330

Budget Impact of Darolutamide + ADT for mCSPC from a United States Payer Perspective

David J. Campbell, PharmD, MS[1]; Bashir Kalayeh, PharmD[2]; Scott D. Ramsey, MD, PhD[1,3]; Manpreet Dhankhar, MS[1,4]; Sreevalsa Appukkuttan, MPH[2]

[1]Curta Inc. Seattle, Washington; [2]Bayer Pharmaceuticals Inc. Whippany, New Jersey. [3]Fred Hutchinson Cancer Center, Seattle, Washington; [4]University of Washington, Seattle, Washington

BACKGROUND

- Prostate cancer is the most common cause of cancer and a leading cause of cancer death among men in the United States[1]
- FDA recently approved darolutamide + androgen deprivation therapy (ADT) for metastatic castration-sensitive prostate cancer (mCSPC) based on efficacy and tolerability demonstrated in the ARANOTE study.[2,3]
- Following this year's FDA approval, there is a need for US healthcare plans to understand the potential economic impact for expanded use of darolutamide + ADT for the treatment of mCSPC.

RESEARCH OBJECTIVE

- To estimate the budget impact of expanded access of darolutamide + ADT from label expansion for the treatment of mCSPC from a US payer perspective.

METHODS

MODEL OVERVIEW

- An Excel-based budget impact model (BIM) was developed to evaluate the incremental costs between a scenario without darolutamide + ADT approval and a scenario with approval and formulary coverage for a hypothetical 1-million-member mixed (commercial and Medicare) US plan (**Figure 1**).
- Real-world utilization of darolutamide + ADT was assessed in the base case and no utilization of darolutamide + ADT was assessed in a scenario analysis for market shares without FDA approval.
- A one-way sensitivity analysis (OWSA) was conducted by varying each input by ±10% to assess parameter uncertainties and explore key model drivers.

MODEL INPUTS AND ASSUMPTIONS

- The annual number of patients with mCSPC eligible for treatment with darolutamide + ADT in a 1 million members in a commercial (78%) / Medicare (22%) health plan was estimated from published data.[1,4-6]
- The estimated number of patients with mCSPC eligible for treatment includes new incident cases of mPC at diagnosis and the annual number of patients that progress from non-metastatic disease.
- Comparators included doublet and triplet intensified regimens with ADT (enzalutamide, apalutamide, abiraterone, abiraterone + docetaxel, and darolutamide + docetaxel).
- Treatment dosing, duration, efficacy, and AE rates were obtained from clinical trial publications and package inserts. Costs were based on the WACs from Red Book® as of January 2025 (**Table 1**).[7-20]
- Administration costs for IV infusions and subcutaneous injections based on the CMS Physician Fee Schedule.[21]

Figure 1. Model Structure

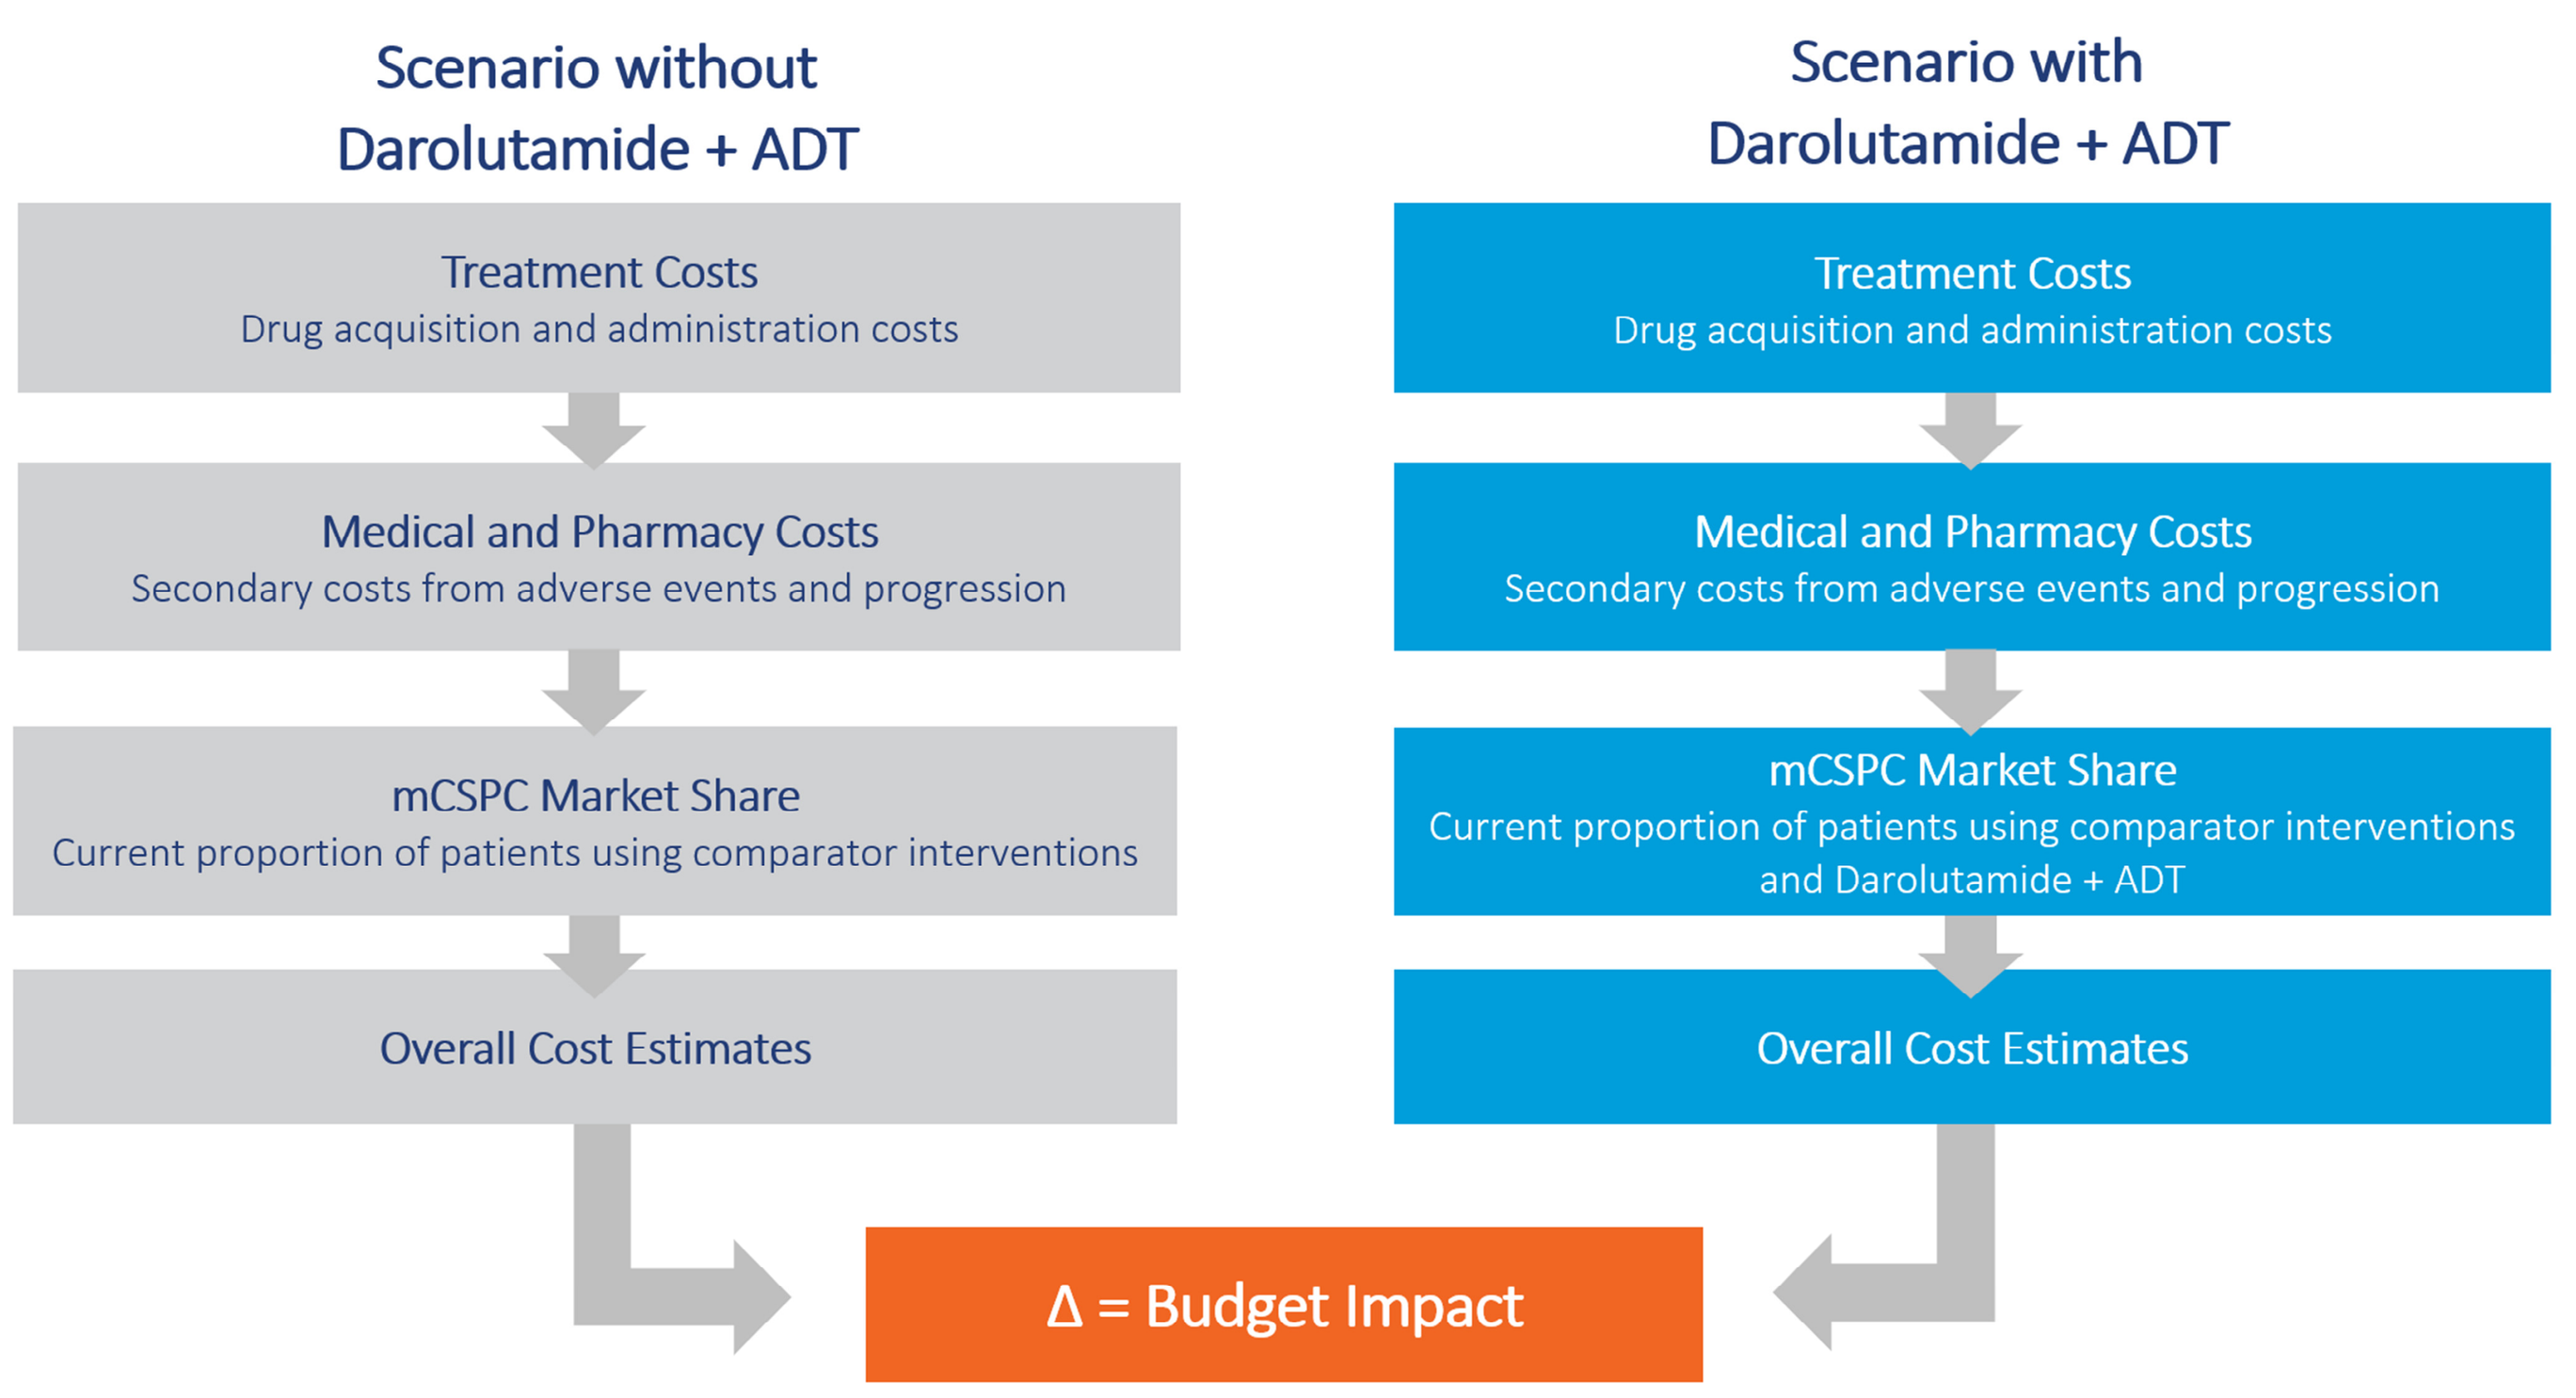
Scenario without Darolutamide + ADT
Treatment Costs
Drug acquisition and administration costs
Medical and Pharmacy Costs
Secondary costs from adverse events and progression
mCSPC Market Share
Current proportion of patients using comparator interventions
Overall Cost Estimates
Scenario with Darolutamide + ADT
Treatment Costs
Drug acquisition and administration costs
Medical and Pharmacy Costs
Secondary costs from adverse events and progression
mCSPC Market Share
Current proportion of patients using comparator interventions and Darolutamide + ADT
Overall Cost Estimates
Δ = Budget Impact

- AE costs were included for grade ≥3 AEs that occurred at a rate of ≥3% for at least one comparator regimen included and were inflation-adjusted to 2024 USD.[2,7-11,20]
- Incremental healthcare resource utilization (HCRU) costs for disease progression were modeled for each comparator with radiographic progression free survival (rPFS) rates at 12 and 24 months from trials.[22]
- Market share of mCSPC treatments was derived from a demand study of expert physicians conducted by Bayer. Darolutamide + ADT are projected to have a market uptake of 9-16% over years 1-3.[23]
 - The study excluded 1st generation androgen receptor inhibitor, ADT monotherapy, and off-label treatments

Table 1. Drug, Administration, Adverse Event and Progression Costs Inputs

Treatment Regimen	Dosing Regimens	Monthly Drug Cost*	Monthly Administration Cost	AE Cost (one-time)	Annual Progression Costs
Darolutamide + ADT	*Darolutamide*: Two 300 mg tablets twice daily	**$15,094**	**$27**	**$2,297**	**$750**
Apalutamide + ADT	*Apalutamide*: 240 mg per day	**$16,175**	**$27**	**$3,112**	**$750**
Enzalutamide + ADT	*Enzalutamide*: 160 mg per day	**$15,768**	**$27**	**$2,966**	**$667**
Abiraterone + ADT	*Abiraterone*: 1,000 mg orally once daily with prednisone 5 mg orally once daily	**$1,925**	**$27**	**$7,611**	**$917**
Abiraterone + Docetaxel + ADT	*Abiraterone*: 1,000 mg orally once daily with prednisone 5 mg orally twice daily *Docetaxel*: 75 mg/m² once every 3 weeks	**$2,292**	**$203**	**$6,678**	**$625**
Darolutamide + Docetaxel + ADT	*Darolutamide*: Two 300 mg tablets twice daily *Docetaxel*: 75 mg/m² once every 3 weeks	**$15,453**	**$203**	**$5,244**	**$584**

*Monthly drug costs including ADT drug costs based on reported wholesale acquisition cost (WAC)

RESULTS

BASE CASE

- Each year an estimated 44 men within a 1-million-member plan will be newly diagnosed or present with mCSPC progression and will receive intensified treatment (**Figure 2**).
- The incremental cost from approval and expanded use of darolutamide + ADT was estimated to be $0.005 to $0.026 in year 1 and in year 3 on a per-member per-month basis (**Figure 3**)
 - Total budget impact costs ranged from $57,709 in year 1 and $309,015 in year 3 (**Table 2**).
- Higher drug acquisition costs in the first year were partially offset by lower AE and disease progression related costs.

Figure 2. mCSPC Model Population

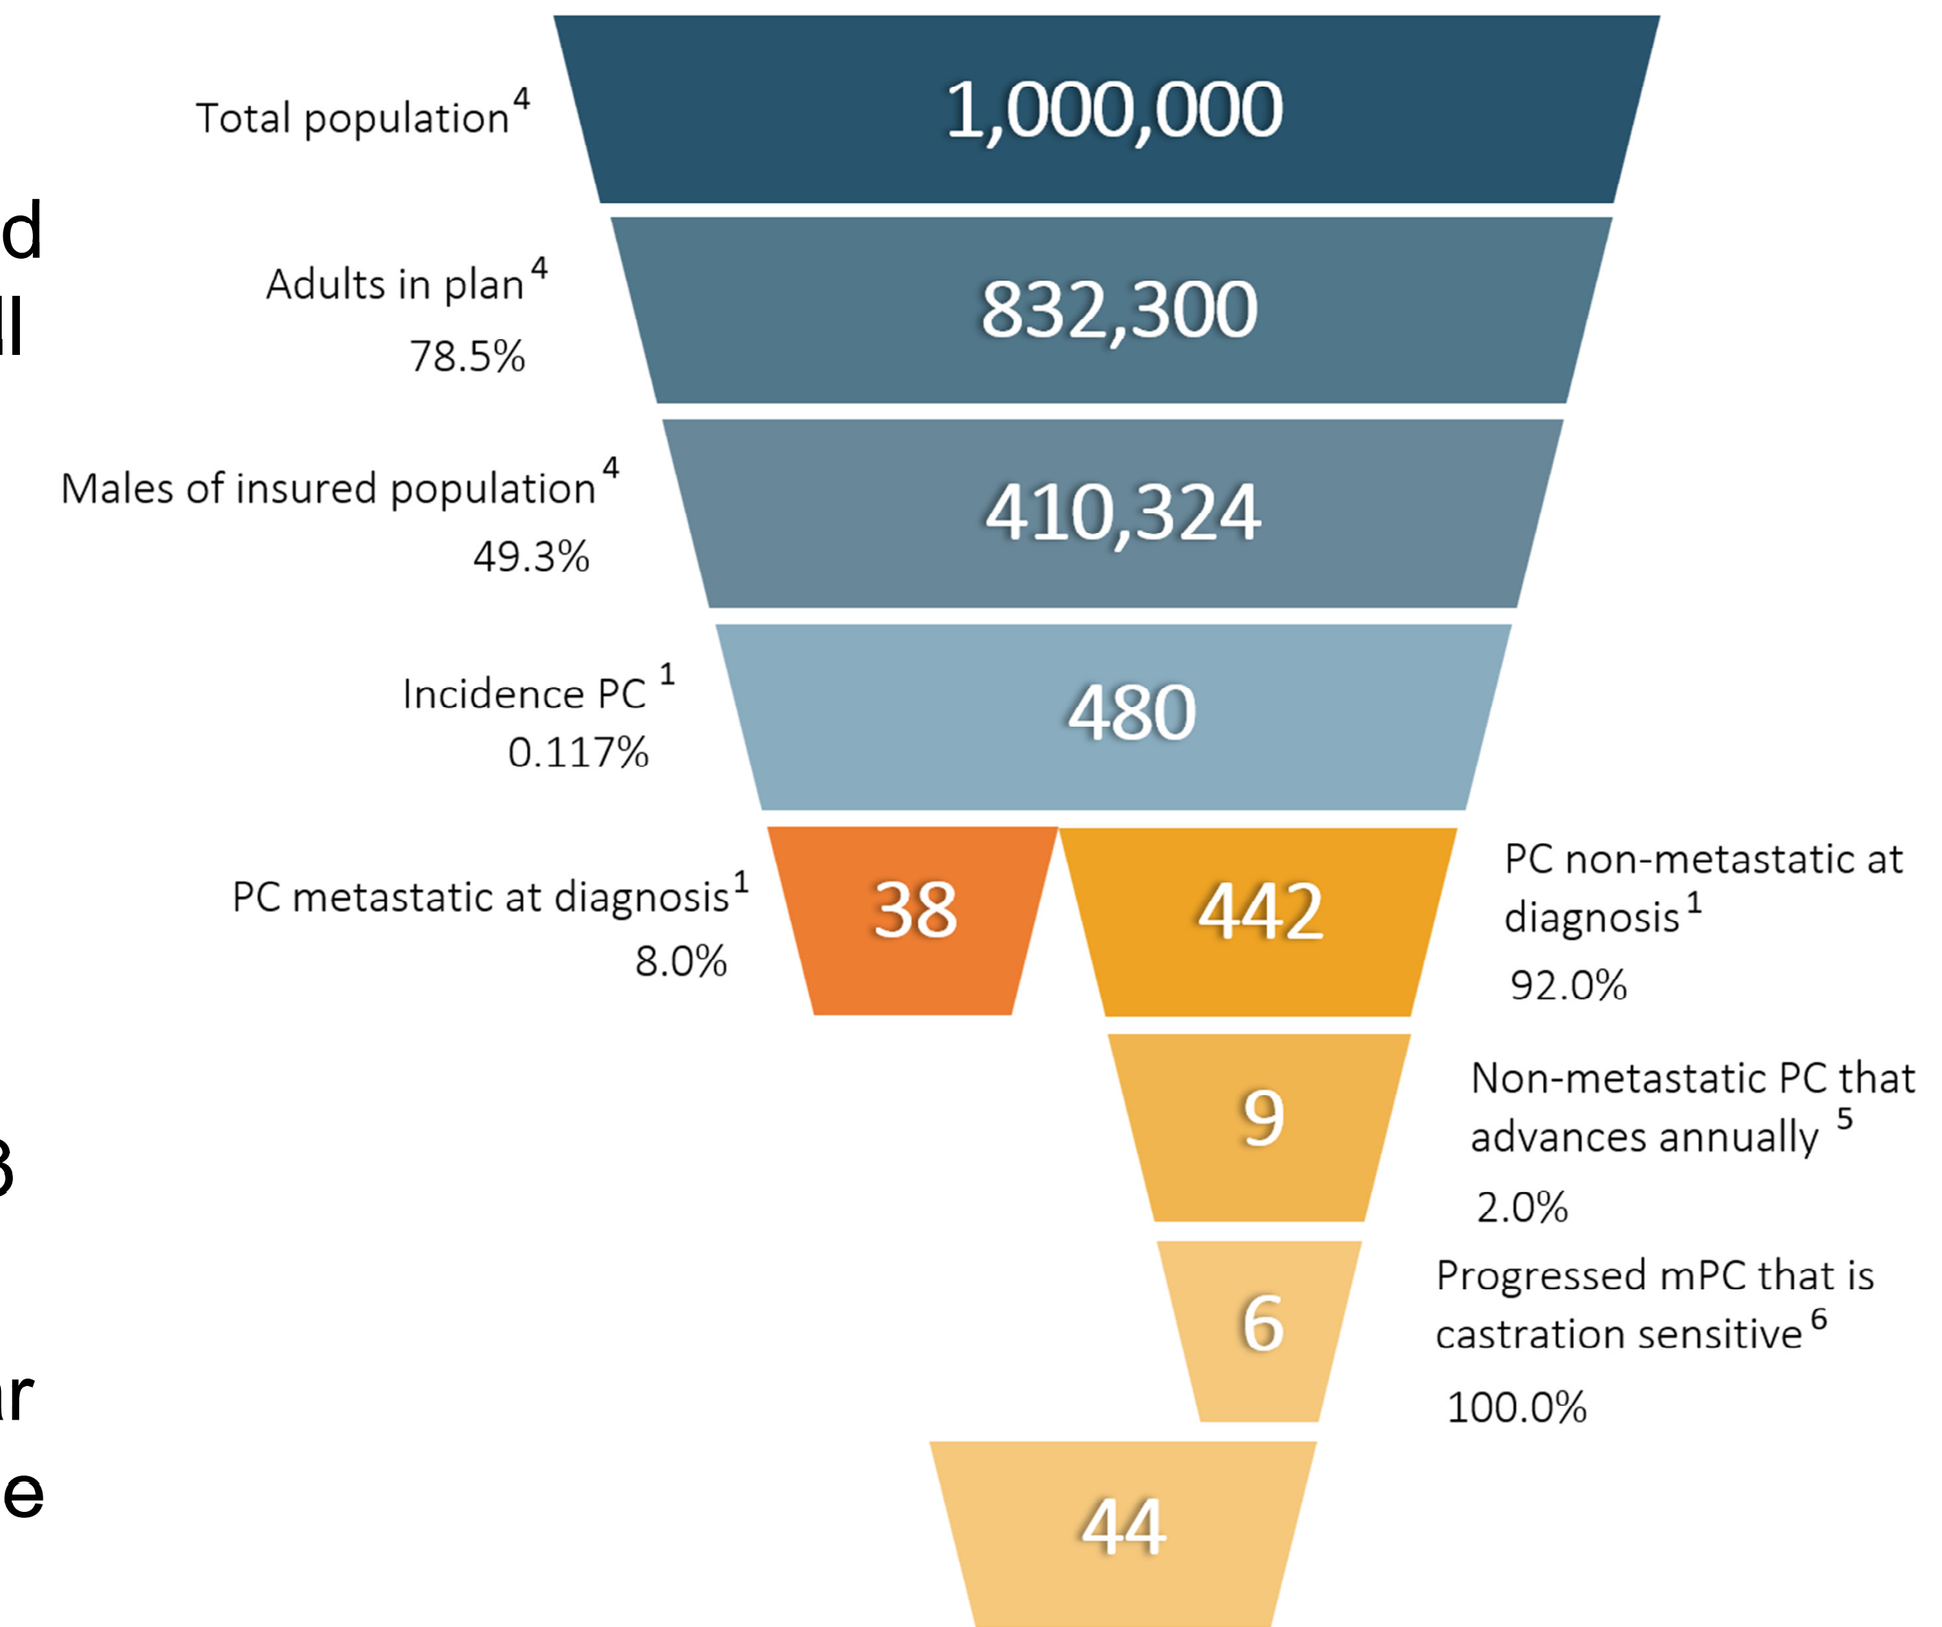
Total population[4]
1,000,000
Adults in plan[4]
78.5%
832,300
Males of insured population[4]
49.3%
410,324
Incidence PC[1]
0.117%
480
PC metastatic at diagnosis[1]
8.0%
38
442
PC non-metastatic at diagnosis[1]
92.0%
9
Non-metastatic PC that advances annually[5]
2.0%
6
Progressed mPC that is castration sensitive[6]
100.0%
44

Table 2. Budget Impact Model Base Case Results

Budget Impact Summary	Year 1	Year 2	Year 3
Δ Total Cost	**$57,709**	**$171,092**	**$309,015**
Treatment costs	**$60,763**	**$179,867**	**$322,258**
Adverse event costs	**-$3,016**	**-$8,867**	**-$13,518**
Disease progression HCRU costs	**-$37**	**$92**	**$274**

Figure 3. PMPM Budget Impact

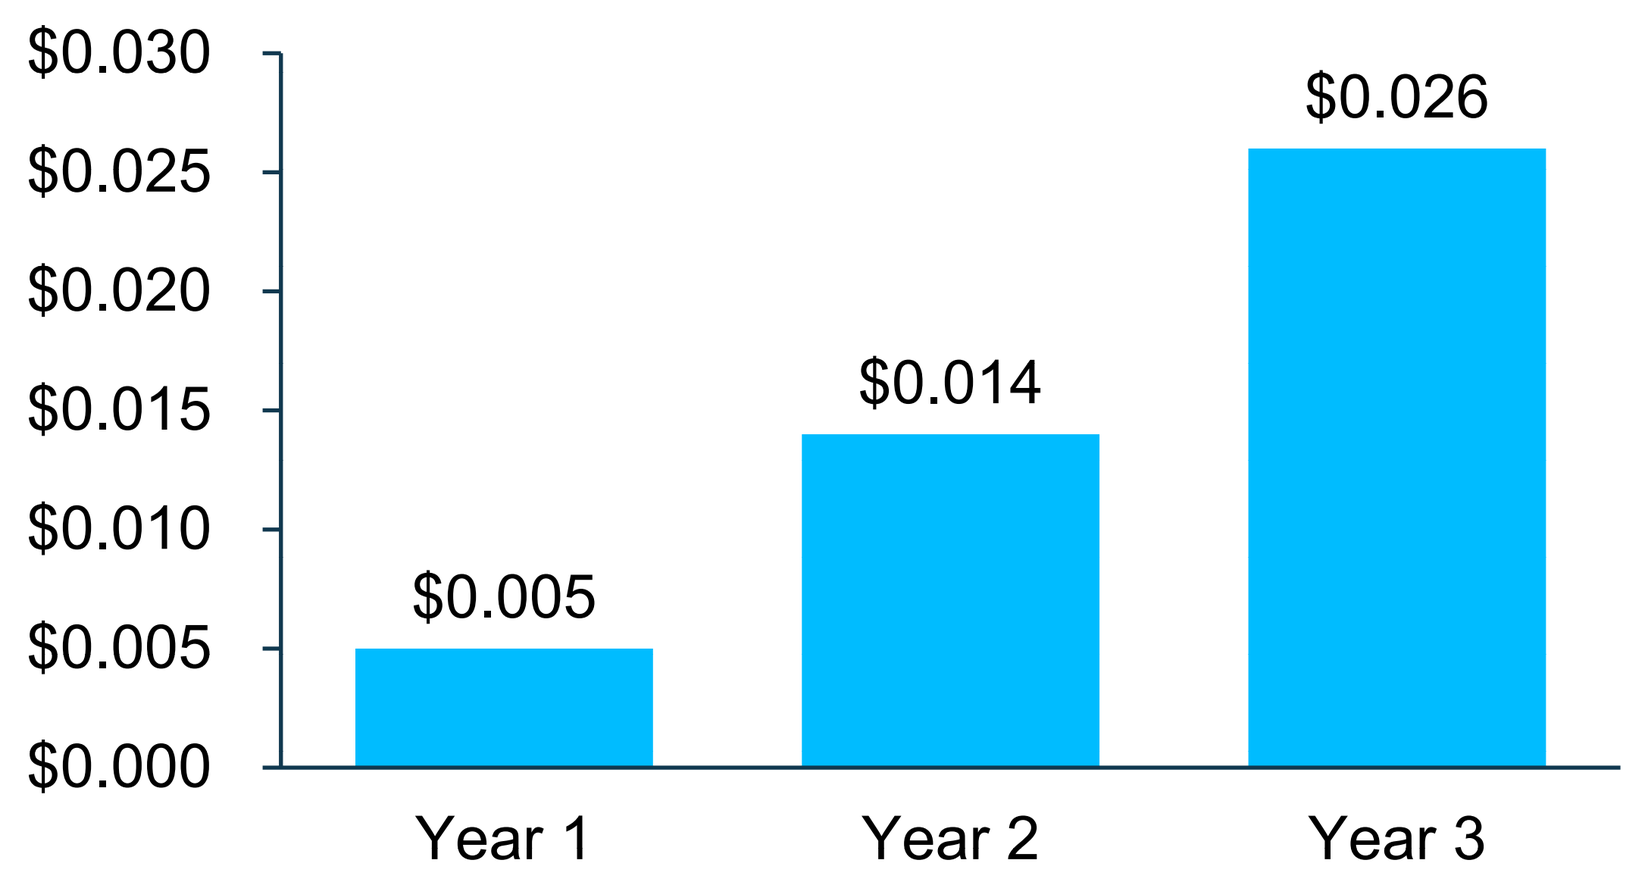
$0.030
$0.025
$0.020
$0.015
$0.010
$0.005
$0.000
$0.005
$0.014
$0.026
Year 1
Year 2
Year 3

SCENARIO ANALYSIS

- No utilization of darolutamide + ADT in the scenario without approval was assessed in a scenario analysis.
- In this scenario the budget impact remained small and ranged from $0.004 to $0.025 PMPM over the 3-year model horizon (**Table 3**).

Table 3. Scenario Analysis Results

Budget Impact Summary	Year 1	Year 2	Year 3
Δ Total Cost	**$44,517**	**$157,900**	**$299,927**
Δ PMPM Cost	**$0.004**	**$0.013**	**$0.025**

LIMITATIONS

- The modeled drug acquisition cost of enzalutamide does not account for the anticipated loss of exclusivity in 2027 and potential reduction to the wholesale acquisition cost.
- Given the use of retrospective studies to inform the model, results may only be generalizable to insured male patients in the US who are 18 or older with prostate cancer.

ONE-WAY SENSITIVITY ANALYSIS

- Across all sensitivity scenario analyses, the approval of darolutamide + ADT and inclusion on formulary had a minimal PMPM budget impact.
- OWSA showed that the budget impact on healthcare costs over a 1-year time horizon were most sensitive to darolutamide brand package cost (**Figure 4**).

Figure 4. One-Way Sensitivity Analysis

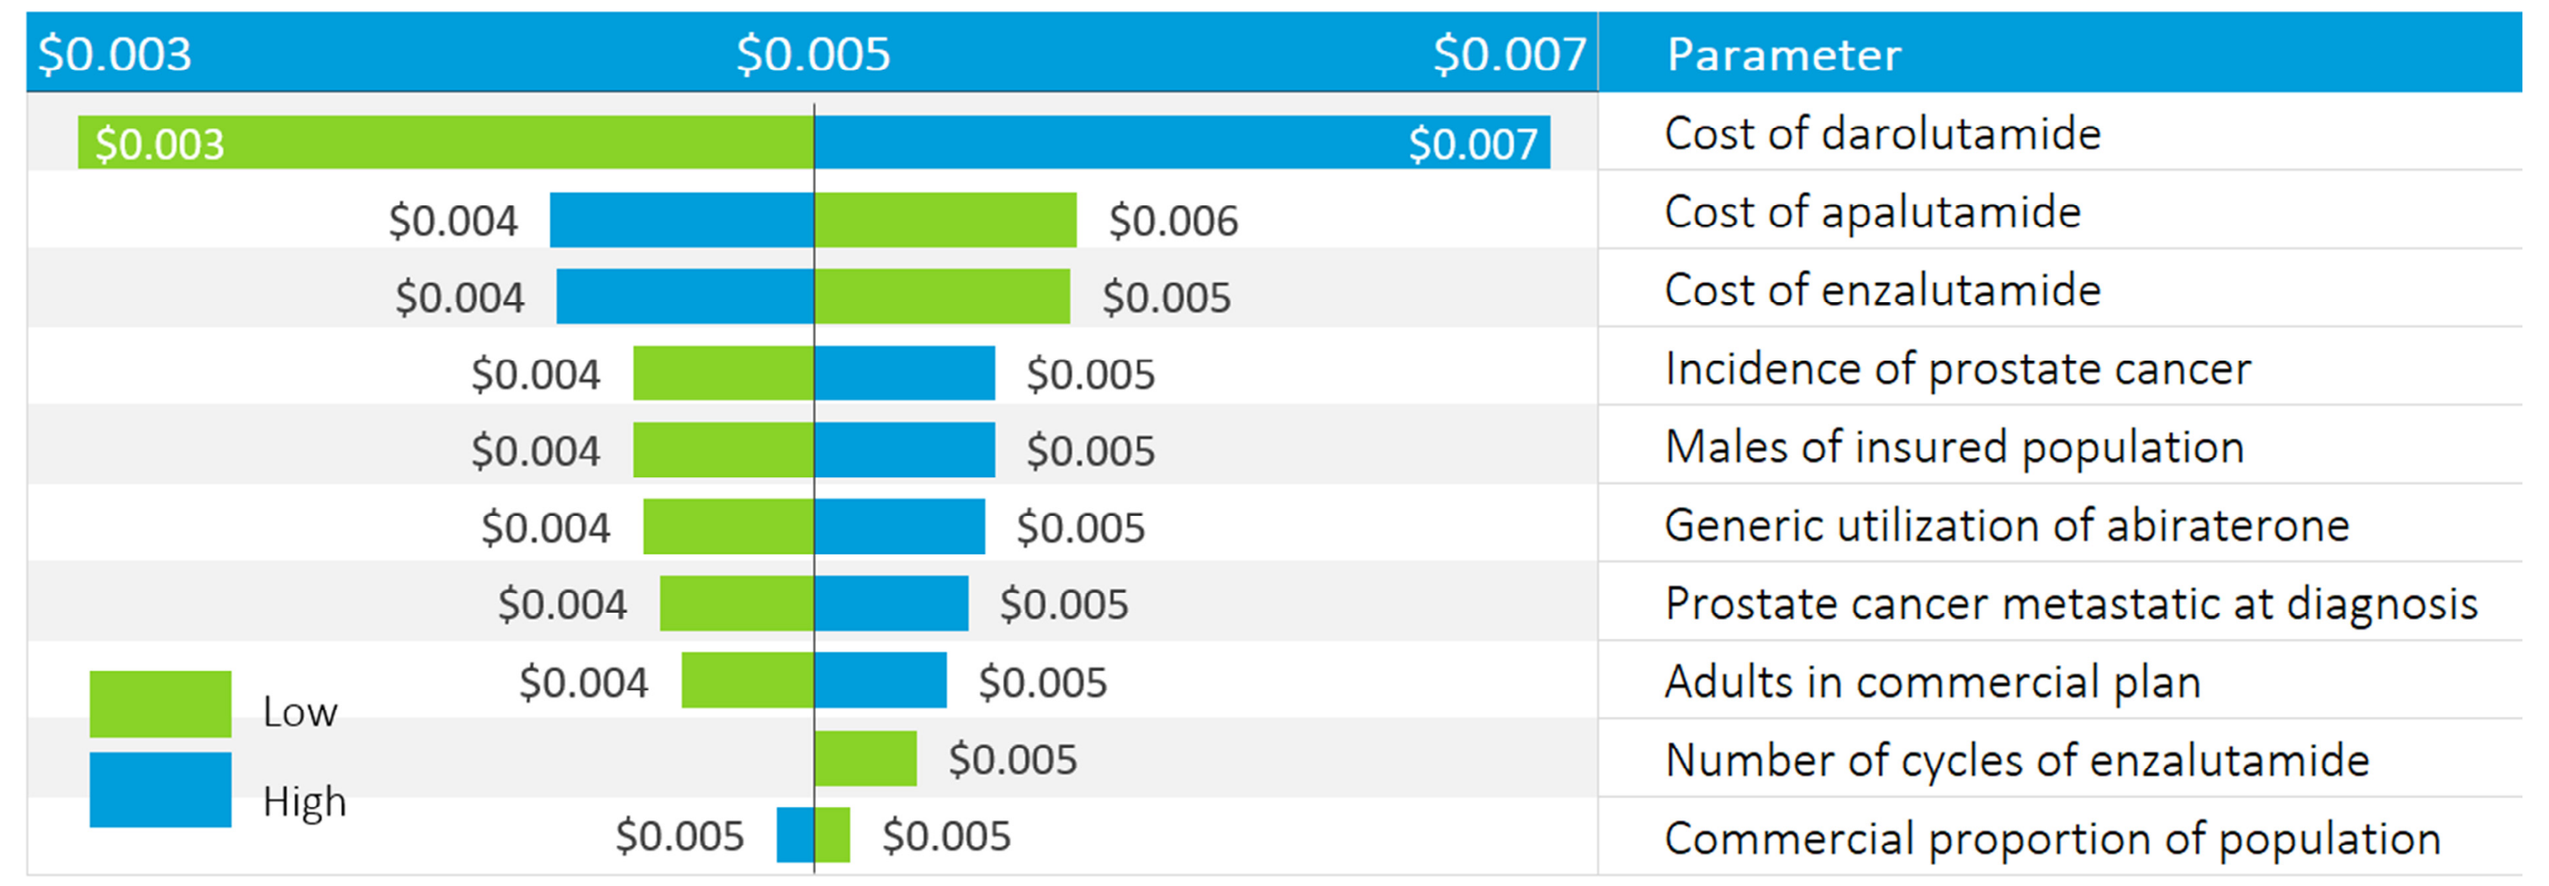
$0.003
$0.005
$0.007
Parameter
$0.003
$0.007
Cost of darolutamide
$0.004
$0.006
Cost of apalutamide
$0.004
$0.005
Cost of enzalutamide
$0.004
$0.005
Incidence of prostate cancer
$0.004
$0.005
Males of insured population
$0.004
$0.005
Generic utilization of abiraterone
$0.004
$0.005
Prostate cancer metastatic at diagnosis
$0.004
$0.005
Adults in commercial plan
$0.005
Number of cycles of enzalutamide
$0.005
$0.005
Commercial proportion of population
Low
High

CONCLUSIONS

- **For US health plans, providing expanded access to darolutamide + ADT for the treatment of adults with mCSPC results in minimal budget impact over a 3-year time horizon.**
- **The budget impact of darolutamide + ADT was partially offset from adverse-event management cost savings.**

Acknowledgments

Research Sponsor: Bayer Healthcare Pharmaceuticals Inc.

References

1. SEER Stat Facts: Prostate cancer. https://seer.cancer.gov/statfacts/html/prost.html. **2.** Saad F, et al. *J Clin Oncol*. 2024 Sep 16:JCO2401798. **3.** Nubeqa (darolutamide). http://labeling.bayerhealthcare.com/-html/products/pi/Nubeqa_PI.pdf. **4.** US Census Bureau. Available at: https://www.census.gov/library/publications/-2024/demo/p60-284.html. **5.** Bill-Axelson A, et al. *N Engl J Med*. 2014 Mar 6;370(10):932-42. **6.** Eule CJ, et al. *Urol Oncol*. 2024 Dec;42(12):447.e17-447.e24. **7.** Armstrong AJ, et al. *J Clin Oncol*. 2022 May 20;40(15):1616-1622. **8.** Chi KN, et al. *N Engl J Med*. 2019 Jul 4;381(1):13-24. **9.** Fizazi K, et al. *N Engl J Med*. 2017 Jul 27;377(4):352-360. **10.** Fizazi K, et al. *Lancet*. 2022 Apr 30;399(10336):1695-1707. **11.** Smith MR, et al. *N Engl J Med*. 2022 Mar 24;386(12):1132-1142. **12.** Casodex (bicalutamide). https://www.accessdata.fda.gov/-drugsatfda_docs/label/2017/020498s028lbl.pdf. **13.** Erleada (apalutamide). https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/ERLEADA-pi.pdf. **14.** Firmagon (degarelix for injection). http://www.ferringusa.com/pi/firmagon. **15.** Lupron (leuprolide acetate for depot suspension). https://www.rxabbvie.com/pdf/lupronuro_pi.pdf. **16.** Xtandi (enzalutamide). https://www.astellas.us/docs/us/12A005-ENZ-WPI.pdf. **17.** Zytiga (abiraterone acetate). https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/ZYTIGA-pi.pdf. **18.** HCUPnet. https://datatools.ahrq.gov/hcupnet/. **19.** Red Book. Available at: https://www.merative.com. Accessed January 2025. **20.** BLS. Consumer Price Index, all items in U.S. city average. Available at: https://www.bls.gov/data/. **21.** CMS. Physician Fee Schedule. Available at: https://www.cms.gov/medicare/physician-fee-schedule/search. Accessed February 2025. **22.** Desmaras H, et al. *Value Health*, Volume 27, Issue 6, S122. **23.** Bayer market share data on file. 2024 US Demand Study of Physicians.

AMCP Nexus, National Harbor, MD, Oct 27-30, 2025

Presented by Bashir Kalayeh; bashir.kalayeh@bayer.com