PEACE-III: A RANDOMIZED, MULTICENTER, OPEN-LABEL PHASE III TRIAL COMPARING ENZALUTAMIDE VERSUS A COMBINATION OF RADIUM 223 AND ENZALUTAMIDE IN ASYMPTOMATIC OR MILDLY SYMPTOMATIC PATIENTS WITH BONE METASTATIC CRPC

Murilo de Almeida Luz[1] on behalf of Silke Gillessen[2], Arghya Choudhury[3], Fred Saad[4], Elena Gallardo[5], Andrey Soares[6], Yohann Loriot[7], Raymond S. McDermott[8], Alejo Rodriguez-Vida[9], P. Isaacson[10], Franco Nolè[11], Felipe Jose Silva Melo Cruz[12], Thierry Roumeguere[13], Gedske Daugaard[14], Rosely Yamamura[15], Frédéric Lecouvet[16], Corneel Coens[17], Beatrice Fournier[18], Bertrand Tombal[19]

[1]Hospital Erasto Gaertner, Curitiba, Brazil; [2]Medical Oncology Department, EOC - Ospedale Regionale Bellinzona e Valli – Istituto Oncologico della Svizzera Italiana (IOSI), Bellinzona, Switzerland; [3]Christie Hospital, The Christie NHS Foundation Trust, Manchester, UK; [4]Urology Department, Centre Hospitalier de Universite to Montréal (CHUM), Montreal, QC, Canada; [5]Oncology Department, Hospital Universitario Parc Taulí, Sabadell, Spain; [6]Oncology Department, Hospital Israelita Albert Einstein (Morumbi) - Pavilhao Vicky e Joseph Safra, Sao Paulo, Brazil; [7]Cancer Medicine Department, Institut Gustave Roussy, Villejuif, France; [8]Department of Oncology, Tallaght University Hospital, Dublin, Ireland; [9]Medical Oncology Department, Hospital del Mar - Parc de Salut Mar, Barcelona, Spain; [10]Department of Oncology, Hospital Moinhos de Vento, Porto Alegre, Brazil; [11]Medical Oncology Dept., IEO - Istituto Europeo di Oncologia IRCCS, Milan, Italy; [12]Medical Oncology, Rede D'Or Sao Luiz, Brasilia, Brazil; [13]Urology Department, Erasme Hospital - Universite Libre de Bruxelles (ULB), Brussels, Belgium; [14]Department of Oncology, Rigshospitalet, Copenhagen, Denmark; [15]Medical Oncology Department, Hospital Beneficencia Portuguesa de Sao Paulo - Mirante, Sao Paulo, Brazil; [16]Radiology, Cliniques Universitaires Saint-Luc (UCLouvain Saint-Luc), Woluwe-Saint-Lambert, Belgium; [17]Statistical Department, EORTC AISBL/IVZW – European Organisation for Research and Treatment of Cancer, Brussels, Belgium; [18]Medical Department, EORTC AISBL/IVZW - European Organisation for Research and Treatment of Cancer, Brussels, Belgium; [19]Urology Department, Cliniques Universitaires Saint-Luc (UCLouvain Saint-Luc), Woluwe-Saint-Lambert, Belgium.

RADIUM 223

INTRODUCTION

Radium 223 dichloride (Ra223) is an alpha particle emitting radionuclide that selectively targets bone metastases and induces double-stranded DNA breaks[1]

In the ALSYMPCA trial, Ra223 plus best standard-of-care[a] has been shown to increase overall survival (OS; HR, 0.7)[2]

No combination of drugs with contemporary life prolonging therapies so far has been proven to increase radiologic progression-free survival (rPFS) and OS in first-line metastatic CRPC (castration-resistant prostate cancer)

OBJECTIVE

To study whether it delays cancer progression compared to enzalutamide alone in patients with bone metastatic CRPC

METHODS

PEACE-III (NCT02194842) is a randomized multicenter phase III study

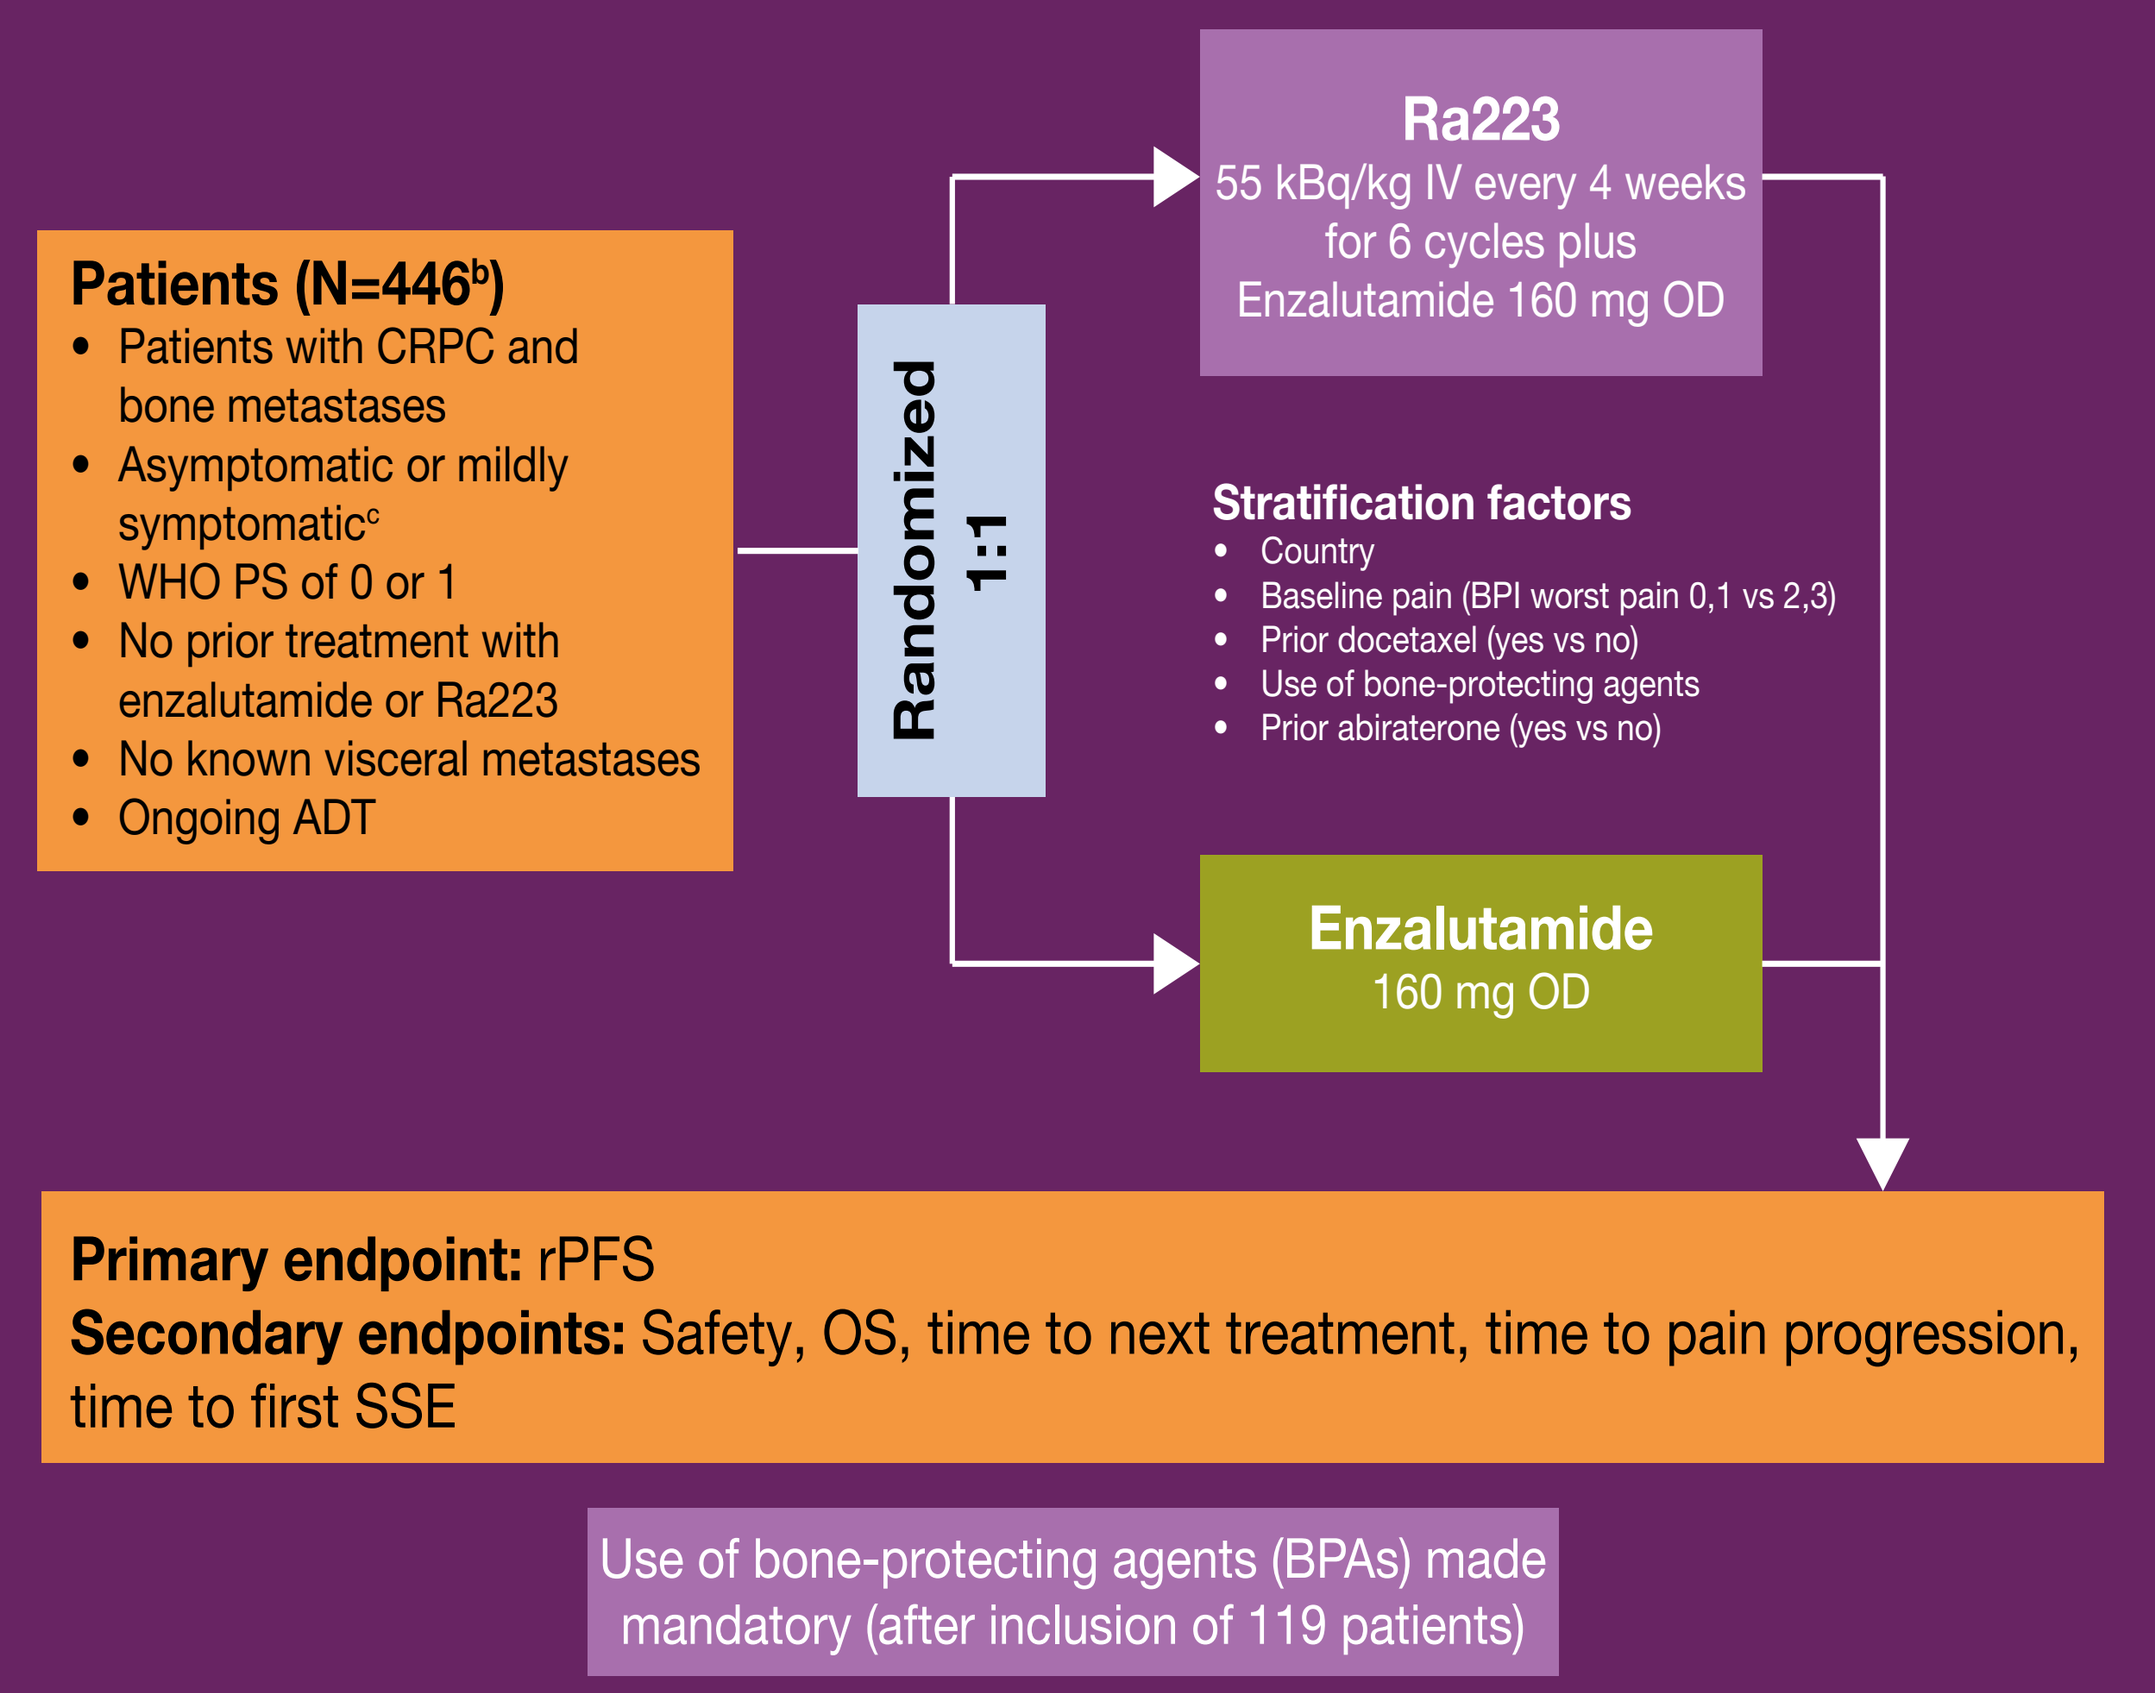
Patients (N=446[b])
• Patients with CRPC and bone metastases
• Asymptomatic or mildly symptomatic[c]
• WHO PS of 0 or 1
• No prior treatment with enzalutamide or Ra223
• No known visceral metastases
• Ongoing ADT
Randomized 1:1
Ra223
55 kBq/kg IV every 4 weeks for 6 cycles plus Enzalutamide 160 mg OD
Stratification factors
• Country
• Baseline pain (BPI worst pain 0,1 vs 2,3)
• Prior docetaxel (yes vs no)
• Use of bone-protecting agents
• Prior abiraterone (yes vs no)
Enzalutamide
160 mg OD
Primary endpoint: rPFS
Secondary endpoints: Safety, OS, time to next treatment, time to pain progression, time to first SSE
Use of bone-protecting agents (BPAs) made mandatory (after inclusion of 119 patients)

[a]Best standard of care was defined as the routine care provided at each center (e.g., local external-beam radiation therapy or treatment with glucocorticoids, antiandrogens, ketoconazole, or estrogens such as diethylstilbestrol or estramustine.
[b]Original target accrual N=560, adapted for slow accrual.
[c]Defined as brief pain inventory WP24 score <4.
This is a EORTC/CTI/CUOG/LACOG/UNICANCER cooperative study.
ADT, androgen-deprivation therapy; IV, intravenous; CRPC, castration-resistant prostate cancer; OD, once daily; OS, overall survival; PSA, prostate-specific antigen; Ra223, radium 223 dichloride; rPFS, radiologic progression-free survival; SSE, symptomatic skeletal event; WHO PS, World Health Organization performance status.

Ra223 improved rPFS vs placebo in men with CRPC and bone metastases (HR 0.7)

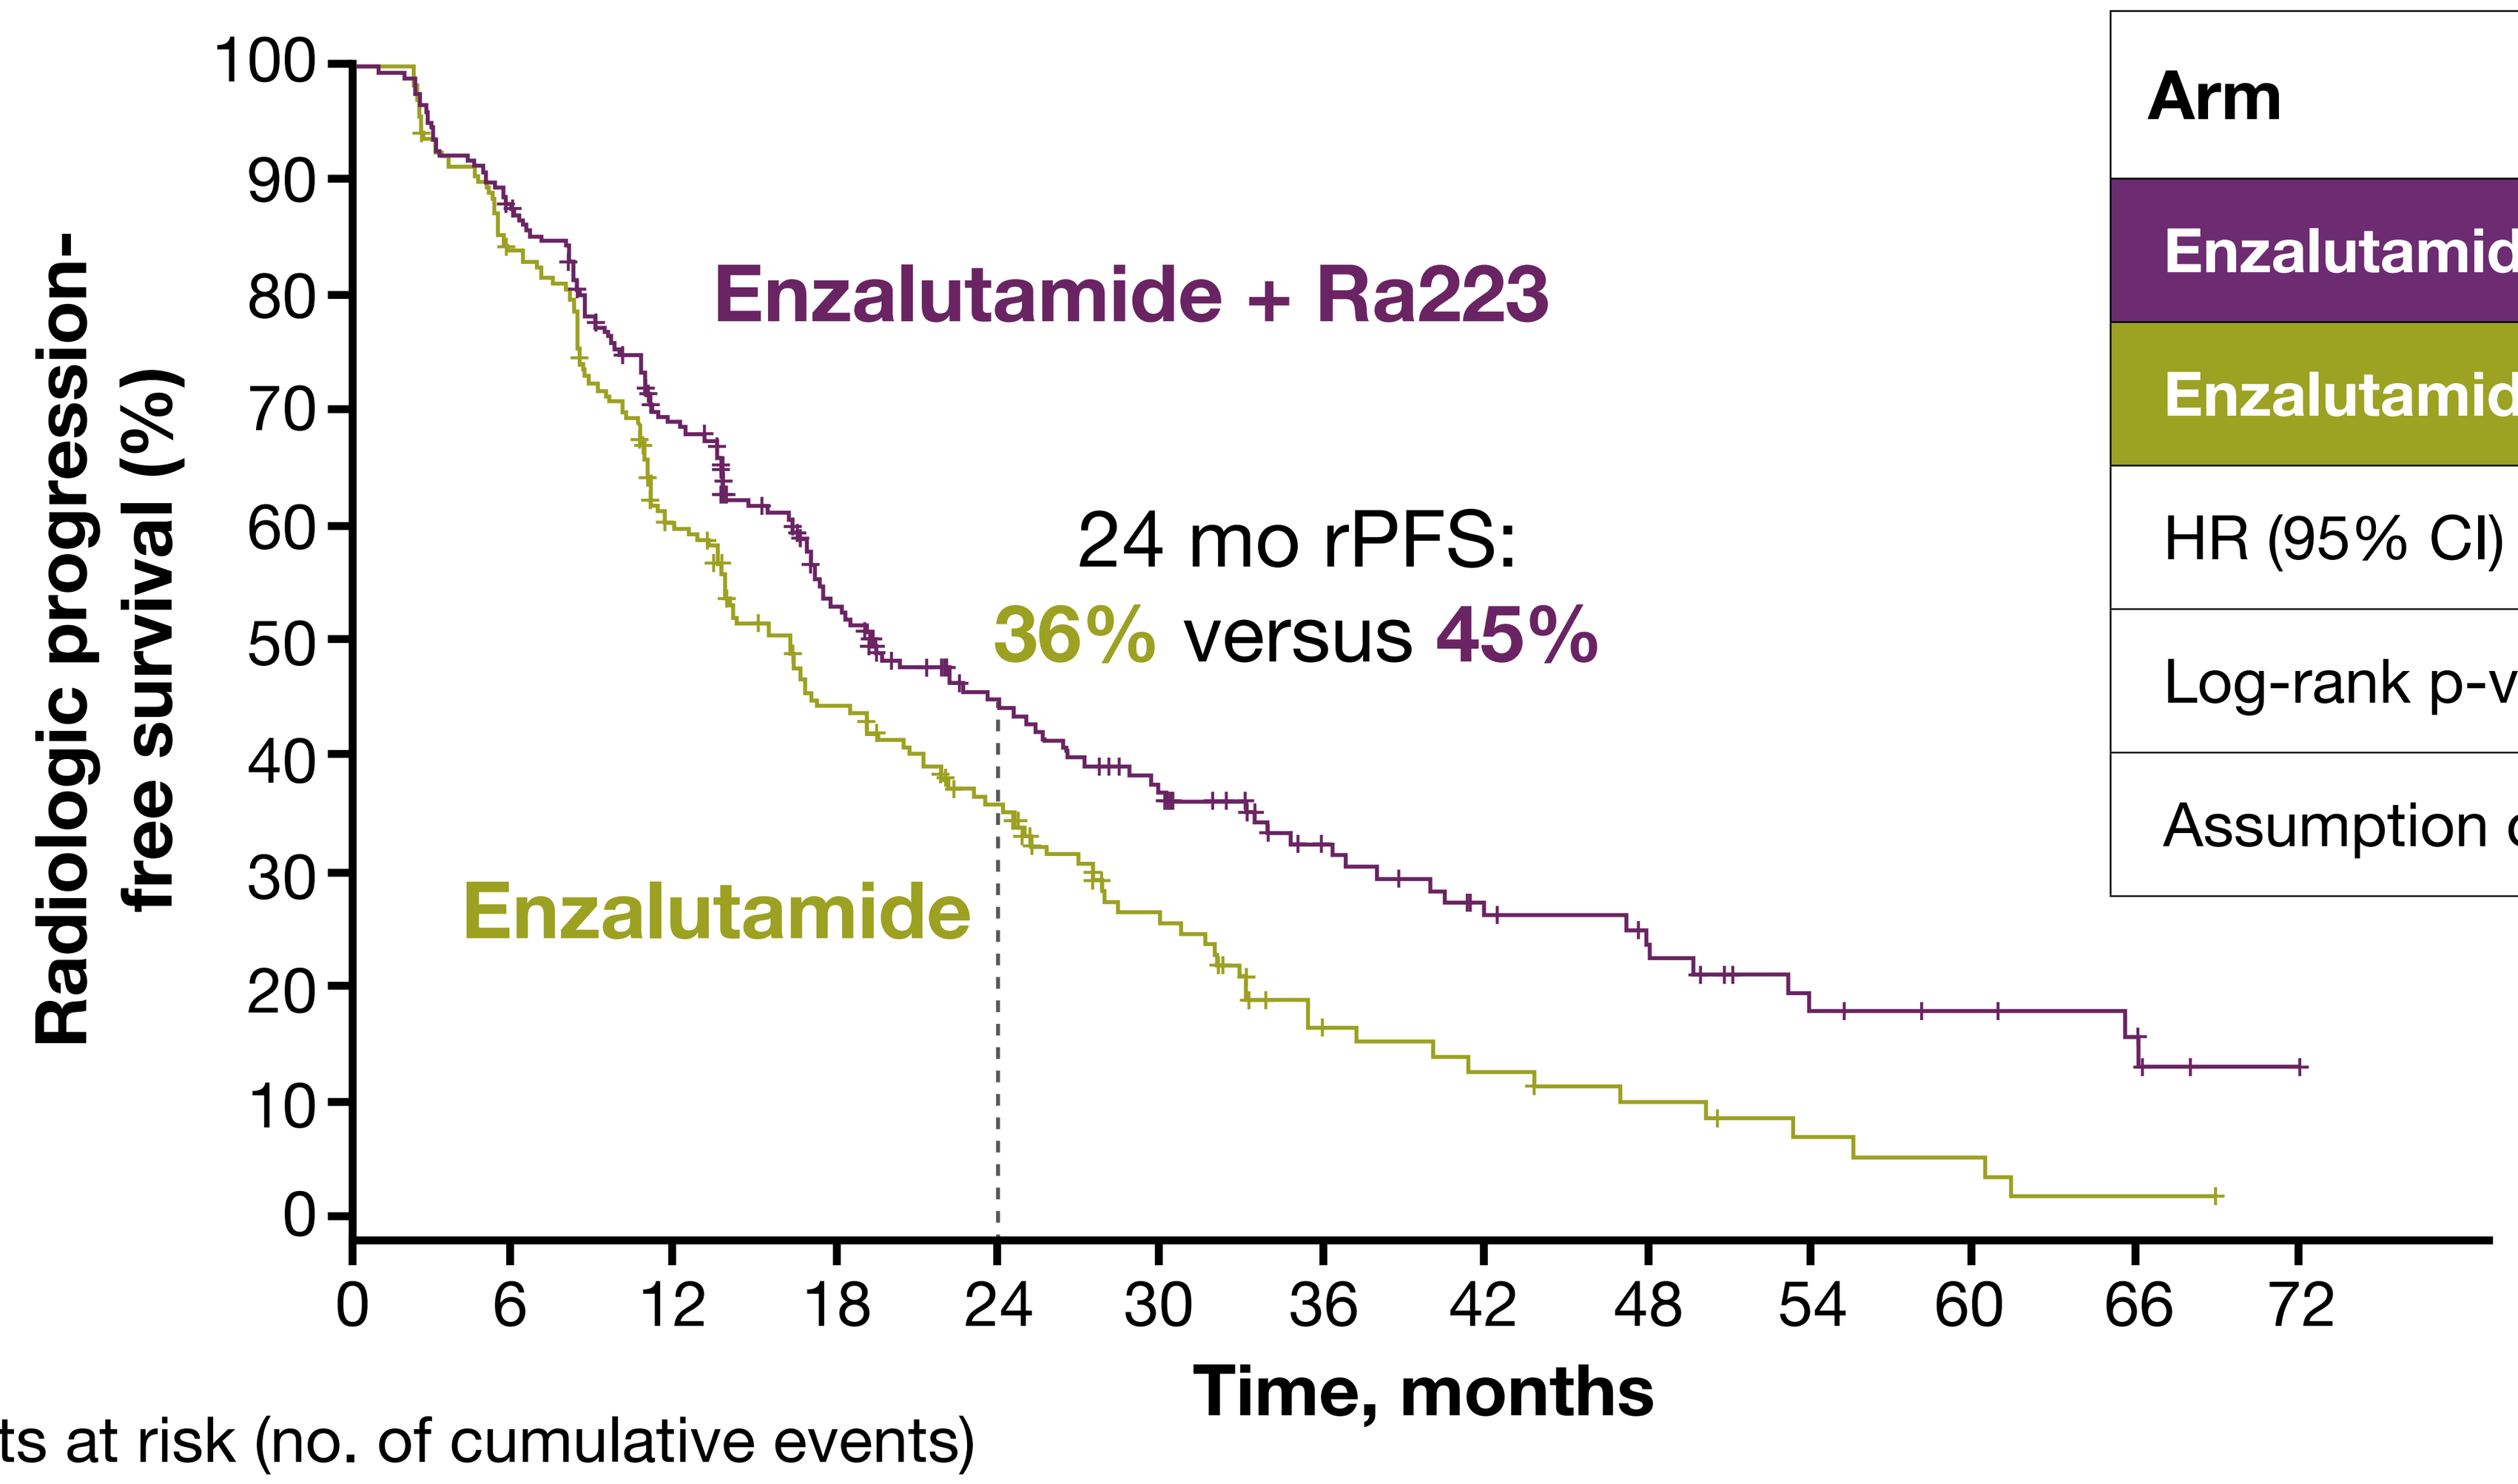
Radiologic progression-free survival (%)
100
90
80
70
60
50
40
30
20
10
0
Enzalutamide + Ra223
24 mo rPFS:
36% versus 45%
Enzalutamide
0 6 12 18 24 30 36 42 48 54 60 66 72
Time, months

Arm	n/N	Median (95% CI)
Enzalutamide + Ra223	139/222	**19.4** (17.1–25.3) mo
Enzalutamide	160/224	**16.4** (13.8–19.2) mo
HR (95% CI)	**0.69** (0.54–0.87)	
Log-rank p-value	**0.0009**	
Assumption of proportional hazard achieved		

Patients at risk (no. of cumulative events)

Enza	224 (0)	122 (84)	52 (128)	13 (150)	7 (155)	3 (158)	0 (160)
Enza + Ra223	222 (0)	138 (65)	64 (107)	32 (123)	19 (131)	9 (135)	3 (137)

CI, confidence interval; CRPC, castration-resistant prostate cancer; HR, hazard ratio; mo, months; Ra223, radium 223 dichloride; rPFS, radiologic progression-free survival.

Risk of death reduced by 31% with enzalutamide + Ra223 versus enzalutamide alone

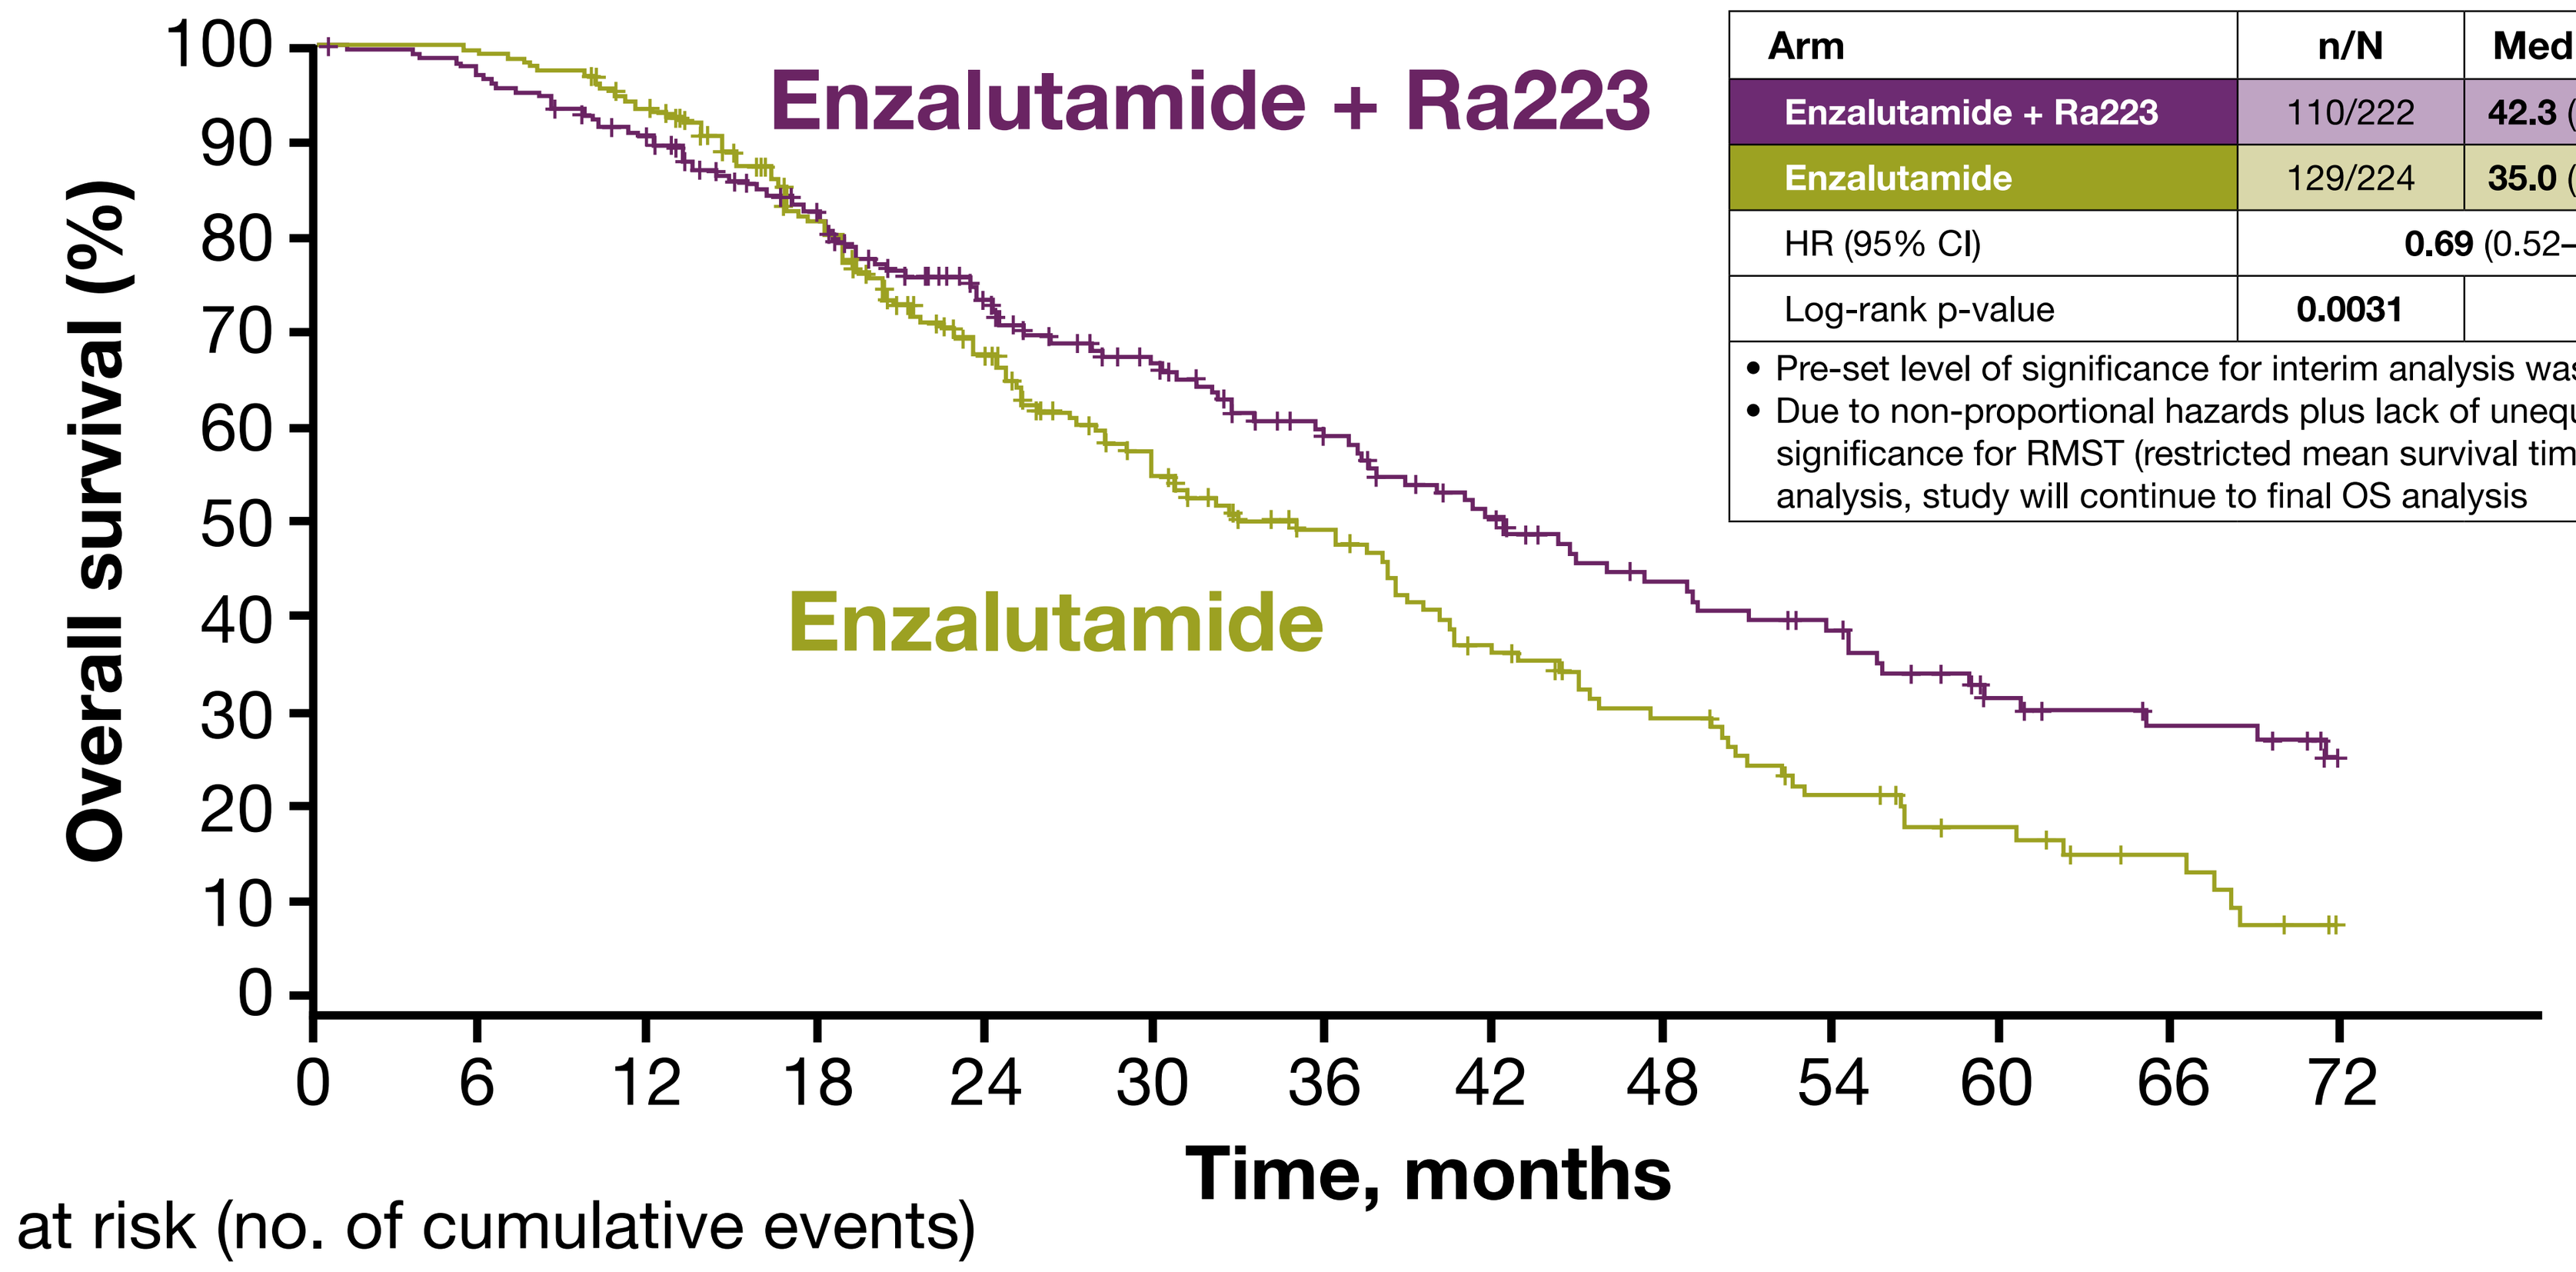
Overall survival (%)
100
90
80
70
60
50
40
30
20
10
0
Enzalutamide + Ra223
Enzalutamide
0 6 12 18 24 30 36 42 48 54 60 66 72
Time, months

Arm	n/N	Median (95% CI)
Enzalutamide + Ra223	110/222	**42.3** (36.8–49.1) mo
Enzalutamide	129/224	**35.0** (28.8–38.9) mo
HR (95% CI)	**0.69** (0.52–0.90)	
Log-rank p-value	**0.0031**	<0.0034

• Pre-set level of significance for interim analysis was ≤0.0034
• Due to non-proportional hazards plus lack of unequivocal significance for RMST (restricted mean survival time) sensitivity analysis, study will continue to final OS analysis

Patients at risk (no. of cumulative events)

Enza	224 (0)	206 (15)	107 (64)	58 (90)	30 (112)	14 (123)	1 (129)
Enza + Ra223	222 (0)	194 (21)	114 (53)	71 (73)	43 (90)	23 (101)	12 (105)

This pre-planned interim analysis was performed at 80% of events.
CI, confidence interval; HR, hazard ratio; OS, overall survival; mo, months; Ra223, radium 223 dichloride.

Safety of enzalutamide + Ra223 versus enzalutamide alone

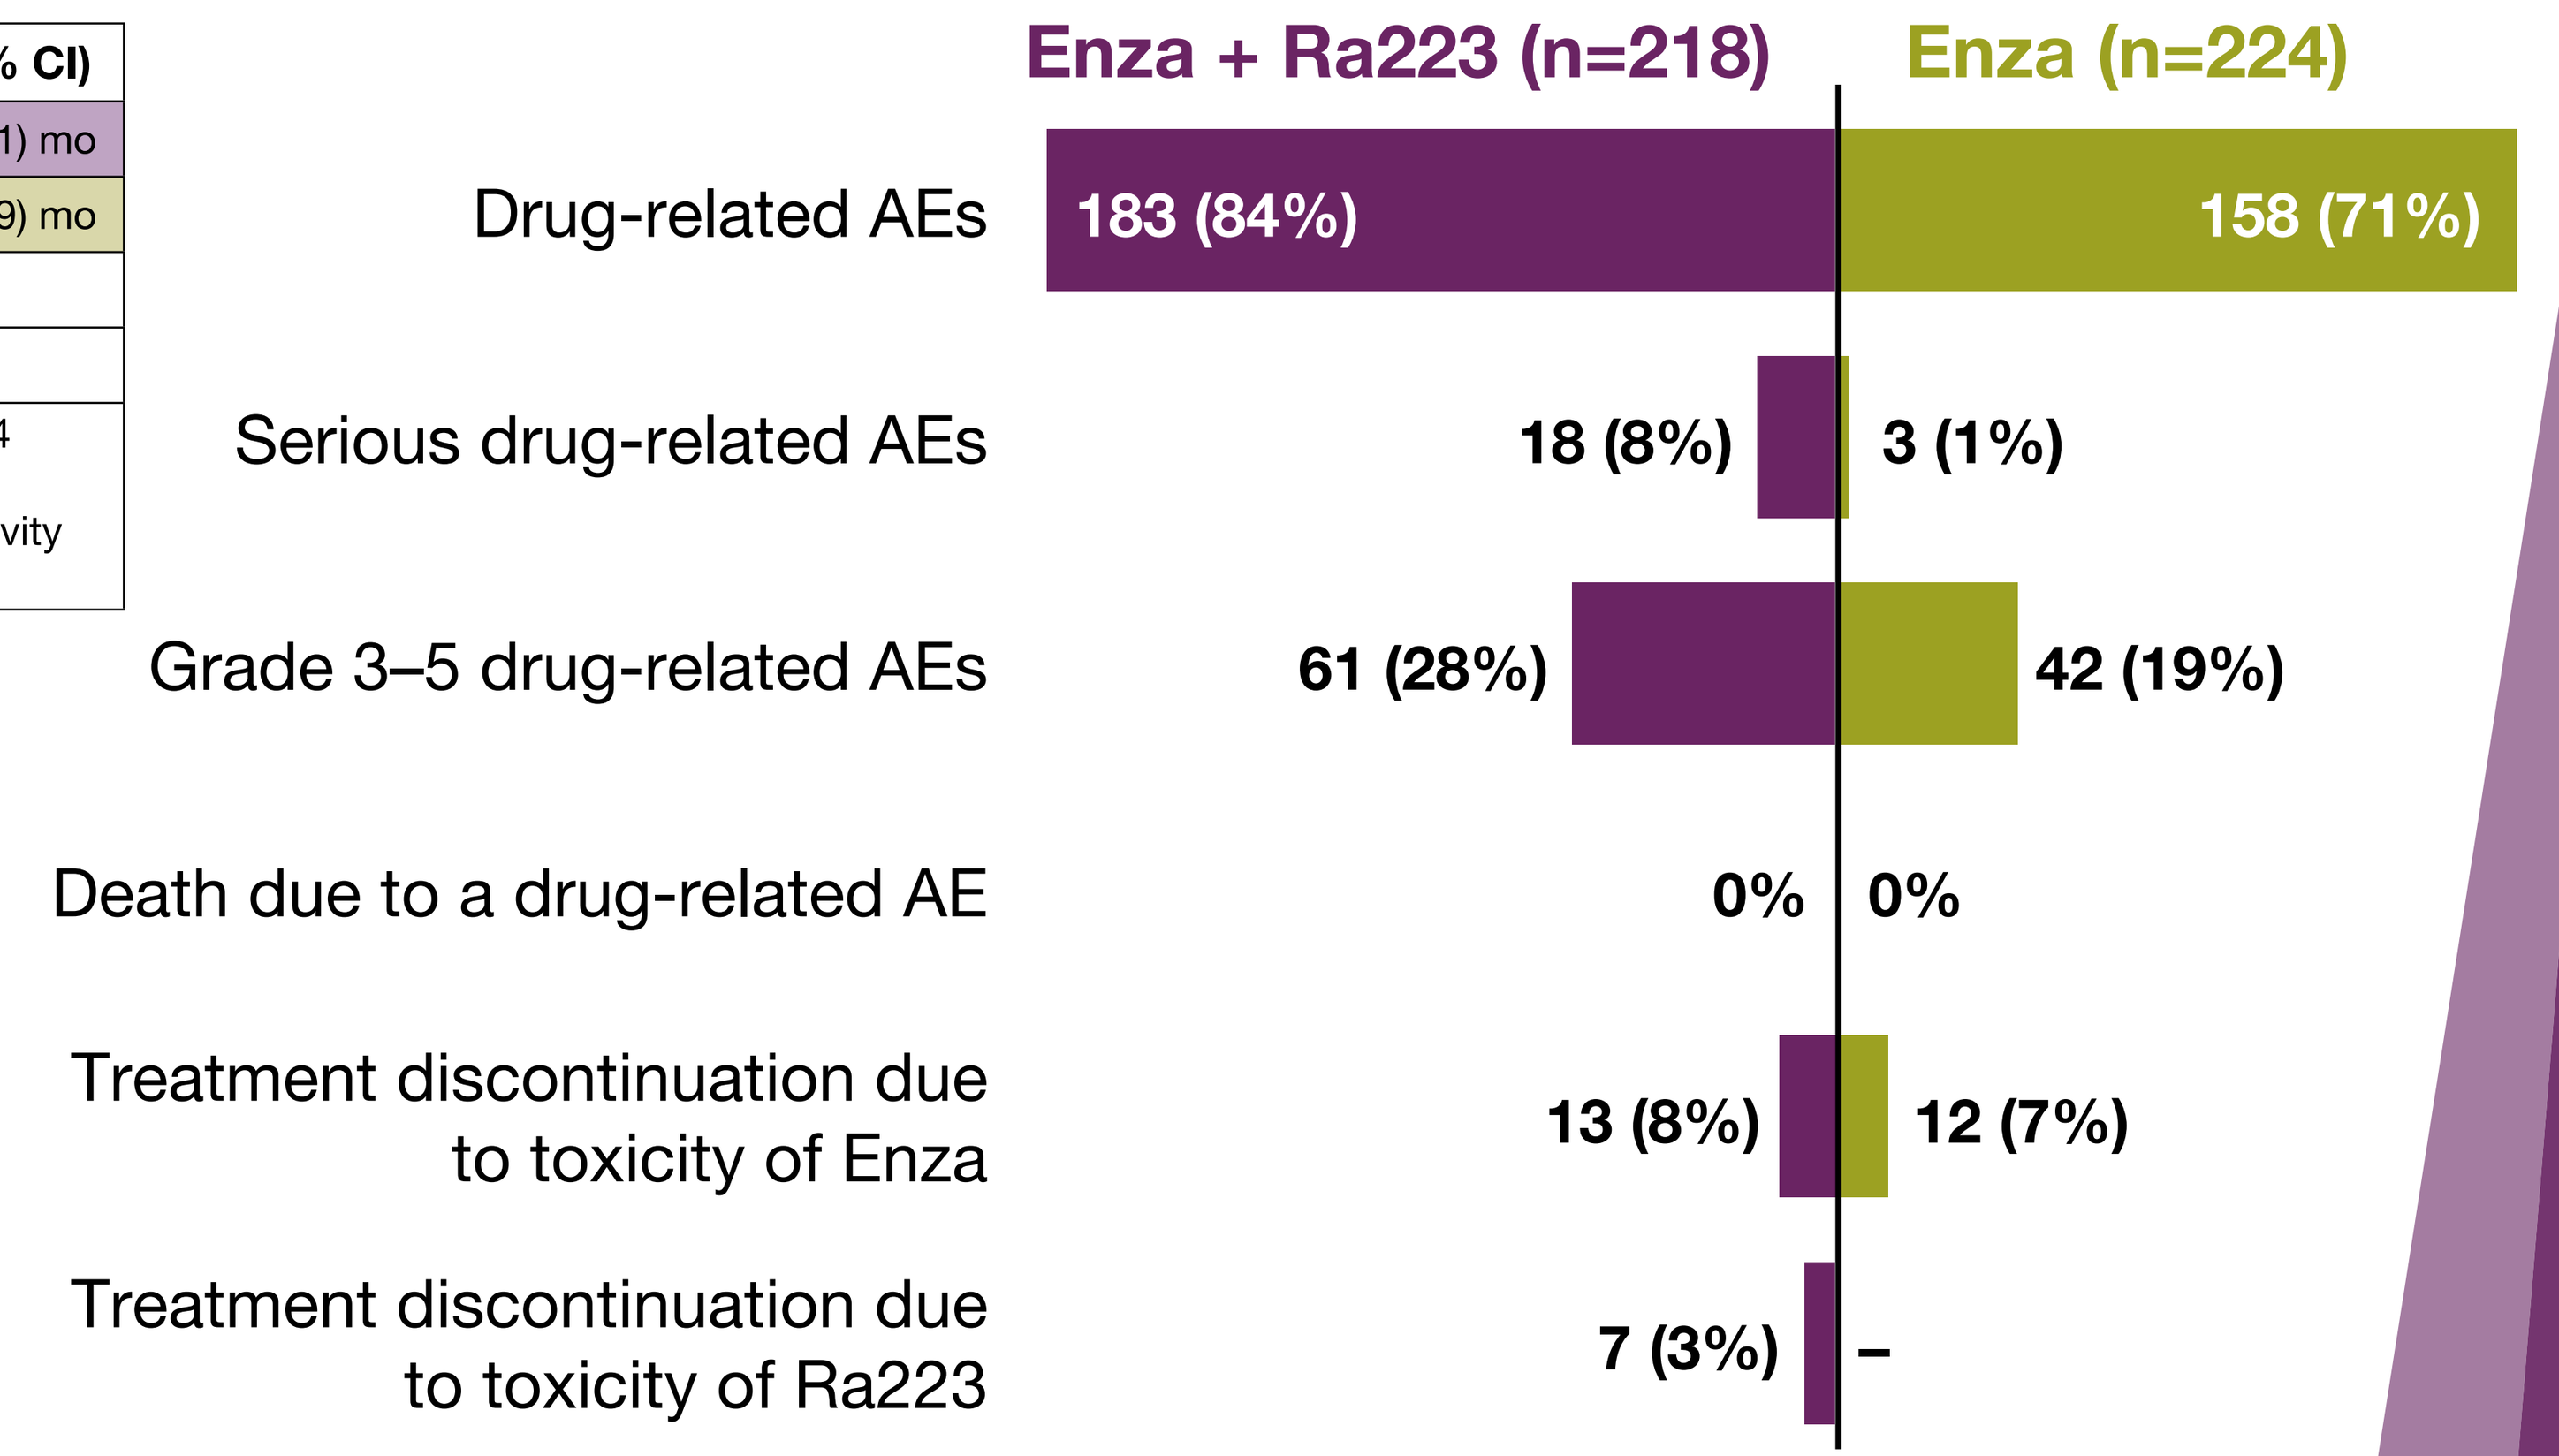
Enza + Ra223 (n=218)
Enza (n=224)
Drug-related AEs
183 (84%)
158 (71%)
Serious drug-related AEs
18 (8%)
3 (1%)
Grade 3–5 drug-related AEs
61 (28%)
42 (19%)
Death due to a drug-related AE
0%
0%
Treatment discontinuation due to toxicity of Enza
13 (8%)
12 (7%)
Treatment discontinuation due to toxicity of Ra223
7 (3%)
–

AE, adverse event; Enza, enzalutamide; Ra223, radium 223 dichloride.

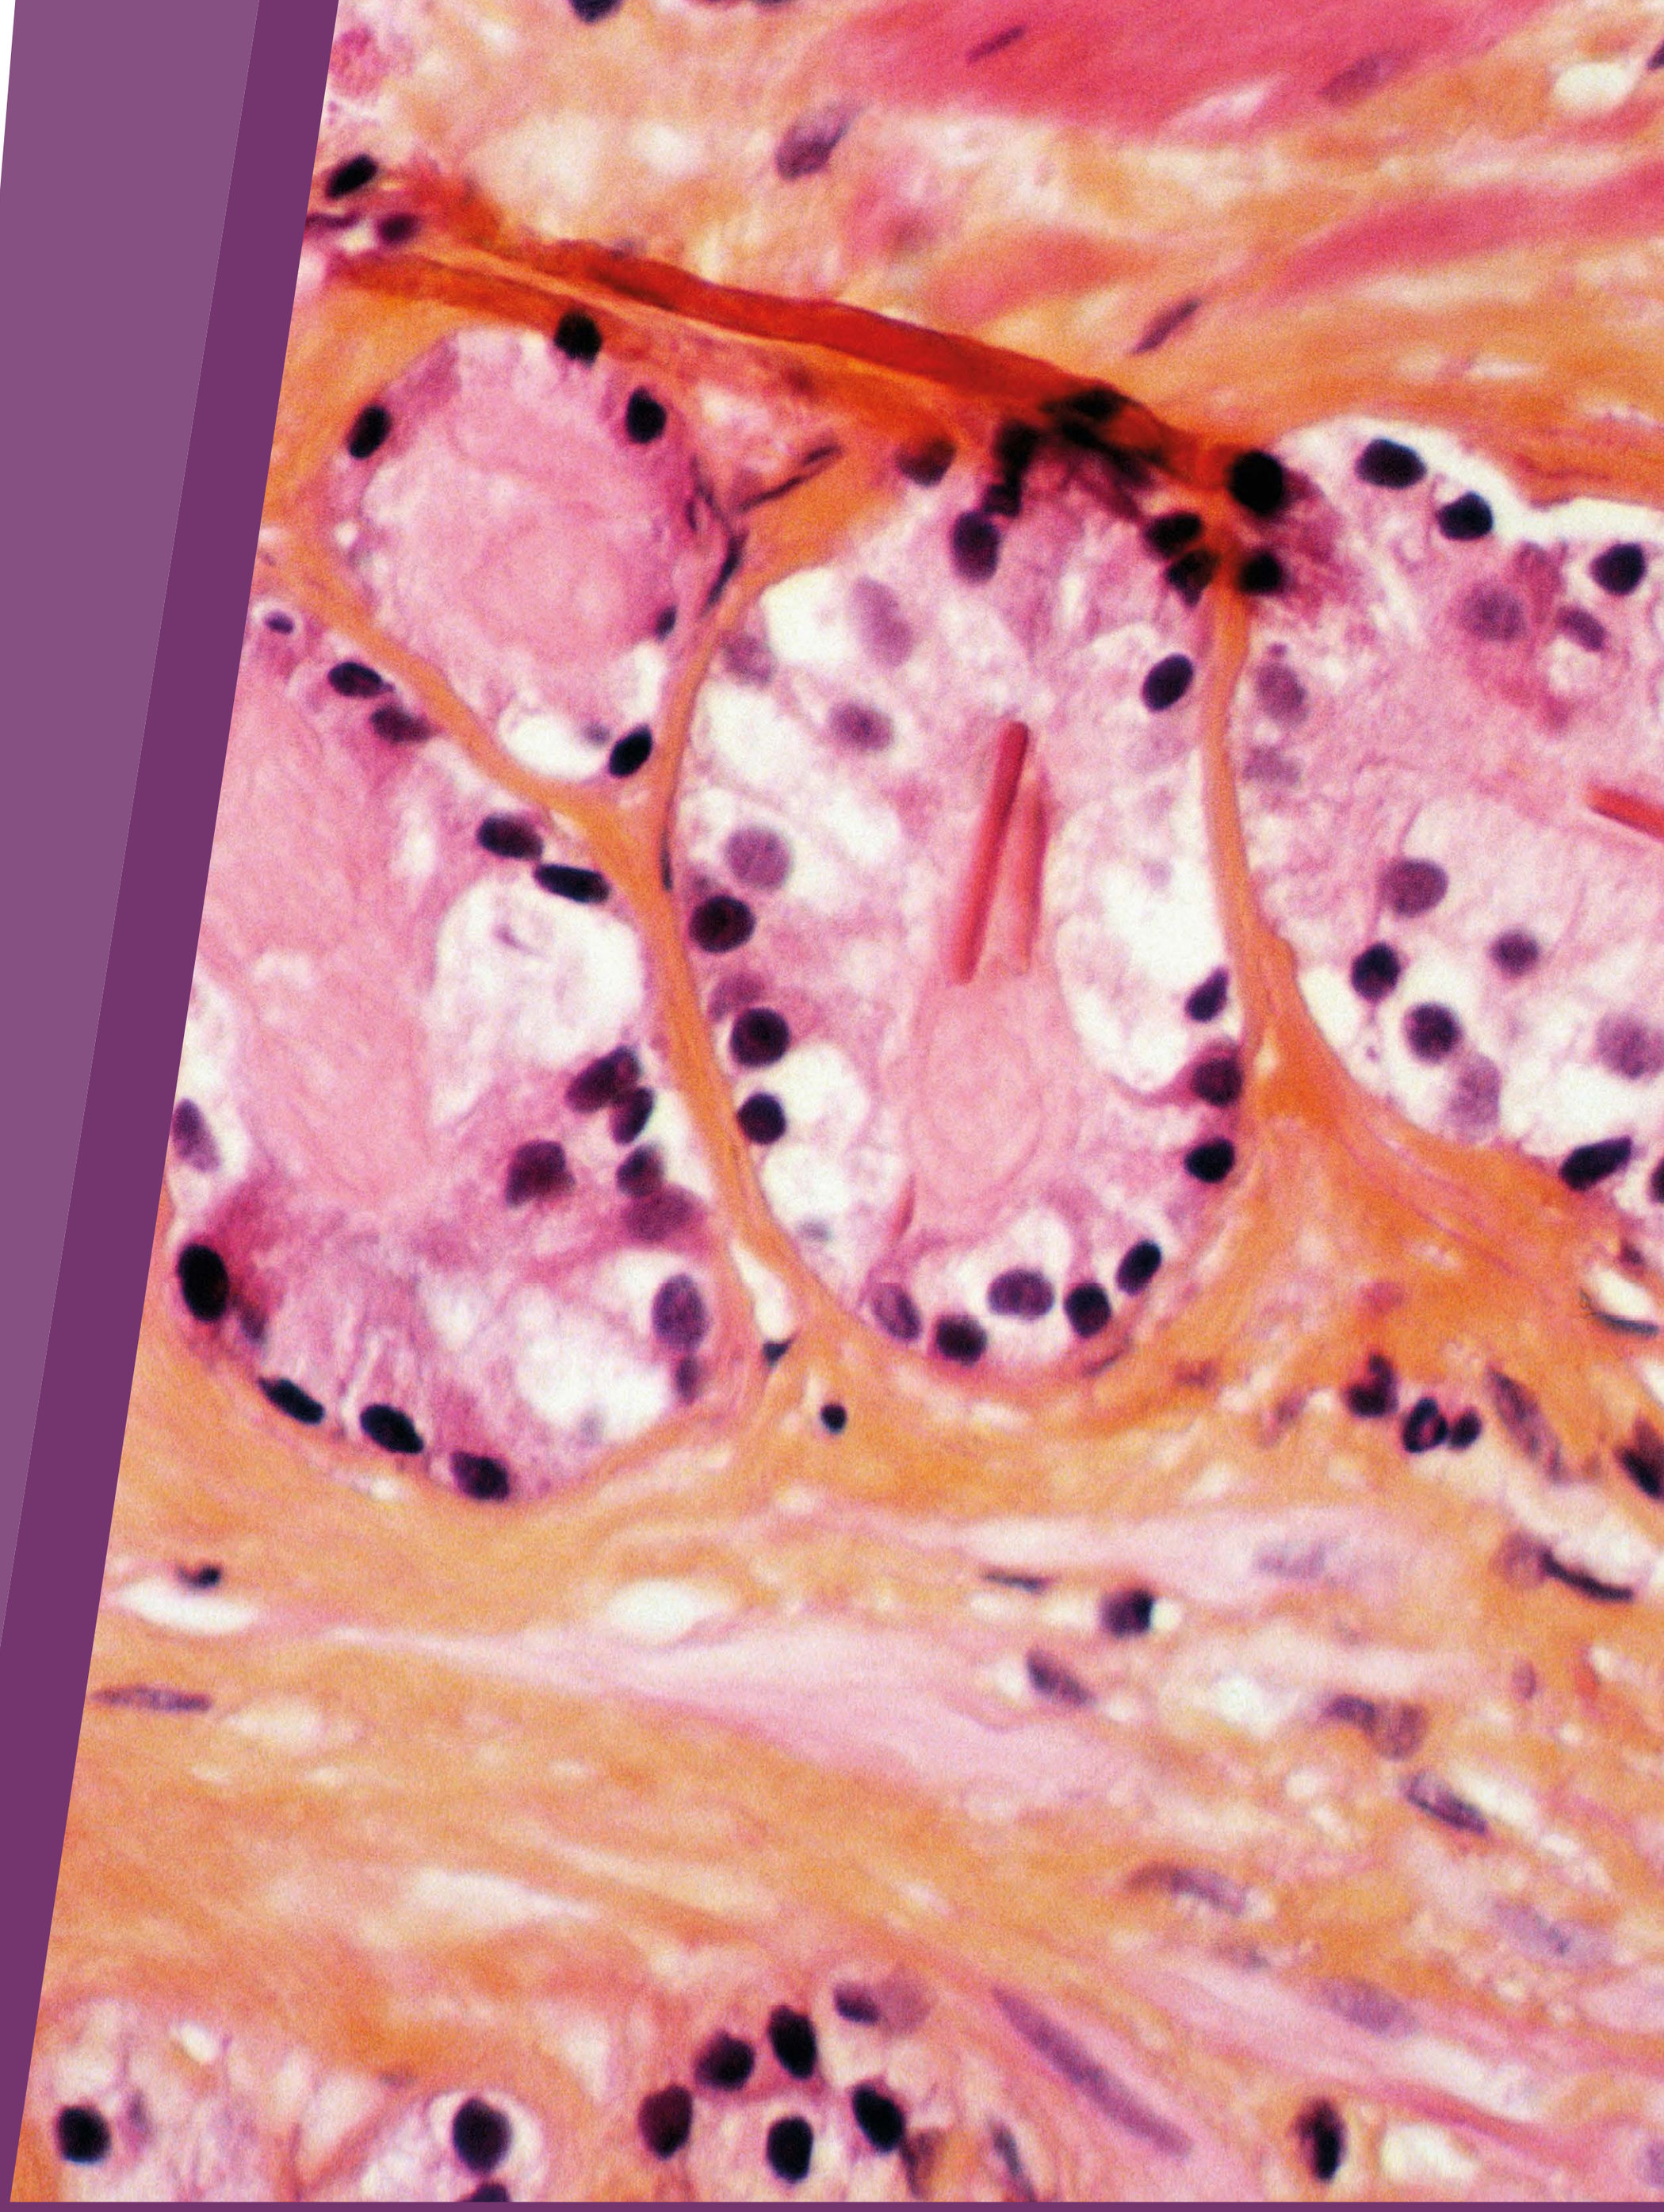

CONCLUSIONS

Adding 6 cycles of Ra223 to enzalutamide significantly improved rPFS (p=0.0009) and OS (p=0.0031) compared with enzalutamide alone

Drug-related ≥ grade 3 adverse events increased from 19% to 28% in the combination arm

These results support the combination of enzalutamide plus Ra223 (plus a bone-protecting agent) as a potential first-line treatment option for patients with CRPC and bone metastases who have not received a prior androgen receptor pathway inhibitor

SCAN ME FOR MORE INFO

Medical writing services were provided by Sidrah Rahman, MSc, of Adelphi Communications Ltd (Bollington, UK), funded by Bayer, in accordance with Good Publication Practice 2022 guidelines.

References: 1. Parker C et al. N Engl J Med 2013. 2. Morris MJ et al. Nat Rev Urol 2019.

Poster presented at the Annual International Prostate Cancer Update 2025, Vail, Colorado, USA; February 09–12, 2025.