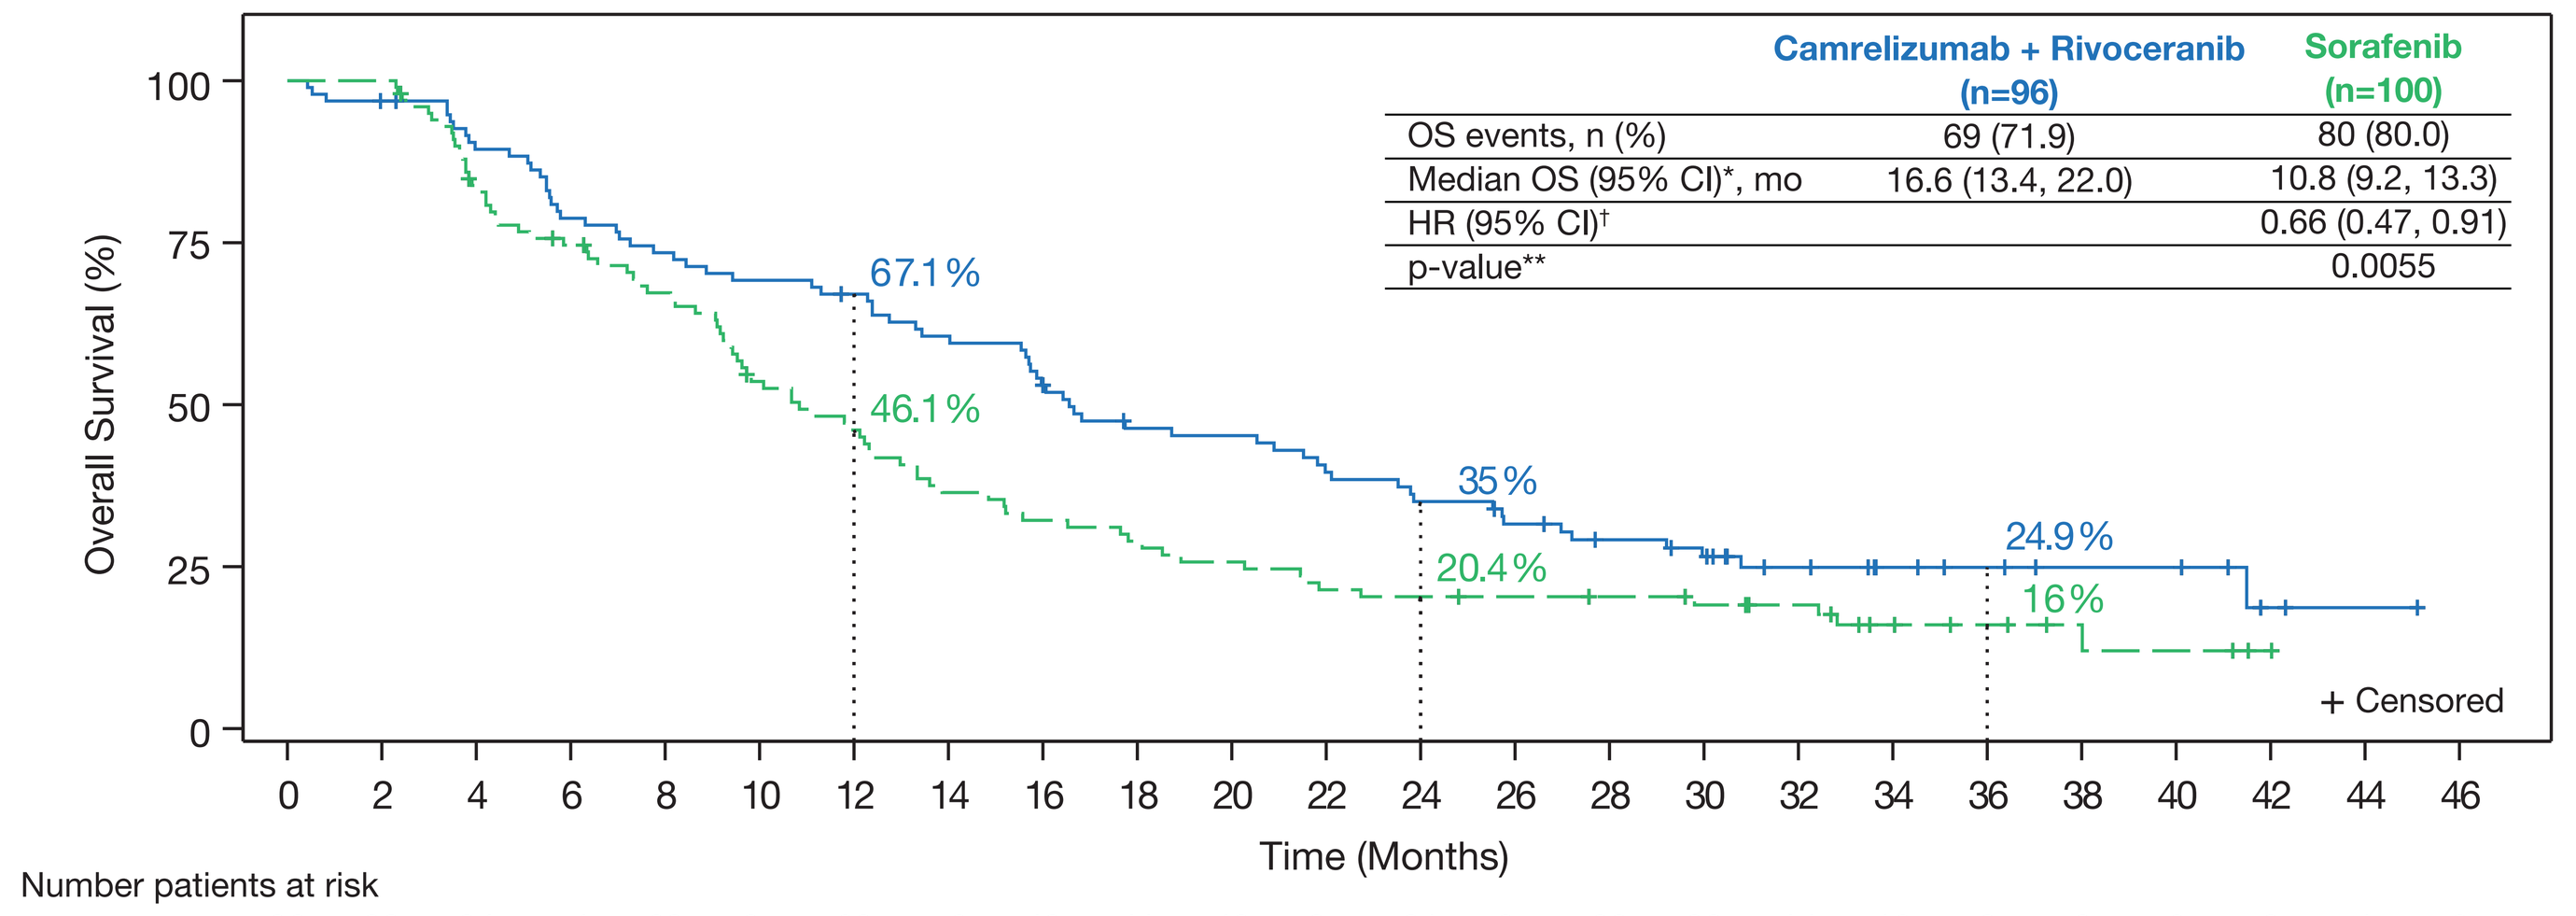
Camrelizumab + Rivoceranib (n=96)
Sorafenib (n=100)
OS events, n (%) 69 (71.9) 80 (80.0)
Median OS (95% CI)*, mo 16.6 (13.4, 22.0) 10.8 (9.2, 13.3)
HR (95% CI)† 0.66 (0.47, 0.91)
p-value** 0.0055
Overall Survival (%)
67.1%
46.1%
35%
20.4%
24.9%
16%
+ Censored
Time (Months)

Number patients at risk

Time (Months)	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32	34	36	38	40	42	44	46
Cam + Rivo	96	92	84	74	69	65	62	56	49	41	40	35	31	27	23	20	14	10	8	6	6	2	1	0
Sorafenib	100	100	82	72	64	50	43	34	30	27	24	20	19	18	17	15	13	8	6	4	3	1	0	

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence) and geographical region (Asia vs countries outside of Asia) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Table 3: Most Common (≥20%) Any Grade or Grade 3-4 (≥5%) TRAEs* for Patients with Baseline AFP <400 ng/mL (Safety Population)

TRAE, n (%)	Rivoceranib + Camrelizumab (N=176)		Sorafenib (N=169)	
	Any Grade	Grade 3-4	Any Grade	Grade 3-4
AST increased	75 (42.6)	27 (15.3)	67 (39.6)	8 (4.7)
ALT increased	64 (36.4)	20 (11.4)	56 (33.1)	6 (3.6)
Platelet count decreased	61 (34.7)	21 (11.9)	59 (34.9)	2 (1.2)
Blood bilirubin increased	59 (33.5)	9 (5.1)	49 (29.0)	2 (1.2)
Proteinuria	40 (22.7)	6 (3.4)	51 (30.2)	3 (1.8)
White blood cell count decreased	37 (21.0)	4 (2.3)	21 (12.4)	3 (1.8)
Neutrophil count decreased	36 (20.5)	9 (5.1)	14 (8.3)	0
GGT increased	35 (19.9)	15 (8.5)	33 (19.5)	14 (8.3)

*TRAEs include adverse events related to both rivoceranib + camrelizumab or sorafenib.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; GGT, gamma-glutamyl transferase; TRAE, treatment-related adverse event.

Figure 3: Progression-free Survival for Patients with Baseline AFP < 400 ng/mL by BIRC Assessment (RECIST v1.1)

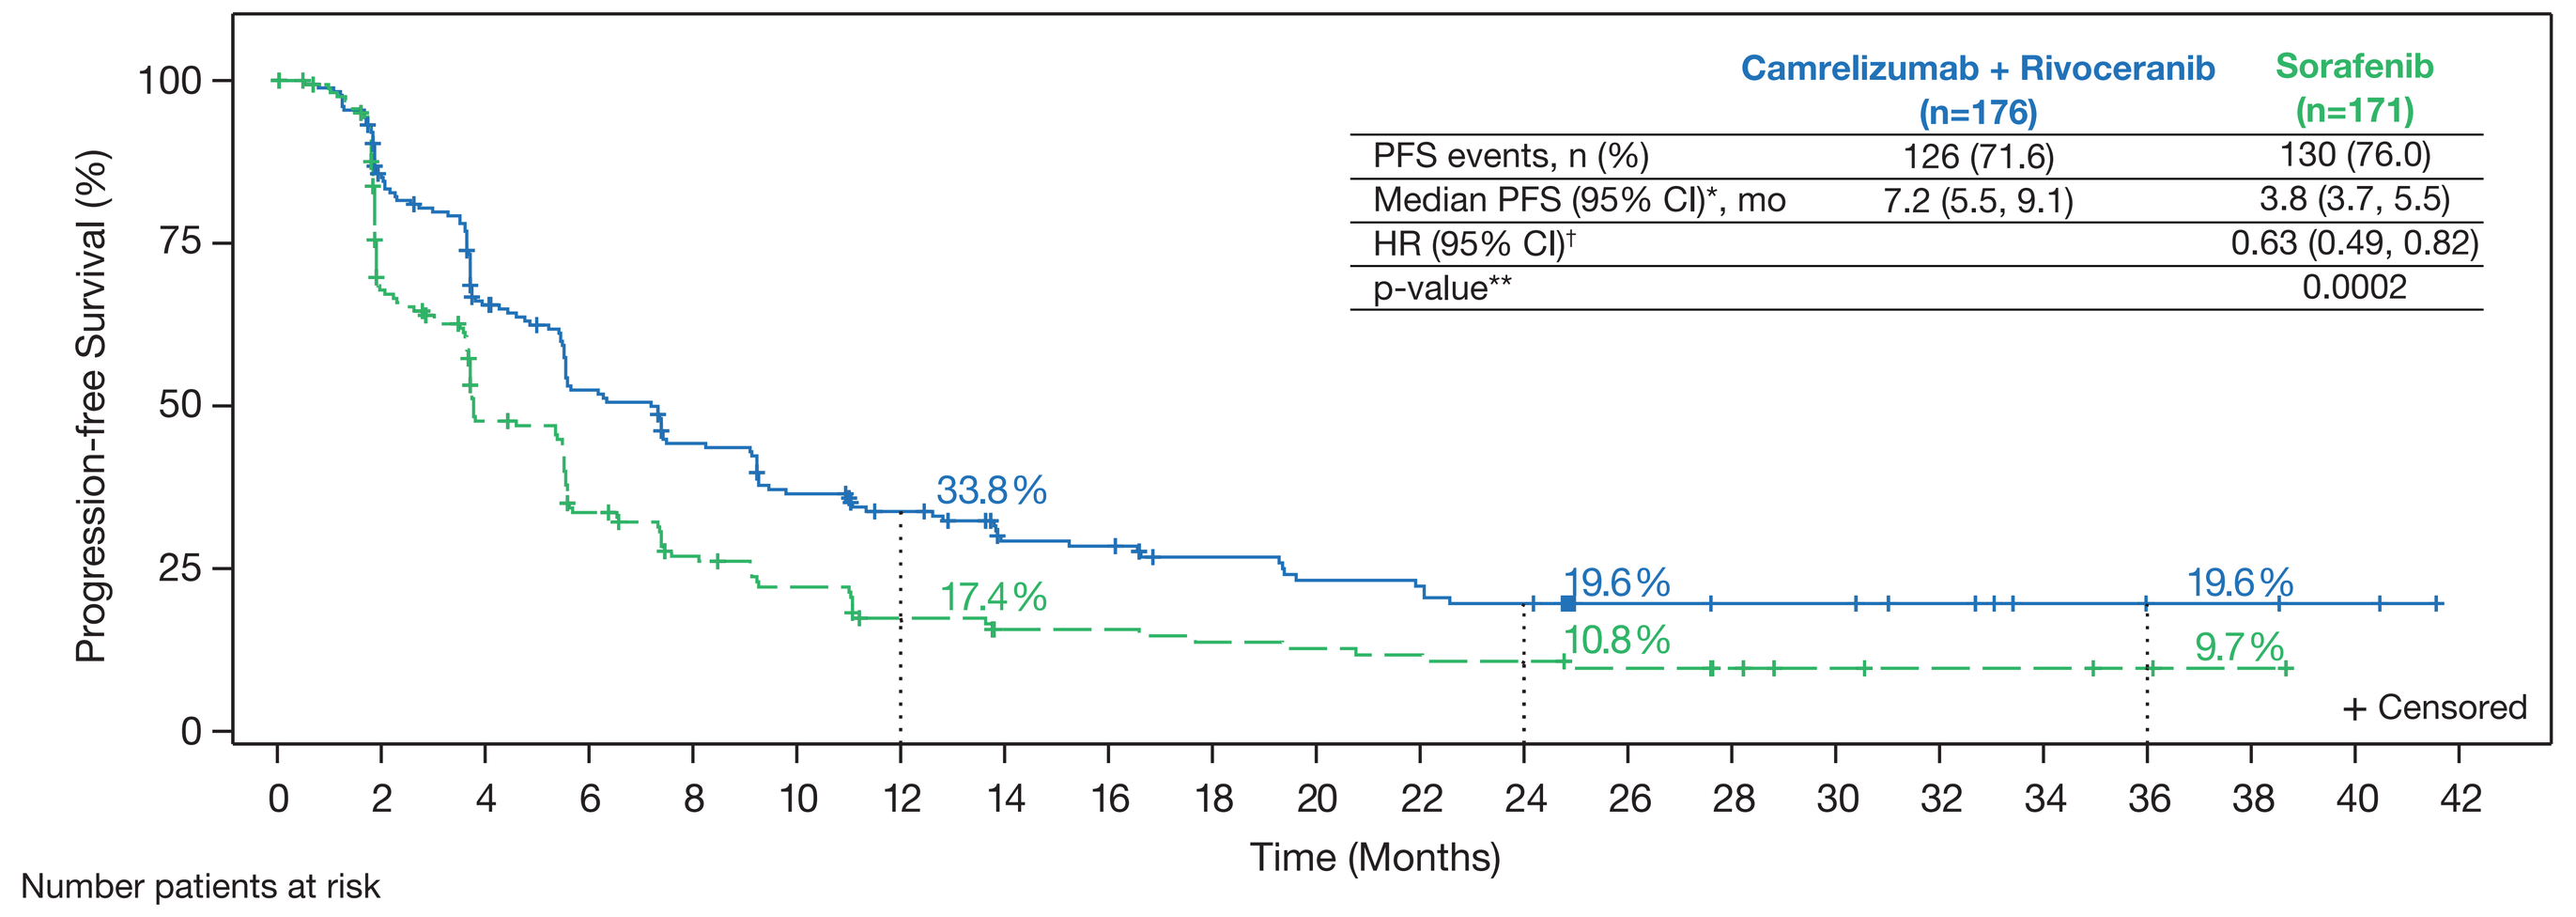
Camrelizumab + Rivoceranib (n=176)
Sorafenib (n=171)
PFS events, n (%) 126 (71.6) 130 (76.0)
Median PFS (95% CI)*, mo 7.2 (5.5, 9.1) 3.8 (3.7, 5.5)
HR (95% CI)† 0.63 (0.49, 0.82)
p-value** 0.0002
Progression-free Survival (%)
33.8%
17.4%
19.6%
10.8%
19.6%
9.7%
+ Censored
Time (Months)

Number patients at risk

Time (Months)	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32	34	36	38	40	42
Cam + Rivo	176	146	108	84	69	56	48	37	36	30	26	25	22	11	9	9	7	4	3	3	2	0
Sorafenib	171	105	69	47	35	28	20	16	16	14	13	12	11	9	7	4	3	3	2	1	0	

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence) and geographical region (Asia vs countries outside of Asia) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Figure 4: Progression-free Survival for Patients with Baseline AFP ≥400 ng/mL by BIRC Assessment (RECIST v1.1)

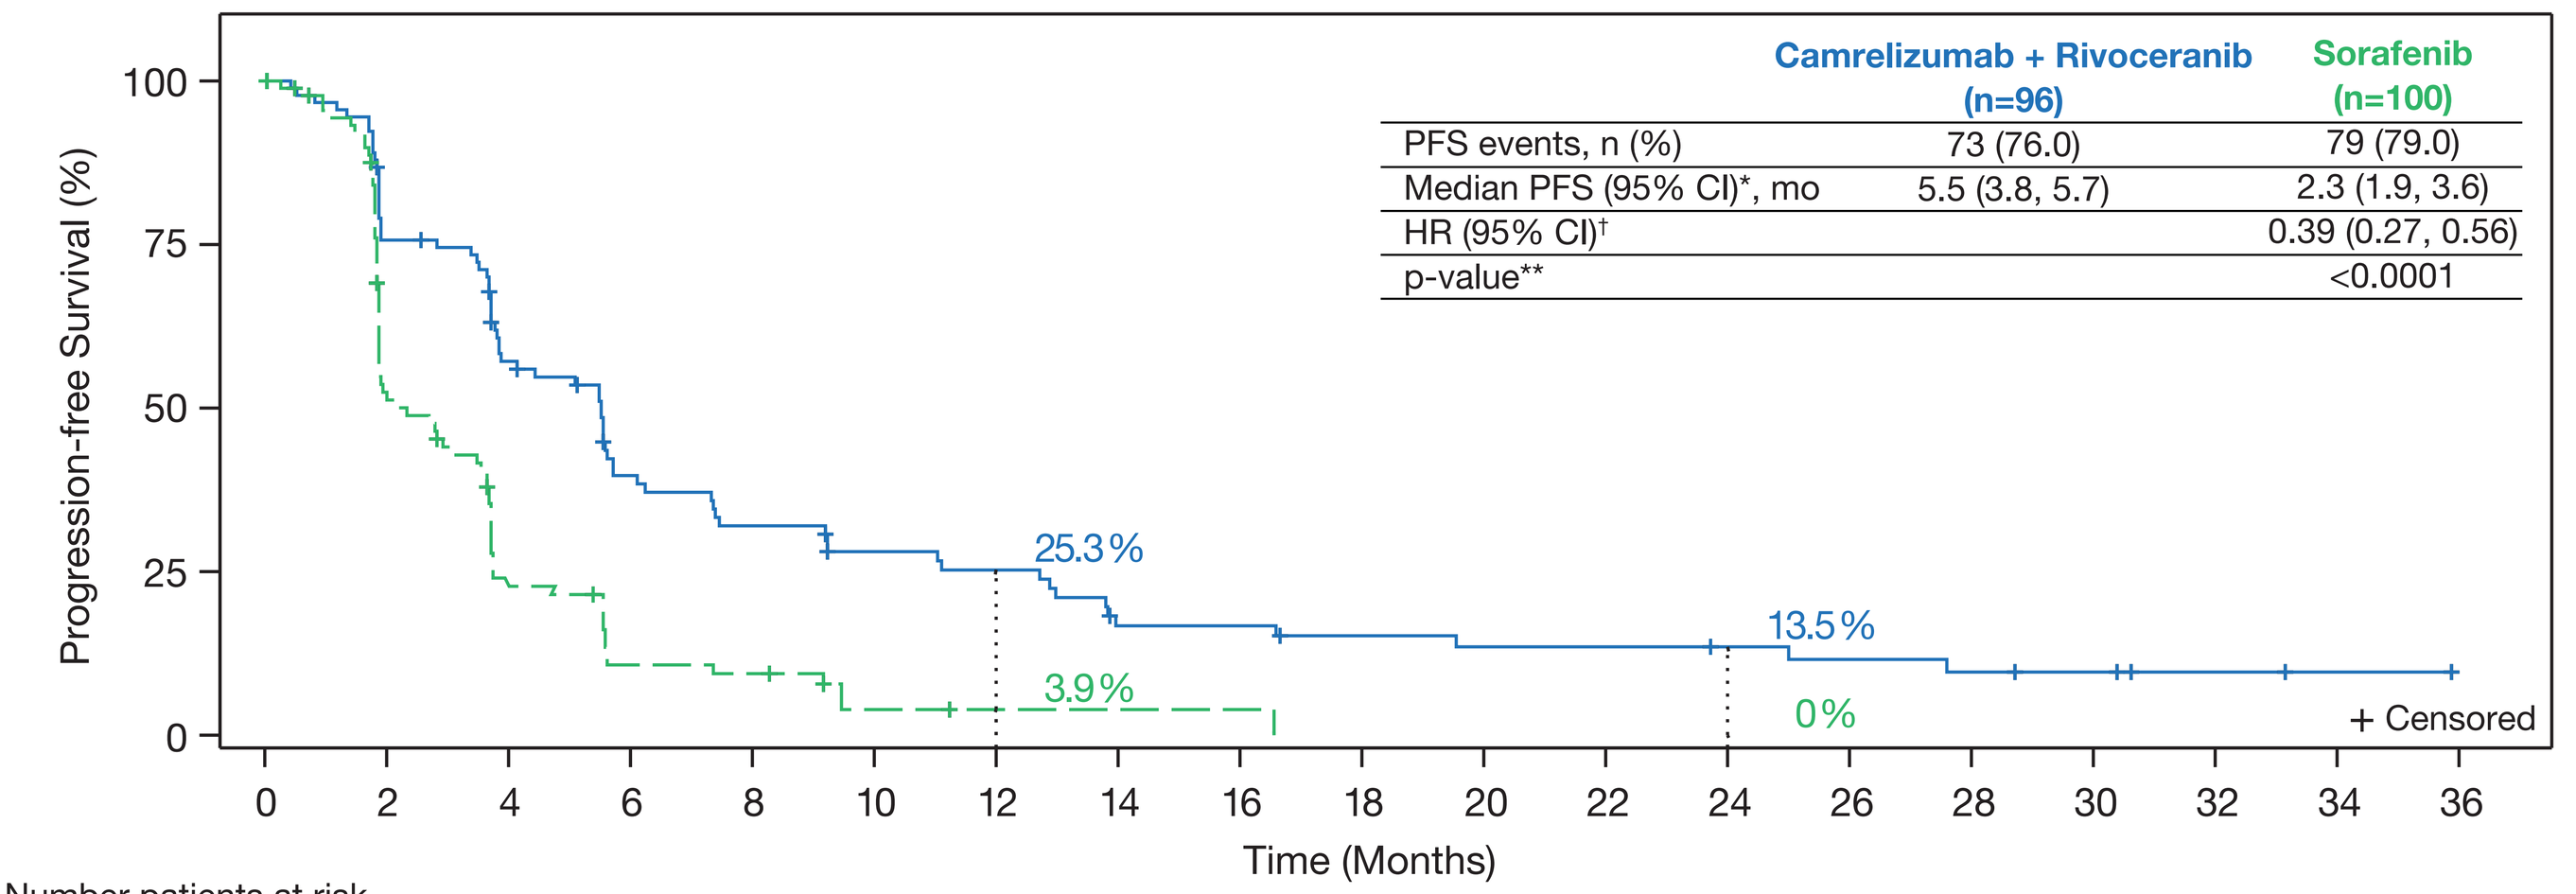
Camrelizumab + Rivoceranib (n=96)
Sorafenib (n=100)
PFS events, n (%) 73 (76.0) 79 (79.0)
Median PFS (95% CI)*, mo 5.5 (3.8, 5.7) 2.3 (1.9, 3.6)
HR (95% CI)† 0.39 (0.27, 0.56)
p-value** <0.0001
Progression-free Survival (%)
25.3%
3.9%
13.5%
0%
+ Censored
Time (Months)

Number patients at risk

Time (Months)	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32	34	36
Cam + Rivo	96	68	48	31	25	20	18	11	11	9	8	8	7	6	5	4	2	1	0
Sorafenib	100	44	18	8	7	2	1	1	1	0									

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence) and geographical region (Asia vs countries outside of Asia) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Table 4: Most Common (≥20%) Any Grade or Grade 3-4 (≥5%) TRAEs* for Patients with Baseline AFP ≥400 ng/mL (Safety Population)

TRAE, n (%)	Rivoceranib + Camrelizumab (N=96)		Sorafenib (N=100)	
	Any Grade	Grade 3-4	Any Grade	Grade 3-4
AST increased	37 (38.5)	9 (9.4)	34 (34.0)	6 (6.0)
ALT increased	29 (30.2)	8 (8.3)	25 (25.0)	2 (2.0)
Blood bilirubin increased	24 (25.0)	6 (6.3)	26 (26.0)	2 (2.0)
GGT increased	13 (13.5)	6 (6.3)	16 (16.0)	5 (5.0)
Platelet count decreased	23 (24.0)	4 (4.2)	31 (31.0)	2 (2.0)

*TRAEs include adverse events related to both rivoceranib + camrelizumab or sorafenib.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; GGT, gamma-glutamyl transferase.

REFERENCES

1. Qin S, et al. *Lancet*. 2023;402(10408):1133-1146.
2. Vogel A, et al. *J Clin Oncol*. 2024;42(suppl_16):abs 4110.
3. Zhang J, et al. *PLoS ONE*. 2020;15(2):e0228857.

AUTHOR INFO

COI for presenting author, Richard Kim: Consulting fees: Taiho, Bayer, Exelixis, Eisai, Roche, Ipsen, Abbvie, Servier, Elevar, Jazz, Takeda.
For questions or additional information, please contact medicalinformation@elevartherapeutics.com.

ACKNOWLEDGMENTS

- This study was funded by Jiangsu Hengrui Pharmaceuticals Co., Ltd. and Elevar Therapeutics. The Phillips Group Oncology Communications Inc. provided professional assistance with poster preparation. Financial support for writing and editorial services was provided by Elevar Therapeutics.

CONCLUSIONS

- Results of this CARES-310 exploratory post-hoc analysis indicate improved mOS and mPFS in the camrelizumab plus rivoceranib arm with baseline serum AFP levels <400 ng/mL or ≥400 ng/mL.
- Safety results showed that increased AST was the most common treatment-related adverse event, regardless of baseline serum AFP level.
- Independent of uHCC prognosis, camrelizumab plus rivoceranib demonstrated improved OS, PFS, and DCR, and if approved, may be a potential first line treatment option.