Treatment Patterns and Preferences in Transthyretin Amyloid Cardiomyopathy in the United States: Results From a Real-World Survey

Lily K. Stern,[1] Jill Waldron,[2] Chelsea P. Renfro,[3] Kayla Johnson,[3] Liana Hennum,[4] Margarita Udall,[4] Andrew M. Rosen,[4] Heather Falvey,[4] Katherine Smethers,[5] Jade Garratt-Wheeldon,[5] Jack Wright[5]

[1]Smidt Heart Institute, Cedars-Sinai Medical Center, Los Angeles, CA, USA; [2]University of Utah Hospital, Salt Lake City, UT, USA; [3]Vanderbilt Specialty Pharmacy, Vanderbilt Health, Nashville, TN, USA; [4]BridgeBio Pharma, Inc., San Francisco, CA, USA; [5]Adelphi Real World, Bollington, UK

BACKGROUND

- **Transthyretin (TTR) amyloid cardiomyopathy (ATTR-CM) is a progressive, life-threatening disease caused by destabilization of TTR tetramers into misfolded monomers that aggregate into toxic amyloid fibrils, which deposit throughout the body, predominantly in the heart[1]**
- **The therapeutic landscape for ATTR-CM in the United States (US) continues to evolve since the first oral TTR stabilizer, tafamidis, was approved in May 2019.[2] Since then, another TTR stabilizer and a new class, TTR silencers, have emerged:**
 - **Acoramidis, an oral TTR stabilizer, was approved in November 2024[3]**
 - **Vutrisiran, an injectable TTR silencer, was approved in March 2025[4]**
- **The role of patients in determining treatment choice is also evolving**
 - **Recent guidance emphasizes shared decision-making (SDM) that incorporates patient preferences and priorities when developing a treatment plan for ATTR-CM[5,6]**
- **As more treatments become available, understanding physician and patient preferences is critical to help inform treatment selection**
- **Here, we describe real-world ATTR-CM treatment patterns, physician satisfaction, and patient administration preferences captured in the Adelphi ATTR Disease Specific Programme™ (DSP) Survey from August 2024 to February 2025**

METHODS

- The Adelphi ATTR DSP is a cross-sectional survey of physicians who treat ATTR-CM and patients diagnosed with ATTR-CM in the US
- Physician-reported demographics, prescribed treatments, treatment satisfaction, and patient-reported treatment administration preferences were analyzed descriptively

Study Design and Data Source

- This was a secondary analysis of the Adelphi ATTR DSP, which consisted of linked physician and patient/caregiver cross-sectional surveys with elements of retrospective data collection
- The DSP methodology has previously been described,[7,8] validated,[9] demonstrated to be representative and consistent over time,[10] and implemented globally across many different disease areas[7] (**Figure 1**)
 - Each physician completed a patient record form for up to their next 10 patients
 - Each patient was then invited to fill out a voluntary self-completion form (the patient could choose not to participate)
 - Participating patients were treated in accordance with usual medical practice; no additional assessments or tests were conducted because of their participation
 - Physician-reported patient data were linked to patient-reported patient data via unique, pseudonymized physician- and patient-coded numbers
- The Adelphi ATTR DSP was conducted between August 2024 and February 2025 in the US

FIGURE 1. Adelphi ATTR DSP Methodology[8,9]

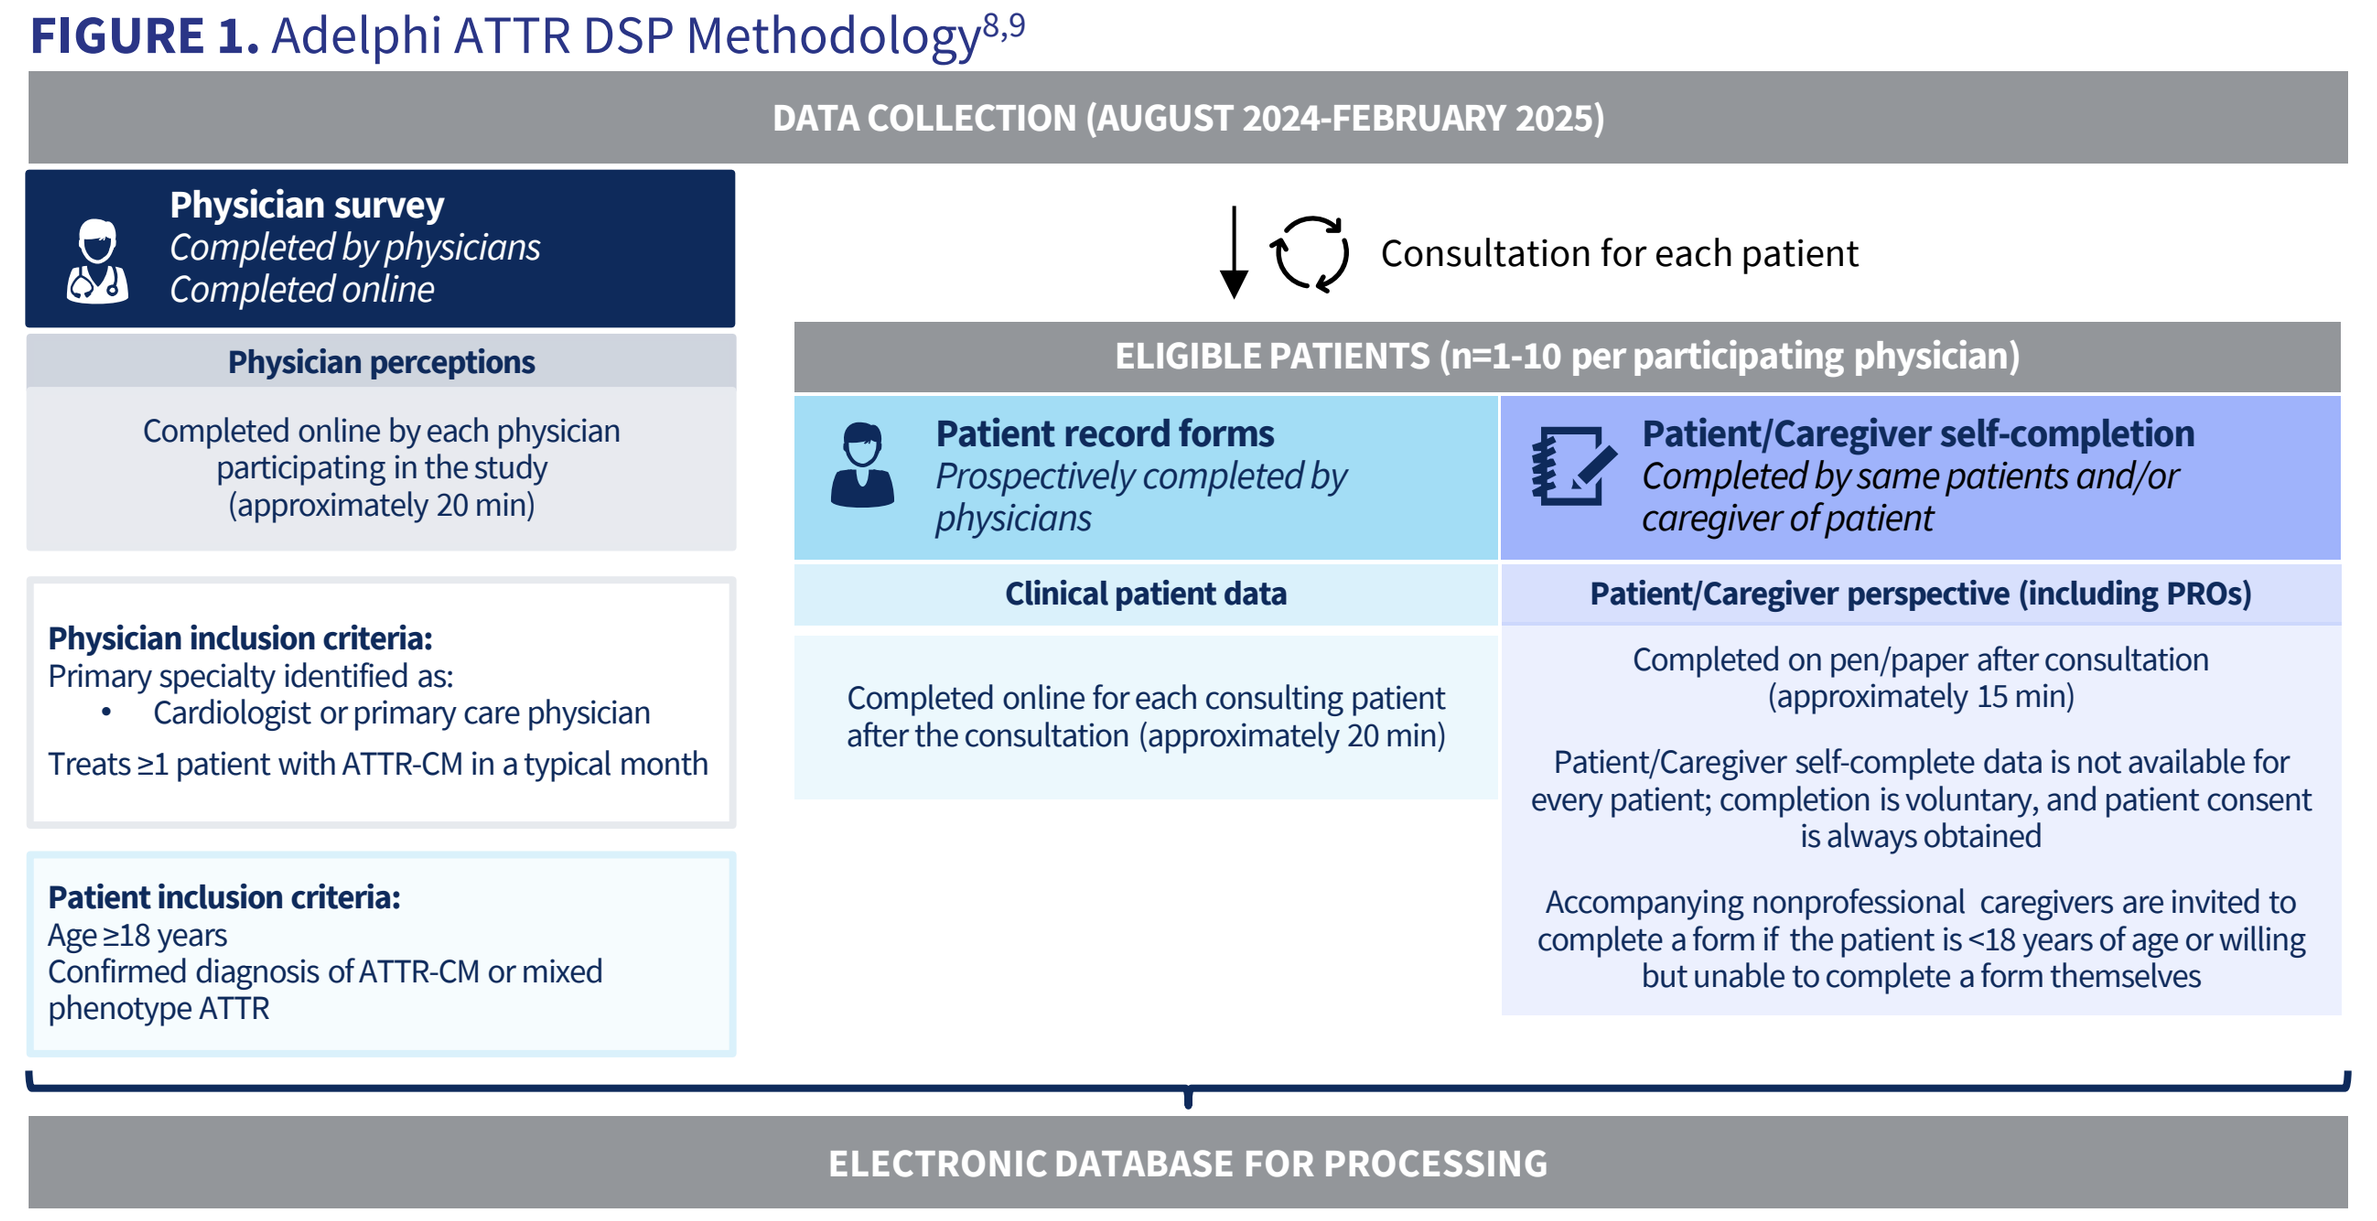
DATA COLLECTION (AUGUST 2024-FEBRUARY 2025)
Physician survey
Completed by physicians
Completed online
Physician perceptions
Completed online by each physician participating in the study (approximately 20 min)
Physician inclusion criteria:
Primary specialty identified as:
Cardiologist or primary care physician
Treats ≥1 patient with ATTR-CM in a typical month
Patient inclusion criteria:
Age ≥18 years
Confirmed diagnosis of ATTR-CM or mixed phenotype ATTR
Consultation for each patient
ELIGIBLE PATIENTS (n=1-10 per participating physician)
Patient record forms
Prospectively completed by physicians
Clinical patient data
Completed online for each consulting patient after the consultation (approximately 20 min)
Patient/Caregiver self-completion
Completed by same patients and/or caregiver of patient
Patient/Caregiver perspective (including PROs)
Completed on pen/paper after consultation (approximately 15 min)
Patient/Caregiver self-complete data is not available for every patient; completion is voluntary, and patient consent is always obtained
Accompanying nonprofessional caregivers are invited to complete a form if the patient is <18 years of age or willing but unable to complete a form themselves
ELECTRONIC DATABASE FOR PROCESSING

ATTR, transthyretin amyloidosis; ATTR-CM, transthyretin amyloid cardiomyopathy; PRO, patient-reported outcome.

Inclusion Criteria

- Eligible physicians were cardiologists or primary care physicians who had treated ≥4 patients with ATTR-CM
 - Patients were eligible if they were treated by a physician participating in the survey, were adult, and had a confirmed diagnosis of ATTR-CM or mixed phenotype (defined as ATTR-CM + ATTR peripheral neuropathy [ATTR-PN] noted in their medical record)
 - Patients with ATTR-PN were excluded

CONCLUSIONS

- **Physicians were not fully satisfied with treatment for >55% of their patients with ATTR-CM, highlighting unmet treatment needs in the period before newer therapies were approved and reached routine clinical use (August 2024-February 2025)**
- **Despite the majority of patients having New York Heart Association class I or II staged disease, a time in the disease history when treatment initiation is critical for optimal long-term benefits, many patients elected not to begin treatment, representing a continued unmet need for patient counseling in the era when SDM is being encouraged[5,6]**
- **The majority of patients (74%) reported some degree of dissatisfaction with their current treatment; all (100%) were most comfortable with oral ATTR-CM therapy**
- **These findings provide real-world insight into patient and physician preferences and priorities for ATTR-CM treatment, which may facilitate SDM and the selection of therapy**
- **Further research is needed to understand patient and physician preferences as more therapeutic options become available for treating ATTR-CM**

METHODS

Study Measures: Demographic and Clinical Characteristics

- Physician surveys, physician-populated patient record forms, and patient self-completion forms captured patient and physician demographics and patient clinical characteristics and preferences

Objectives

- To describe treatment patterns and associated outcomes, including:
 - Frequency of prescription for the proportion of patients prescribed ATTR-CM treatment at the time of the survey. Among those not prescribed treatment at the time of the survey, the reasoning was captured
 - Levels of satisfaction with current treatment, as reported by both physicians and patients, as well as reasoning for dissatisfaction
- To describe patient-reported levels of comfort receiving treatment across modes of administration

Analysis

- All statistical analyses were descriptive in nature (ie, there was no hypothesis testing)

RESULTS

Physicians and Patients

- Of the 34 participating physicians, the majority were general cardiologists or heart failure specialists (55.8%). Among their overall ATTR caseload, physicians consulted a mean (SD) of 21.6% (33.4%) patients in an academic hospital, 7.1% (14.0%) in a community hospital, and 71.3% (34.0%) in an office
- Among 256 patients treated by the participating physicians, the mean (SD) age was 63.9 y (13.2), 60.9% were male, and 55.9% were White. ATTR phenotype was 79.7% cardiac and 20.3% mixed. Median (interquartile range) disease duration (n=164) was 1.9 y (1.3-3.0) (**Table 1**); 58 of these patients provided self-reported data

TABLE 1. Patient Demographic and Clinical Characteristics

Characteristic	Patients N	Patients Value
Age, mean (SD), y	256	63.9 (13.2)
Biological sex,[a] n (%)	256	
Male		156 (60.9)
Female		100 (39.1)
Intersex		0
Body mass index, mean (SD), kg/m²	255	25.4 (3.9)
Race/Ethnicity,[b] n (%)	256	
White		143 (55.9)
Black or African American		81 (31.6)
South or Central American Native		13 (5.1)
East or Southeast Asian		9 (3.5)
Middle Eastern or North African		6 (2.3)
South Asian (Indian subcontinent)		3 (1.2)
American Indian, Indigenous American, or Alaska Native		1 (0.4)
Other		1 (0.4)
NYHA class at time of survey, n (%)	256	
I		56 (21.9)
II		139 (54.3)
III		51 (19.9)
IV		10 (3.9)
Patient treated at an amyloidosis clinic, n (%)	256	49 (19.1)
ATTR phenotype	256	
ATTR-CM		204 (79.7)
Mixed phenotype[c]		52 (20.3)
Disease type, n (%)	180	
Hereditary ATTR-CM		56 (31.1)
Wild-type ATTR-CM		20 (11.1)
Unconfirmed (no genetic test performed)		104 (57.8)
Diagnostic journey and disease history from onset of signs/symptoms, median (IQR), y		
Time from onset to first consultation	159	0.2 (0.0-0.3)
Time from first consultation to diagnosis	175	0.3 (0.1-0.5)
Time from diagnosis to initiation of treatment	172	0.2 (0.1-0.6)
Time from diagnosis to survey	256	1.4 (0.8-2.2)
Time from onset to survey	164	1.9 (1.3-3.0)

[a]As stated in the patient's medical record. [b]Survey question was, "How would you describe the patient?"; multiple selections were allowed. [c]Confirmed diagnosis of ATTR-CM or mixed phenotype (defined as ATTR-CM + ATTR-PN noted in their medical record).
ATTR, transthyretin amyloidosis; ATTR-CM, transthyretin amyloid cardiomyopathy; ATTR-PN, transthyretin amyloid polyneuropathy; IQR, interquartile range; NYHA, New York Heart Association.

RESULTS

TABLE 2. Treatment History Among Patients With ATTR-CM

Characteristic	Patients N	Patients Value
Patient has received treatment for ATTR symptoms, n (%)	256	
Yes, currently prescribed treatment		155 (60.5)
No, but has previously been prescribed treatment		17 (6.6)
No, has never received any prescribed treatment		84 (32.8)
Number of distinct treatment lines ever received	256	
Mean (SD)		0.7 (0.6)
At least 1 treatment line, n (%)		172 (67.2)
More than 1 treatment line, n (%)		14 (5.5)
Number of distinct treatment lines ever received among patients receiving treatment at time of survey	155	
Mean (SD)		1.1 (0.4)
More than 1 treatment line, n (%)		10 (6.5)
Reason why patient has never received treatment,[a] n (%)	84	
Patient does not want treatment for their ATTR		30 (35.7)
Patient is waiting to start their first treatment regimen		22 (26.2)
Patient's medical insurance did not cover the cost of treatment		17 (20.2)
Patient not eligible for treatment		17 (20.2)
Patient newly diagnosed and will be prescribed treatment at future consultation		4 (4.8)
Other (specify)		2 (2.4)
Reason why patient is not receiving treatment at time of survey,[a] n (%)	17	
Patient requested to stop treatment		9 (52.9)
Patient is waiting to start a new treatment regimen		4 (23.5)
Patient did not respond adequately to prior treatment		3 (17.6)
Patient stopped treatment due to side effects		3 (17.6)
Other (specify)		2 (11.8)
Patient's physician would have preferred to use a different disease-modifying treatment than that prescribed, n (%)	145	5 (3.4)

[a]Multiple selections could be made; percentages may not sum up to 100%.
ATTR, transthyretin amyloidosis; ATTR-CM, transthyretin amyloid cardiomyopathy.

- Treatment was received by 60.5% of patients, 6.6% had prior treatment, and 32.8% were treatment naive (**Table 2**)

Treatment Patterns of US Survey Respondents (August 2024-February 2025)

- Among patients receiving treatment at the time of the survey (N=155), the TTR stabilizer, tafamidis, was the most frequently prescribed (n=129 [83.2%]); no patients in this survey had received acoramidis (**Figure 2**)
- While no TTR silencer was approved for ATTR-CM at the time of this study, a few options were available and approved for use in ATTR-PN; some TTR silencer use was noted (16.8% patisiran, 1.9 % vutrisiran, and 1.3% inotersen) (**Figure 2**)

FIGURE 2. Treatment at Time of Survey Among Patients Treated for ATTR-CM (n=155)[a]

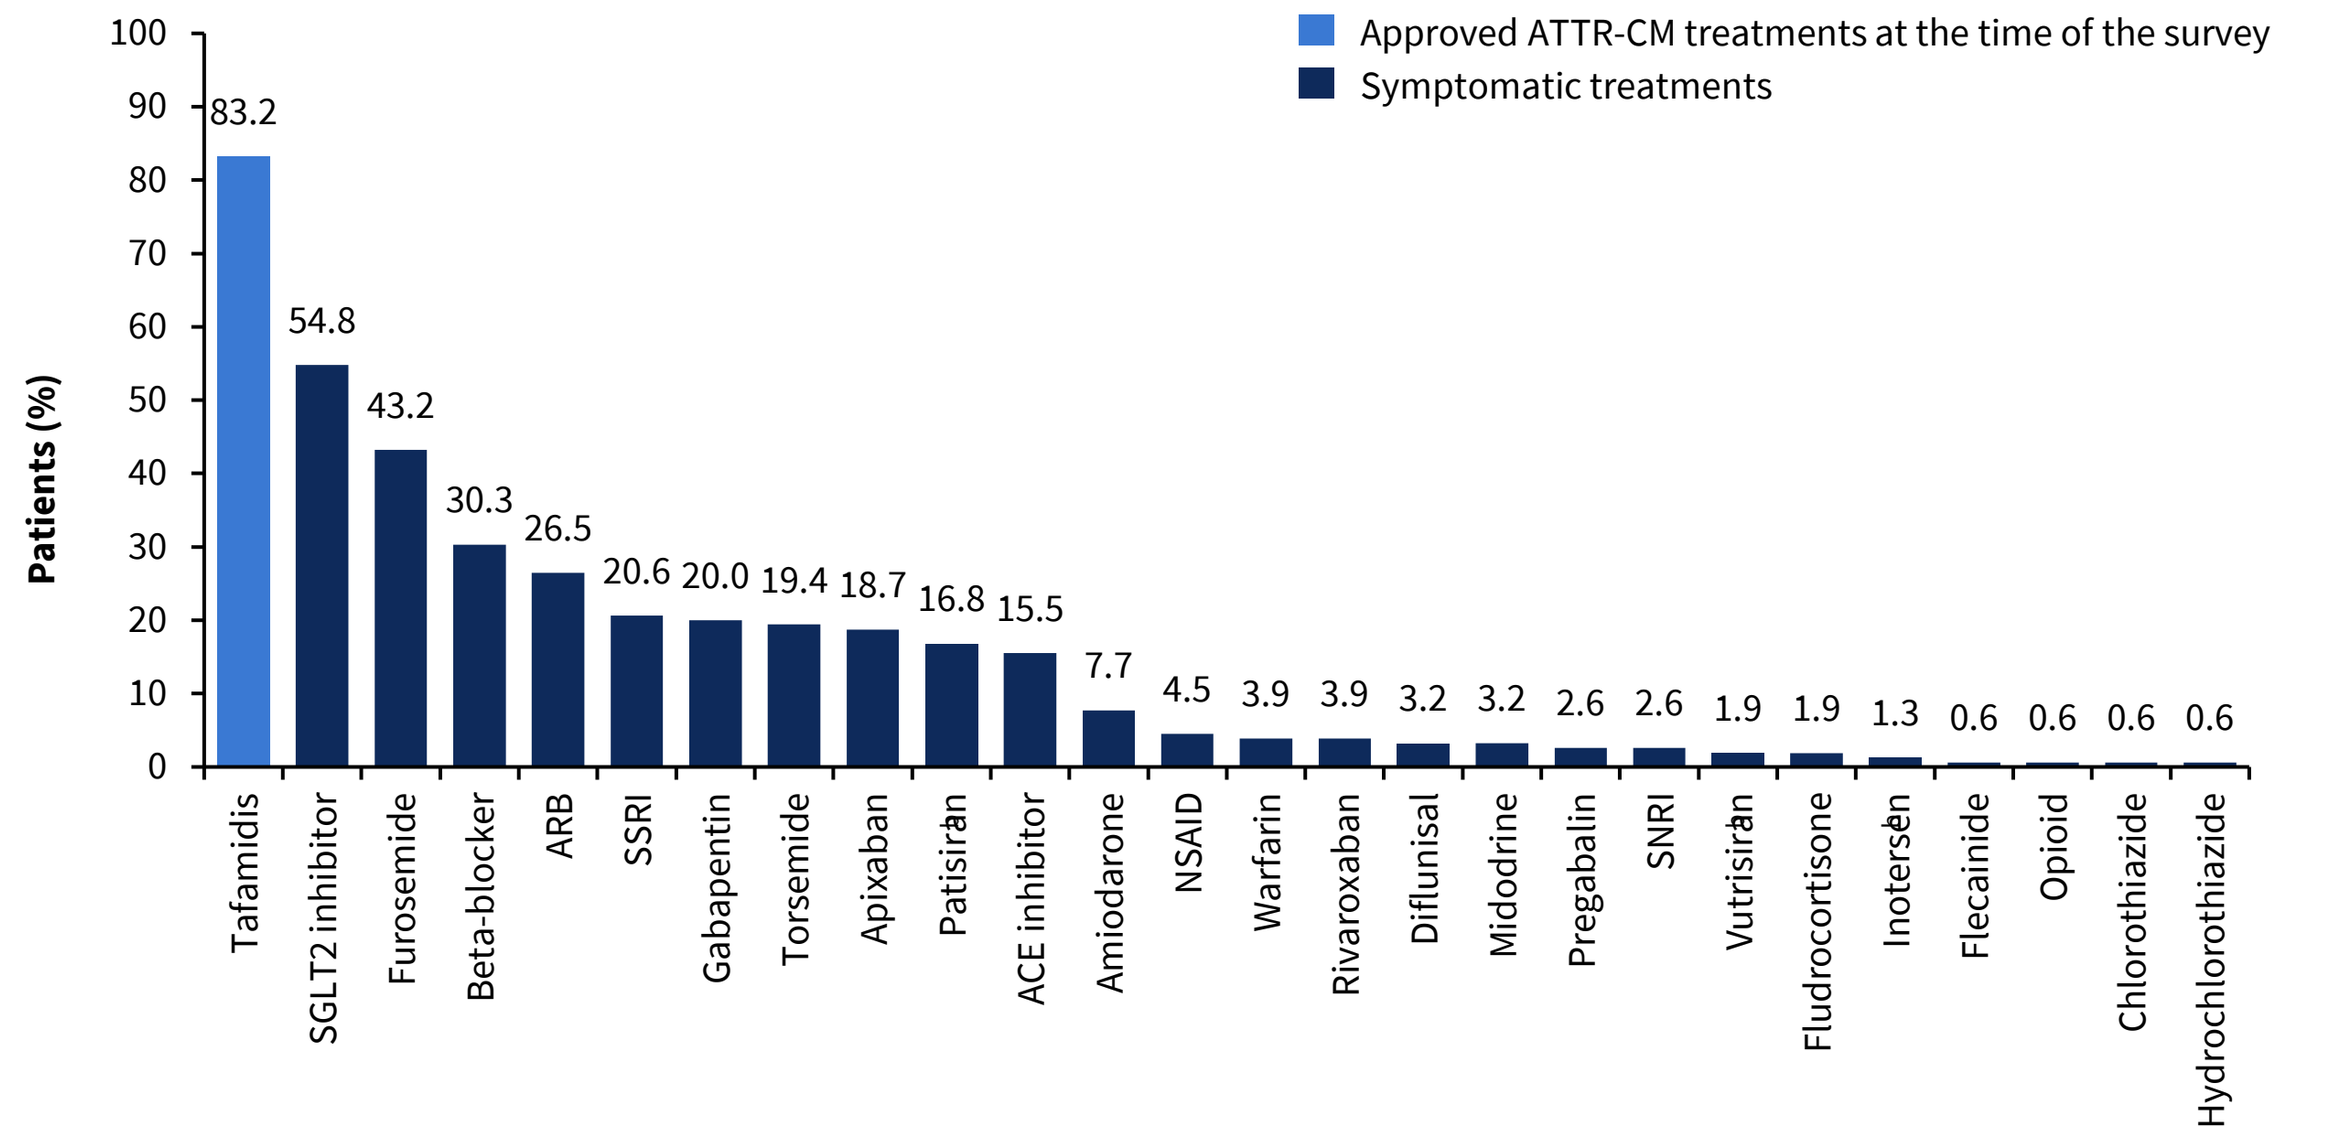
Approved ATTR-CM treatments at the time of the survey
Symptomatic treatments
Patients (%)
0
10
20
30
40
50
60
70
80
90
100
83.2
54.8
43.2
30.3
26.5
20.6
20.0
19.4
18.7
16.8
15.5
7.7
4.5
3.9
3.9
3.2
3.2
2.6
2.6
1.9
1.9
1.3
0.6
0.6
0.6
0.6
Tafamidis
SGLT2 inhibitor
Furosemide
Beta-blocker
ARB
SSRI
Gabapentin
Torsemide
Apixaban
Patisiran[b]
ACE inhibitor
Amiodarone
NSAID
Warfarin
Rivaroxaban
Diflunisal
Midodrine
Pregabalin
SNRI
Vutrisiran[b]
Fludrocortisone
Inotersen[b]
Flecainide
Opioid
Chlorothiazide
Hydrochlorothiazide

[a]If a patient received >1 treatment, that patient was counted once for each treatment they received. [b]Approved for ATTR-PN at the time of the survey.
ACE, angiotensin-converting enzyme; ARB, angiotensin receptor blocker; ATTR-CM, transthyretin amyloid cardiomyopathy; ATTR-PN, transthyretin amyloid polyneuropathy; NSAID, nonsteroidal anti-inflammatory drug; SGLT2, sodium-glucose cotransporter 2; SNRI, serotonin norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

FIGURE 3. Physician-Reported (A) and Patient-Reported (B) Satisfaction With Current Prescribed Treatment for ATTR-CM, and Reasons Why Physician/Patient Was Not Entirely Satisfied With This Treatment[1]

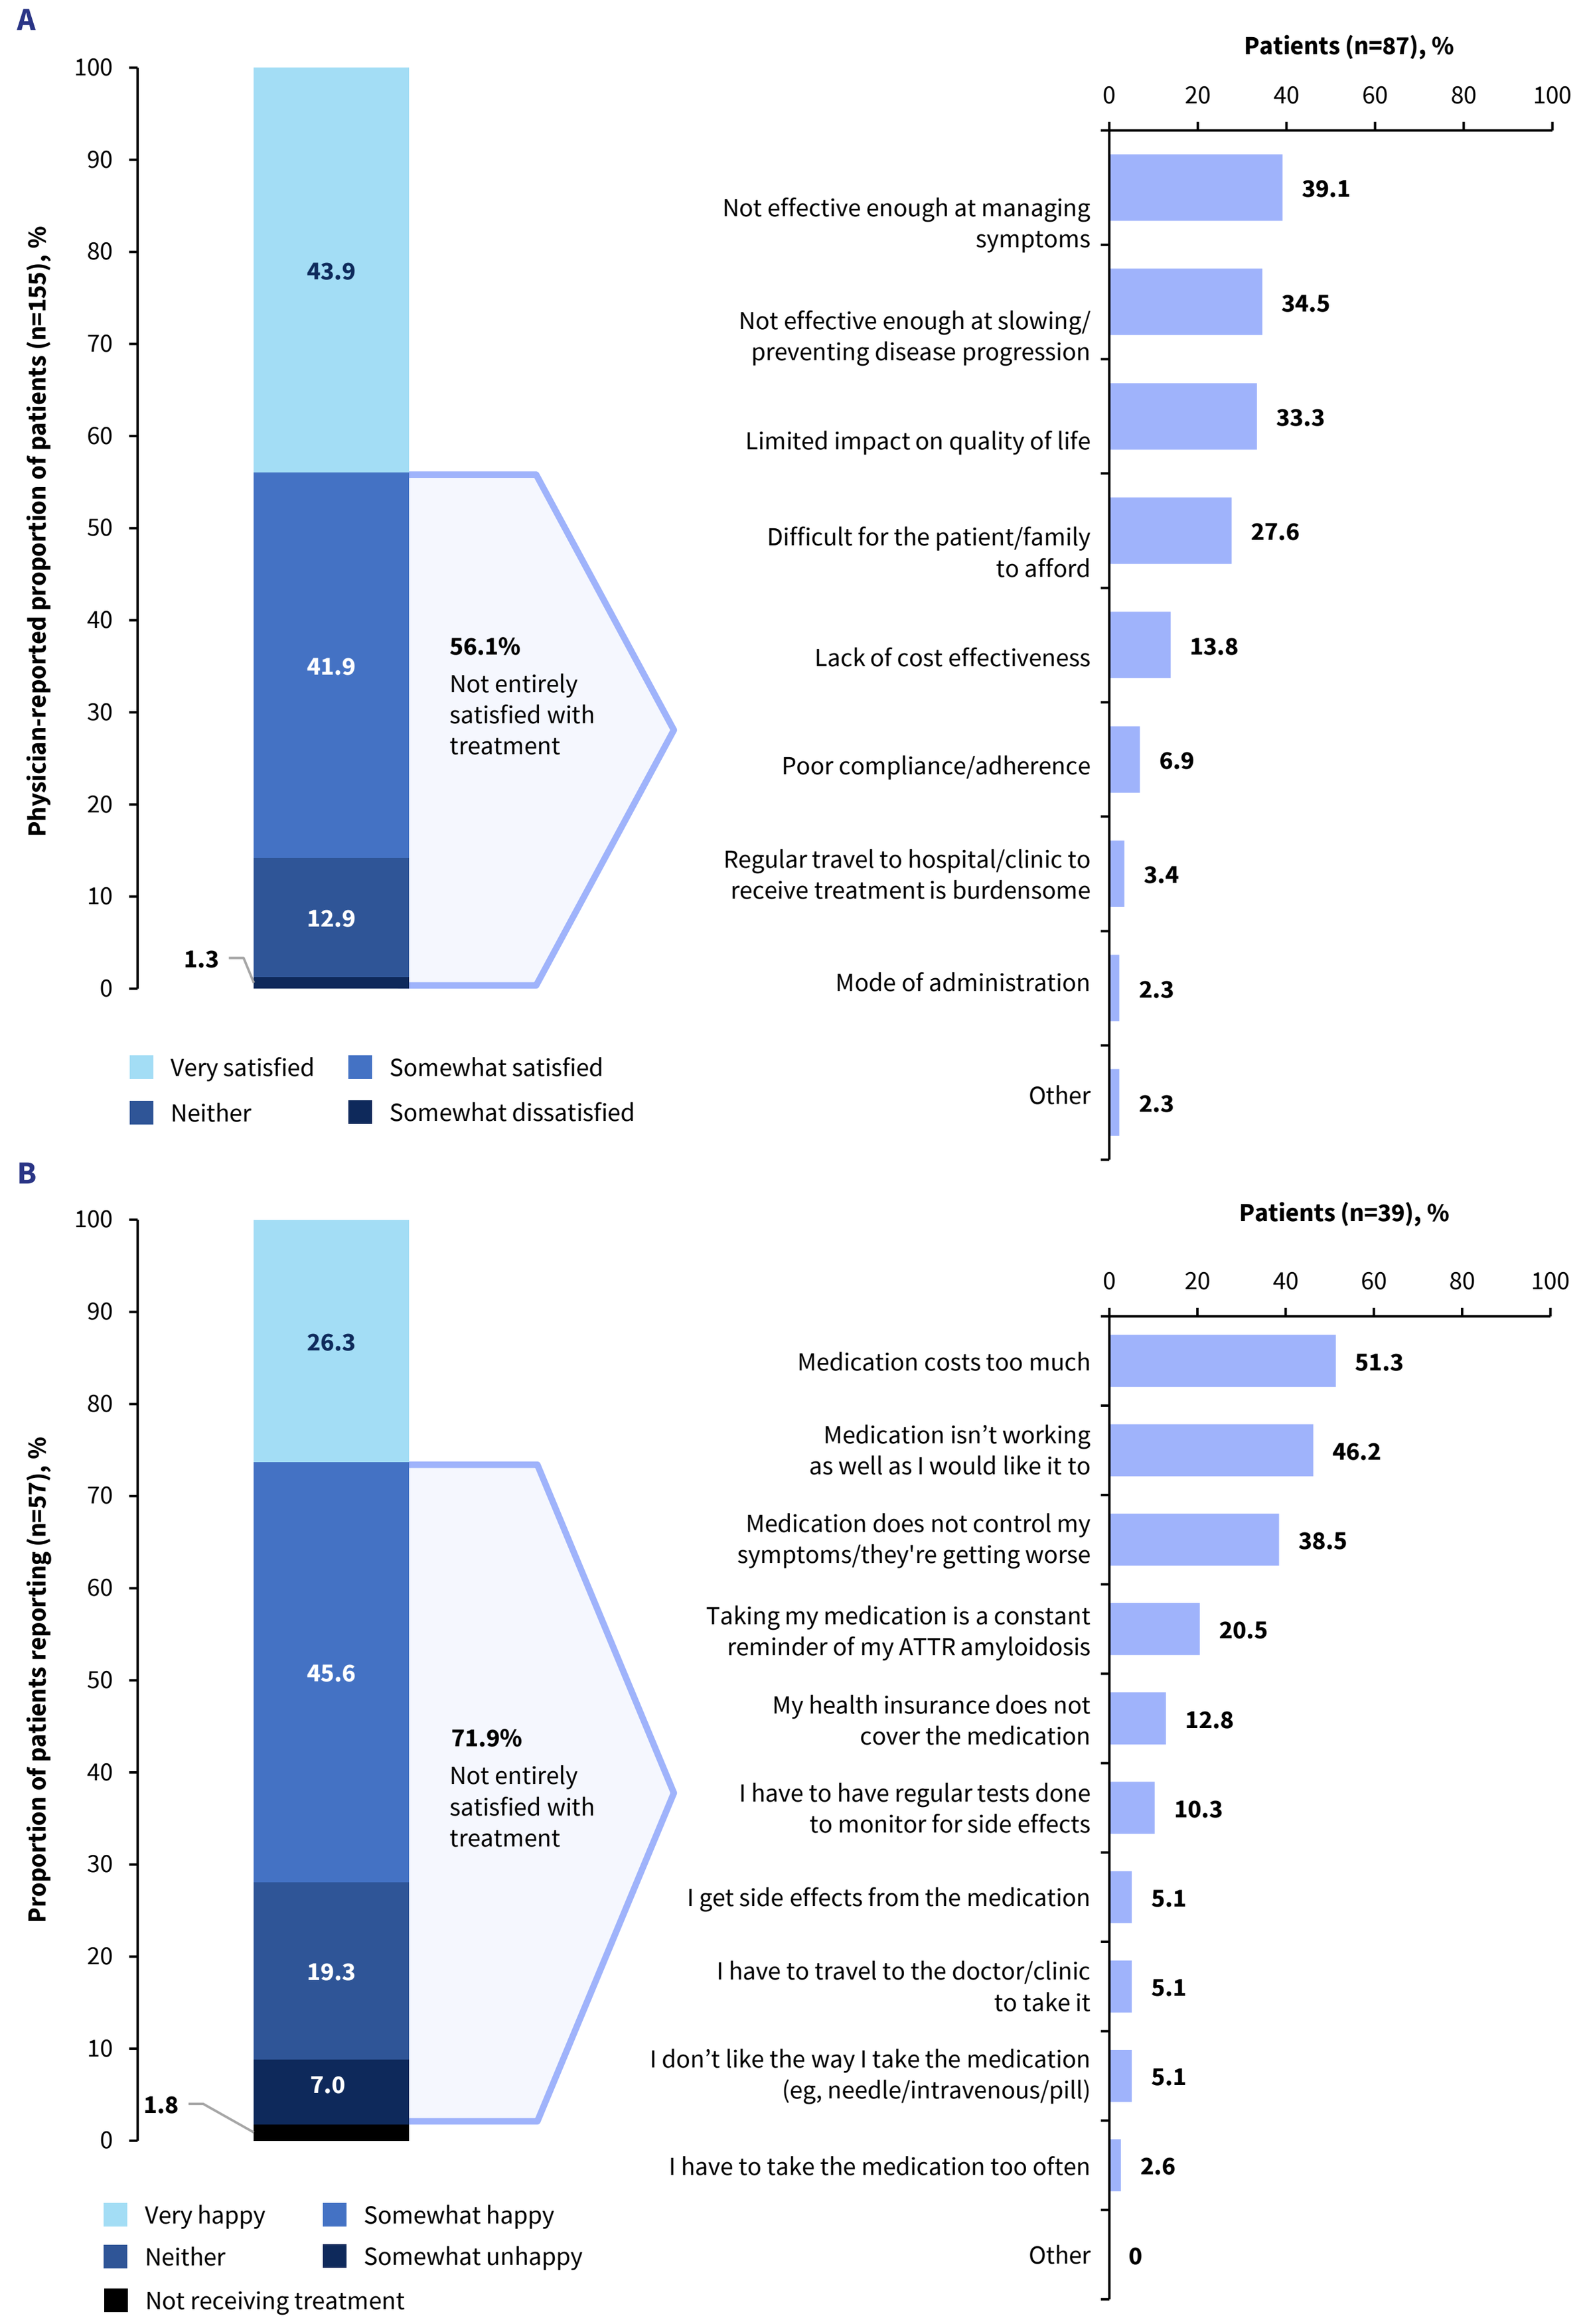
A
Physician-reported proportion of patients (n=155), %
43.9
41.9
12.9
1.3
56.1%
Not entirely satisfied with treatment
Very satisfied
Somewhat satisfied
Neither
Somewhat dissatisfied
Patients (n=87), %
0
20
40
60
80
100
Not effective enough at managing symptoms
39.1
Not effective enough at slowing/preventing disease progression
34.5
Limited impact on quality of life
33.3
Difficult for the patient/family to afford
27.6
Lack of cost effectiveness
13.8
Poor compliance/adherence
6.9
Regular travel to hospital/clinic to receive treatment is burdensome
3.4
Mode of administration
2.3
Other
2.3
B
Proportion of patients reporting (n=57), %
26.3
45.6
19.3
7.0
1.8
71.9%
Not entirely satisfied with treatment
Very happy
Somewhat happy
Neither
Somewhat unhappy
Not receiving treatment
Patients (n=39), %
Medication costs too much
51.3
Medication isn't working as well as I would like it to
46.2
Medication does not control my symptoms/they're getting worse
38.5
Taking my medication is a constant reminder of my ATTR amyloidosis
20.5
My health insurance does not cover the medication
12.8
I have to have regular tests done to monitor for side effects
10.3
I get side effects from the medication
5.1
I have to travel to the doctor/clinic to take it
5.1
I don't like the way I take the medication (eg, needle/intravenous/pill)
5.1
I have to take the medication too often
2.6
Other
0

ATTR, transthyretin amyloidosis; ATTR-CM, transthyretin amyloid cardiomyopathy.

RESULTS

- Physicians were less than "very satisfied" with treatment for 56.1% of patients due to (**Figure 3A**):
 - Level of symptom management (39.1%)
 - Limited impact on disease progression (34.5%)
 - Limited impact on quality of life (33.3%)
 - Difficulty affording treatment (27.6%)
- The majority (71.9%) of patients reported being less than "very happy" with their current treatment due to (**Figure 3B**):
 - Expense (51.3%)
 - Expected better improvement (46.2%)
 - Symptoms persist or exacerbate (38.5%)

FIGURE 4. Patient-Reported Preference With Regard to Mode of Administration of Treatment for ATTR-CM

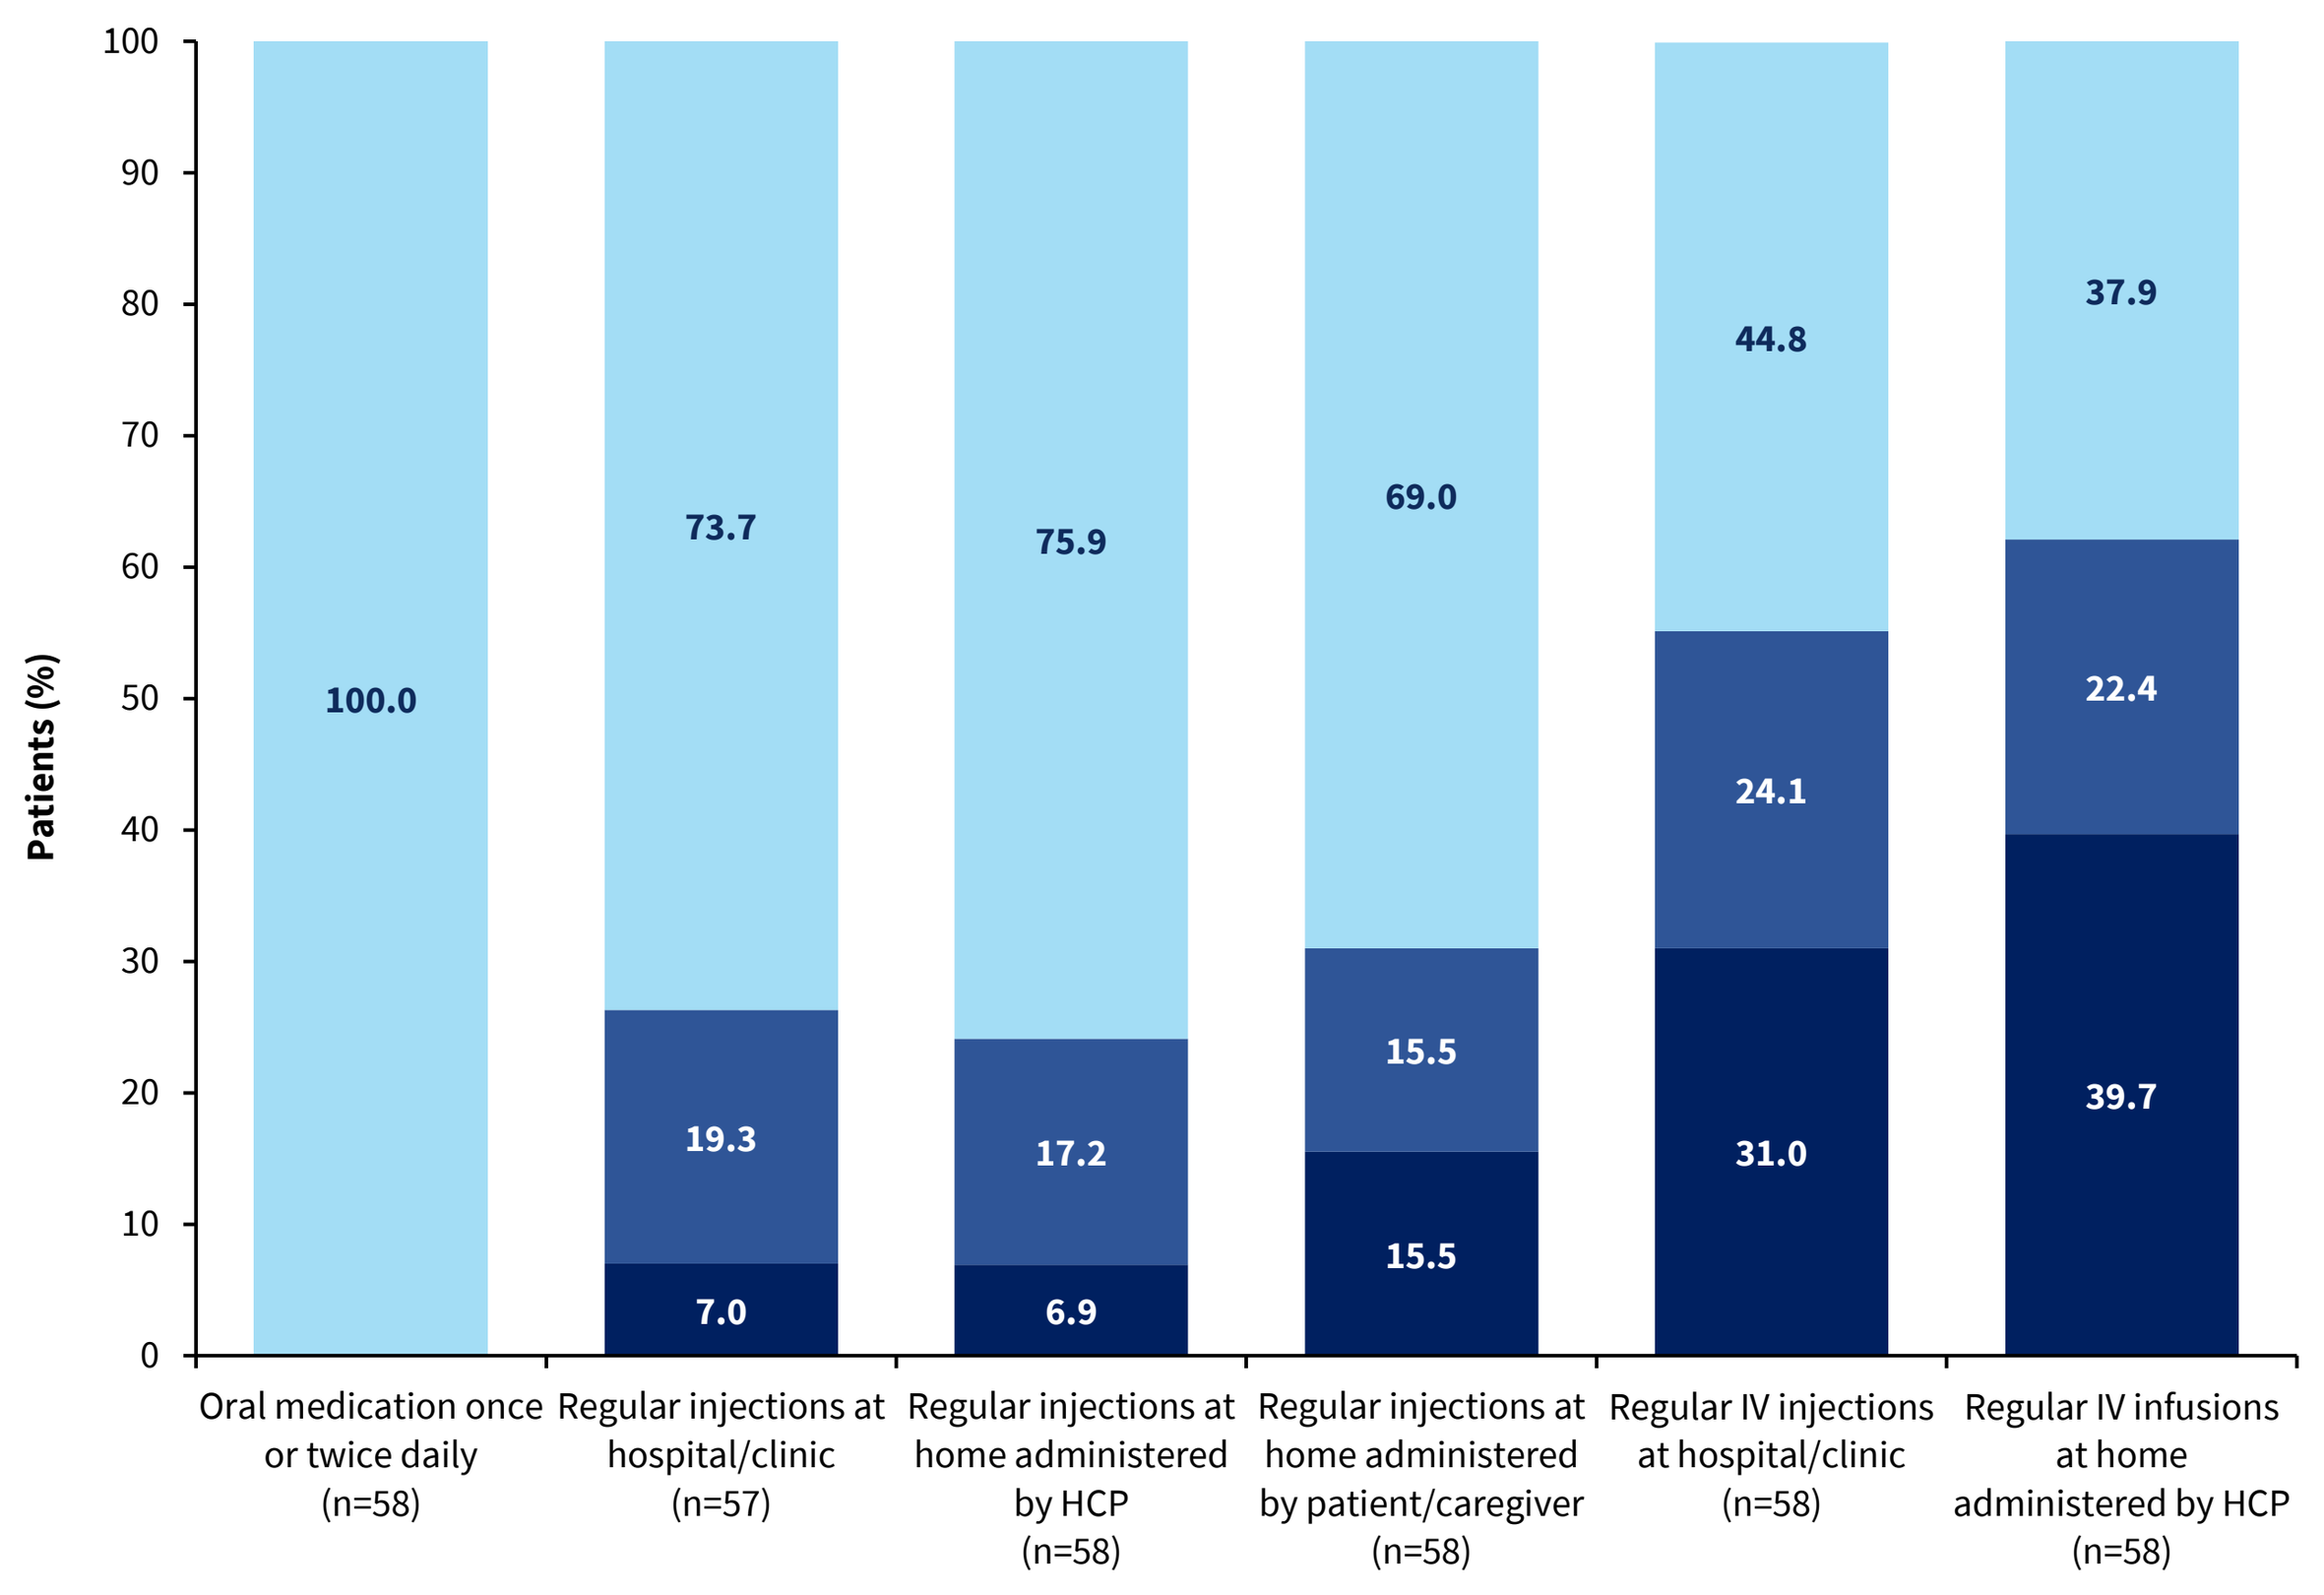
Patients (%)
100.0
73.7
19.3
7.0
75.9
17.2
6.9
69.0
15.5
15.5
44.8
24.1
31.0
37.9
22.4
39.7
Oral medication once or twice daily (n=58)
Regular injections at hospital/clinic (n=57)
Regular injections at home administered by HCP (n=58)
Regular injections at home administered by patient/caregiver (n=58)
Regular IV injections at hospital/clinic (n=58)
Regular IV infusions at home administered by HCP (n=58)
Completely or somewhat comfortable
Neutral
Completely or somewhat uncomfortable

ATTR-CM, transthyretin amyloid cardiomyopathy; HCP, healthcare professional; IV, intravenous.

- All patients (100% [n=58]) were "completely" or "somewhat" comfortable with once- or twice-daily medication. Patients "completely" or "somewhat" comfortable receiving injectable treatment ranged from 69.0% to 75.9%, and receiving intravenously from 37.9% to 44.8% (**Figure 4**)

DISCUSSION

Between August 2024 and February 2025:

- One-third of patients had never been prescribed treatment for ATTR despite receiving an ATTR-CM diagnosis; of these, one-quarter were awaiting treatment start
- Common reasons for lack of treatment satisfaction among both physicians and patients involved suboptimal symptom management and high cost of treatment
- Overall, less than half of physicians were very satisfied with treatment and less than one-third of patients were very happy with treatment
- Patients were most comfortable with the prospect of oral therapy

LIMITATIONS

- This is a survey whose cross-sectional design does not allow for determination of causal relationships and is limited in sample size
- Though consecutive patient inclusion reduces selection bias, the DSP reflects a pragmatic physician sample and pseudo-random patient participation
- Retrospective reporting may introduce recall bias, mitigated by physician access to medical records and short recall periods in the patient self-completion form
- This study occurred during a time period when the second oral TTR stabilizer, acoramidis, was recently approved but not yet available nor part of routine clinical practice, and the TTR silencer, vutrisiran, had not yet been approved for ATTR-CM. Future studies are needed to assess treatment patterns and preferences in a more contemporary cohort

FUNDING: This study used data from the Adelphi Real World ATTR DSP, an independently conducted study wholly owned by Adelphi Real World. BridgeBio Pharma, Inc. is one of the multiple subscribers to the Adelphi ATTR DSP and funded this study.
ETHICAL APPROVAL: All DSPs were performed in compliance with local regulations, and patients provided their informed consent prior to participation; all data were deidentified and pseudonymized.
ACKNOWLEDGMENTS: The authors thank all the patients and caregivers for their participation in this study and acknowledge the collaboration and commitment of all investigators and their research support staff. Under the guidance of the authors, additional medical writing and editorial assistance were provided by Jeremy J. Henriques, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA), funded by BridgeBio Pharma, Inc. Editorial support was provided by Erin Mathern, MBA, CMPP, of BridgeBio Pharma, Inc.

DISCLOSURES: LKS: Researcher for Pfizer and Intellia Therapeutics; advisor for BridgeBio Pharma, Inc. and Pfizer; and recipient of honorarium from Alnylam Pharmaceuticals. **JW:** Speaker bureau and advisory board participant for BridgeBio Pharma, Inc. and Alnylam Pharmaceuticals.

REFERENCES: 1. Ruberg FL, et al. *J Am Coll Cardiol.* 2019;73(22):2872-2891. **2.** Vyndaquel/Vyndamax (tafamidis). Package insert. Pfizer, Inc.; 2023. **3.** ATTRUBY (acoramidis). Package insert. BridgeBio Pharma, Inc.; 2024. **4.** AMVUTTRA (vutrisiran). Package insert. Alnylam, Inc.; 2025. **5.** Kittleson MM, et al. *J Am Coll Cardiol* 2025;87(5):549-565. **6.** Do M, et al. *J Am Geriatr Soc.* Published online January 16, 2026; doi:10.1111/jgs.70299. **7.** Anderson P, et al. *Curr Med Res Opin.* 2023;39(12):1707-1715. **8.** Anderson P, et al. *Curr Med Res Opin.* 2008;24(11):3063-3072. **9.** Babineaux SM, et al. *BMJ Open.* 2016;6(8):e010352. **10.** Higgins V, et al. *Diabetes Metab Syndr Obes.* 2016;9:371-380.

PRESENTED AT THE 75TH ANNUAL SCIENTIFIC SESSION OF THE AMERICAN COLLEGE OF CARDIOLOGY; MARCH 28-30, 2026; NEW ORLEANS, LA, USA.