Third Interim Subgroup Analysis of the Effectiveness and Safety of Damoctocog Alfa Pegol in Patients with Hemophilia A Treated Every 5 or Every 7 Days: Results from the Real-World HEM-POWR Study

Publication No. PB1263

Mark T. Reding,[1] María Teresa Alvarez Román,[2] Giancarlo Castaman,[3] Maissaa Janbain,[4] Tadashi Matsushita,[5] Karina Meijer,[6]* Kathrin Schmidt,[7] Johannes Oldenburg[8]

[1]University of Minnesota Medical Center, Minneapolis, Minnesota, US; [2]Hospital Universitario La Paz, Madrid, Spain; [3]Careggi University Hospital, Florence, Italy; [4]Tulane School of Medicine, New Orleans, Louisiana, US; [5]Nagoya University Hospital, Nagoya, Japan; [6]University Medical Center Groningen, Groningen, the Netherlands; [7]Bayer, Berlin, Germany; [8]University Clinic Bonn, Bonn, Germany

Contact details for presenting author: Karina Meijer, k.meijer@umcg.nl

CONCLUSION

- **Extended dosing intervals of damoctocog alfa pegol of every 5 days (E5D) and every 7 days (E7D) continue to demonstrate effectiveness and an acceptable safety profile in previously-treated patients (PTPs) with hemophilia A in real-world clinical settings.**
- **Patients treated with damoctocog alfa pegol E5D and E7D demonstrated improvements in annualized bleeding rate (ABR) and joint health compared with prior to initiation of damoctocog alfa pegol, suggesting extended dosing regimens may be a viable treatment approach in real-world scenarios.**
- **The safety profile of E5D and E7D damoctocog alfa pegol were consistent with standard dosing regimens, with no study-drug-related treatment-emergent adverse events or discontinuations reported.**

AIMS

- **To explore the effectiveness and safety of E5D and E7D prophylactic damoctocog alfa pegol dosing in the ongoing real-world HEM-POWR study (NCT03932201).**

INTRODUCTION

- The standard of care for hemophilia A is prophylaxis with Factor VIII (FVIII) replacement therapy. Compared with established standard half-life therapies, extended half-life (EHL) FVIII products may lessen treatment burden and improve adherence for patients with hemophilia A by reducing dosing frequency.[1]
- Damoctocog alfa pegol (BAY 94-9027, Jivi®) is a PEGylated B-domain-deleted recombinant FVIII EHL product approved for the treatment of PTPs aged ≥12 years with hemophilia A.[2,3]
- In the US, the recommended routine prophylactic dosing regimen is twice weekly. Based on occurrence of bleeding episodes, the regimen can be adjusted to E5D or further adjusted to the individual as required.[4]
- Previous interim analyses of the ongoing HEM-POWR study have reported on the real-world safety and effectiveness of any dosing regimen of damoctocog alfa pegol[5,6]; prior data have also been presented for extended dosing regimens.[7]
- This poster will present updated real-world analysis of extended dosing regimens in PTPs with hemophilia A.

METHODS

- HEM-POWR is a Phase 4 ongoing, prospective, multinational, observational, open-label, cohort study of damoctocog alfa pegol in PTPs with hemophilia A.
- This third interim analysis of the HEM-POWR study includes a subgroup of PTPs who were prescribed damoctocog alfa pegol E5D or E7D.
- The primary endpoint was ABR; secondary endpoints included joint health and safety, reported as treatment-emergent adverse events (TEAEs).
- Baseline characteristics and endpoints were reported as descriptive statistics. Data were collated from patient diaries and physician records and described in a full analysis set (FAS) and safety analysis set (SAF). Ethical approval was obtained for all sites.
- Patients included in the SAF had ≥1 study dose in the observation period and provided informed consent; the FAS was defined as patients who fulfilled all inclusion criteria with a documented first study drug dose and ≥1 infusion during the observation period.

RESULTS

- At data cut-off (August 17, 2022), 161 patients were included in the interim HEM-POWR study FAS; 54 patients received either E5D (n=31) or E7D (n=23) regimens at initial visit.
- Baseline characteristics were similar between groups. Median (Q1, Q3) observation period for the E5D group was 323.0 (183.0, 464.0) and for the E7D group was 331.0 (152.0, 428.0) days in the FAS **(Table 1).**

Table 1: BASELINE DEMOGRAPHICS AND CHARACTERISTICS FOR PATIENTS RECEIVING E5D AND E7D DAMOCTOCOG ALFA PEGOL DOSING REGIMENS (FAS)

Characteristic	E5D (n=31)	E7D (n=23)
Observation period, days, median (Q1, Q3)	323.0 (183.0, 464.0)	331.0 (152.0, 428.0)
Male, n (%)	31 (100.0)	23 (100.0)
Race, n (%)		
White	17 (54.8)[a]	7 (30.4)[b]
Black or African American	1 (3.2)	0
Asian	9 (29.0)	14 (60.9)
Native Hawaiian or other Pacific islander	0	1 (4.4)
Age at enrollment, years, median (Q1, Q3)	28 (21.0, 45.0)	38 (32.0, 58.0)
Country of recruitment, n (%)		
Canada	1 (3.2)	1 (4.4)
Denmark	3 (9.7)	1 (4.4)
Germany	6 (19.4)	3 (13.0)
Italy	3 (9.7)	0
Japan	4 (12.9)	12 (52.2)
Spain	1 (3.2)	2 (8.7)
Sweden	1 (3.2)	0
Taiwan	4 (12.9)	2 (8.7)
USA	8 (25.8)	2 (8.7)
Disease severity at diagnosis, n (%)		
Non-severe	3 (9.7)[c]	6 (26.1)
Severe	27 (87.1)	17 (73.9)
Family history of hemophilia, yes, n (%)	19 (61.3)[d]	7 (30.4)[e]
Time from diagnosis to initial visit, years, median (Q1, Q3)	29.9 (21.5, 45.3)	35.8 (31.4, 57.2)

[a]missing data for 4 (12.9%) patients; [b]missing data for 1 (4.4%) patient; [c]missing data for 1 (3.2%) patient; [d]missing data for 1 (3.2%) patients; [e]missing data for 2 (8.7%) patients.
E5D, every 5 days; E7D, every 7 days; FAS, Full Analysis Set; Q1, first quartile; Q3, third quartile; SD, standard deviation.

- At baseline, the proportion of patients on E5D dosing with severe and non-severe (moderate and mild) hemophilia A was 87.1% (n=27/31) and 9.7% (moderate n=2/31, mild n=1/31; one missing), respectively. The proportion of patients on E7D dosing regimen with severe and non-severe hemophilia A was 73.9% (n=17/23) and 26.1% (moderate n=5/23, mild n=1/23), respectively.
- For the E5D dosing regimen, the total median (mean, SD) ABR during the observation period was 0.0 (0.8, 1.2); ABR for the E7D group was 0.0 (1.8, 5.5) **(Figure 1).**

Figure 1: ABR PRIOR TO INITIATION OF DAMOCTOCOG ALFA PEGOL AND DURING THE OBSERVATION PERIOD FOR THE E5D (A) AND E7D (B) GROUP (FAS)

A. E5D (n=31)

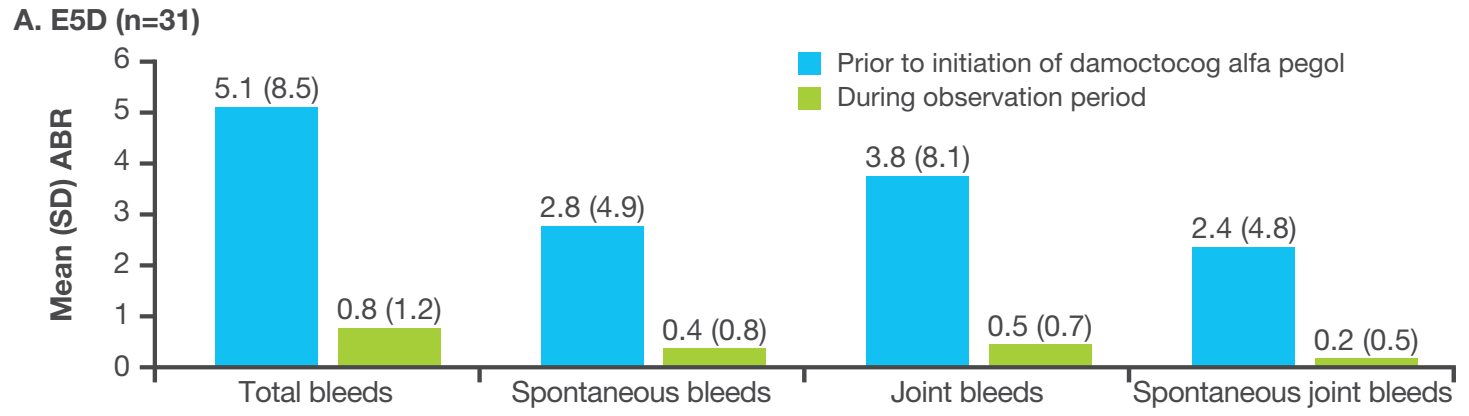
Mean (SD) ABR
0
1
2
3
4
5
6
Prior to initiation of damoctocog alfa pegol
During observation period
5.1 (8.5)
0.8 (1.2)
2.8 (4.9)
0.4 (0.8)
3.8 (8.1)
0.5 (0.7)
2.4 (4.8)
0.2 (0.5)
Total bleeds
Spontaneous bleeds
Joint bleeds
Spontaneous joint bleeds

	Total bleeds	Spontaneous bleeds	Joint bleeds	Spontaneous joint bleeds
Prior to initiation of damoctocog alfa pegol, median (Q1, Q3)	3.0 (0.0, 5.0)	1.0 (0.0, 4.0)	1.0 (0.0, 4.0)	0.0 (0.0, 3.0)
During observation period, median (Q1, Q3)	0.0 (0.0, 1.8)	0.0 (0.0, 0.0)	0.0 (0.0, 0.9)	0.0 (0.0, 0.0)

B. E7D (n=23)*

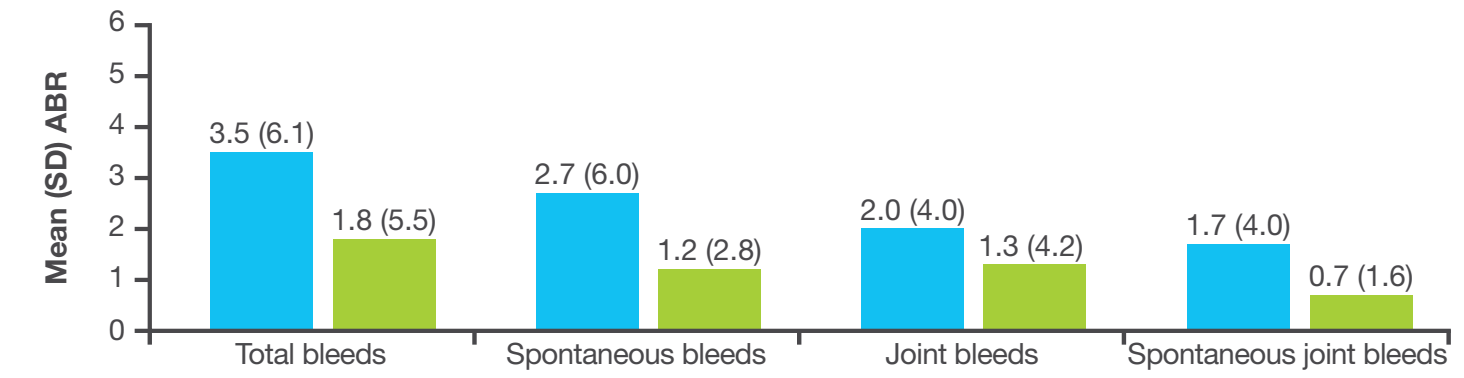
Mean (SD) ABR
0
1
2
3
4
5
6
3.5 (6.1)
1.8 (5.5)
2.7 (6.0)
1.2 (2.8)
2.0 (4.0)
1.3 (4.2)
1.7 (4.0)
0.7 (1.6)
Total bleeds
Spontaneous bleeds
Joint bleeds
Spontaneous joint bleeds

	Total bleeds	Spontaneous bleeds	Joint bleeds	Spontaneous joint bleeds
Prior to initiation of damoctocog alfa pegol, median (Q1, Q3)	1.5 (0.0, 3.0)	1.0 (0.0, 3.0)	0.0 (0.0, 3.0)	0.0 (0.0, 2.0)
During observation period, median (Q1, Q3)	0.0 (0.0, 1.2)	0.0 (0.0, 1.1)	0.0 (0.0, 0.9)	0.0 (0.0, 0.9)

Data missing for 1 (4.3%) patient prior to damoctocog alfa pegol initiation. Data from 12 months prior to initiation were the average number of bleeds over 12 months; data during the observation period were calculated based on an annualized rate.
ABR, annualized bleeding rate; E5D, every 5 days; E7D, every 7 days; FAS, full analysis set; SD, standard deviation.

- In the E5D and E7D subgroups, the median (mean, SD) total number of bleeds within 12 months prior to damoctocog alfa pegol initiation was 3.0 (5.1, 8.5) and 1.5 (3.5, 6.1).
- The median (mean, SD) difference in ABR during the observation period with damoctocog alfa pegol compared with prior to initiation was –1.5 (–4.3, 8.2) for the E5D dosing regimen and –1.2 (–1.7, 2.2) for the E7D regimen **(Figure 2)**.

Figure 2: DIFFERENCE IN ABR DURING OBSERVATION PERIOD FROM 12 MONTHS PRIOR TO DAMOCTOCOG ALFA PEGOL INITIATION (FAS)

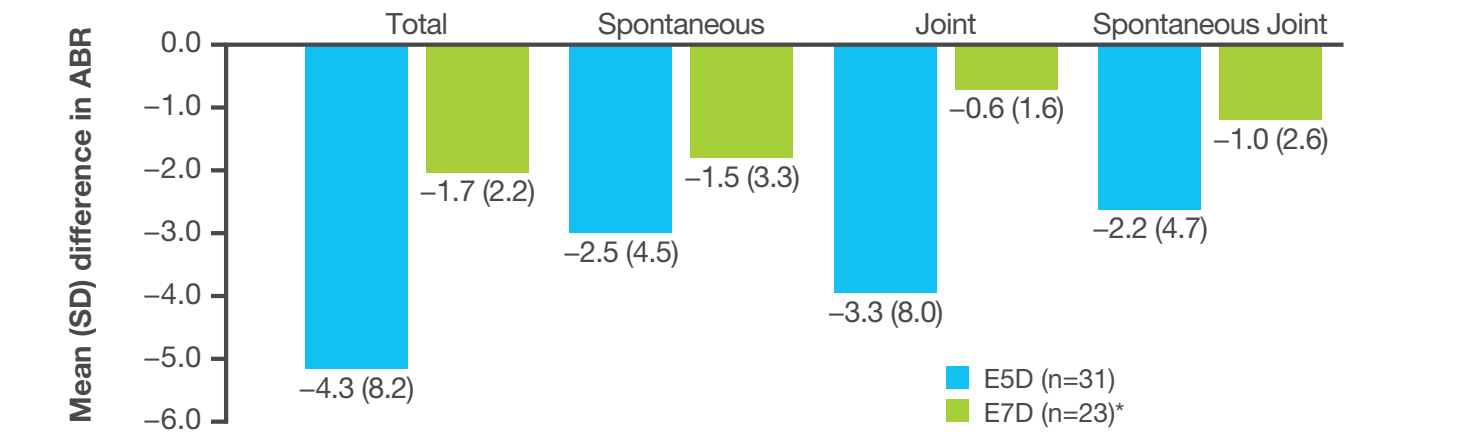
Mean (SD) difference in ABR
0.0
–1.0
–2.0
–3.0
–4.0
–5.0
–6.0
Total
Spontaneous
Joint
Spontaneous Joint
–4.3 (8.2)
–1.7 (2.2)
–2.5 (4.5)
–1.5 (3.3)
–3.3 (8.0)
–0.6 (1.6)
–2.2 (4.7)
–1.0 (2.6)
E5D (n=31)
E7D (n=23)*

	Total	Spontaneous	Joint	Spontaneous joint
E5D, median (Q1, Q3)	–1.5 (–4.0, 0.0)	–1.0 (–3.0, 0.0)	–0.6 (–2.8, 0.0)	0.0 (–1.8, 0.0)
E7D, median (Q1, Q3)*	–1.2 (–3.0, 0.0)	–0.5 (–2.2, 0.0)	0.0 (–1.2, 0.0)	0.0 (–1.1, 0.0)

*Data missing for 1 (4.3%) patient prior to damoctocog alfa pegol initiation. ABR, annualized bleeding rate; E5D, every 5 days; E7D, every 7 days; FAS, full analysis set; SD, standard deviation.

- The median (mean, SD) number of affected joints per patient prior to damoctocog alfa pegol initiation was 1.0 (1.5, 2.0) in the E5D group and 1.0 (1.3, 2.0) in the E7D group. At first follow-up*, the number of affected joints was 0.0 (0.7, 1.6) for E5D and 0.0 (0.8, 1.8) for E7D.
- The proportion of patients with no affected joints prior to damoctocog alfa pegol was 45.2% (n=14/31) in the E5D group, compared with 73.7% (n=14/19) at follow-up window 1. In the E7D group, 47.8% (n=11/23) of patients had no affected joints prior to damoctocog alfa pegol, compared with 66.7% (n=10/15) at follow-up window 1 **(Figure 3).**
- In the SAF, the proportion of patients experiencing a TEAE was 29.4% (n=15/51) for the E5D group and 13.2% (n=5/38) for E7D. No study-drug-related TEAEs, discontinuations, or deaths were reported **(Table 2).**

Figure 3: NUMBER OF PATIENTS WITH NO AFFECTED JOINTS BY DOSING REGIMEN (FAS)

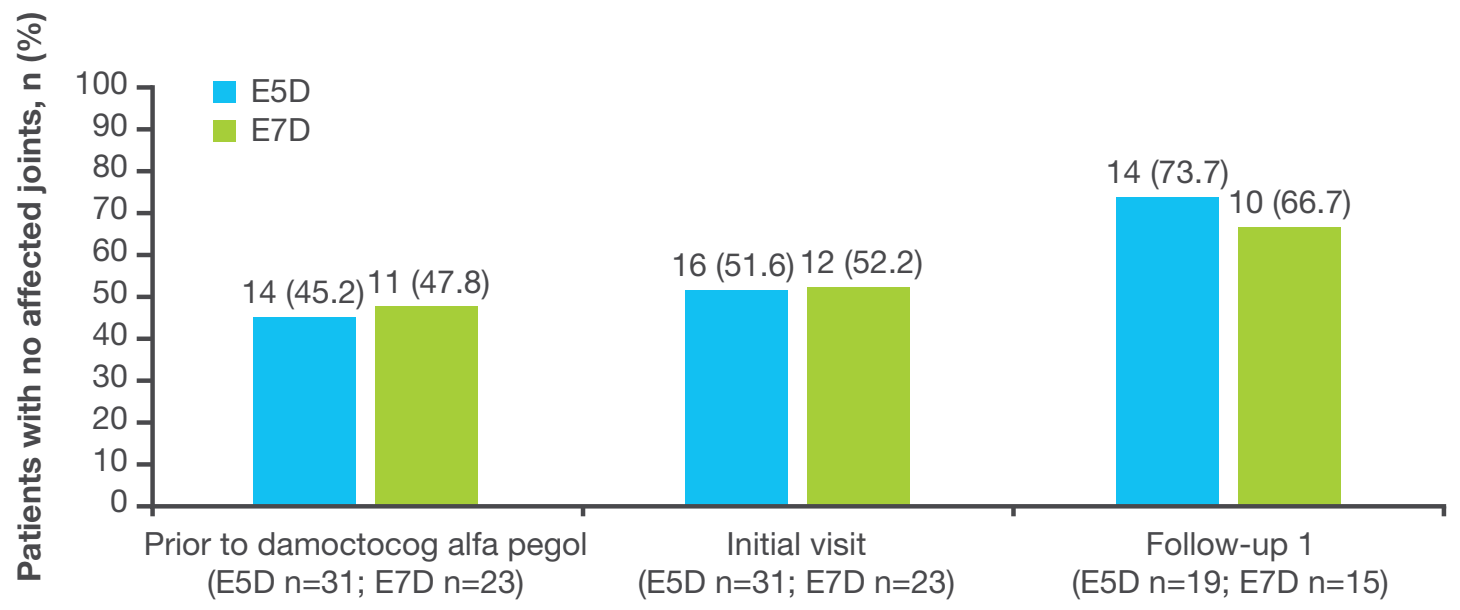
Patients with no affected joints, n (%)
0
10
20
30
40
50
60
70
80
90
100
E5D
E7D
14 (45.2)
11 (47.8)
16 (51.6)
12 (52.2)
14 (73.7)
10 (66.7)
Prior to damoctocog alfa pegol (E5D n=31; E7D n=23)
Initial visit (E5D n=31; E7D n=23)
Follow-up 1 (E5D n=19; E7D n=15)

Follow-up windows are defined as 180-day intervals (±90 days). Baseline is initial visit, Follow-up window 1 (Days 90 - <270).
ABR, annualized bleeding rate; E5D, every 5 days; E7D, every 7 days; FAS, full analysis set; SD, standard deviation.

Table 2: SUMMARY OF TEAES IN THE SAF

Characteristics	E5D (n=51)	E7D (n=38)
Any TEAE, n (%)	15 (29.4)	5 (13.2)
Study-drug-related TEAE	1 (2.0)	0
Any TEAE leading to treatment regimen change	3 (5.9)	3 (7.9)
Any TEAE leading to discontinuation of treatment	0	0
Any TEAE leading to inhibitor development	0	0
TEAE-related death	0	0
Any serious TEAE, n (%)	1 (2.0)	1 (2.6)
Study-drug-related serious TEAE	0	0
Serious TEAE leading to treatment regimen change	0	1 (2.6)
Any serious TEAE leading to discontinuation of treatment	0	0
Any serious TEAE leading to inhibitor development	0	0
Serious TEAE-related death	0	0

SAF, Safety Analysis Set; TEAE, treatment emergent adverse event.

*Not all patients providing information about affected joints at initial visit provided information during follow-up. At first follow-up, 19 patients in the E5D and 15 patients in the E7D group reported information on affected joints.

Disclosures

MTR: receipt of institutional research support from Bayer and BioMarin, member of advisory boards and/or speaker bureaus for Bayer, CSL Behring, Novo Nordisk, Sanofi Genzyme, Takeda, and HEMA Biologics. **MTAR:** speaker in advisory boards and sponsored symposia with Novo Nordisk, Bayer, Takeda, Roche, Pfizer, Octapharma, Amgen, Novartis, CSL Behring, and Sobi. **GC:** speaker at satellite symposia during scientific meetings for Bayer, Grifols, LFB, Roche, Sobi, Novo Nordisk, Werfen, and Kedrion, member of steering committee of UniQure, participant of advisory boards for Alexion, Bayer, BioMarin, Takeda, CSL Behring, LFB, Novo Nordisk, Pfizer, Roche, Sanofi, Sobi, and UniQure, consultant for Roche. **MJ:** member of speaker bureau for Takeda, BioMarin, CSL Behring, and Sanofi, consultancy for Takeda, CSL Behring, Sanofi, BioMarin, Genentech, and Octapharma, member of Bayer steering committee. **TM:** member of advisory boards for Takeda, Bayer, Novo Nordisk, Chugai and Pfizer, receipt of educational and investigational support from Chugai and Novo Nordisk, received honoraria from Takeda, Bayer, Sanofi, Chugai, CSL Behring, JB Pharma, KMB Pharma, Novo Nordisk, Octapharma, and Sysmex. **KM:** receipt of speaker fees from Bayer and Alexion, participation in trial steering committee for Bayer, receipt of consulting fees from UniQure. **KS:** employee of Bayer. **JO:** reimbursed for attending symposia/congresses and/or received honoraria and/or funds for research from Bayer, Biogen Idec, Biomarin, Biotest, Chugai, CSL Behring, Freeline, Grifols, LFB, Novo Nordisk, Octapharma, Pfizer, Roche, Sanofi, Spark Therapeutics, Swedish Orphan Biovitrum and Takeda.

Acknowledgments

The authors would like to acknowledge Martin Sanabria of Bayer, for his contributions to the HEM-POWR study. Editorial support (in the form of writing assistance, including development of the initial draft based on author direction, collating authors' comments and grammatical editing) was provided by Jason Foreman of Fishawack Communications Ltd UK, part of Fishawack Health, and was funded by Bayer. The authors would like to acknowledge all clinical sites and investigators that participated in the study. This study was funded by Bayer.

References

1. Santagostino E, et al. *Haemophilia*. 2020;26:e55-e65.
2. Reding MT, et al. *J Thromb Haemost*. 2017;15:411-419.
3. Jivi®. Summary of Product Characteristics. https://www.ema.europa.eu/en/medicines/human/EPAR/jivi. Accessed April 4, 2023.
4. Jivi®. Package insert. https://www.fda.gov/media/115934/download. Accessed April 4, 2023
5. Reding M, Alvarez Román M, Sanabria M, et al. [abstract presented at ISTH 2021]. https://abstracts.isth.org/abstract/first-interim-analysis-from-theobservational-hem-powr-study-evaluating-effectiveness-and-safety-of-real-world-treatment-with-damoctocog-alfa-pegol-in-previously-treatedpatients-with-hemophilia-a/. Accessed April 4, 2023.
6. Reding M, Alvarez Román M, Sanabria M, et al. [abstract presented at ISTH 2022]. https://abstracts.isth.org/abstract/annualised-bleeding-rate-andjoint-health-second-interim-analysis-of-the-real-world-hem-powr-study-evaluating-the-effectiveness-of-damoctocog-alfa-pegol-in-patients-withhaemophilia-a/. Accessed April 4, 2023.
7. Reding MT, et al. *Blood*. 2022;140 (Suppl 1):5612-5614.

Presented at ISTH 2023 congress, 24-28 June